Final analysis of the phase 3 TOURMALINE-MM1 study investigating ixazomib + lenalidomide-dexamethasone (Rd) vs placebo-Rd in relapsed and/or refractory multiple myeloma patients

EP963

Paul G. Richardson,[1] Shaji K. Kumar,[2] Tamás Masszi,[3] Norbert Grzasko,[4,5] Nizar J. Bahlis,[6] Markus Hansson,[7] Luděk Pour,[8] Irwindeep Sandhu,[9] Peter Ganly,[10] Bartrum W. Baker,[11] Sharon R. Jackson,[12] Anne-Marie Stoppa,[13] Peter Gimsing,[14] Laurent Garderet,[15] Cyrille Touzeau,[16] Francis K. Buadi,[2] Jacob P. Laubach,[1] Michele Cavo,[17] Mohamed Darif,[18] Richard Labotka,[18] Deborah Berg,[18] Philippe Moreau[16]

[1]Dana-Farber Cancer Institute, Boston, MA, USA; [2]Division of Hematology, Mayo Clinic, Rochester, MN, USA; [3]3rd Department of Internal Medicine, Semmelweis University, Budapest, Hungary; [4]Department of Experimental Hematooncology, Medical University of Lublin, Lublin, Poland; [5]Center of Oncology of the Lublin Region St. Jana z Dukli, Lublin, Poland; [6]Arnie Charbonneau Cancer Research Institute, University of Calgary, Calgary, Canada; [7]Department of Hematology, Skåne University Hospital, Lund, and Sahlgrenska Academy, Göteborg, Sweden; [8]Hematology and Oncology, University Hospital Brno, Brno, Czech Republic; [9]Cross Cancer Institute, University of Alberta, Edmonton, Canada; [10]Department of Haematology, Christchurch Hospital, Christchurch, New Zealand; [11]Department of Haematology, Palmerston North Hospital, Palmerston North, New Zealand; [12]Department of Haematology, Middlemore Hospital, Auckland, New Zealand; [13]Department of Hematology, Institut Paoli-Calmettes, Marseille, France; [14]Department of Hematology, University Hospital Rigshospitalet, Copenhagen, Denmark; [15]Hôpital Pitié-Salpêtrière, Paris, France; [16]University Hospital Hôtel Dieu, Nantes, France; [17]IRCCS Azienda Ospedaliero-Universitaria di Bologna, Istituto di Ematologia "Seràgnoli", Bologna, Italy; [18]Millennium Pharmaceuticals, Inc., Cambridge, MA, USA, a wholly owned subsidiary of Takeda Pharmaceutical Company Limited

Background

- An increasing number of treatment options are becoming available for RRMM,[1,2] resulting in marked improvements in OS.[3]
- However, there remains a need for effective and tolerable therapies for:
 - Patients who continue to relapse or are refractory to treatment
 - Hard-to-treat subgroups, e.g., elderly/frail, patients with high disease burden, patients with high-risk cytogenetics.[2]
- PIs are an established backbone of therapy;[1,4] longer- vs shorter-term PI-based therapy may prolong PFS and OS.[5]
 - Longer-term use of parenteral PIs may be limited by treatment burden and the need for clinic/hospital visits.[6,7]
 - Thus, there remains a need for all-oral regimens with manageable toxicity that can delay progression in RRMM patients.

Ixazomib

- Ixazomib, the first oral PI,[8] is approved in combination with Rd for the treatment of MM patients who have received ≥1 prior therapy.[9,10]
 - The phase 3 TOURMALINE-MM1 study demonstrated a statistically significant improvement in PFS with ixazomib-Rd vs placebo-Rd in RRMM patients (median 20.6 vs 14.7 months; HR 0.74; p=0.01), with limited additional toxicity.[11]

Objective

- Here we report the final analysis for OS in the ITT population of TOURMALINE-MM1 (a key secondary endpoint), and OS in subgroups of interest.

Methods and disposition

- TOURMALINE-MM1 (NCT01564537) was a randomized, double-blind, phase 3 study of ixazomib-Rd vs placebo-Rd in adult patients with RRMM (**Figure 1**).

Figure 1. TOURMALINE-MM1 study design and patient disposition at final OS analysis

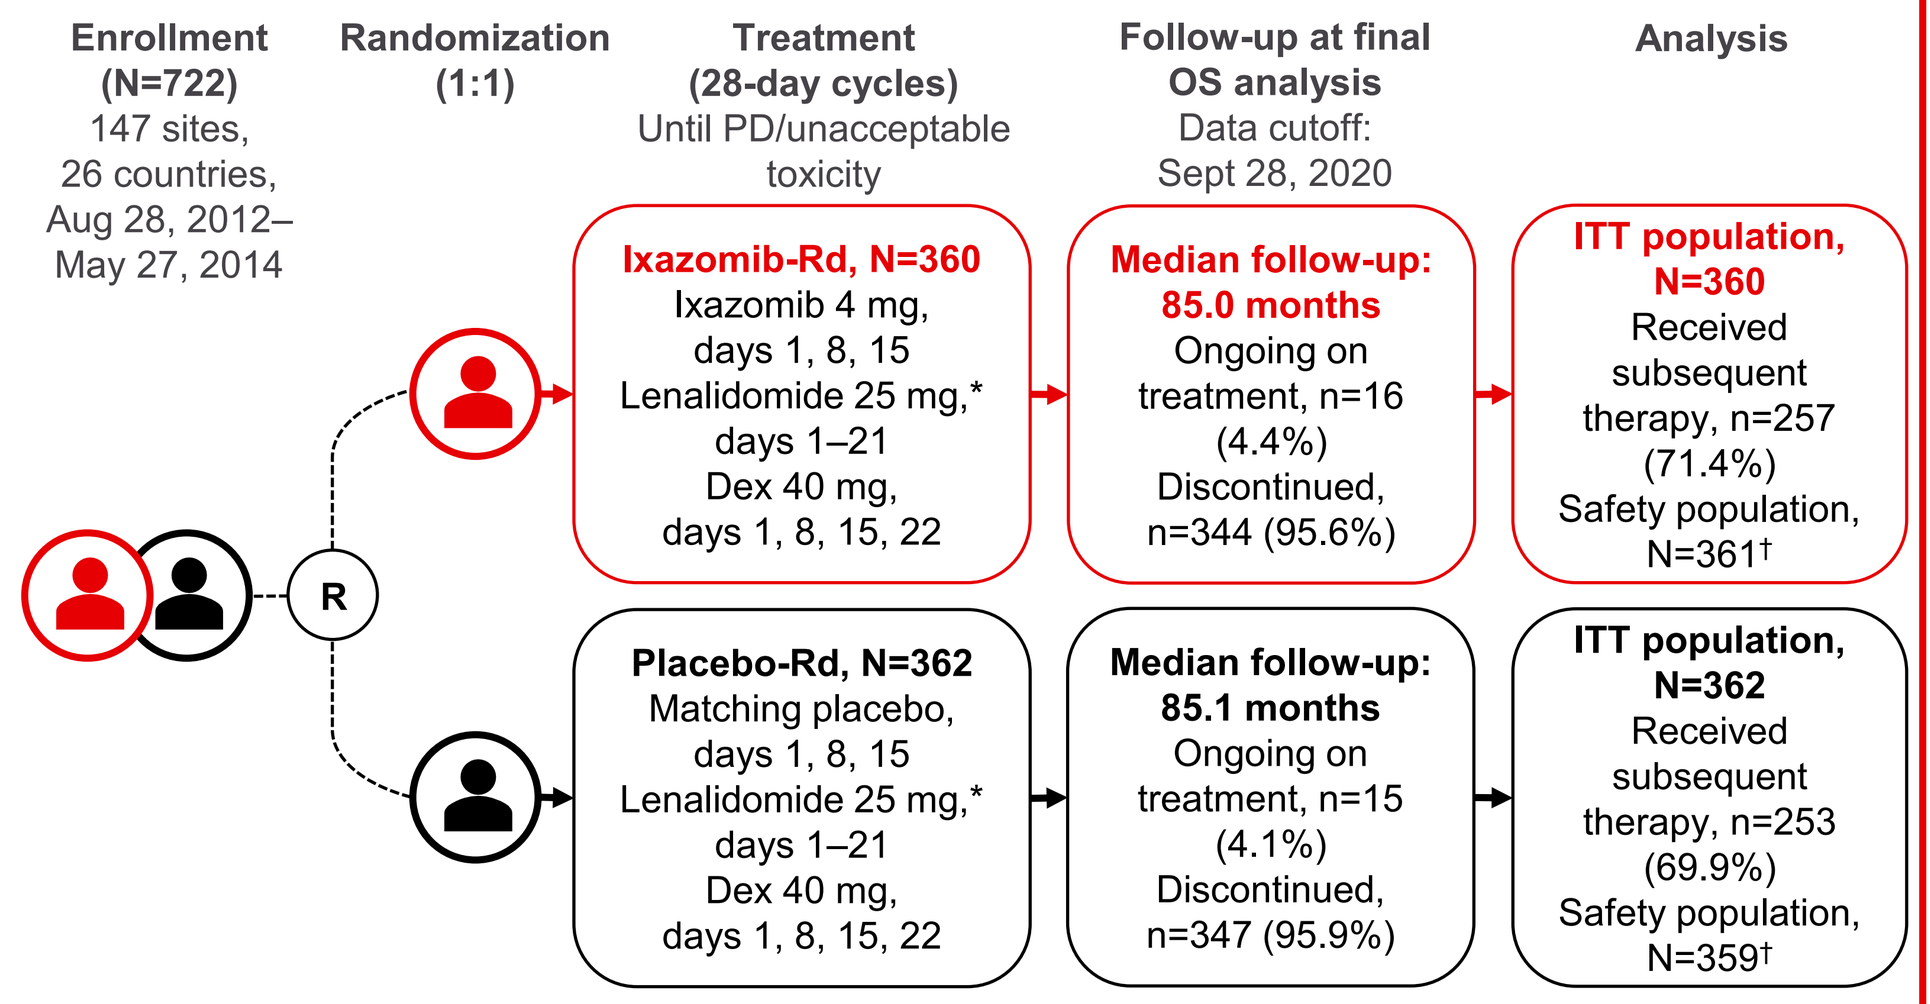

Enrollment (N=722)
147 sites, 26 countries, Aug 28, 2012– May 27, 2014
Randomization (1:1)
Treatment (28-day cycles)
Until PD/unacceptable toxicity
Follow-up at final OS analysis
Data cutoff: Sept 28, 2020
Analysis
R
Ixazomib-Rd, N=360
Ixazomib 4 mg, days 1, 8, 15
Lenalidomide 25 mg,* days 1–21
Dex 40 mg, days 1, 8, 15, 22
Median follow-up: 85.0 months
Ongoing on treatment, n=16 (4.4%)
Discontinued, n=344 (95.6%)
ITT population, N=360
Received subsequent therapy, n=257 (71.4%)
Safety population, N=361†
Placebo-Rd, N=362
Matching placebo, days 1, 8, 15
Lenalidomide 25 mg,* days 1–21
Dex 40 mg, days 1, 8, 15, 22
Median follow-up: 85.1 months
Ongoing on treatment, n=15 (4.1%)
Discontinued, n=347 (95.9%)
ITT population, N=362
Received subsequent therapy, n=253 (69.9%)
Safety population, N=359†

Key eligibility criteria:
- RRMM
- Measurable disease
- 1–3 prior therapies
- ECOG PS 0–2

Stratification:
- Prior therapies: 1 vs 2/3
- Prior PI exposure: yes vs no
- ISS disease stage: I/II vs III

Key endpoints	Exploratory ad-hoc analyses
Primary: PFS	Subsequent therapy
Key secondary:	TTNT
OS (ITT, α=0.0462)	OS according to
OS in patients with del(17p)	subsequent therapy

*10 mg for patients with reduced CrCl according to local prescribing information. †Two patients in the ixazomib-Rd arm did not receive treatment and 3 in the placebo-Rd arm erroneously received ≤2 doses of ixazomib and were included in the ixazomib-Rd arm in the safety population.

Results

Patient characteristics

- Patient demographics and baseline characteristics were well balanced between arms (**Table 1**).

Table 1. Key baseline characteristics (ITT population)

Characteristic	Ixazomib-Rd (N=360)	Placebo-Rd (N=362)
Median age, years (range)	66 (38–91)	66 (30–89)
Male, n (%)	207 (58)	202 (56)
White, n (%)	312 (87)	303 (84)
ISS stage I / II / III at study entry, n (%)	226 (63) / 89 (25) / 45 (13)	233 (64) / 86 (24) / 43 (12)
Cytogenetics, n (%)		
High risk – del(17p), t(4;14), t(14;16)	75 (21)	62 (17)
Standard risk	200 (56)	216 (60)
Not available	85 (24)	84 (23)
Expanded high risk – del(17p), t(4;14), t(14;16), amp1q21	155 (43)	154 (43)
1 / 2 / 3 prior therapies, n (%)*	224 (62) / 97 (27) / 39 (11)	217 (60) / 111 (31) / 34 (9)
Prior stem cell transplant, n (%)	212 (59)	199 (55)
Prior PI therapy, n (%)	249 (69)	253 (70)
Prior bortezomib	248 (69)	250 (69)
Prior IMiD therapy, n (%)	193 (54)	204 (56)

*Number of prior therapies determined by the sponsor in a blinded medical review of data.

Final OS analysis: ITT population

- After a median follow-up for OS of 85 months:
 - 484 (67%) patients had died
 - 229 (32%) were alive at last OS follow-up.
- Median OS was 53.6 vs 51.6 months with ixazomib-Rd vs placebo-Rd (**summary panel**).
- OS was not statistically significantly different between arms (HR 0.939, p=0.495).
- On two prespecified sensitivity analyses for OS, to adjust for the confounding effect of subsequent therapies (MSM and IPCW methods), HRs were 0.68 and 0.70, respectively.

Final OS analysis: Prespecified subgroups

- Lower HRs, indicating a larger magnitude of OS benefit with ixazomib-Rd vs placebo-Rd, were seen in various prespecified subgroups of patients with adverse-risk characteristics as shown in **Figure 2** (patients with high-risk cytogenetics) and **Figure 3** (shaded subgroups plus patients with del(17p) [HR 0.916; data not shown in figure]).
- OS benefit was also seen in patients with standard-risk cytogenetics (**Figure 3**).

QUESTION

Does the significant improvement seen in PFS with ixazomib-Rd vs placebo-Rd in patients with RRMM after 1–3 prior therapies translate into an OS benefit?

INVESTIGATION

Randomization (1:1) N=722 — Stratified by prior PI exposure; ISS disease stage; lines of prior therapy — Ixazomib-Rd (N=360) / Placebo-Rd (N=362) — Treat in 28-day cycles until PD or unacceptable toxicity

RESULTS

Key secondary endpoint: OS

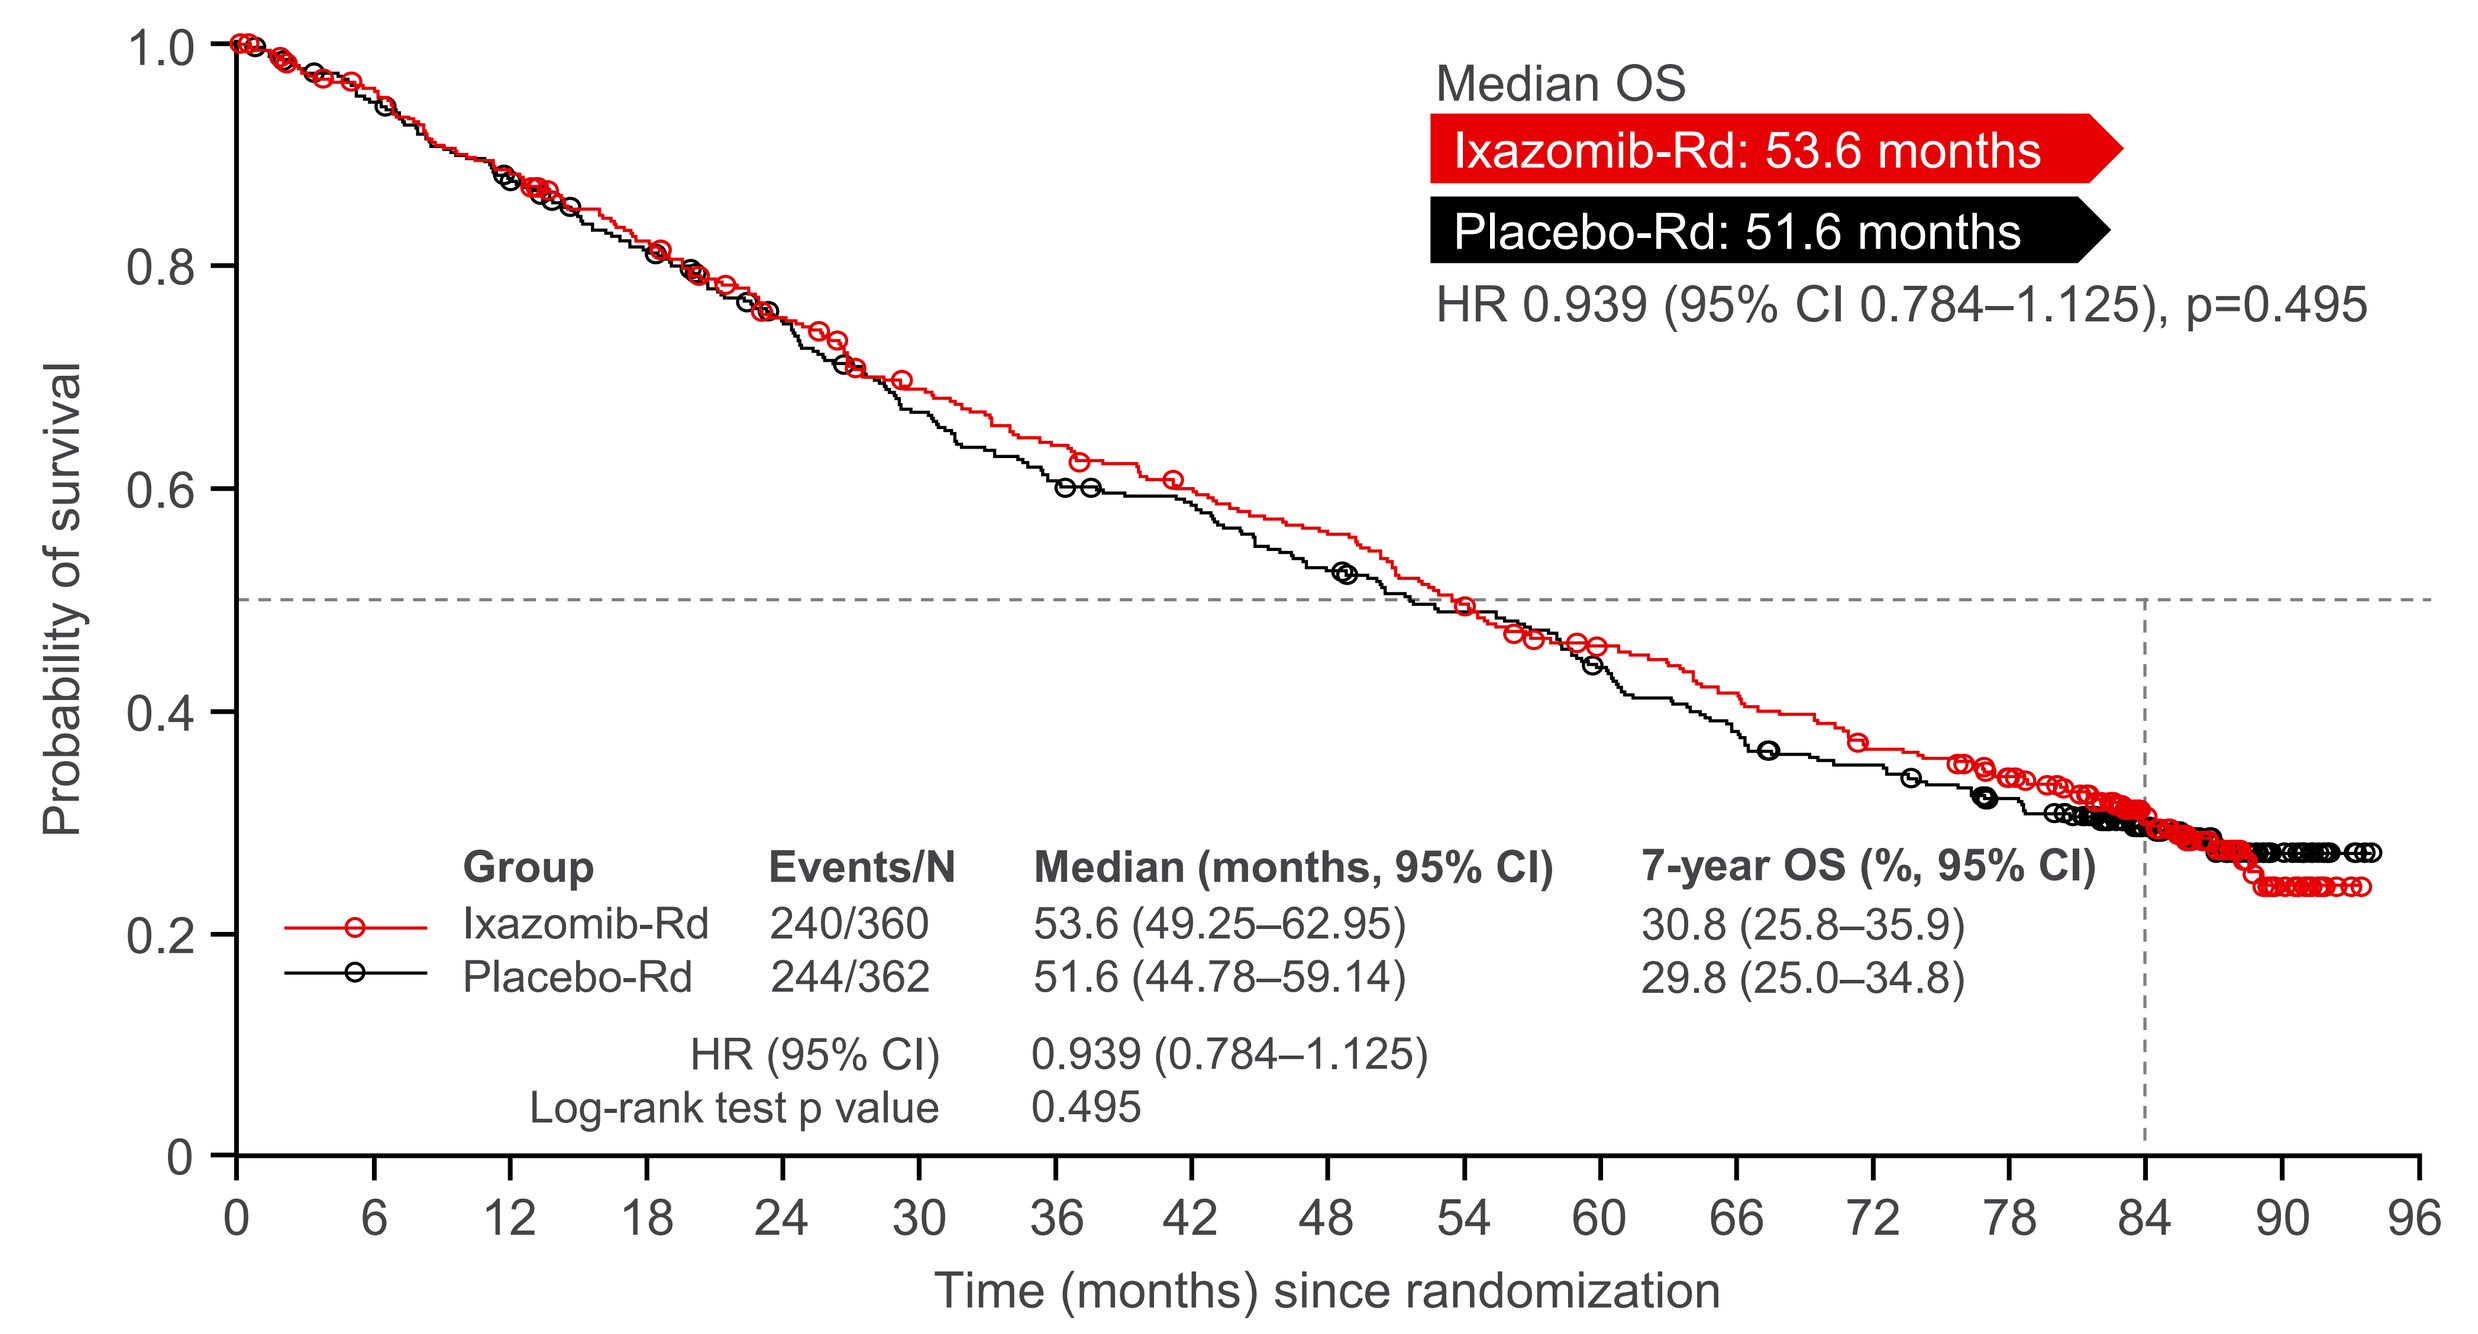

Median OS
Ixazomib-Rd: 53.6 months
Placebo-Rd: 51.6 months
HR 0.939 (95% CI 0.784–1.125), p=0.495
Probability of survival
Group | Events/N | Median (months, 95% CI) | 7-year OS (%, 95% CI)
Ixazomib-Rd | 240/360 | 53.6 (49.25–62.95) | 30.8 (25.8–35.9)
Placebo-Rd | 244/362 | 51.6 (44.78–59.14) | 29.8 (25.0–34.8)
HR (95% CI) 0.939 (0.784–1.125)
Log-rank test p value 0.495
Time (months) since randomization

Exploratory ad-hoc analysis: Any subsequent therapy

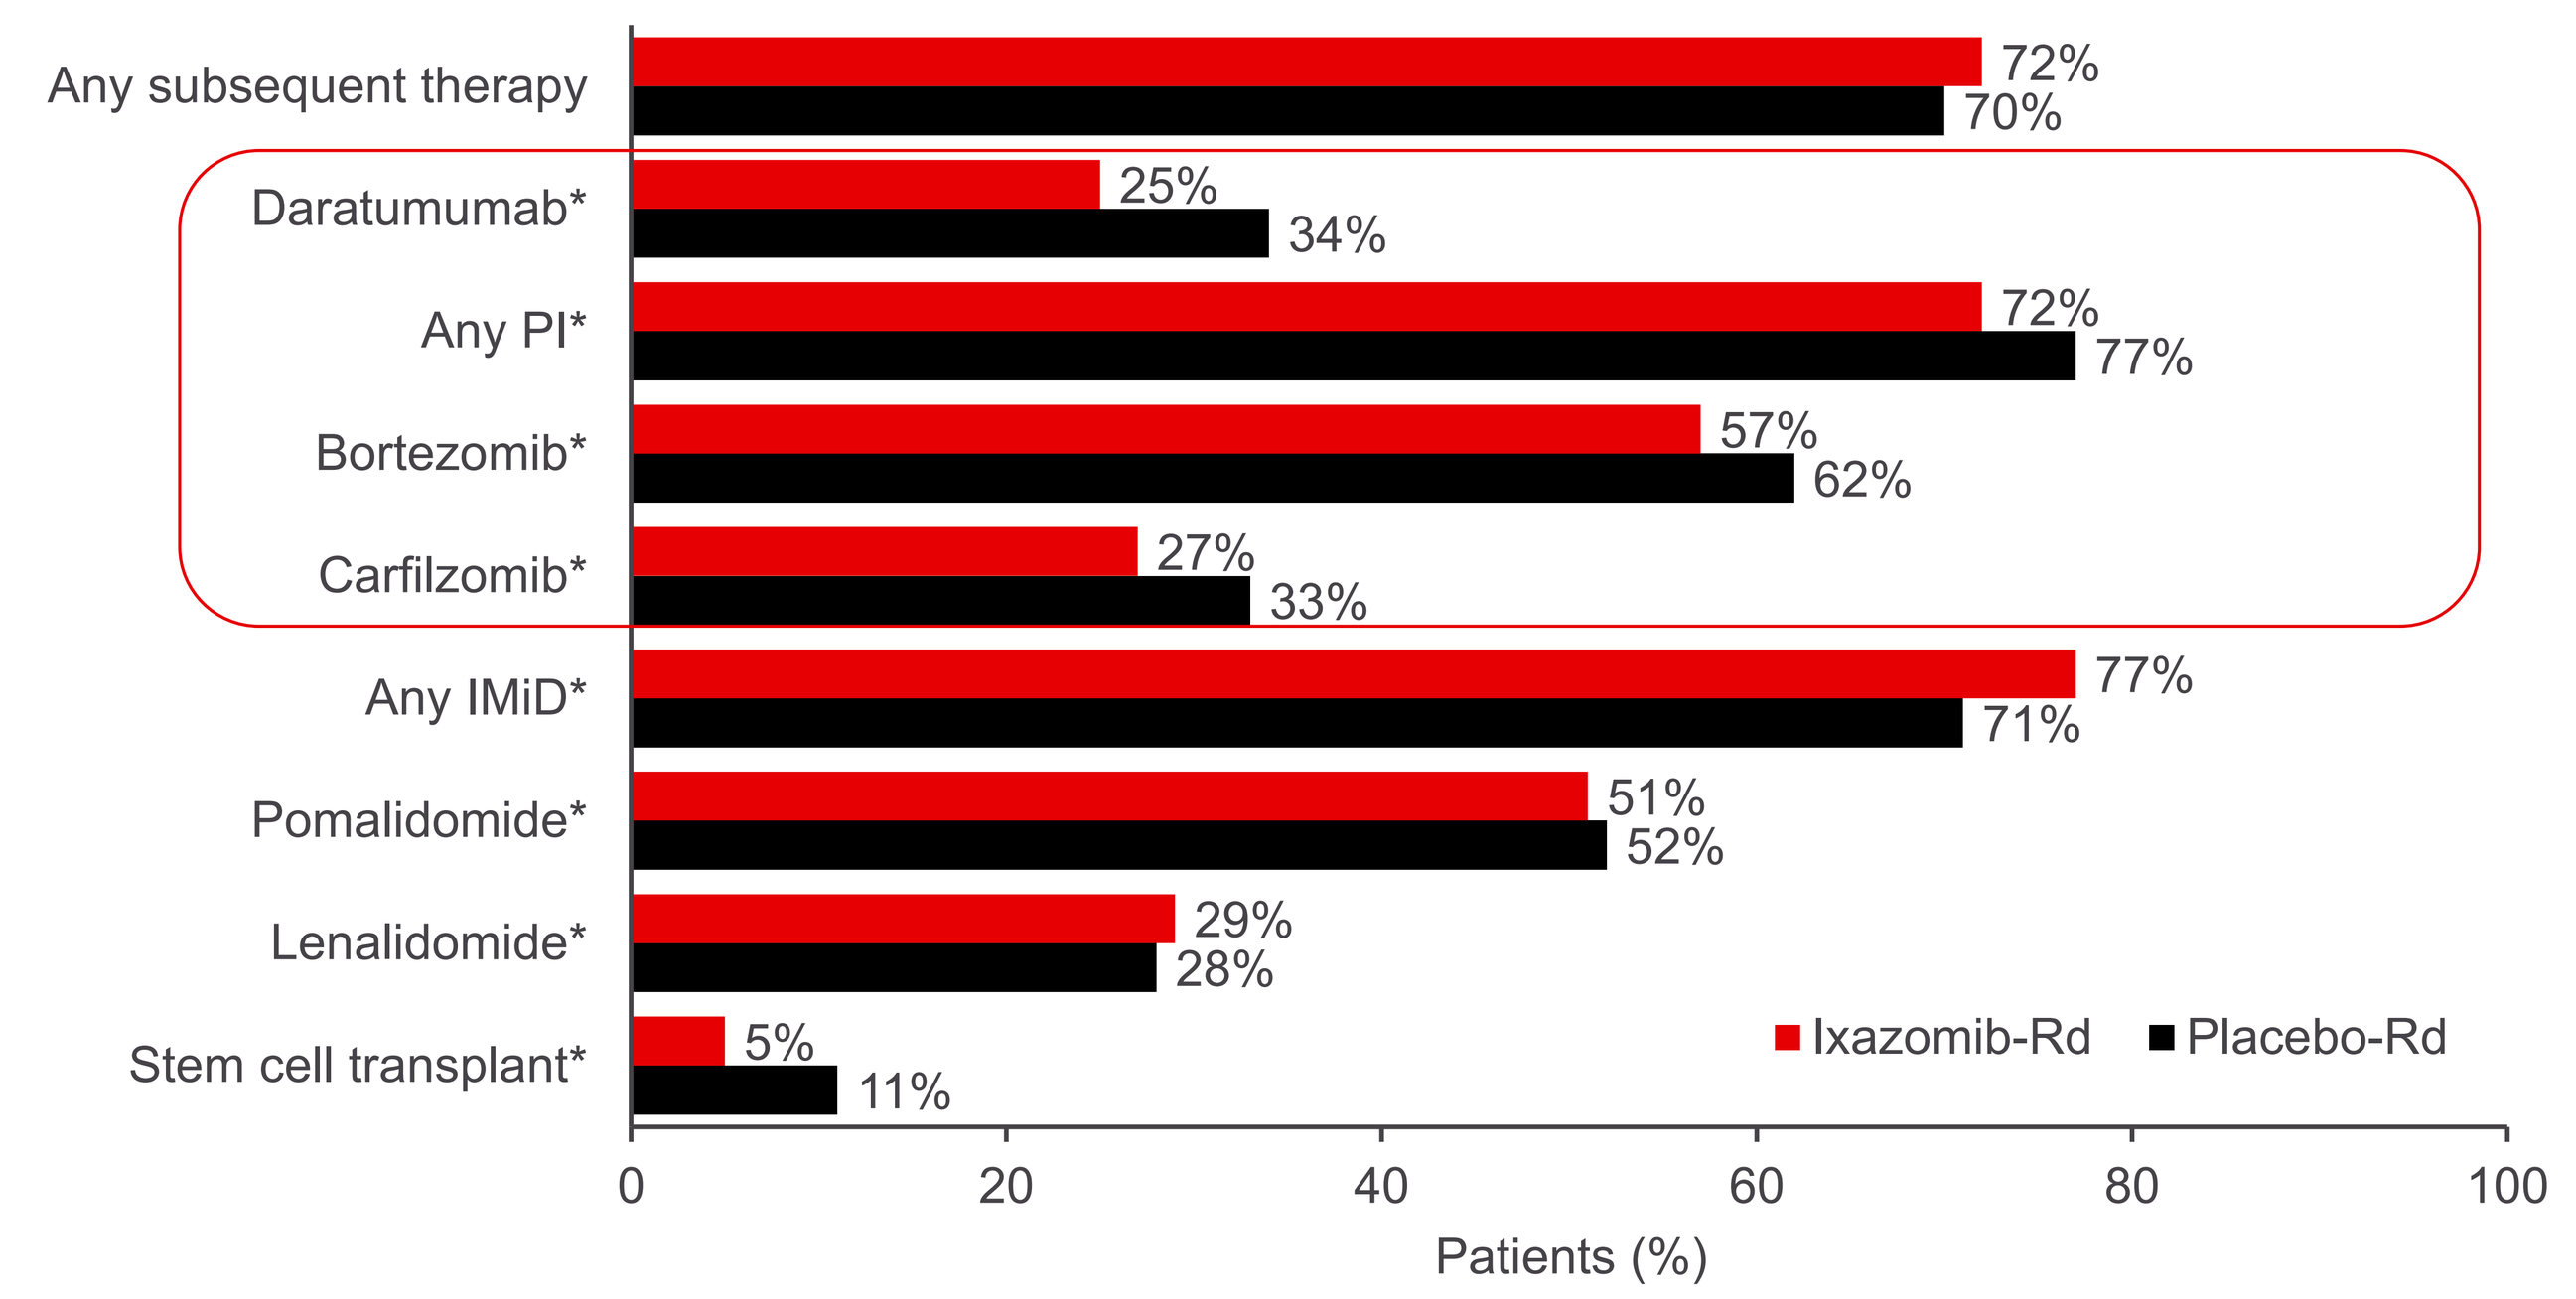

Any subsequent therapy 72% 70%
Daratumumab* 25% 34%
Any PI* 72% 77%
Bortezomib* 57% 62%
Carfilzomib* 27% 33%
Any IMiD* 77% 71%
Pomalidomide* 51% 52%
Lenalidomide* 29% 28%
Stem cell transplant* 5% 11%
Ixazomib-Rd
Placebo-Rd
Patients (%)

*Number of patients with ≥1 subsequent therapy (ixazomib-Rd, n=259; placebo-Rd, n=251) used as the denominator for percentages.

CONCLUSIONS

The slight OS trend in favor of ixazomib-Rd in the ITT population was not statistically significant, but OS interpretation was confounded by imbalances between arms in subsequent therapies

Figure 2. OS with ixazomib-Rd vs placebo-Rd in patients with high-risk cytogenetics

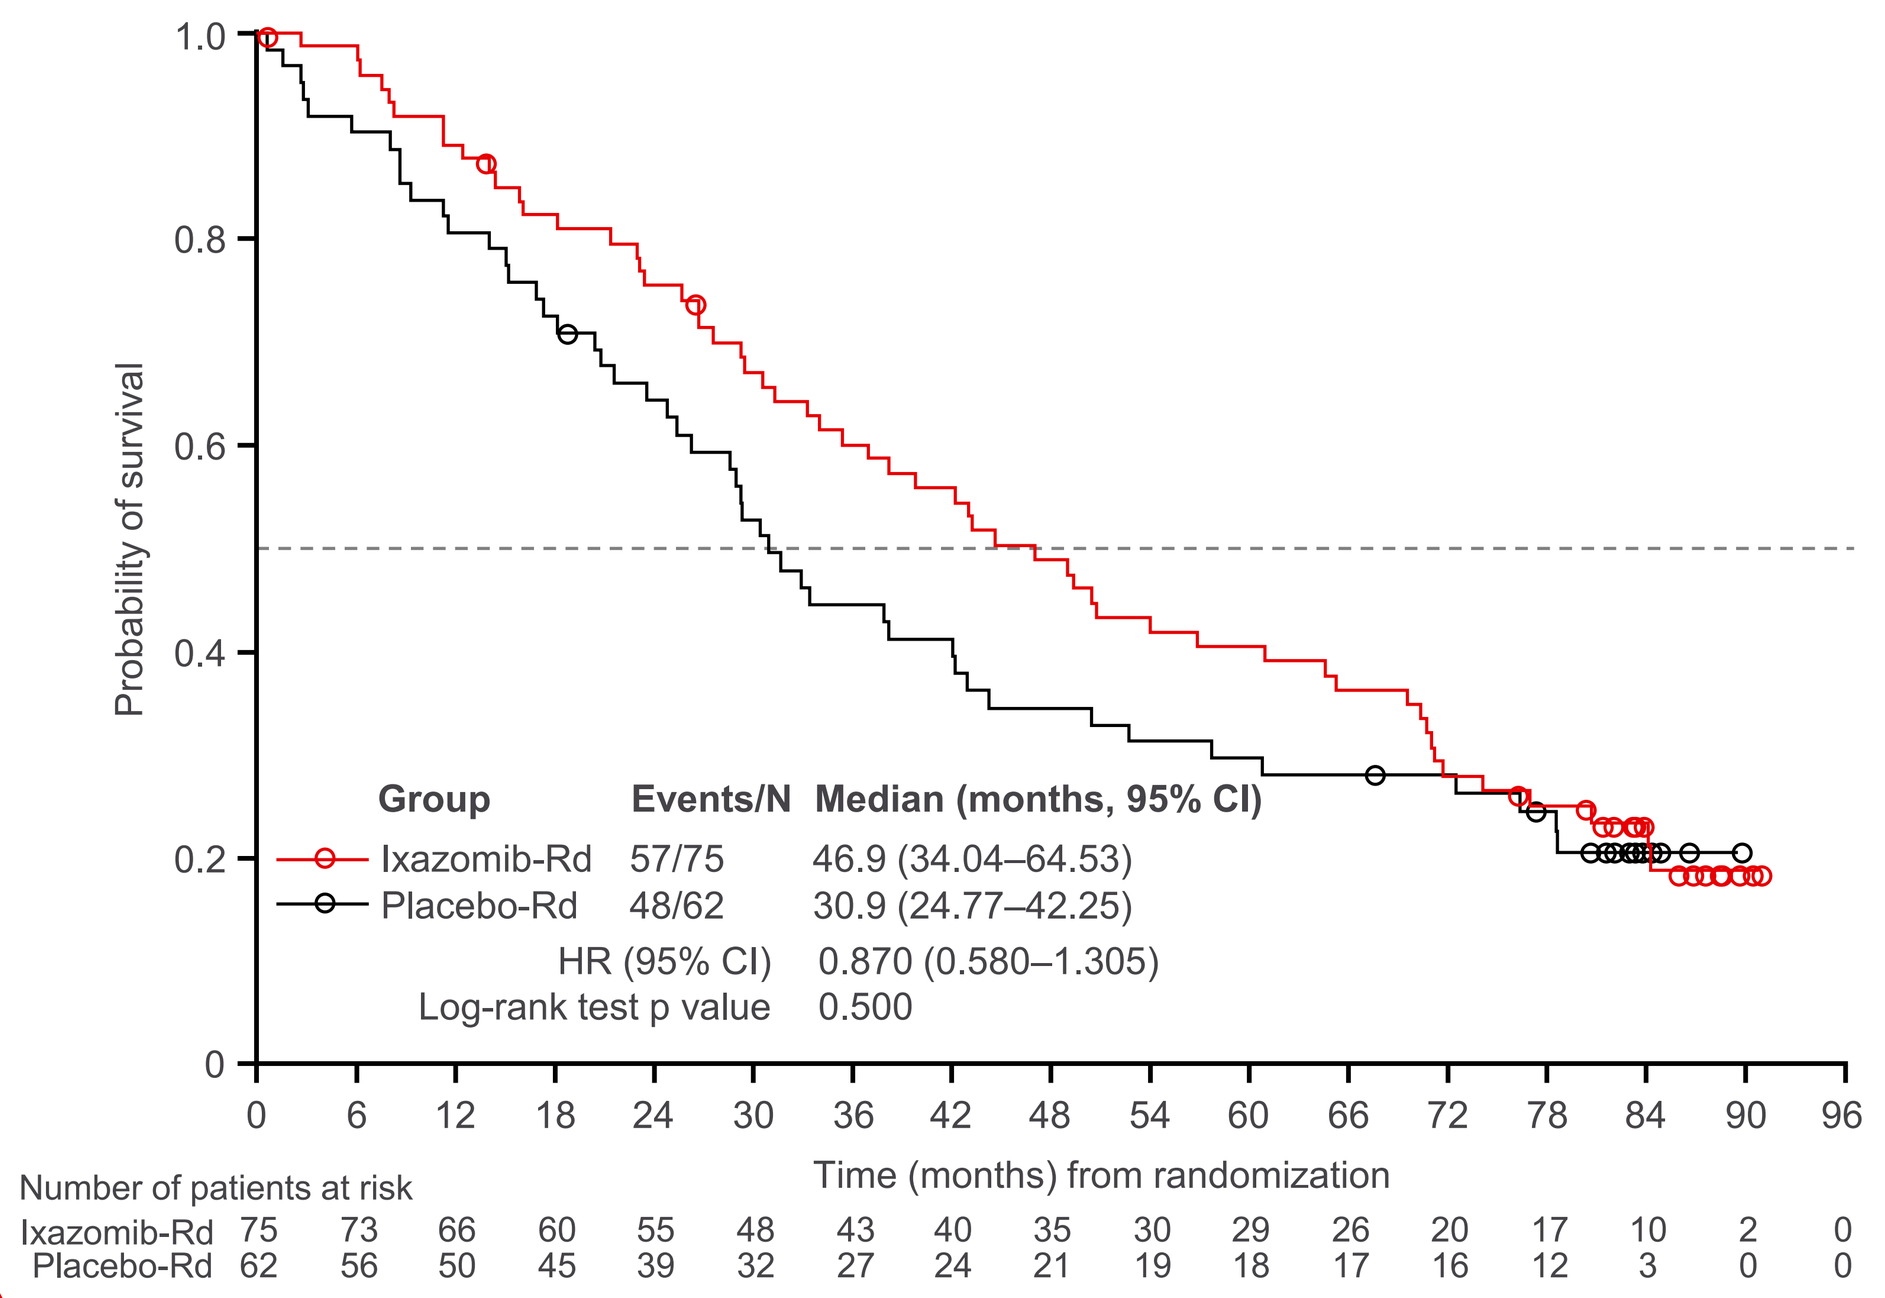

Probability of survival
Group | Events/N | Median (months, 95% CI)
Ixazomib-Rd | 57/75 | 46.9 (34.04–64.53)
Placebo-Rd | 48/62 | 30.9 (24.77–42.25)
HR (95% CI) 0.870 (0.580–1.305)
Log-rank test p value 0.500
Time (months) from randomization
Number of patients at risk
Ixazomib-Rd 75 73 66 60 55 48 43 40 35 30 29 26 20 17 10 2 0
Placebo-Rd 62 56 50 45 39 32 27 24 21 19 18 17 16 12 3 0 0

Figure 3. Forest plot of OS with ixazomib-Rd vs placebo-Rd in prespecified patient subgroups

Variable	Subgroup	Events, n/N Ixazomib-Rd	Events, n/N Placebo-Rd	Median survival (months) Ixazomib-Rd	Median survival (months) Placebo-Rd	HR	95% CI
All patients	ALL (N=722)	240 / 360	244 / 362	53.6	51.6	0.939	(0.784–1.125)
Age, years	≤65 (n=344)	106 / 168	111 / 176	54.9	60.4	1.079	(0.822–1.416)
	>65–75 (n=270)	93 / 145	85 / 125	56.7	46.9	0.757	(0.559–1.027)
	>75 (n=108)	41 / 47	48 / 61	36.9	37.9	1.068	(0.670–1.704)
ISS stage	I or II (n=632)	202 / 314	207 / 318	59.5	57.7	0.970	(0.798–1.178)
	III (n=90)	38 / 46	37 / 44	27.6	23.6	0.779	(0.487–1.247)
Cytogenetic risk	High risk (n=137)	57 / 75	48 / 62	46.9	30.9	0.870	(0.580–1.305)
	Standard (n=416)	123 / 200	140 / 216	56.9	55.8	0.875	(0.684–1.118)
	Expanded high risk (n=309)	112 / 155	117 / 154	44.6	33.4	0.862	(0.660–1.124)
Prior therapies (IVRS)	1 (n=425)	136 / 212	140 / 213	54.3	58.3	1.016	(0.801–1.287)
	2 or 3 (n=297)	104 / 148	104 / 149	53.0	43.0	0.845	(0.642–1.114)
PI	Exposed (n=503)	173 / 250	169 / 253	53.0	55.8	0.994	(0.803–1.231)
	Naive (n=219)	67 / 110	75 / 109	55.4	50.4	0.817	(0.585–1.142)
Prior IMiD therapy	Exposed (n=397)	130 / 193	138 / 204	52.4	50.5	0.918	(0.717–1.176)
	Naive (n=325)	110 / 167	106 / 158	54.3	56.6	0.945	(0.721–1.240)
Relapsed or refractory	Relapsed (n=557)	186 / 278	188 / 279	53.9	57.7	1.000	(0.815–1.227)
	Refractory (n=80)	21 / 40	30 / 40	51.0	32.0	0.680	(0.375–1.236)
	Relapsed and refractory (n=85)	33 / 42	26 / 43	54.3	44.2	0.920	(0.531–1.593)
Refractory to any prior line	Yes (n=165)	54 / 82	56 / 83	52.2	35.5	0.794	(0.538–1.172)
	No (n=557)	186 / 278	188 / 279	53.9	57.7	1.000	(0.815–1.227)
Refractory to last prior line	Yes (n=117)	36 / 59	40 / 58	49.0	35.5	0.742	(0.460–1.198)
	No (n=605)	204 / 301	204 / 304	54.3	56.9	1.003	(0.824–1.221)
Thalidomide refractory	Yes (n=89)	26 / 39	36 / 50	42.2	31.7	0.781	(0.461–1.322)
	No (n=633)	214 / 321	208 / 312	54.3	57.7	0.975	(0.804–1.183)

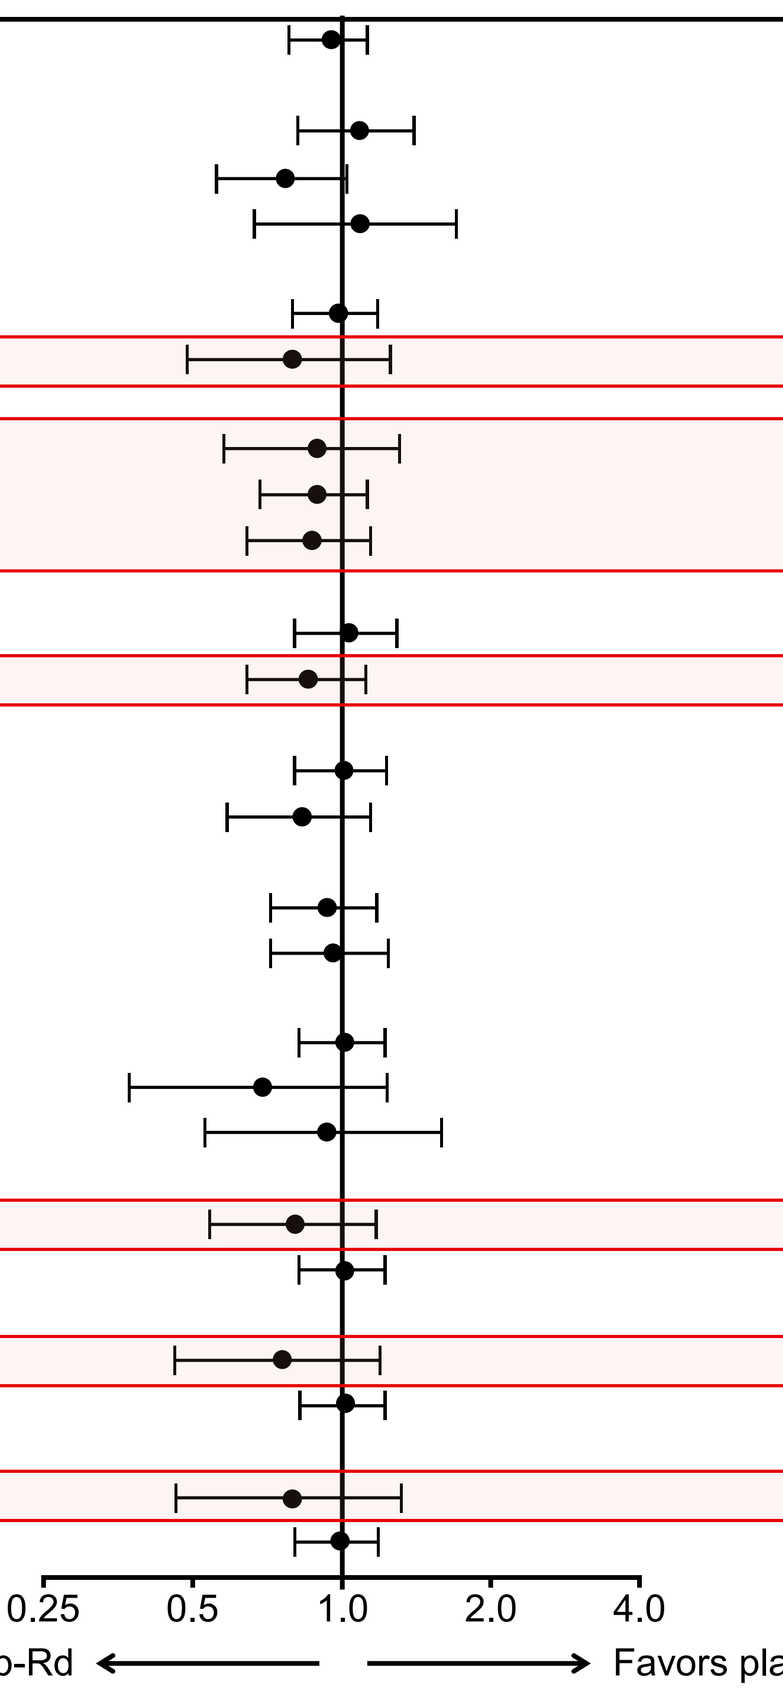

0.25 0.5 1.0 2.0 4.0
Favors ixazomib-Rd
Favors placebo-Rd

Exploratory ad-hoc analysis: Next-line and subsequent therapy

- Median TTNT (ITT population) was 29.7 and 26.9 months in the ixazomib-Rd and placebo-Rd arms, respectively (HR 0.917).
 - 72% and 70% of patients, respectively, received subsequent therapy.
- There were imbalances between groups in subsequent therapies received; patients progressed earlier on placebo-Rd (shorter PFS/TTP)[11], received a higher number of subsequent therapies than those on ixazomib-Rd (median 3 vs 2), and received subsequent PIs, daratumumab, and other treatments more frequently (**summary panel** and **Figure 4**).

Figure 4. Subsequent therapies received by patients in the ixazomib-Rd and placebo-Rd groups (safety population) as part of next-line therapy

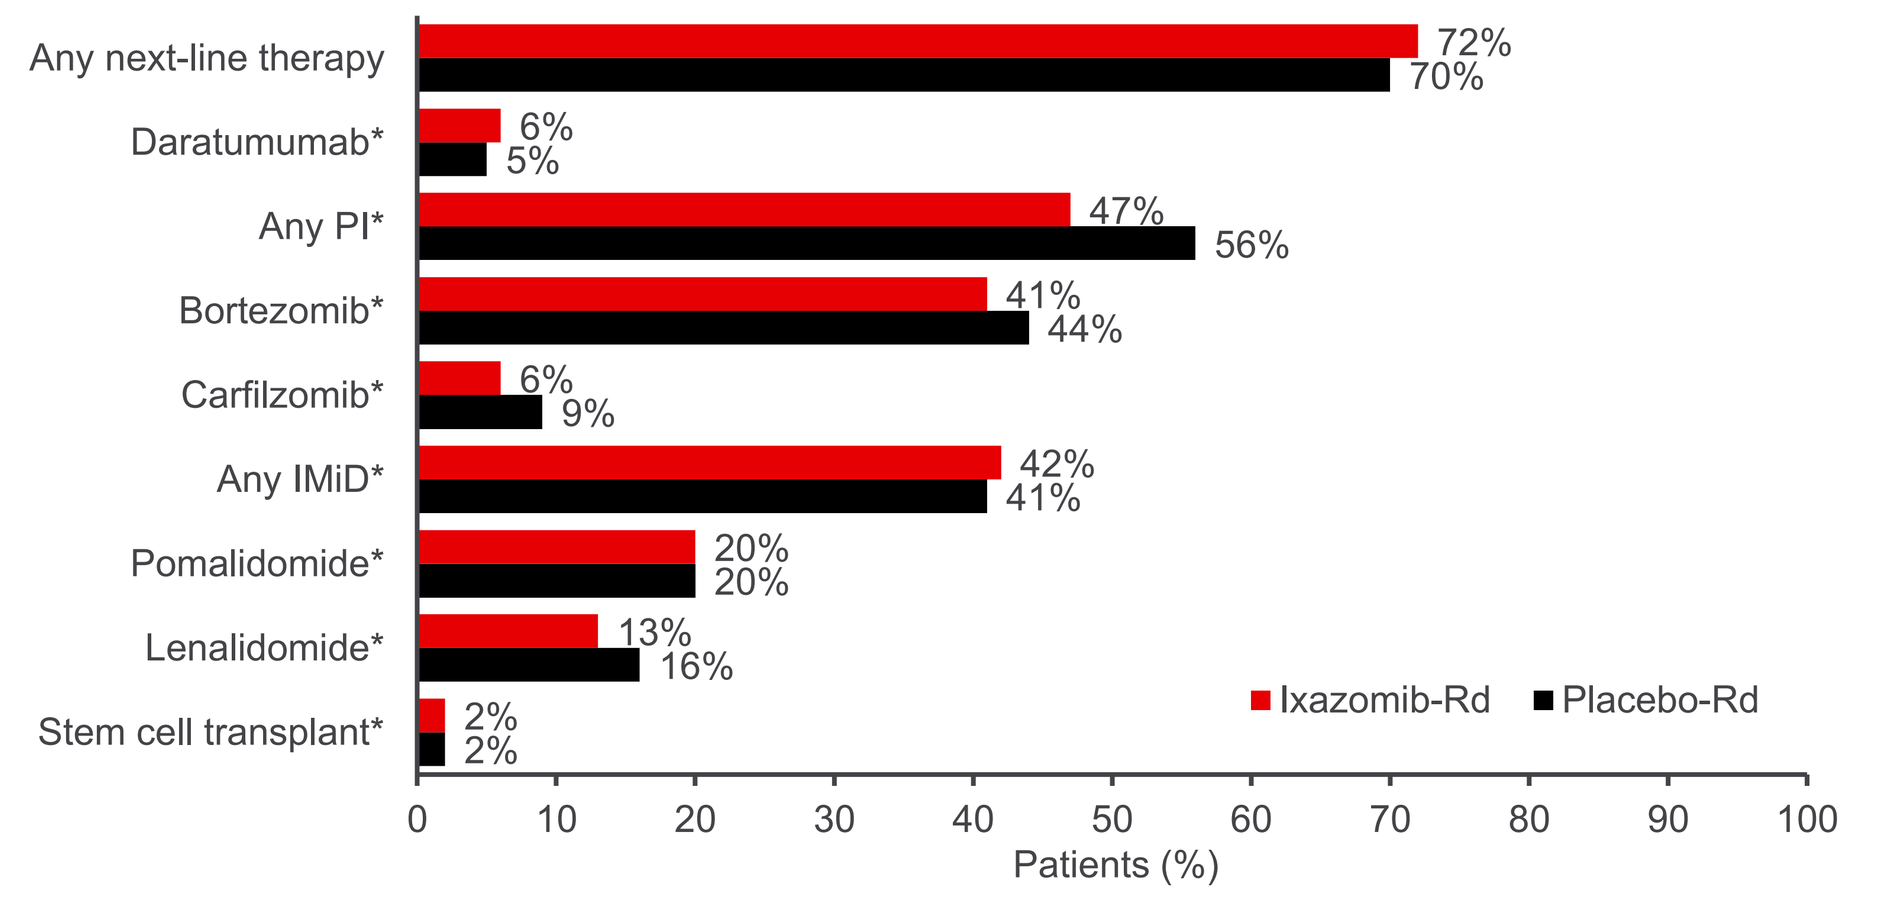

Any next-line therapy 72% 70%
Daratumumab* 6% 5%
Any PI* 47% 56%
Bortezomib* 41% 44%
Carfilzomib* 6% 9%
Any IMiD* 42% 41%
Pomalidomide* 20% 20%
Lenalidomide* 13% 16%
Stem cell transplant* 2% 2%
Ixazomib-Rd
Placebo-Rd
Patients (%)

*Number of patients with ≥1 subsequent therapy (ixazomib-Rd, n=259; placebo-Rd, n=251) used as the denominator for percentages.

Exploratory ad-hoc analysis: OS according to subsequent therapy

- Patients in the ixazomib-Rd vs placebo-Rd arms received subsequent daratumumab less frequently and later in the course of the study.
 - Among patients receiving daratumumab in any subsequent line, median OS was 78.9 vs 83.4 months (HR 1.15); in those not receiving subsequent daratumumab, median OS was 49.2 vs 35.5 months (HR 0.83).
- A similar differential OS benefit was seen between patients who received a PI as next-line therapy and those who did not (**Figure 5**).

Figure 5. OS with ixazomib-Rd vs placebo-Rd in (A) patients who received a PI-based next-line therapy and (B) the remaining patients in the ITT population

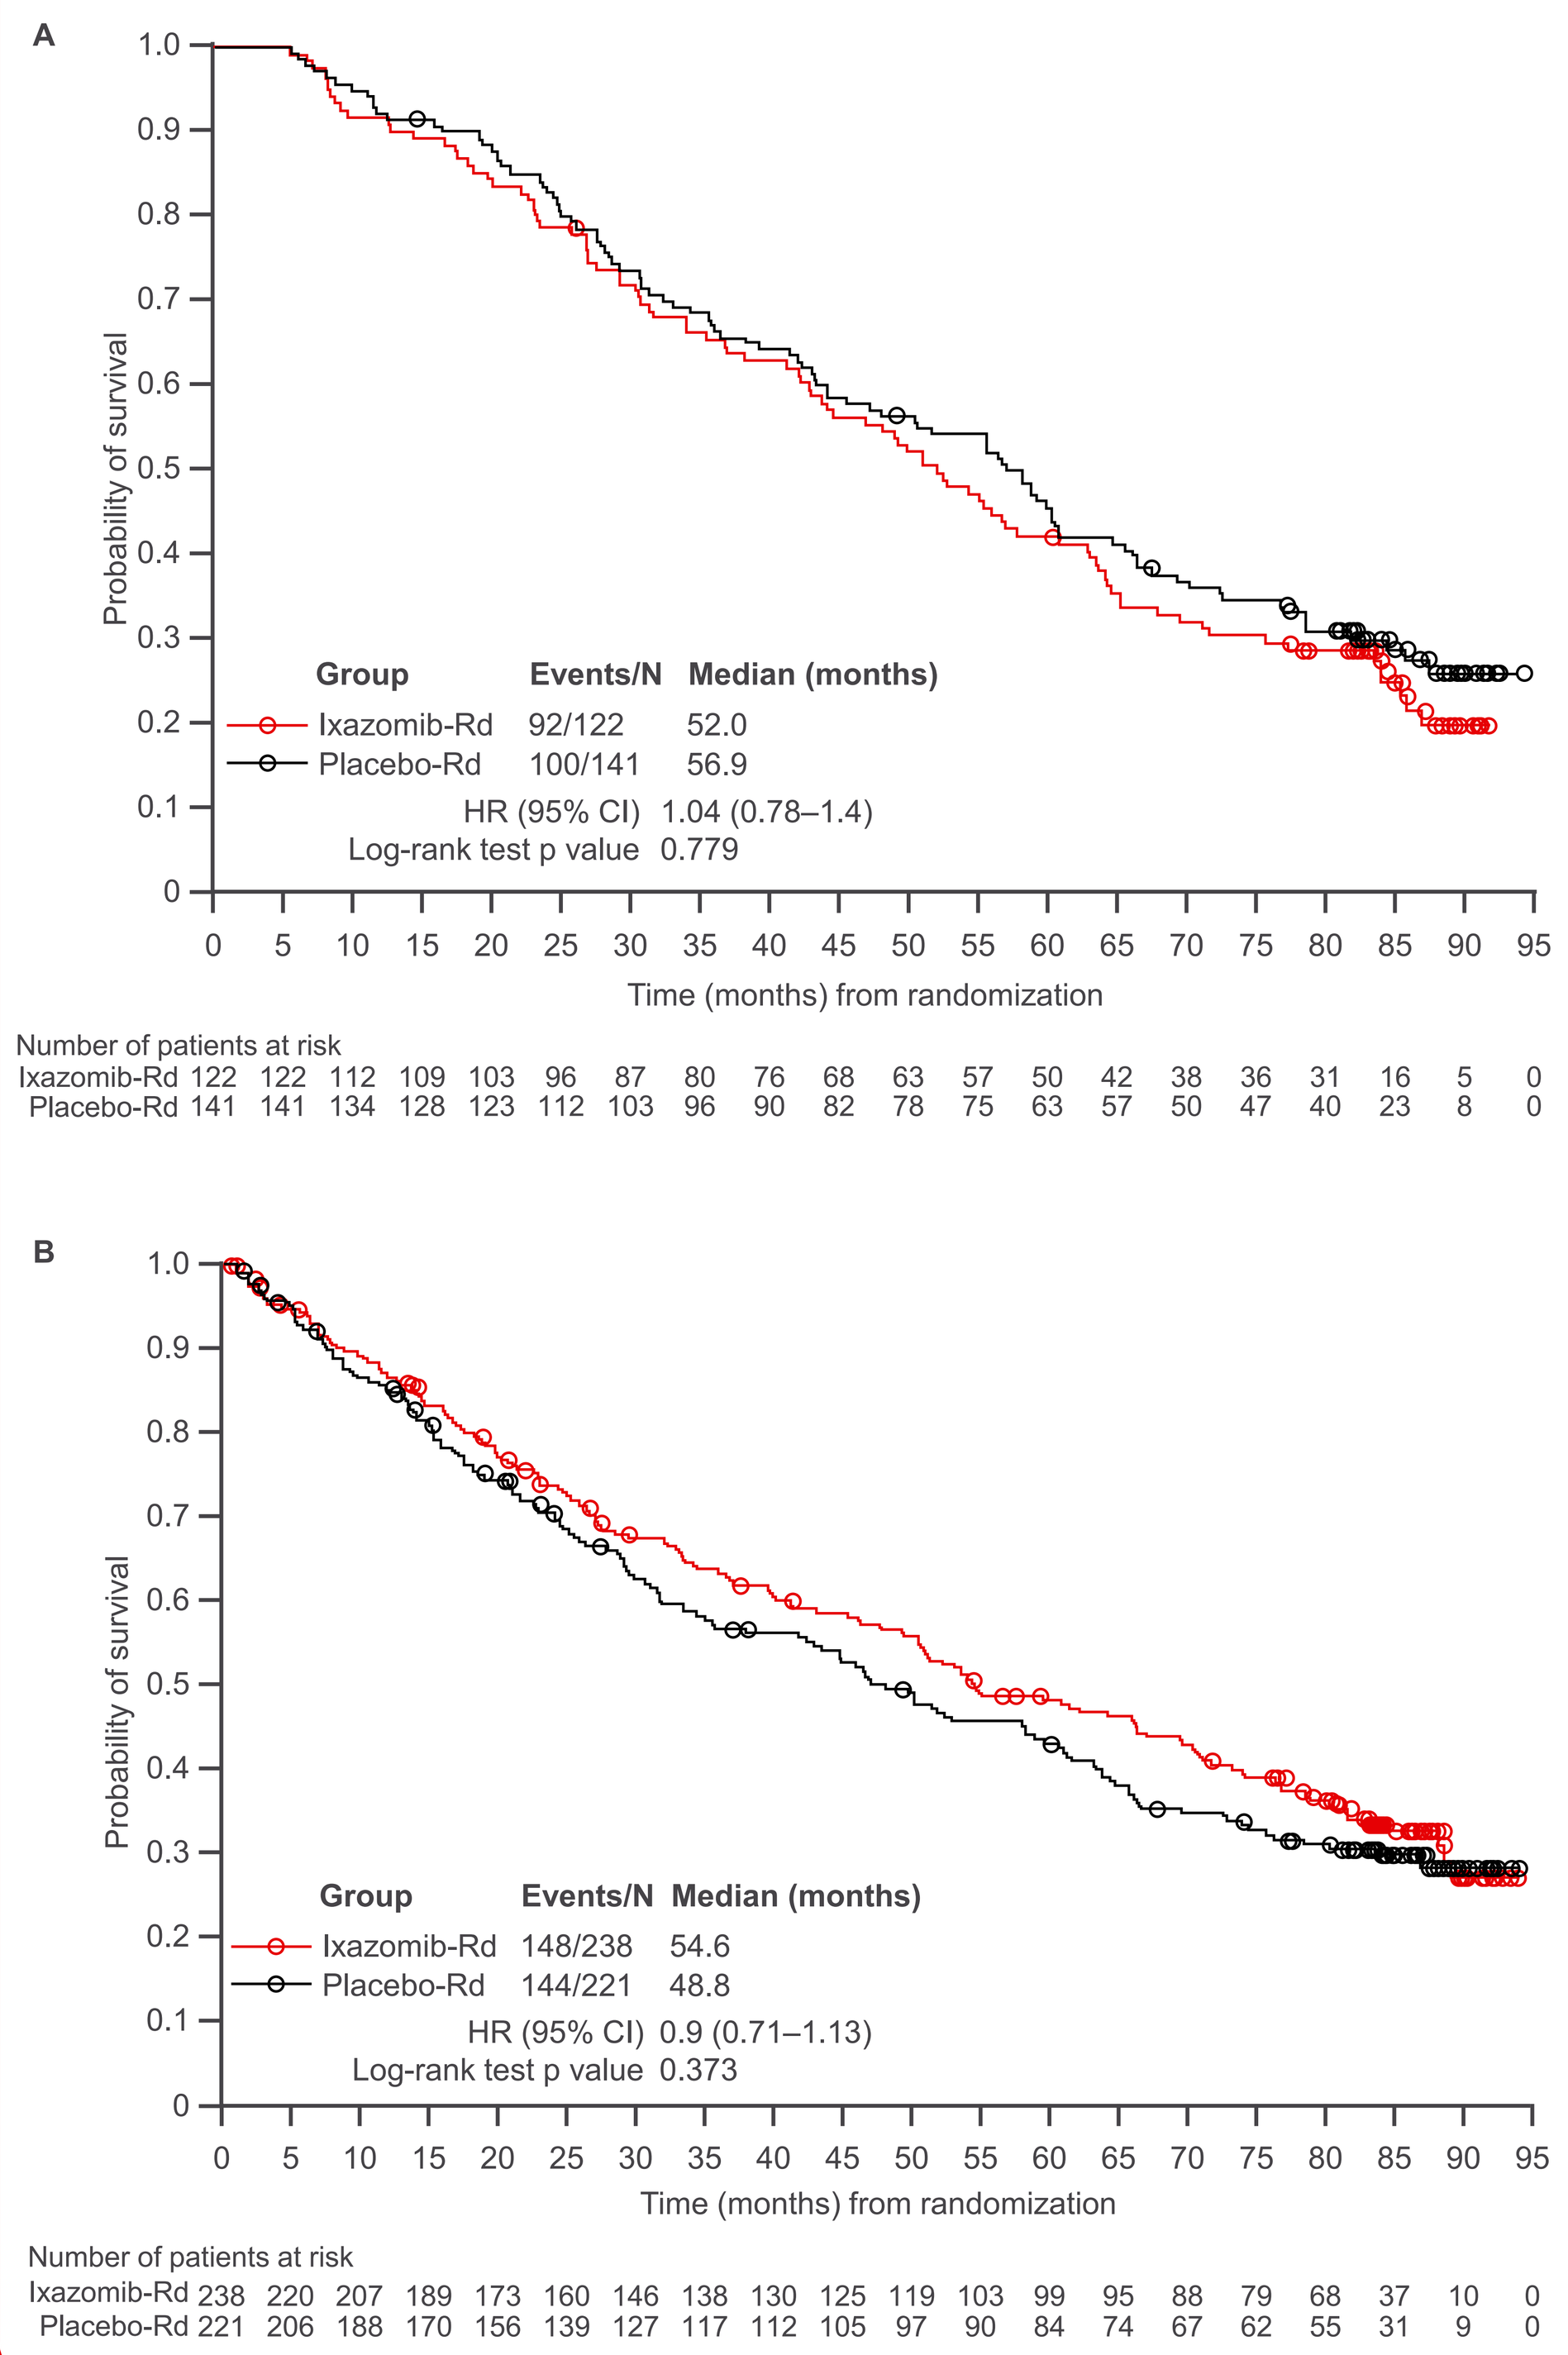

A
Probability of survival
Group | Events/N | Median (months)
Ixazomib-Rd | 92/122 | 52.0
Placebo-Rd | 100/141 | 56.9
HR (95% CI) 1.04 (0.78–1.4)
Log-rank test p value 0.779
Time (months) from randomization
Number of patients at risk
Ixazomib-Rd 122 122 112 109 103 96 87 80 76 68 63 57 50 42 38 36 31 16 5 0
Placebo-Rd 141 141 134 128 123 112 103 96 90 82 78 75 63 57 50 47 40 23 8 0
B
Probability of survival
Group | Events/N | Median (months)
Ixazomib-Rd | 148/238 | 54.6
Placebo-Rd | 144/221 | 48.8
HR (95% CI) 0.9 (0.71–1.13)
Log-rank test p value 0.373
Time (months) from randomization
Number of patients at risk
Ixazomib-Rd 238 220 207 189 173 160 146 138 130 125 119 103 99 95 88 79 68 37 10 0
Placebo-Rd 221 206 188 170 156 139 127 117 112 105 97 90 84 74 67 62 55 31 9 0

- The double-blind nature of TOURMALINE-MM1 confounded OS interpretation.
- Choice of next-line therapy was affected by blinding (**Table 2**).
 - Similar proportions of patients who remained blinded received next-line PI-based and non-PI-based therapy in both arms.
 - In the small number of patients unblinded at PD, PI-based therapy was less likely to be selected following ixazomib-Rd vs placebo-Rd.
- Next-line PI-based therapy was potentially less effective in patients progressing on ixazomib-Rd vs placebo-Rd.

Table 2. Next-line therapy received following ixazomib-Rd and placebo-Rd according to whether patients were unblinded or not

Blinding status at next-line therapy	Ixazomib-Rd (N=257)	Placebo-Rd (N=253)
Patients remaining blinded, n	236	216
PI-based next-line therapy, n (%)	117 (50)	111 (51)
Non-PI-based next-line therapy, n (%)	119 (50)	105 (49)
Patients who were unblinded, n	21	37
PI-based next-line therapy, n (%)	5 (24)	30 (81)
Non-PI-based next-line therapy, n (%)	16 (76)	7 (19)

Treatment exposure, safety, and QoL

- Patients received a median of 18 (range 1–99) and 16 (range 1–100) cycles of ixazomib-Rd and placebo-Rd, respectively.
- No new or additional safety concerns were evident during the 7-year follow-up period, beyond those reported previously.[11,12]
- The rate of new primary malignancies was similar in both arms: ixazomib-Rd 10%, placebo-Rd 12%.
- There were no apparent differences in QoL measures between arms; addition of ixazomib to Rd did not negatively impact patients' QoL.
- Approximately half the patients in each arm reported ≥10-point (MID) increases in the EORTC QLQ-C30 GHS/QoL domain over the course of treatment: ixazomib-Rd 49%, placebo-Rd 52%.

Conclusions

- This final analysis of TOURMALINE-MM1, after a median follow-up of >7 years, reports the longest median OS data seen to date in phase 3 studies of Rd-based combinations in RRMM.[13–16]
 - Median OS approached 4.5 years in both arms.
- Although the slight trend (OS HR 0.939) in favor of ixazomib-Rd in the ITT population was not statistically significant, greater magnitudes of OS benefit (lower HRs) were seen in prespecified subgroups with adverse prognostic factors, including high-risk cytogenetics.
- OS interpretation was confounded by the double-blind nature of the study and by imbalances between arms in subsequent therapies, including extensive use of PIs and daratumumab.
 - Prespecified sensitivity analyses for OS, to adjust for the impact of subsequent therapies, resulted in HRs (0.68, 0.70) supportive of a more substantial OS benefit with ixazomib-Rd.
- The evolving treatment landscape has increased OS for RRMM patients,[1–3] but the availability of multiple active salvage therapies may limit the ability to demonstrate an OS benefit in clinical trials in early-relapse RRMM.
- These findings show that ixazomib-Rd continues to represent an important option for patients with RRMM.
- **These findings are now published in the *Journal of Clinical Oncology*.**
 - **The paper is freely available via the following link: https://ascopubs.org/doi/abs/10.1200/JCO.21.00972**

References

1. Kumar SK, et al. J Natl Compr Canc Netw 2019;17:1154–65.
2. Moreau P, et al. Lancet Oncol 2021;22:e105–18.
3. Branagan A, et al. JCO Oncol Pract 2020;16:5–14.
4. Gandolfi S, et al. Cancer Metastasis Rev 2017;36:561–84.
5. Jimenez-Zepeda VH, et al. Ann Hematol 2017;96:431–9.
6. Richardson PG, et al. Blood Cancer J 2018;8:109.
7. Terpos E, et al. Blood Cancer J 2021;11:40.
8. Gupta N, et al. Clin Pharmacokinet 2019;58:431–49.
9. NINLARO® (ixazomib) capsules, for oral use. US Prescribing Information, revised 02/2020.
10. NINLARO: EPAR – Product Information. Last updated 03/2021.
11. Moreau P, et al. N Engl J Med 2016;374:1621–34.
12. Kumar S, et al. Br J Haematol 2017;178:571–82.
13. Bahlis NJ, et al. Leukemia 2020;34:1875–84.
14. Dimopoulos MA, et al. Leukemia 2009;23:2147–52.
15. Dimopoulos MA, et al. Cancer 2018;124:4032–43.
16. Siegel DS, et al. J Clin Oncol 2018;36:728–34.

Abbreviations

CI, confidence interval; CrCl, creatinine clearance; Dex, dexamethasone; ECOG PS, Eastern Cooperative Oncology Group performance status; EORTC, European Organization for the Research and Treatment of Cancer; GHS, Global Health Status; HR, hazard ratio; IMiD, immunomodulatory drug; IPCW, inverse probability of censoring weighted; ISS, International Staging System; ITT, intention-to-treat; IVRS, interactive voice response system (randomization stratification); MID, minimal important difference; MM, multiple myeloma; MSM, marginal structural models; OS, overall survival; PD, progressive disease; PFS, progression-free survival; PI, proteasome inhibitor; QLQ-C30, Quality of Life Questionnaire Core-30; QoL, quality of life; Rd, lenalidomide-dexamethasone; RRMM, relapsed/refractory MM; TTNT, time to next therapy; TTP, time to progression.

Acknowledgments

The authors would like to thank the patients who took part in this study and their families, the investigators and staff at all clinical sites for TOURMALINE-MM1, and the members of the independent data monitoring committee and independent review committee.

Medical writing support for the development of this poster, under the direction of the authors, was provided by Jenny Wilkinson, PhD, and Steve Hill, PhD, of Ashfield MedComms, an Ashfield Health company, funded by Millennium Pharmaceuticals, Inc., a wholly owned subsidiary of Takeda Pharmaceutical Company Limited, and complied with the Good Publication Practice-3 (GPP3) guidelines (Battisti WP, et al. Ann Intern Med 2015;163:461–4).

Disclosures

This study was funded by Millennium Pharmaceuticals, Inc., Cambridge, MA, USA, a wholly owned subsidiary of Takeda Pharmaceutical Company Limited.

PGR: Membership on an entity's advisory council or committee for Oncopeptides, Celgene/BMS, Takeda, Janssen, Karyopharm, Sanofi, SecuraBio, and GSK; Research funding or grants from Oncopeptides, Celgene/BMS, and Takeda. **SKK**: Consulting/Advisory Board participation (with no personal payments) for AbbVie, Amgen, BMS, Janssen, Roche-Genentech, Takeda, KITE, AstraZeneca, and (with personal payment) Oncopeptides, Beigene, and Antagene; Research funding for clinical trials to the institution from AbbVie, Amgen, BMS, Carsgen, Janssen, KITE, Merck, AstraZeneca, Novartis, Roche-Genentech, Takeda, and Tenebio. **TM:** Membership on an entity's advisory council or committee for AbbVie, BMS, Janssen-Cilag, Novartis, Pfizer, and Takeda. **NG**: No conflicts to declare. **NJB**: Honoraria and membership on an entity's advisory council or committee for Celgene, BMS, Amgen, Janssen, Sanofi, AbbVie, Karyopharm, Takeda, Genentech, Pfizer, and GSK; Research funding or grants from Celgene. **MH**: No conflicts to declare. **LP**: No conflicts to declare. **IS**: Honoraria from Celgene/BMS, Amgen, Takeda, Gilead, Beigene, and Janssen. **PGan**: No conflicts to declare. **BWB**: No conflicts to declare. **SRJ**: Honoraria from AbbVie. **AMS**: Honoraria from Celgene. **PGim**: No conflicts to declare. **LG**: Membership on an entity's advisory council or committee for Amgen, BMS, Takeda, Janssen, and Sanofi. **CT**: Honoraria and membership on an entity's advisory council or committee for Takeda. **FKB**: No conflicts to declare. **JPL**: No conflicts to declare. **MC**: Honoraria from Janssen, BMS, Amgen, Sanofi, GSK, and Takeda. **MD**: Employment with and consulting fees from Millennium Pharmaceuticals, Inc., Cambridge, MA, USA, a wholly owned subsidiary of Takeda Pharmaceutical Company Limited. **RL and DB**: Employment with Millennium Pharmaceuticals, Inc., Cambridge, MA, USA, a wholly owned subsidiary of Takeda Pharmaceutical Company Limited. **PM**: Honoraria from Janssen, Celgene, AbbVie, Sanofi, Takeda, and Amgen.

Electronic poster presentation at the virtual 2021 Congress of the European Hematology Association (EHA), June 9–17, 2021; for questions or comments please contact Dr Paul G. Richardson: Paul_Richardson@dfci.harvard.edu