A phase 1 study of TAK-164, an anti-GCC antibody-drug conjugate, in patients with advanced gastrointestinal cancers expressing GCC

Richard Kim,[1] James M. Cleary,[2] Alexis D. Leal,[3] Aparna Parikh,[4] David P. Ryan,[4] Shining Wang,[5] Brittany Bahamon,[5] Neeraj Gupta,[5] Aaron Moss,[6] Joanna Pye,[5] Harry Miao,[5] Johanna Bendell[7]

[1]Moffitt Cancer Center, Tampa, FL, USA; [2]Dana-Farber Cancer Institute, Harvard Medical School, Boston, MA, USA; [3]Division of Medical Oncology, University of Colorado School of Medicine, Aurora, CO, USA; [4]Massachusetts General Hospital Cancer Center, Harvard Medical School, Boston, MA, USA; [5]Millennium Pharmaceuticals, Inc., Cambridge, MA, USA, a wholly owned subsidiary of Takeda Pharmaceutical Company Limited; [6]Audentes Therapeutics, Inc., San Francisco, CA, USA; [7]Sarah Cannon Research Institute/Tennessee Oncology, Nashville, TN, USA

Background

- GCC is a transmembrane cell surface receptor expressed throughout the GI tract,[1,2] that plays a key role in mucosal homeostasis,[3] genomic integrity and cell proliferation[4,5]
- GCC is expressed in various GI tumors,[6] and in colorectal tumors GCC expression is higher than in healthy matched tissue[7,8]
- IV administration of GCC-targeting agents is expected to be preferentially delivered to GCC-positive tumor tissue, without affecting healthy tissue,[9] since GCC expression in healthy cells is confined to the apical region and thus the luminal side of the mucosa[10]
- TAK-164 is a novel ADC comprising a human IgG1 monoclonal antibody specifically targeting GCC, conjugated to a DNA-damaging alkylating agent by a peptide linker[11]
- Preclinical data have shown that TAK-164 has cytotoxic and antitumor activity in GCC-expressing cells and xenograft mouse models[11]
- This first in-human, phase 1 study evaluated the safety, tolerability, MTD/RP2D, and PK of TAK-164, in adult patients with GCC-positive GI malignancies

Tumor antigen:[1,2]
- GCC
 - Transmembrane cell surface receptor
 - Restricted expression profile throughout the GI tract

Antibody[11]
- 5F9 – fully human IgG1
 - High affinity
 - Well characterized PK

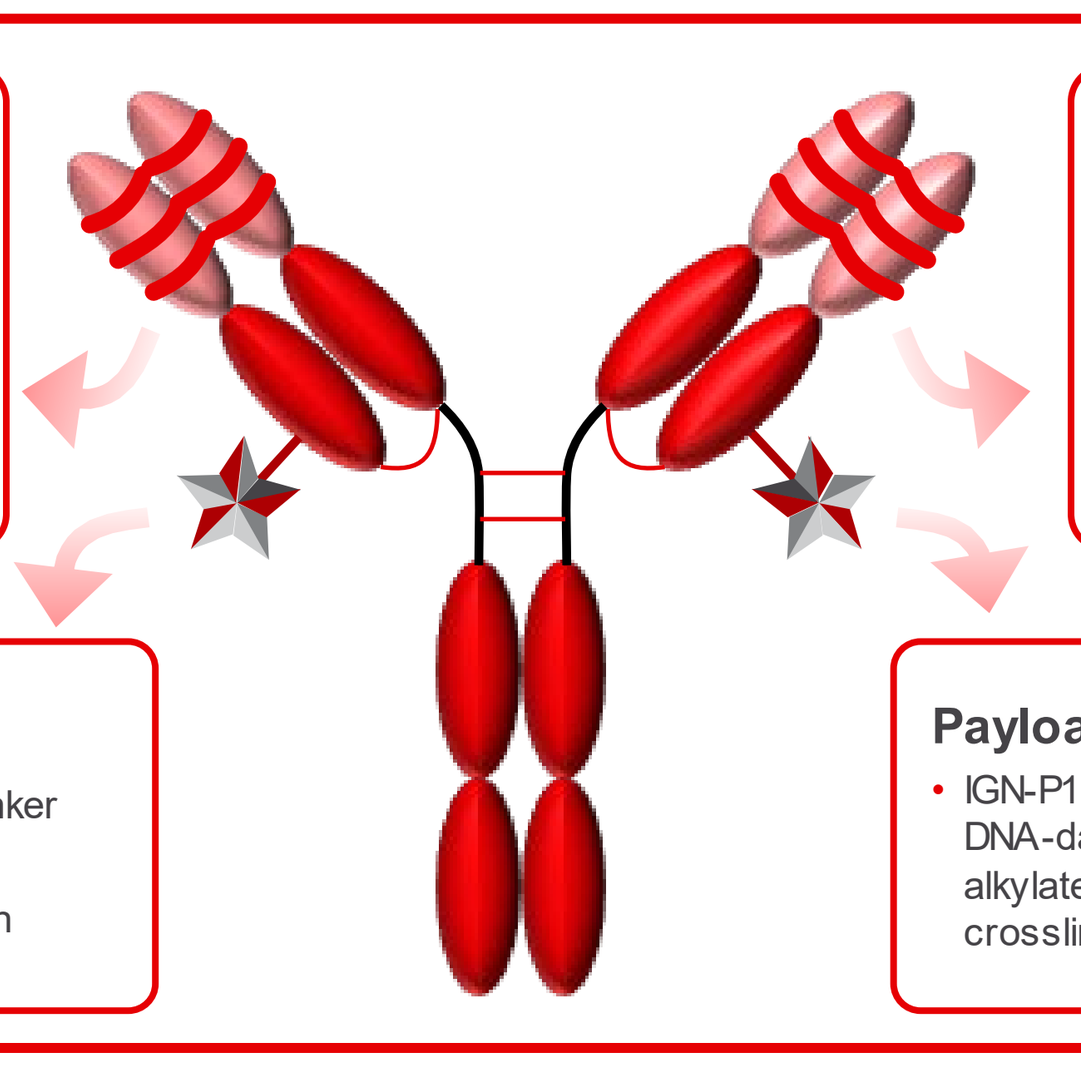

Linker[11]
- Cleavable peptide linker
 - Lysine conjugation
 - Stable in circulation

Payload[11]
- IGN-P1; a novel DNA-damaging agent that alkylates, but does not crosslink DNA

QUESTION

What is the safety and tolerability of TAK-164, a novel, human IgG1 mAb targeting GCC, conjugated to a DNA-damaging alkylating agent by a peptide linker?

INVESTIGATION

Patients with GCC-positive (H score ≥10) GI cancers (N=31)

TAK-164 IV Q3W
Dose escalation from 0.004 mg/kg

Treat until PD, discontinuation due to unacceptable toxicity, or withdrawal of consent

RESULTS

RP2D

- No DLTs in cycle 1
- Three patients had DLTs during cycle 2
- TAK-164 capped at 0.19 mg/kg due to hepatic toxicity
- RP2D 0.064 mg/kg

Most common related TEAEs

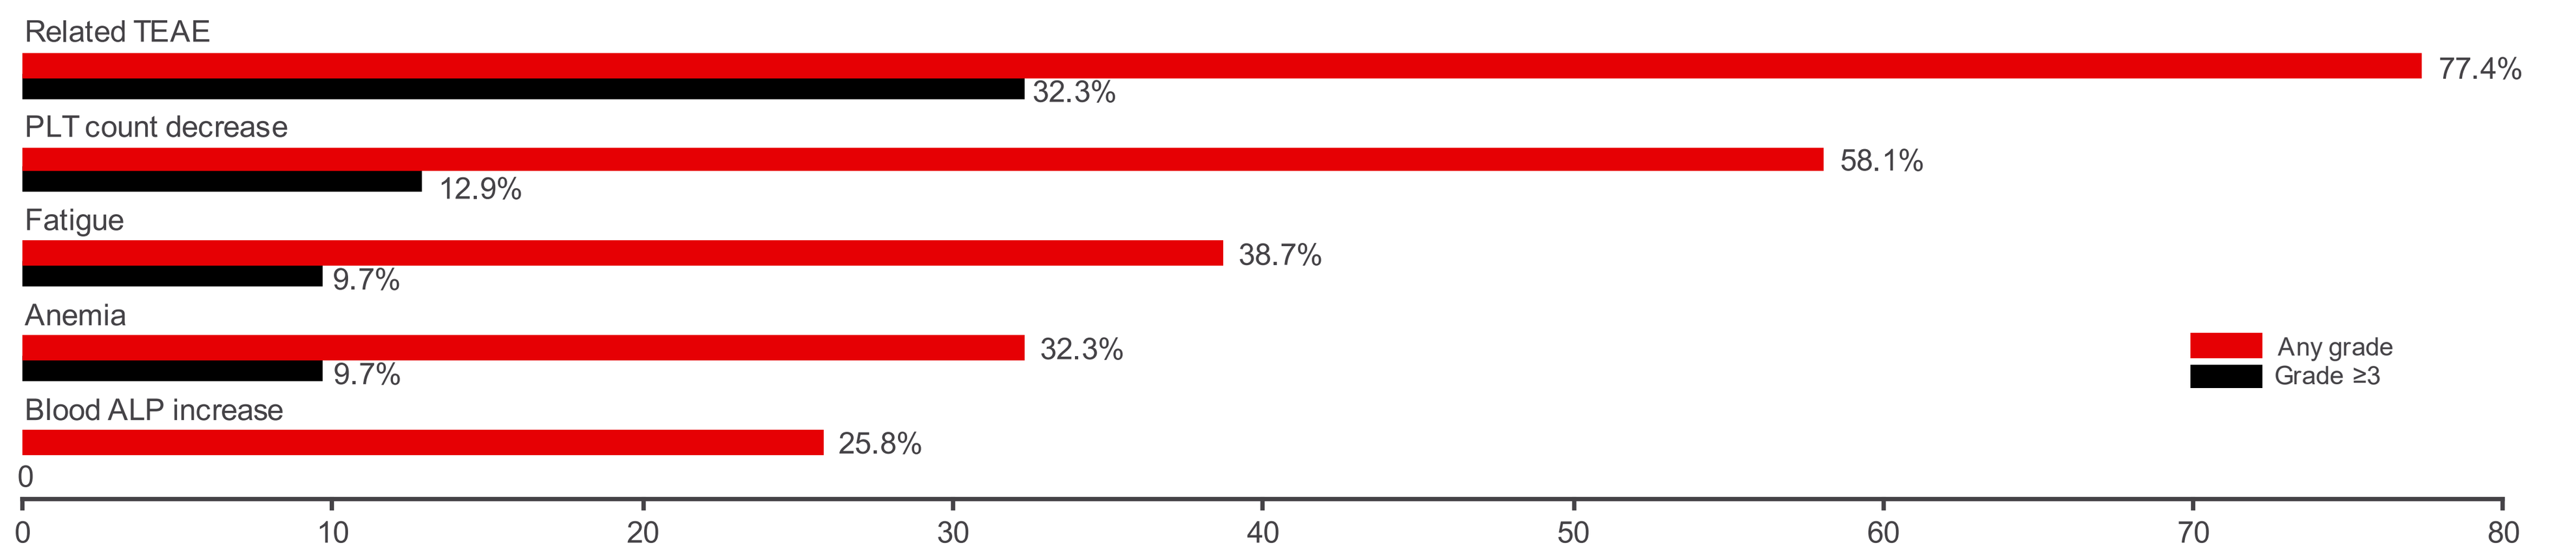
Related TEAE
77.4%
32.3%
PLT count decrease
58.1%
12.9%
Fatigue
38.7%
9.7%
Anemia
32.3%
9.7%
Blood ALP increase
25.8%
0
Any grade
Grade ≥3
0 10 20 30 40 50 60 70 80

Response

- One unconfirmed PR (patient with low baseline GCC expression who received 0.008 mg/kg)
- Eleven patients had SD

CONCLUSIONS

TAK-164 appeared to have a manageable safety profile up to 0.064 mg/kg. Hepatic toxicity was identified as a potential risk.
The RP2D was considered insufficient to derive significant clinical benefit.

Methods

Eligibility criteria

Key inclusion criteria
Patients aged ≥18 years with histologically or cytologically confirmed GCC-positive (H-score ≥10 as indicated by IHC) GI cancers, including metastatic colorectal carcinoma, gastric carcinoma, esophageal, small intestine and pancreatic cancers
Patients for whom standard treatment was no longer effective, or not available
ECOG PS 0–1
Completion of chemotherapy, biologic therapy, immunotherapy, or radiotherapy at least 4 weeks prior to enrollment
Adequate bone marrow function, defined as an absolute neutrophil count of ≥1.5 x 10^9/L, platelet count ≥100 x 10^9/L and hemoglobin ≥9 g/dL
Adequate renal function defined as creatinine clearance ≥60 mL/min
Adequate hepatic function, defined as total bilirubin ≤1.5 x upper limit of normal, serum AST and ALT must have been ≤2.5 x upper limit normal (AST and ALT could be elevated up to 3 x upper limit normal if the elevation could be reasonably ascribed to the presence of metastatic disease in liver), and serum albumin ≥3.0 g/dL
Key exclusion criteria
Patients who received anticancer chemotherapy or biologic therapy, or treatment with an experimental anticancer agent within 28 days of first TAK-164 dose
Patients with chronic or active infection requiring systemic therapy
Diagnosis of a symptomatic CNS malignancy or metastasis
Diagnosed or treated for another malignancy within 2 years before administration of the first dose of study drug, or previously diagnosed with another malignancy and have any evidence of residual disease

Figure 1. Open-label, dose-escalation phase 1 study design

Patients with GCC-positive (H-score ≥10) GI cancers for whom standard treatment was no longer effective, or not available
↓
IV TAK-164 on day 1 of 21-day cycles (Q3W)
↓
Dose escalation via a Bayesian model of mTPI method
Starting dose: 0.004 mg/kg
↓
Treatment administered until disease progression, unacceptable toxicity, or withdrawal from the study
↓
End of treatment*

*Defined as 30 days after the last TAK-164 dose, or prior to the start of subsequent antineoplastic therapy.

Objectives

- Primary objective:
 - Safety and tolerability, MTD/RP2D of TAK-164
- Secondary objectives:
 - PK
 - Immunogenicity
 - Efficacy
- Exploratory objectives:
 - Evaluate the pharmacodynamics effect of TAK-164 in tumor and/or liquid biopsies

Assessments

- TEAEs were assessed after administration of the first dose of TAK-164 and through 30 days after the last dose of TAK-164 and were graded using the NCI-CTCAE version 5
- Response was assessed based on the modified RECIST criteria version 1.1, at screening, every 2 cycles, and EOT
- PK samples were collected pre-dose and post-dose on day 1 and on days 2, 3, 4, 8, and 15 of cycles 1 and 2, as well as pre-dose and post-dose on day 1 of cycles 3+
- A semi-quantitative IHC assay was used to determine GCC expression in archival tissue or pre-treatment biopsies

Results

Patients

- A total of 31 patients were enrolled at 5 sites in the USA
- Patients received TAK-164 at the following doses **(Table 1)**

Table 1. Enrollment by TAK-164 dose

	TAK-164 dose, mg/kg										
	0.004	0.008	0.016	0.032	0.064	0.12	0.16	0.19	0.25	0.32	Total
n	1	1	1	5	7	7	2	3	3	1	31

- Patient baseline demographics and disease characteristics are summarized in **Table 2**
- All patients discontinued from the study due to:
 - Disease progression (54.8%)
 - Withdrawal (19.4%)
 - Symptomatic deterioration (19.4%)
 - AE (6.5%)

Table 2. Patient baseline demographics and disease characteristics

Characteristic	N=31
Median age, years (range)	58 (32–72)
Female, n (%)	18 (58.1)
ECOG PS 0 / 1 / 2, n (%)	14 (45.2) / 16 (51.6) / 1 (3.2)
Tumor type, n (%)	
Colorectal carcinoma	25 (80.6)
Esophageal carcinoma	2 (6.5)
Gallbladder carcinoma	1 (3.2)
Mucinous adenocarcinoma of unknown primary	1 (3.2)
Peritoneum	1 (3.2)
Omentum, ovarian	1 (3.2)
Disease Stage at study entry, III / IV, n (%)	1 (3.2) / 30 (96.8)
MSI high / MSS / unknown, n (%)	5 (16.1) / 17 (54.8) / 9 (29.0)
KRAS mutant / wild-type / unknown, n (%)	16 (51.6) / 13 (41.9) / 2 (6.5)
Median number of prior lines of therapy (range)	4 (2–9)

Dose-escalation and determination of RP2D

- No DLTs were reported during cycle 1, allowing dose escalation to proceed to 0.32 mg/kg
- Three patients reported DLTs during cycle 2 **(Table 3)**

Table 3. DLTs reported during dose-escalation

Dose level	N	N with DLTs during cycle 2	Description of DLTs
0.19 mg/kg	3	1	Grade 3 pyrexia and grade 5 hepatic failure
0.25 mg/kg	3	2	Grade 3 nausea, grade 4 platelet count decreased, and grade 4 neutrophil count decreased (n=1) Grade 4 hepatic failure and grade 4 platelet count decreased (n=1)

- As a result of hepatic toxicity events, the maximum TAK-164 dose was capped at 0.19 mg/kg, and additional patients were enrolled at lower doses to determine the RP2D
- Based on a holistic assessment of all data, the RP2D was determined as 0.064 mg/kg

Safety

- All patients (N=31) were included in the safety population
- Patients received a median of 2 (1–8) treatment cycles
- Median treatment duration was 6 (3–27) weeks
- Median TAK-164 relative dose intensity was 100% (60–102)
- A summary of the TAK-164 safety profile is shown by dose level in **Table 4**
- The most frequently reported (in ≥2 patients) any-grade and grade ≥3 TEAEs related to TAK-164 are shown in **Figure 2**
- One patient was positive for ADA at baseline and post-baseline, with a titer lower than the minimum required dilution of 81, demonstrating that observed toxicity was not due to ADA

Table 4. TAK-164 safety profile

n (%)	0.004 (n=1)	0.008 (n=1)	0.016 (n=1)	0.032 (n=5)	0.064 (n=7)	0.12 (n=7)	0.16 (n=2)	0.19 (n=3)	0.25 (n=3)	0.32 (n=1)	Total (N=31)
TEAEs	1 (100)	1 (100)	1 (100)	5 (100)	7 (100)	7 (100)	2 (100)	3 (100)	3 (100)	1 (100)	31 (100)
Related	1 (100)	0	1 (100)	5 (100)	6 (85.7)	5 (71.4)	1 (50.0)	2 (66.7)	2 (66.7)	1 (100)	24 (77.4)
Grade ≥3 TEAEs	0	1 (100)	1 (100)	3 (60.0)	1 (14.3)	5 (71.4)	1 (50.0)	3 (100)	3 (100)	0	18 (58.1)
Related	0	0	1 (100)	1 (20.0)	1 (14.3)	3 (42.9)	1 (50.0)	1 (33.3)	2 (66.7)	0	10 (32.3)
TEAEs leading to discontinuation*	0	0	0	0	0	2 (28.6)	0	0	1 (33.3)	0	3 (9.7)
TEAEs leading to dose reduction	0	0	0	0	1 (14.3)	1 (14.3)	0	0	0	0	2 (6.5)
TEAEs leading to dose delay	0	0	0	2 (40.0)	0	1 (14.3)	0	1 (33.3)	1 (33.3)	0	5 (16.1)
SAEs	0	1 (100)	0	3 (60.0)	0	3 (42.9)	1 (50.0)	2 (66.7)	2 (66.7)	0	12 (38.7)
Related SAEs	0	0	0	0	0	1 (14.3)	1 (50.0)	1 (33.3)	2 (66.7)	0	5 (16.1)

Column heads: TAK-164 dose, mg/kg.

*All TEAEs leading to discontinuation were related to TAK-164.

Figure 2. Any-grade and grade ≥3 TEAEs related to TAK-164, reported in ≥2 patients

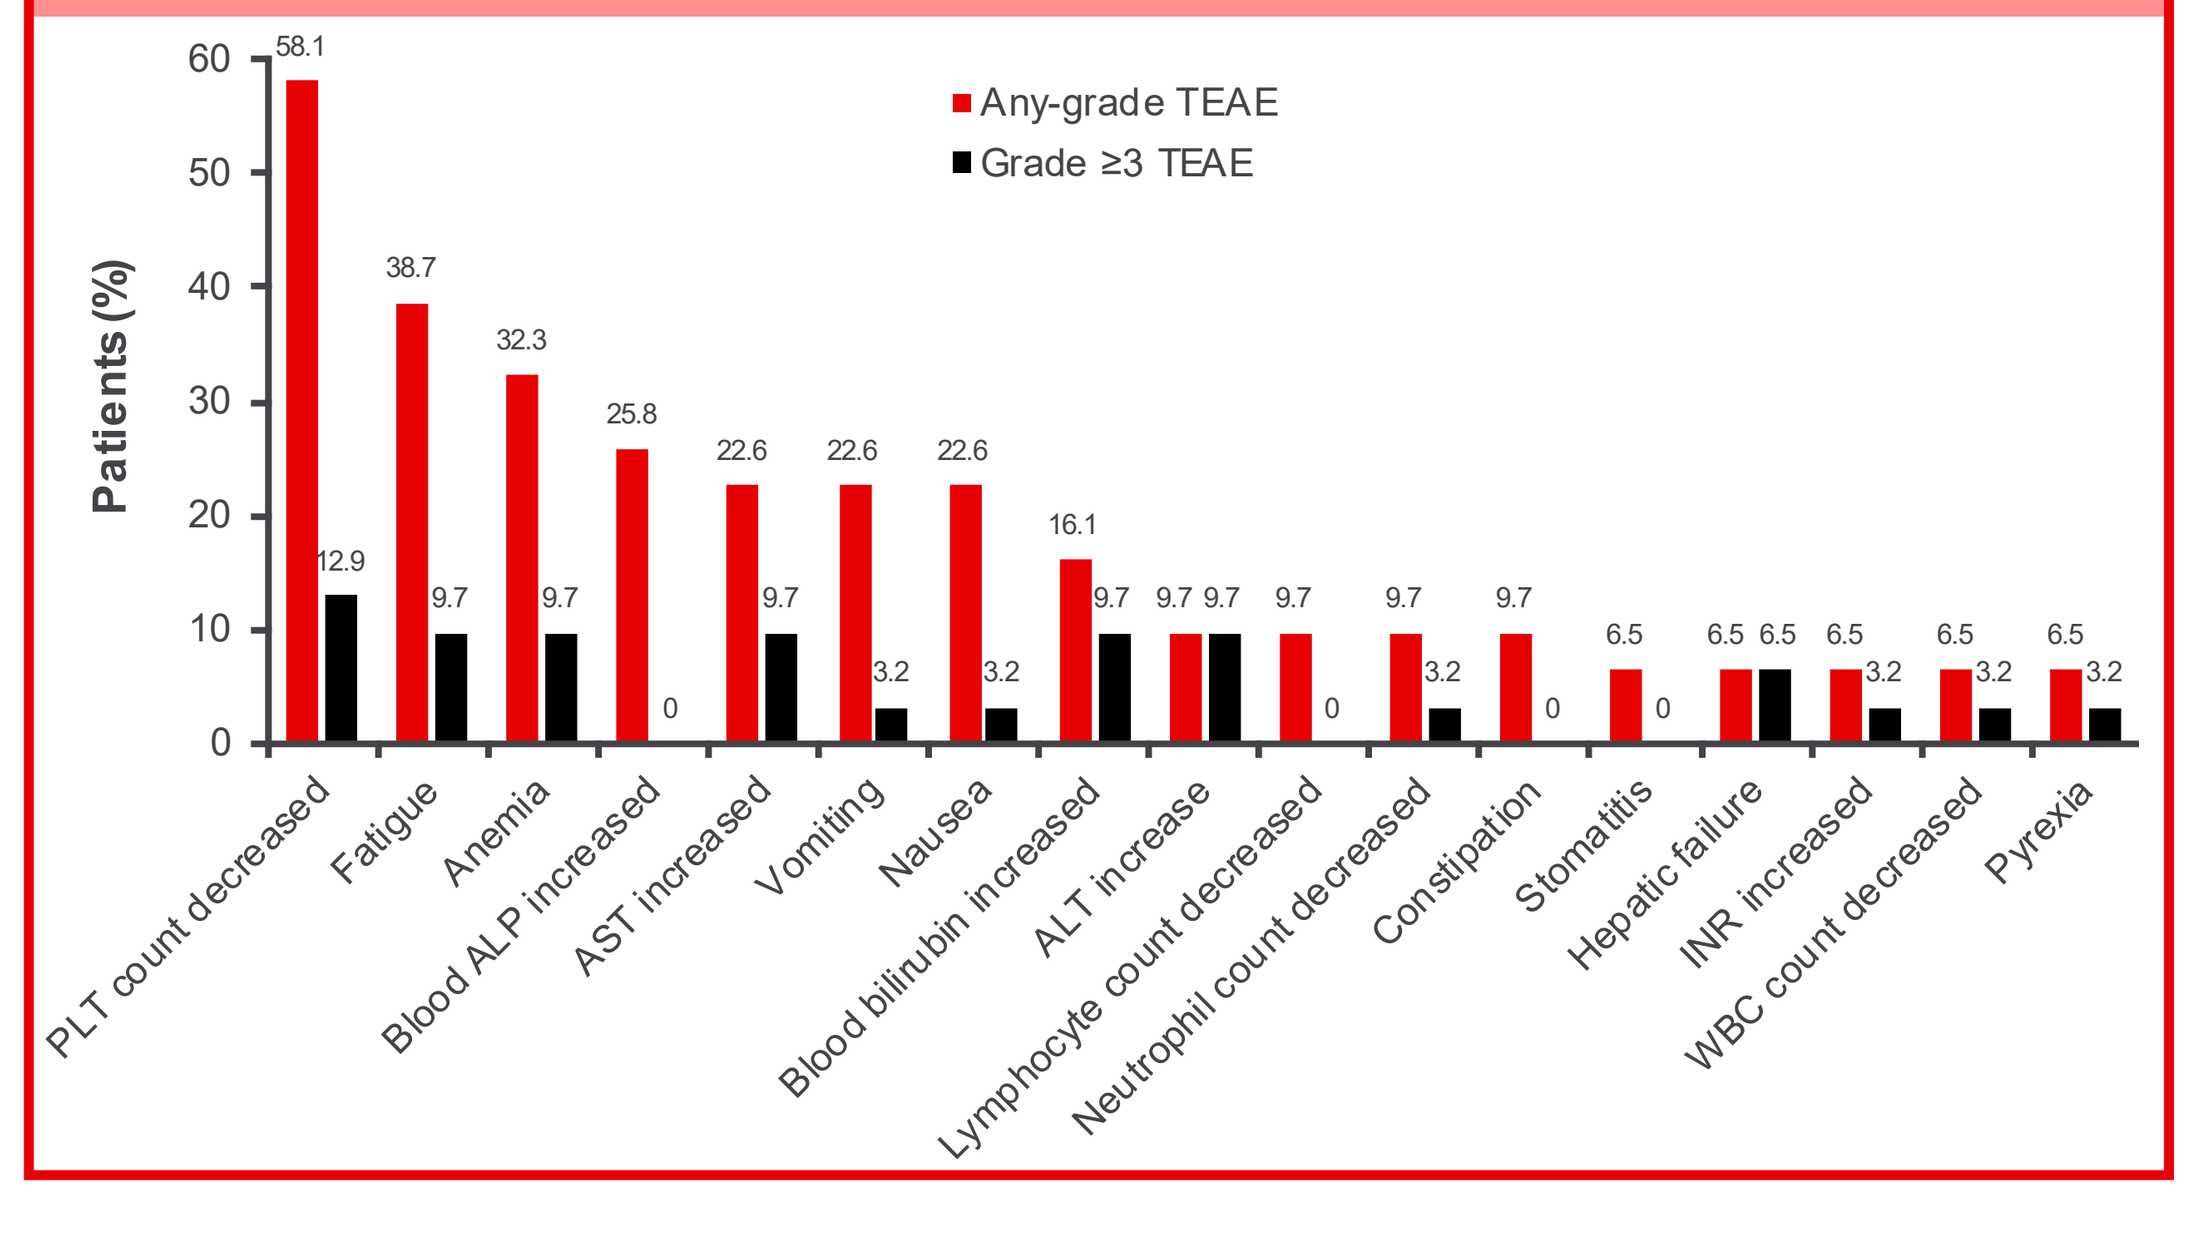
Any-grade TEAE
Grade ≥3 TEAE
Patients (%)
PLT count decreased 58.1 12.9
Fatigue 38.7 9.7
Anemia 32.3 9.7
Blood ALP increased 25.8 0
AST increased 22.6 9.7
Vomiting 22.6 3.2
Nausea 22.6 3.2
Blood bilirubin increased 16.1 9.7
ALT increase 9.7 9.7
Lymphocyte count decreased 9.7 0
Neutrophil count decreased 9.7 3.2
Constipation 9.7 0
Stomatitis 6.5 0
Hepatic failure 6.5 6.5
INR increased 6.5 3.2
WBC count decreased 6.5 3.2
Pyrexia 6.5 3.2

- Hepatic toxicity was identified as a safety signal during dose escalation **(Table 5)**

Table 5. Observed hepatic toxicity events related to TAK-164 (safety population, N=31)

n (%)	TAK-164-related TEAE	TAK-164-related grade ≥3 TEAE	TAK-164-related SAE
AST increased	7 (22.6)	3 (9.7)	0
Blood bilirubin increased	5 (16.1)	3 (9.7)	1 (3.2)
ALT increased	3 (9.7)	3 (9.7)	1 (3.2)
Hepatic failure	2 (6.5)	2 (6.5)	2 (6.5)

PK analysis

- TAK-164 concentration–time profiles **(Figure 3)** were consistent with an IV infusion, with peak concentrations generally occurring at (or near) the time of end of the infusion
- The disposition phase appeared to be bi-phasic **(Figure 3)** and there was an increase in TAK-164 exposure (C_{max} and AUC_{0-inf}) with increasing dose **(Figure 4)**
- The mean terminal disposition half-life of TAK-164 ranged from 63.5 to 159 hours
- Overall, there were no meaningful differences in the PK of TAK-164 during cycle 1 and cycle 2 after 2 doses with Q3W administration

Figure 3. TAK-164 plasma concentration–time profiles at 0.004–0.32 mg/kg Q3W. Semi-log plot grouped by dose and segregated by cycle

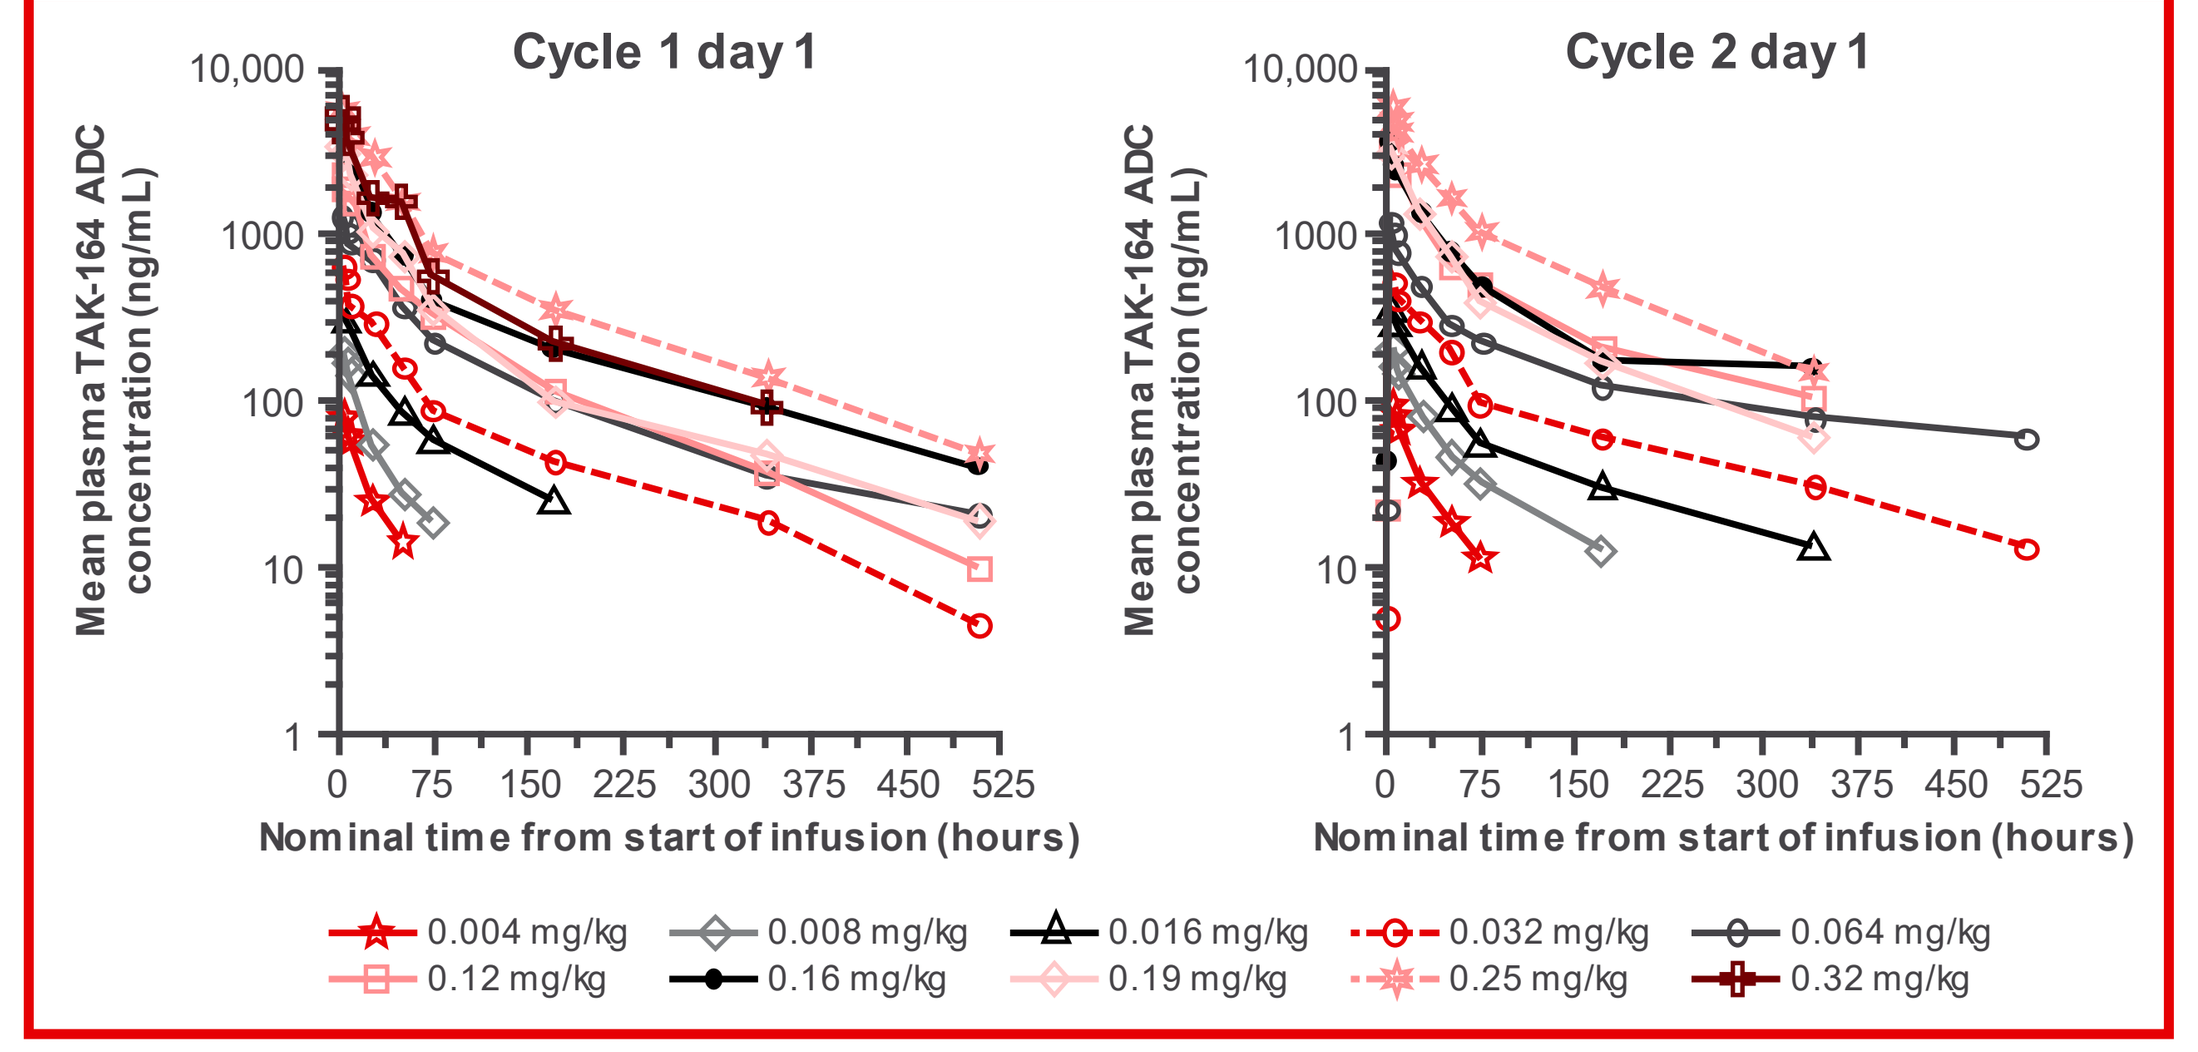
Cycle 1 day 1
Cycle 2 day 1
Mean plasma TAK-164 ADC concentration (ng/mL)
Nominal time from start of infusion (hours)
0.004 mg/kg
0.008 mg/kg
0.016 mg/kg
0.032 mg/kg
0.064 mg/kg
0.12 mg/kg
0.16 mg/kg
0.19 mg/kg
0.25 mg/kg
0.32 mg/kg

Figure 4. TAK-164 C_{max} and AUC_{0-inf} dose proportionality plots, grouped by dose and segregated by cycle

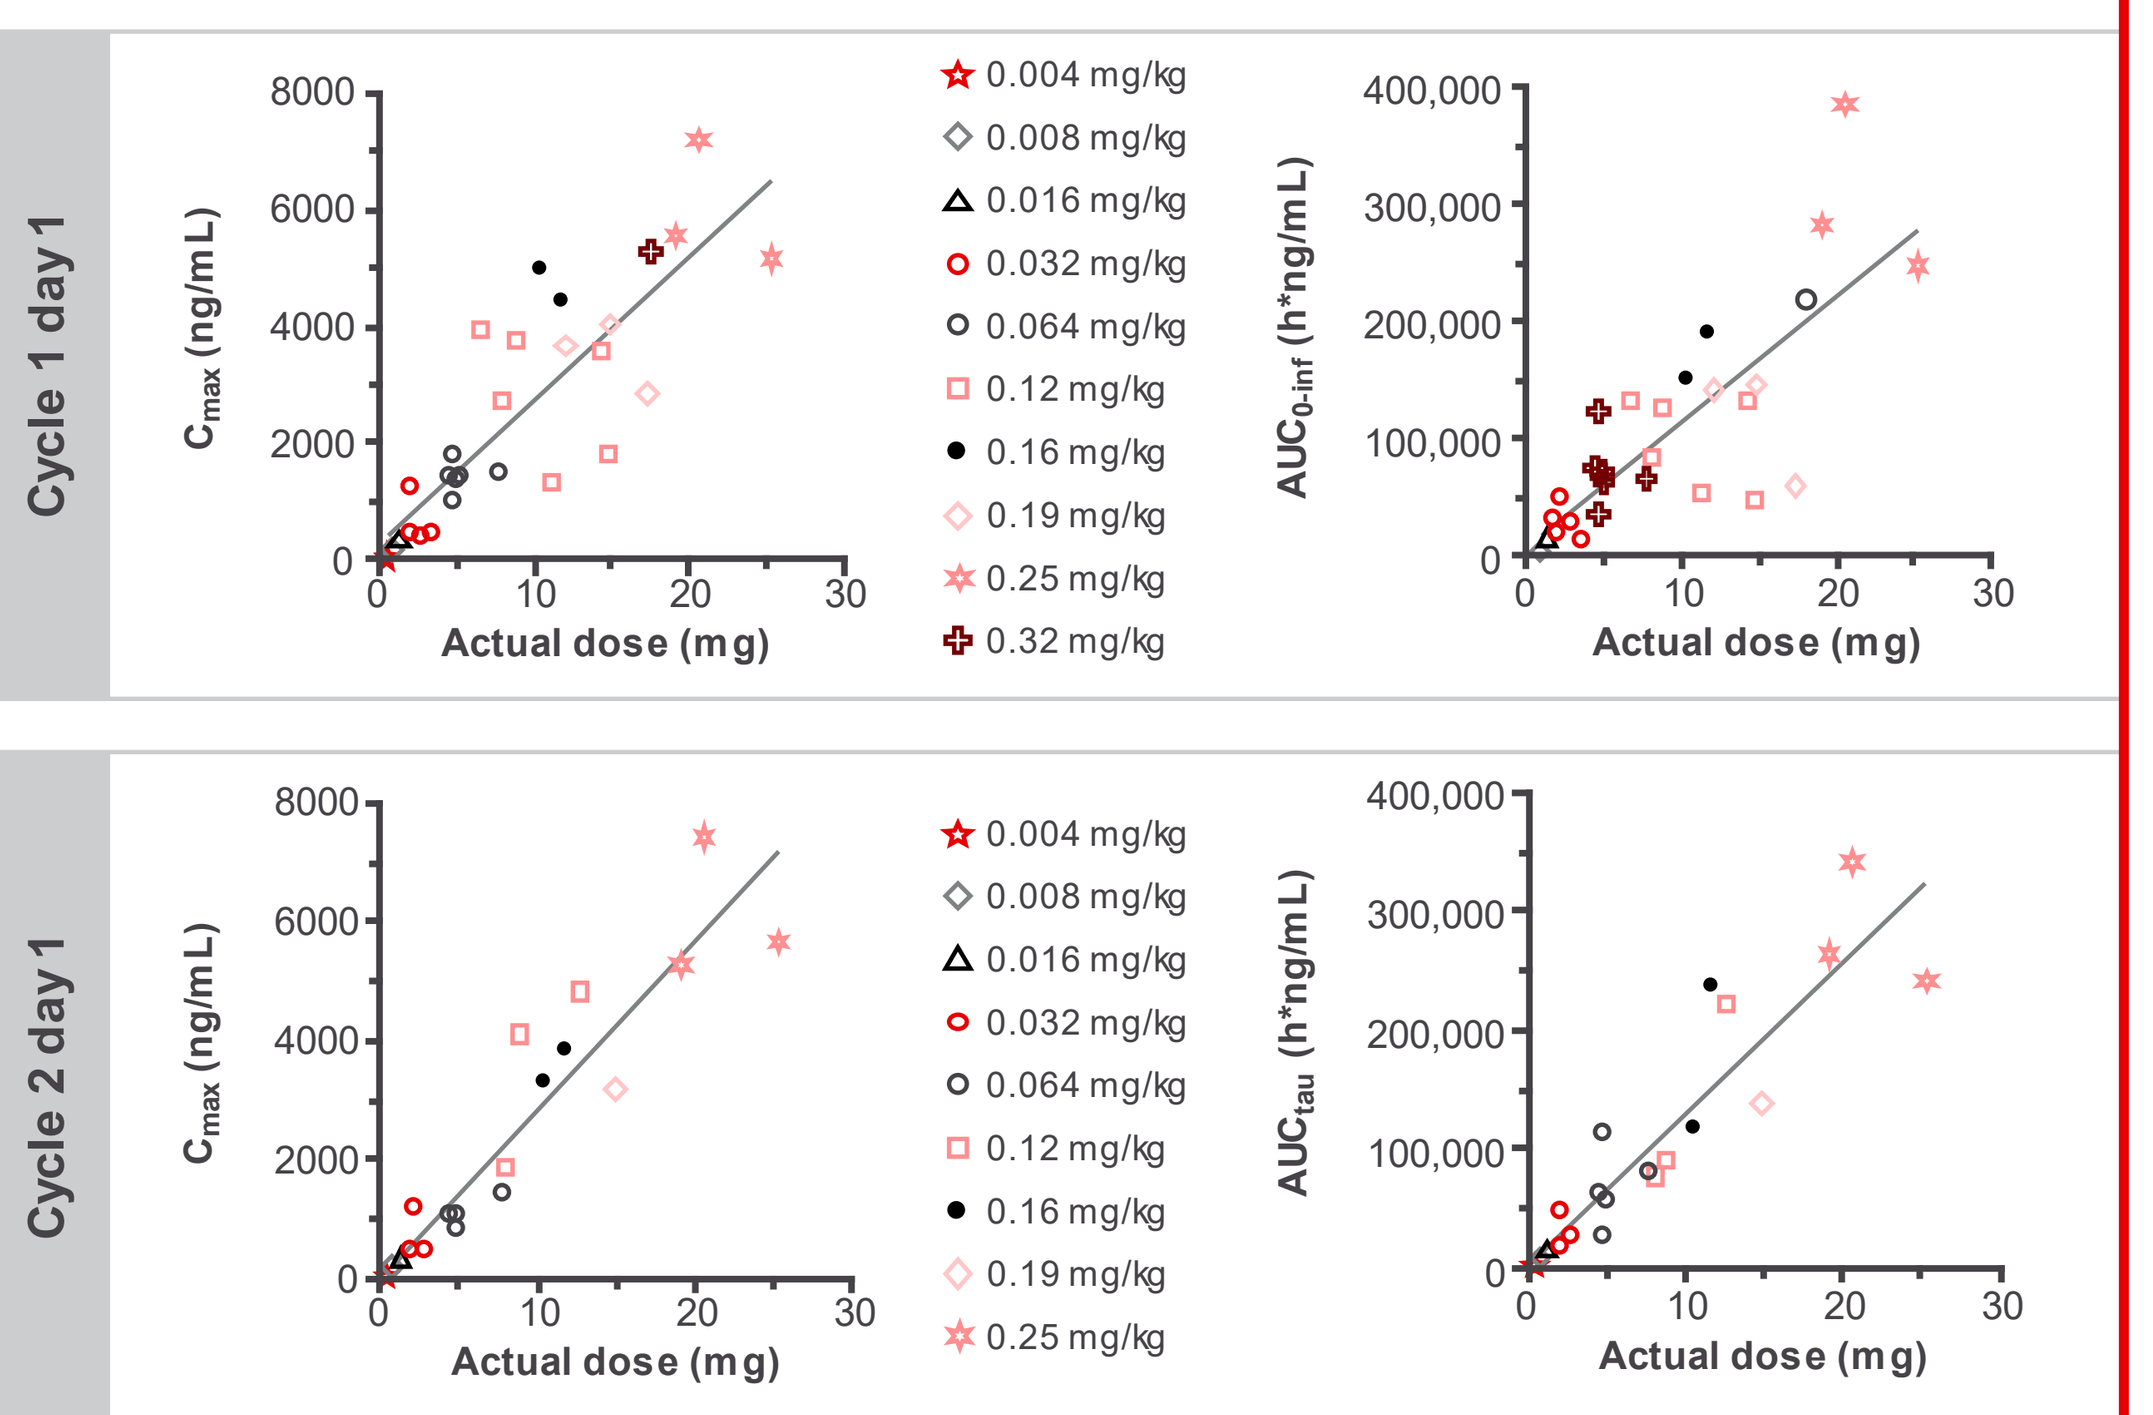
Cycle 1 day 1
Cycle 2 day 1
C_{max} (ng/mL)
AUC_{0-inf} (h*ng/mL)
AUC_{tau} (h*ng/mL)
Actual dose (mg)
0.004 mg/kg
0.008 mg/kg
0.016 mg/kg
0.032 mg/kg
0.064 mg/kg
0.12 mg/kg
0.16 mg/kg
0.19 mg/kg
0.25 mg/kg
0.32 mg/kg

- A relationship between clearance and body weight was observed, suggesting that body weight-based dosing is appropriate for TAK-164 **(Figure 5)**

Figure 5. TAK-164 clearance according to bodyweight

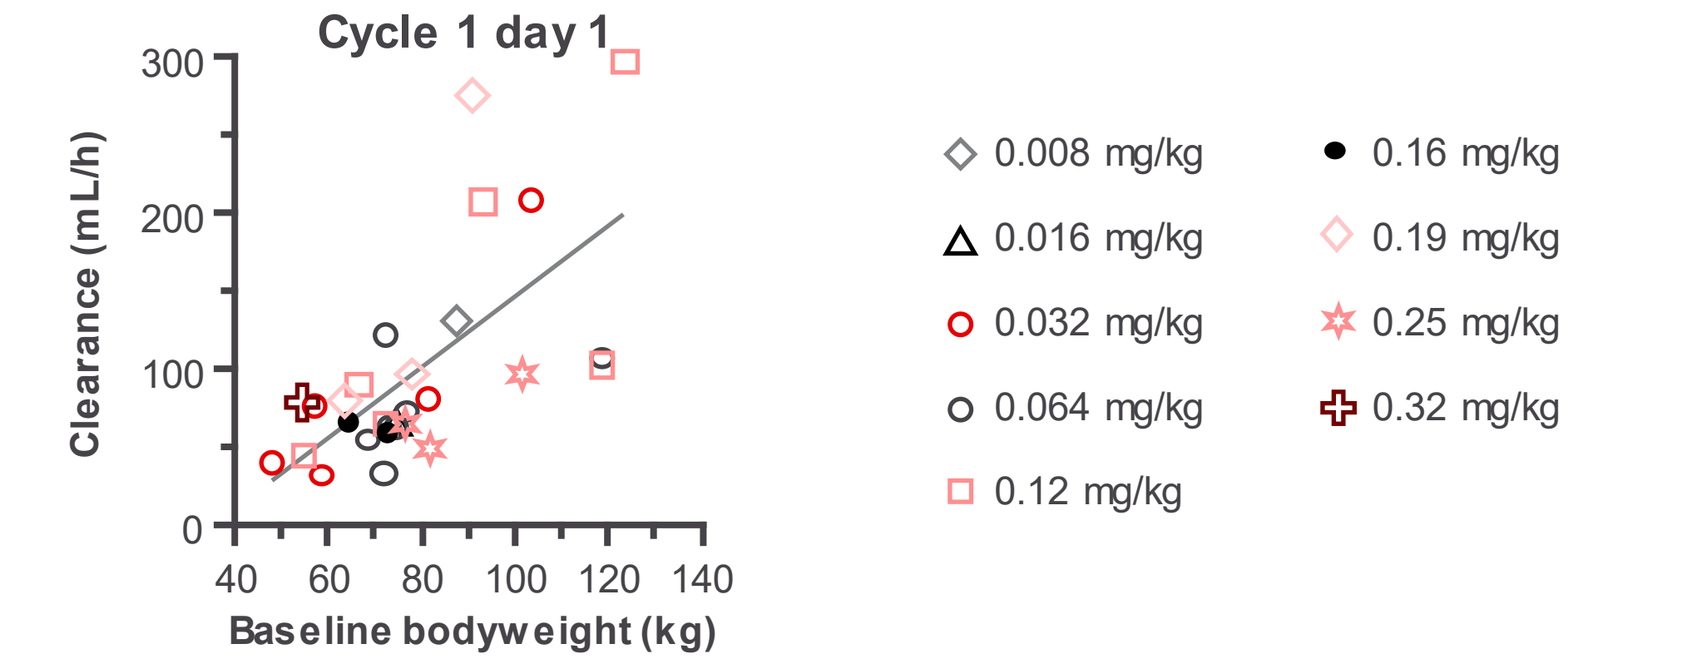
Cycle 1 day 1
Clearance (mL/h)
Baseline bodyweight (kg)
0.008 mg/kg
0.016 mg/kg
0.032 mg/kg
0.064 mg/kg
0.12 mg/kg
0.16 mg/kg
0.19 mg/kg
0.25 mg/kg
0.32 mg/kg

Target engagement

- Paired biopsies from two patients were available for γH2AX staining to assess target engagement:
 - Images from a patient receiving 0.19 mg/kg demonstrated target engagement with limited γH2AX staining present at baseline, and clear induction of γH2AX staining present following TAK-164 treatment **(Figure 6)**
 - Images from a patient receiving 0.064 mg/kg did not demonstrate target engagement, likely as a result of significant DNA damage visible at baseline

Figure 6. Baseline and post-treatment biopsy imaging for γH2AX staining, from a patient receiving 0.19 mg/kg TAK-164

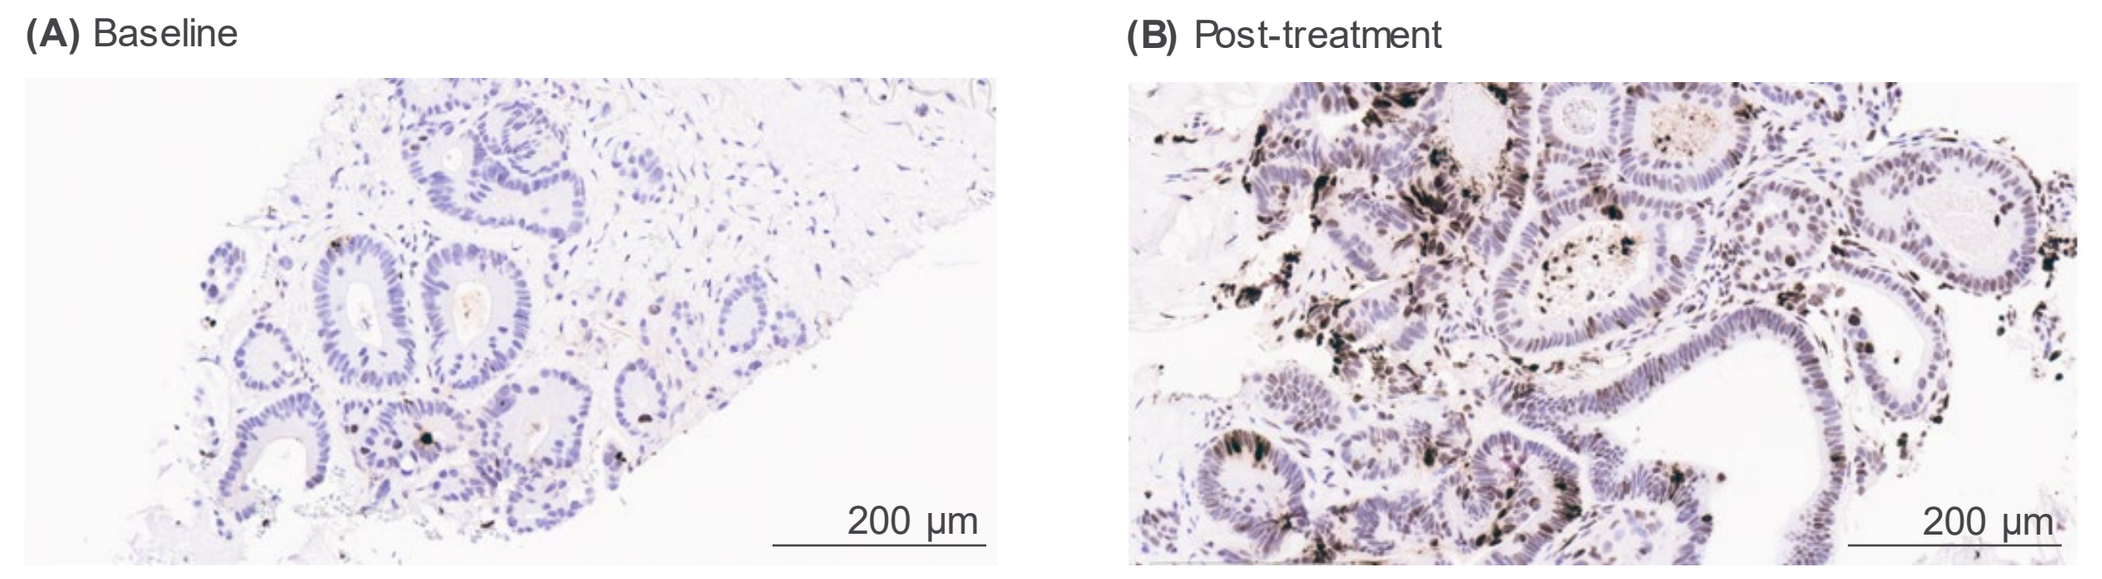
(A) Baseline
(B) Post-treatment
200 μm
200 μm

Response

- Overall, 25 patients were evaluable for response
- One patient diagnosed with colon cancer, with low baseline GCC expression who received 5 cycles of 0.008 mg/kg TAK-164 reported an unconfirmed PR at cycle 4, giving an ORR of 4%
- Eleven patients (44%) had a best overall response of SD giving a disease control rate of 48%

Conclusions

- TAK-164 appeared to have a manageable safety profile up to 0.064 mg/kg in patients with advanced GI cancers
- Hepatic toxicity was identified as a potential risk
- The mean terminal disposition half-life of TAK-164 ranged from 63.5 to 159 hours
- The RP2D was determined as 0.064 mg/kg; however, this dose appears to be insufficient and does not appear to be clinically effective

References

1. Krause WJ, et al. J Anat 1994;184:407–17.
2. Vaandrager AB. Mol Cell Biochem 2002;230:73–83.
3. Han X, et al. PLoS One 2011;6:e16139.
4. Li P, et al. Gastroenterology 2007;133:599–607.
5. Li P, et al. Am J Pathol 2007;171:1847–58.
6. Danaee H, et al. PLoS One 2017;12:e0189953.
7. Witek ME, et al. Clin Cancer Res 2005;11:8549–56.
8. Liu Y, et al. Oncotarget 2017;8:38841–9.
9. Gallery M, et al. PLoS One 2018;13:e0191046.
10. Almenoff JS, et al. Mol Microbiol 1993;8:865–73.
11. Abu-Yousif AO, et al. Mol Cancer Ther 2020;19:2079–88.

Abbreviations

ADA, anti-drug antibody; ADC, antibody-drug conjugate; AE, adverse event; ALP, alkaline phosphatase; ALT, alanine aminotransferase; AST, aspartate aminotransferase; AUC_{0-inf}, area under the curve 0-inf; C_{max}, maximum serum concentration; CNS, central nervous system; DLT, dose-limiting toxicity; ECOG PS, Eastern Cooperative Oncology Group Performance Status; EOT, end of treatment; GCC, guanylyl cyclase C; GI, gastrointestinal; IgG1, immunoglobulin G1; IHC, immunohistochemistry; INR, international normalized ratio; IV, intravenous; KRAS, Kirsten rat sarcoma viral oncogene homolog; mAb, monoclonal antibody; MSI, microsatellite instability status; MSS, microsatellite stable; MTD, maximum tolerated dose; mTPI, modified toxicity probability interval; NCI-CTCAE, National Cancer Institute Common Terminology Criteria for Adverse Events; ORR, overall response rate; PD, progressive disease; PK, pharmacokinetics; PLT, platelet; PR, partial response; Q3W, once every 3 weeks; RECIST, Response Evaluation Criteria in Solid Tumors; RP2D, recommended phase 2 dose; SAE, serious adverse event; SD, stable disease; TEAE, treatment-emergent adverse event; WBC, white blood cell.

Acknowledgments

- This study is funded by Millennium Pharmaceuticals, Inc., Cambridge, MA, USA, a wholly owned subsidiary of Takeda Pharmaceutical Company Limited
- We thank all patients and their families, and investigators at all clinical sites for their participation in the study
- Medical writing support for the development of this poster, under the direction of the authors, was provided by Selma Rayon, MA, and Helen Wilkinson, PhD, of Ashfield MedComms, an Ashfield Health company, funded by Millennium Pharmaceuticals, Inc., a wholly owned subsidiary of Takeda Pharmaceutical Company Limited, and complied with the Good Publication Practice 3 guidelines (Battisti WP, et al. Ann Intern Med 2015;163:461–4)

Disclosures

RK reports consulting/advisory role for Bristol-Myers Squibb, Lilly, Bayer, and Array BioPharma; speaker's bureau for Lilly; honoraria from Bristol-Myers Squibb and Lilly; and research funding (received by institution) from Bayer, Eisai and Bristol-Myers Squibb. **JMC** reports consulting/advisory role for Bristol-Myers Squibb; travel/accommodation/expenses from Roche, Agios, and Bristol-Myers Squibb; research funding from Merck, Tesaro, AstraZeneca, and Esperas Pharma; and research funding (received by institution) from Abbvie, Merus, Roche/Genentech, and Bristol-Myers Squibb. **ADL** reports research funding (received by institution) from Aduro-Biotech, Takeda, Bristol Myers Squibb, Exelixis, Corcept Therapeutics, and Hutchinson MediPharma. **AP** reports consulting/advisory role for Pure Tech, Foundation Medicine, Lilly, and Natera; and research funding (received by institution) from Plexxikon, Bristol-Myers-Squibb, Genentech, Guardant Health, Array BioPharma, Lilly, Novartis Pharmaceuticals, and Tesaro. **DPR** reports consulting/advisory role at MPM Capital, Oncorus, Gritstone Oncology, Maverick Therapeutics, TCR2 Therapeutics, and Twentyeight-Seven Therapeutics; expert testimony for Boehringer Ingelheim; patents/royalties/other intellectual property at McGraw Hill Chapter Royalties and John Hopkins University Press; stock/other ownership interests for MPM Capital, Acworth Pharmaceuticals, Thrive Earlier Detection Corp, and Exact Sciences; honoraria from UptoDate and Research to Practice; research funding (received by institution) from Stand up to Cancer; and other relationship with TCR2 Therapeutics. **AM** reports employment with Audentes Therapeutics, Inc. and Certara USA. **SW, BB, NG, and JP** report employment with Takeda. **HM** reports employment with and stock/other ownership interests for Takeda. **JB** reports employment with Takeda; consulting or advisory role (funding received by institution) for Gilead Sciences, Genentech/Roche, Bristol-Myers Squibb, Five Prime Therapeutics, Lilly, Merck, MedImmune, Celgene, EMD Serono, Taiho Pharmaceutical, Macrogenics, GlaxoSmithKline, Novartis, OncoMed, Leap Therapeutics, TG Therapeutics, AstraZeneca, Boehringer Ingelheim, Daiichi Sankyo, Bayer, Incyte, Apexigen, Array BioPharma, Sanofi, ARMO BioSciences, Ipsen, Merrimack, Oncogenex, FORMA Therapeutics, Arch Oncology, Prelude Therapeutics, Phoenix Biotech, Cyteir, Molecular Partners, Innate Pharma, Torque, Tizona Therapeutics, Inc., Janssen, Tolero Pharmaceuticals, TD2, Amgen, Seattle Genetics, Moderna Therapeutics, Tanabe Research, Beigene, Continuum Clinical, Cerulean Pharma, Kyn Therapeutics, Bicycle Therapeutics, Relay Therapeutics, Evelo Therapeutics, and Fusion Pharmaceuticals; travel/accommodation/expenses from Merck, Roche/Genentech, Celgene, Daiichi Sankyo, Gilead Sciences, Bristol-Myers Squibb, Lilly, MedImmune, Taiho Pharmaceutical, Novartis, OncoMed, Boehringer Ingelheim, ARMO BioSciences, Ipsen, and FORMA Therapeutics; and research funding (received by institution) from Lilly, Genentech/Roche, Incyte, Gilead Sciences, Bristol-Myers Squibb, Leap Therapeutics, AstraZeneca/MedImmune, Boston Biomedical, GlaxoSmithKline, Novartis, Array BioPharma, Taiho Pharmaceutical, Celgene, OncoMed, Daiichi Sankyo, Bayer, Apexigen, Kolltan Pharmaceuticals, SynDevRx, Merck, Macrogenics, Five Prime Therapeutics, EMD Serono, TG Therapeutics, Boehringer Ingelheim, Forty Seven, Stem CentRx, Onyx, Sanofi, Takeda, Abbott/AbbVie, Eisai, Celldex, Agios, ARMO BioSciences, CytomX Therapeutics, Nektar, Ipsen, Merrimack, Tarveda Therapeutics, Tyrogenex, Oncogenex, Marshall Edwards, Pieris Pharmaceuticals, Mersana, Calithera Biosciences, Blueprint Medicines, Gritstone Oncology, Evelo Therapeutics, FORMA Therapeutics, Forty Seven, EMD Serono, Merus, Jacobio, eFFECTOR Therapeutics, Novocure, Sorrento Therapeutics, Arrys Therapeutics, TRACON Pharma, Sierra Oncology, Innate Pharma, Prelude Therapeutics, Arch Oncology, Harpoon Therapeutics, Phoenix Biotech, Unum Therapeutics, Vyriad, Cyteir, Molecular Partners, ADC Therapeutics, Torque, Tizona Therapeutics, Inc., Janssen, Amgen, BeiGene, Pfizer, Millennium Pharmaceuticals, ImClone Systems, Acerta Pharma, Rgenix, Bellicum Pharmaceuticals, Arcus Biosciences, Gossamer Bio, Seattle Genetics, Tempest Therapeutics, Shattuck Labs, Synthorx, Revolution Medicines, Bicycle Therapeutics, Zymeworks, Relay Therapeutics, Evelo Therapeutics, Scholar Rock, NGM Biopharmaceuticals, Numab, AtlasMedx, Treadwell Therapeutics, IGM, MabSpace Biosciences, Hutchison MediPharma, Repare Therapeutics, NeoImmuneTech, Regeneron, PureTech, G1 Therapeutics, Erasca, Inc., Rubius Therapeutics, Pionyr, Loxo/Lilly, BioNTech AG, and Elicio Therapeutics.

To request a mobile-friendly version of this poster, scan this QR code or visit: http://tago.ca/1asco

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® and the author of this poster.

Electronic poster presentation at the virtual 2021 Annual Meeting of the American Society of Clinical Oncology (ASCO), June 4–8, 2021. Contact information: [richard.kim@moffitt.org].