Fruquintinib plus best supportive care for patients with metastatic colorectal cancer: characterization of patients who had an overall survival of ≥10 months in the FRESCO-2 study

142

Stefan Kasper,[1] Andrea Sartore-Bianchi,[2] Violaine Randrian,[3] Chiara Cremolini,[4] Dirk Arnold,[5] **Arvind Dasari**,[6] Cathy Eng,[7] Sara Lonardi,[8] Elena Elez,[9] Takayuki Yoshino,[10] Alberto F. Sobrero,[11] James C. Yao,[6] Howard S. Hochster,[12] Eric Van Cutsem,[13] David Tougeron,[3] Ziji Yu,[14] Qi Dong,[14] William R. Schelman,[15] François Ghiringhelli[16]

[1]University Hospital Essen, Essen, Germany; [2]Università degli Studi di Milano, Milan, Italy; [3]Poitiers University Hospital, Poitiers, France; [4]University of Pisa, Pisa, Italy; [5]Asklepios Tumorzentrum Hamburg, Hamburg, Germany; [6]University of Texas MD Anderson Cancer Center, Houston, TX, USA; [7]Vanderbilt Ingram Cancer Center, Nashville, TN, USA; [8]Veneto Institute of Oncology IOV-IRCCS, Padua, Italy; [9]Vall d'Hebron Institute of Oncology, Barcelona, Spain; [10]National Cancer Center Hospital East, Chiba, Japan; [11]Azienda Ospedaliera San Martino, Genoa, Italy; [12]Rutgers Cancer Institute of New Jersey, New Brunswick, NJ, USA; [13]University Hospitals Gasthuisberg / Leuven & KU Leuven, Leuven, Belgium; [14]Takeda Development Center Americas, Inc. (TDCA), Lexington, MA, USA; [15]HUTCHMED, Florham Park, NJ, USA; [16]Centre Georges-François Leclerc, Dijon, France

Background

- Up to 70% of patients with colorectal cancer (CRC) will experience metastatic disease (mCRC), either at diagnosis or over the course of their treatment[1,2]
- The prognosis for patients with mCRC is poor, with a 5-year relative survival rate of approximately 14%[3]
- Fruquintinib is a highly selective, oral tyrosine kinase inhibitor of all 3 vascular endothelial growth factor receptors (VEGFR -1, -2, and -3) that was approved by the United States Food and Drug Administration in November 2023[4], and in Europe[5,6] and Japan[7] in 2024 for previously treated mCRC, regardless of biomarker status
- The FRESCO-2 study met its primary endpoint demonstrating significantly improved overall survival (OS) with fruquintinib + best supportive care (BSC) versus placebo + BSC among patients with heavily pretreated mCRC[8]
- The median OS of patients who received fruquintinib in FRESCO-2 was 7.4 months, but a proportion of patients were able to gain longer survival benefits[8]
- We conducted a subgroup analysis of these patients to assess baseline characteristics and safety data using a cut-off of ≥10 months OS with fruquintinib treatment

Methods

- FRESCO-2 was a global phase 3 study conducted across 14 countries[8]
- Eligible patients had received all standard treatments including fluoropyrimidine, oxaliplatin, and irinotecan chemotherapy; anti-vascular endothelial growth factor therapy; anti-epidermal growth factor receptor therapy (if *RAS* wild type); and had disease progression on, or had been intolerant to trifluridine-tipiracil (TAS-102) and/or regorafenib[8]
- Hazard ratios (HRs) and 95% confidence intervals (CIs) between the two treatment arms were calculated from a stratified Cox proportional hazard model with the treatment group as the only covariate in the model[8]
- Landmark survival (OS and progression-free survival [PFS]) was assessed at 6 and 9 months
- Comparison of patient characteristics between the OS ≥10 months subgroup and the intent-to-treat (ITT) population was descriptive only
- The FRESCO-2 ITT population included all patients who were randomly assigned to a treatment group; the safety population included all patients who received at least one dose of fruquintinib or placebo[8]

Question

What are the baseline characteristics of patients treated with fruquintinib in FRESCO-2 who had an OS ≥10 months, compared with the ITT population?

Study design

FRESCO-2: (NCT04322539)
Previously treated patients with mCRC from North America, Europe, Japan, and Australia (**N=691**)

R 2:1

Fruquintinib (n=461)
5 mg PO, QD + BSC, 28-day cycle
(3 weeks on, 1 week off)

Placebo (n=230)
5 mg PO, QD + BSC, 28-day cycle
(3 weeks on, 1 week off)

Treatment until progressive disease or unacceptable toxicity

Primary endpoint: OS
Secondary endpoints: PFS, ORR, DCR, and safety

Subgroup analysis: **Baseline characteristics and safety data of patients treated with fruquintinib who had an OS ≥10 months compared with the ITT population**

Results

FRESCO-2 OS (ITT population)[8]

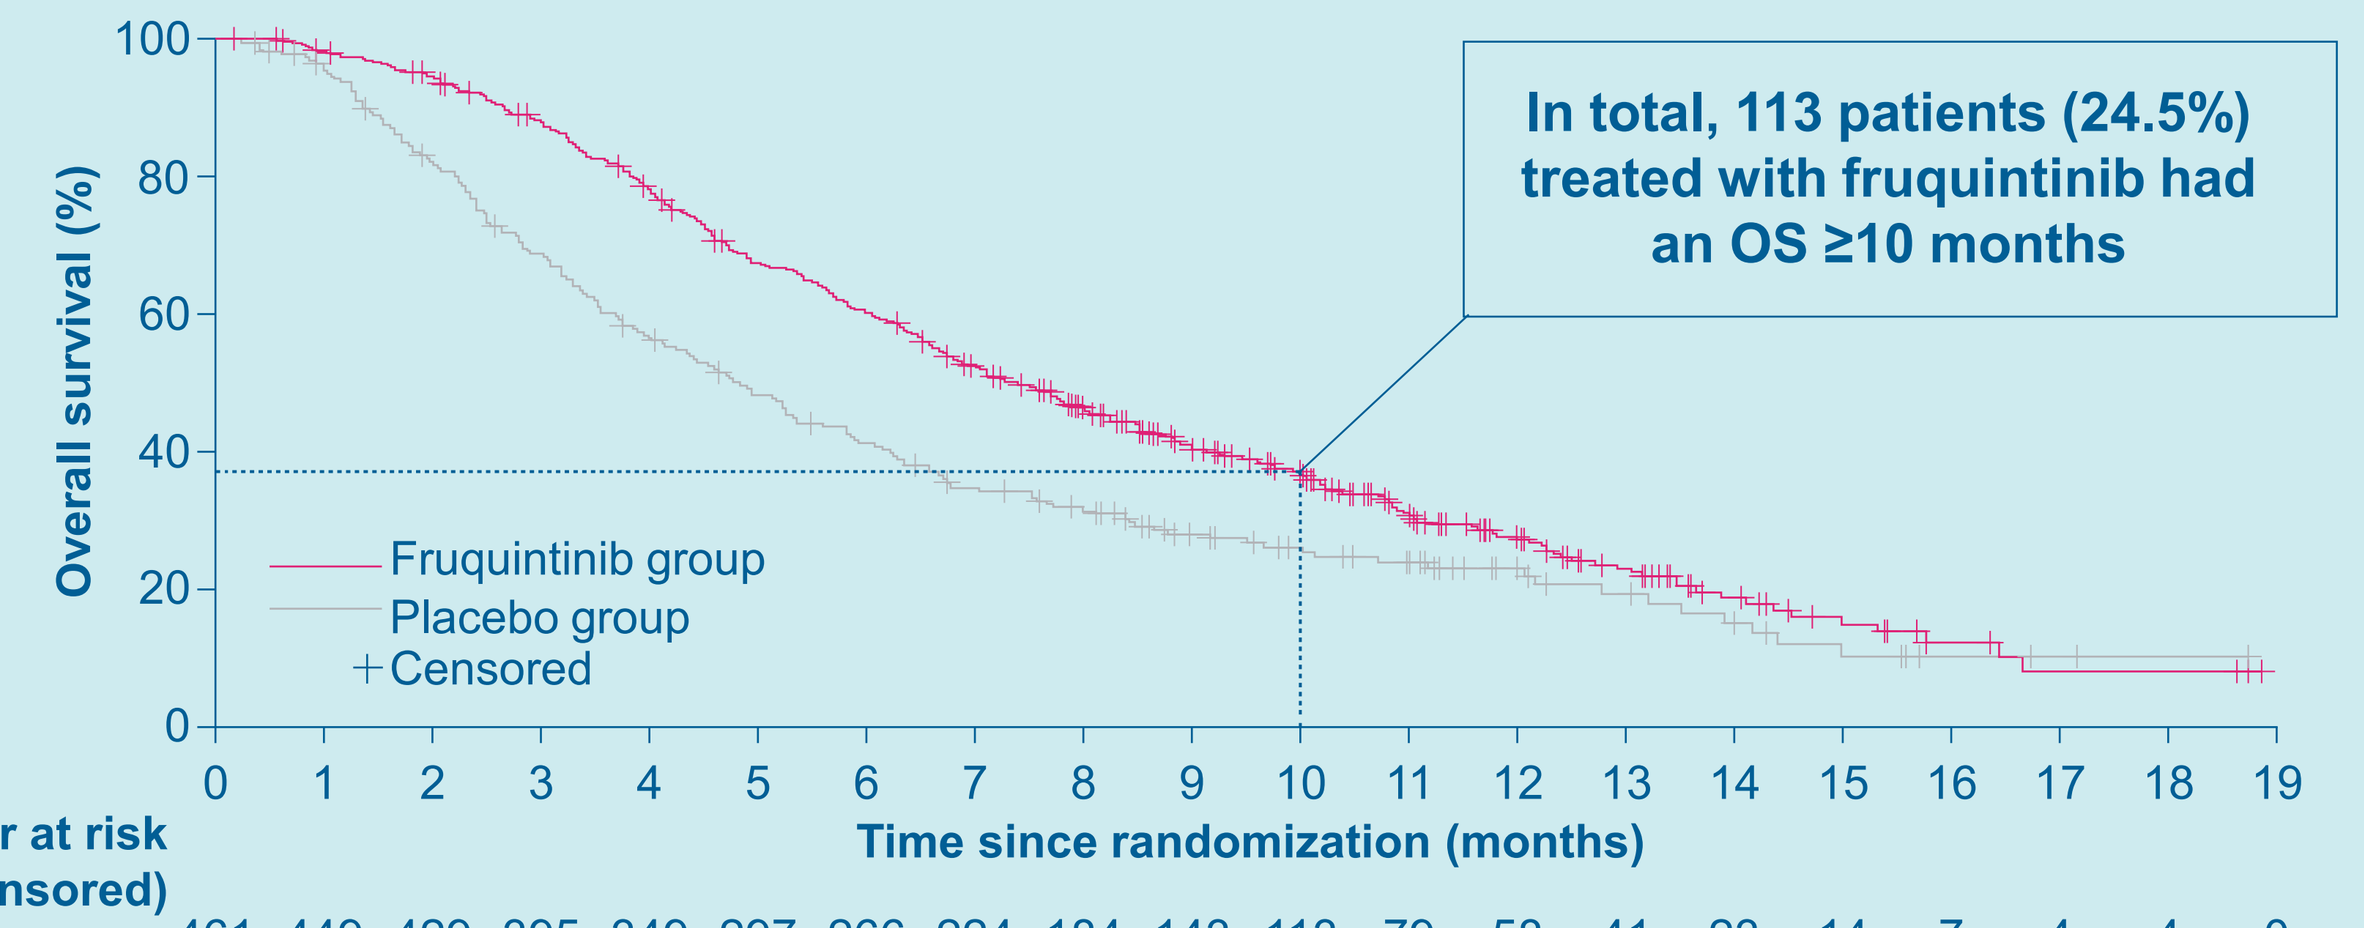
Overall survival (%)
100
80
60
40
20
0
0 1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17 18 19
Time since randomization (months)
Fruquintinib group
Placebo group
Censored
In total, 113 patients (24.5%) treated with fruquintinib had an OS ≥10 months

Number at risk (number censored)

	0	1	2	3	4	5	6	7	8	9	10	11	12	13	14	15	16	17	18	19
Fruquintinib group	461 (0)	449 (4)	429 (3)	395 (5)	349 (2)	297 (4)	266 (0)	224 (7)	184 (15)	143 (21)	113 (17)	79 (18)	58 (13)	41 (8)	23 (12)	14 (5)	7 (5)	4 (1)	4 (0)	0 (4)
Placebo group	230 (0)	216 (4)	184 (2)	153 (1)	125 (1)	105 (2)	89 (1)	73 (2)	63 (3)	45 (12)	37 (5)	31 (3)	20 (10)	15 (2)	10 (2)	6 (1)	3 (3)	2 (1)	1 (1)	0 (1)

Baseline characteristics of patients from FRESCO-2 treated with fruquintinib with an OS ≥10 months and in the ITT

	Fruquintinib	
Patient characteristic	**Patients with OS ≥10 months (n=113)**	**ITT population (n=461)**
No liver metastases at baseline, n (%)	47 (41.6)	122 (26.5)
ECOG PS 0 at baseline, n (%)	61 (54.0)	196 (42.5)

Key conclusions

A higher proportion of patients with ≥10 months OS with fruquintinib had an absence of liver metastases and an ECOG PS of 0 at baseline than in the ITT population

Results

Landmark survival

- In the FRESCO-2 ITT population, the primary endpoint of OS was significantly improved with fruquintinib + BSC versus placebo + BSC (HR: 0.66; 95% CI: 0.55–0.80; p<0.001)
 - OS rates (95% CIs) at 6 months were 60.4% (55.9–64.9%) in the fruquintinib group versus 41.5% (35.0–48.0%) in the placebo group
 - OS rates at 9 months were 41.1% (36.4–45.8%) in the fruquintinib group versus 28.2% (22.1–34.3%) in the placebo group
- The key secondary endpoint of PFS was also significantly improved with fruquintinib + BSC versus placebo + BSC (HR: 0.32; 95% CI: 0.27–0.39; p<0.001)
 - PFS rates (95% CIs) for fruquintinib versus placebo were 23.8% (19.7–28.0%) versus 1.1% (0–2.6%) at 6 months and 11.3% (8.1–14.6%) versus 0.5% (0–1.6%) at 9 months

Patients with an OS ≥10 months

- Of 461 patients who received fruquintinib in FRESCO-2, 113 (24.5%) had an OS ≥10 months (range 10.0–18.9)
- Median (range) duration of fruquintinib treatment was 6.3 (0.7–19.1) months for patients with an OS ≥10 months versus 3.1 (0.3–19.1) months for the ITT population
- Patients with an OS ≥10 months received a median (range) of 7 (1–20) treatment cycles versus 3 (1–20) treatment cycles for patients in the ITT population
- Baseline characteristics were generally balanced between patients with an OS ≥10 months and the ITT population (**Table 1**)
- A higher proportion (>10% difference) of patients with an OS ≥10 months had no liver metastases and an Eastern Cooperative Oncology Group performance status (ECOG PS) of 0 at baseline compared with the ITT population
- Median time since first diagnosis of CRC and median duration of mCRC was around five months longer for patients in the OS ≥10 months subgroup compared with the ITT population (**Table 1**)

Table 1. Baseline characteristics of patients treated with fruquintinib + BSC in FRESCO-2

	Patients with an OS ≥10 months (n=113)	ITT population (n=461)
Mean (SD) age, years	62.9 (10.4)	62.2 (10.4)
Female, n (%)	56 (49.6)	216 (46.9)
Race, n (%)		
White	91 (80.5)	367 (79.6)
Asian	7 (6.2)	43 (9.3)
Black or African American	3 (2.7)	13 (2.8)
Other*	12 (10.6)	5 (1.1)
ECOG PS, n (%)		
0	61 (54.0)	196 (42.5)
1	52 (46.0)	265 (57.5)
Median (range) time since first diagnosis of CRC, months	52.0 (10.1–242.4)	47.2 (6.0–242.4)
Primary tumor location at first diagnosis, n (%)		
Colon	58 (51.3)	279 (60.5)
Rectum	40 (35.4)	143 (31.0)
Both	15 (13.3)	39 (8.5)
Primary tumor site at first diagnosis, n (%)		
Colon left	48 (42.5)	192 (41.6)
Colon right	18 (15.9)	97 (21.0)
Colon left and right	0	4 (0.9)
Liver metastases, yes / no, n (%)	66 (58.4) / 47 (41.6)	339 (73.5) / 122 (26.5)
Median (range) duration of mCRC, months	42.7 (10.1–121.0)	37.9 (6.0–192.8)
≤18 months, n (%)	6 (5.3)	37 (8.0)
>18 months, n (%)	107 (94.7)	424 (92.0)
Prior lines of therapy for mCRC, n (%)		
≤3	30 (26.5)	125 (27.1)
>3	83 (73.5)	336 (72.9)
Prior TAS-102, n (%)	53 (46.9)	240 (52.1)
Prior regorafenib, n (%)	14 (12.4)	40 (8.7)
Prior TAS-102 and regorafenib, n (%)	46 (40.7)	181 (39.3)

*Includes American Indian or Alaska Native, Native Hawaiian or Other Pacific Islander, Other as recorded on the Demographics electronic case report form, and patients with multiple races selected. SD, standard deviation.

Safety

- The proportion of patients experiencing a grade ≥3 treatment-emergent adverse event (TEAE) was similar between patients receiving fruquintinib with ≥10 months OS and patients in the overall FRESCO-2 safety population (61.9% vs 62.7%; **Table 2**)
- A higher proportion of patients in the OS ≥10 months subgroup had TEAEs leading to dose modification compared with the FRESCO-2 safety population (95.6% vs 70.8%)
- However, a lower proportion of patients in the OS ≥10 months subgroup experienced TEAEs leading to treatment discontinuation versus the overall FRESCO-2 safety population (14.2% vs 20.4%)
- A higher proportion of patients with an OS ≥10 months experienced treatment-emergent adverse events of special interest (AESIs) compared with the overall FRESCO-2 safety population (94.7% vs 80.7%; **Table 2**)

Table 2. Overview of TEAEs for patients treated with fruquintinib + BSC in FRESCO-2

n (%)	Patients with an OS ≥10 months (n=113)	FRESCO-2 safety population* (n=456)
Any TEAE	112 (99.1)	451 (98.9)
Grade ≥3	70 (61.9)	286 (62.7)
Leading to dose reduction	45 (39.8)	110 (24.1)
Leading to dose interruption	63 (55.8)	213 (46.7)
Leading to treatment discontinuation	16 (14.2)	93 (20.4)
Leading to death	1 (0.9)	48 (10.5)
Treatment-related TEAEs	108 (95.6)	395 (86.6)
Grade ≥3	49 (43.4)	164 (36.0)
Leading to dose reduction	40 (35.4)	93 (20.4)
Leading to dose interruption	43 (38.1)	134 (29.4)
Leading to treatment discontinuation	13 (11.5)	45 (9.9)
Serious TEAEs	32 (28.3)	171 (37.5)
Grade ≥3	32 (28.3)	162 (35.5)
Treatment-emergent AESI	107 (94.7)	368 (80.7)

*Five patients randomized to the fruquintinib arm did not receive fruquintinib treatment.

Conclusions

- This exploratory analysis showed that baseline characteristics were generally balanced between patients with an OS ≥10 months and the ITT population; however, a higher proportion of patients receiving fruquintinib with an OS ≥10 months had an absence of liver metastases and ECOG PS 0 at baseline compared with the ITT population
- As may be expected due to the longer overall duration of treatment, patients receiving fruquintinib with an OS ≥10 months required more dose modifications compared with the overall FRESCO-2 safety population; however, these patients were able to continue treatment for longer with fewer discontinuations due to TEAEs
- Fruquintinib is a novel treatment option that demonstrates significantly improved survival compared with placebo that is clinically meaningful, as evidenced by some patients gaining ≥10 months OS benefit

References

1. Vatandoust S, et al. *World J Gastroenterol.* 2015;21:11767–76
2. Van Cutsem E, et al. *Ann Oncol.* 2009;20(Suppl 4):iv61–iv63
3. International Agency for Research on Cancer. Age-standardized 5-year net survival, both sexes, age (15–99), colon cancer, 2010–2014. Accessed Nov 14, 2024. https://gco.iarc.fr/survival/survmark/visualizations/viz7/?mode=%22bar%22&sub_mode=%22all%22&groupby=%22country%22&period=%225%22&cancer=%22COLO%22&country=%22Australia%22&gender=%220%22&stage=%22SEER%22&age_group=%2215-99%22&show_ci=true
4. FRUZAQLA™ (fruquintinib) Prescribing Information. Published Nov 2023. Takeda Pharmaceuticals America, Inc., Lexington, MA, USA
5. FRUZAQLA™ (fruquintinib) Summary of Product Characteristics. EMEA/H/C/005979. Published Jul 2024. Takeda Pharmaceuticals International AG Ireland, Dublin, Ireland
6. MHRA Public Assessment Report. FRUZAQLA (fruquintinib). Published Oct 2024. Takeda UK Ltd
7. Takeda. 2024. Available online: https://www.takeda.com/newsroom/newsreleases/2024/takeda-receives-approval-for-fruzaqla-in-japan-for-the-treatment-of-unresectable-advanced-or-recurrent-colorectal-cancer/ (accessed Nov 14, 2024)
8. Dasari A, et al. *Lancet.* 2023;402:41–53

Acknowledgments

This study was funded by HUTCHMED. The authors would like to thank all patients and their families, as well as all investigators for their valuable contributions to this study. Medical writing support for the development of this poster, under the direction of the authors, was provided by Rebecca Cunningham, PhD, of Ashfield MedComms, an Inizio Company, funded by Takeda, and complied with the Good Publication Practice (GPP) guidelines (DeTora LM, et al. *Ann Intern Med.* 2022;175:1298–304).

Disclosures

AD: consulting or advisory role for Personalis, Hutchison MediPharma, Illumina/GRAIL, Takeda, Sirtris Pharmaceuticals; travel, accommodations, expenses from Hutchison MediPharma, Takeda; research funding (to institution) from Eisai, Hutcison MediPharma, Merck, Guardant Health, XENCOR, Natera.

All remaining author disclosures can be accessed by scanning the QR code.

To view an electronic version of this poster, scan this Quick Response (QR) code or visit: https://tiny.one/6iW18066Sj.

Copies of this poster obtained through QR code are for personal use only and may not be reproduced without permission from the congress and the author of this poster.

Presented at ASCO Gastrointestinal Cancers Symposium, January 23–25, 2025, San Francisco, CA, USA. For questions or comments, please contact Dr Arvind Dasari: adasari@mdanderson.org.