Real-World Outcomes of 2L ALK TKIs Following 1L Brigatinib for Patients With *ALK*+ NSCLC From the ALTA-1L Trial

Myung-Ju Ahn[1], Angelo Delmonte[2], Sharmistha Ghosh[3], Maximilian Hochmair[4,5], Tsung-Ying Yang[6], James Chih-Hsin Yang[7], Ji-Youn Han[8], Karin Holmskov Hansen[9], Yanyu Wu[10], Yin Wan[10], Huamao Mark Lin[10], Julian Kretz[11*], Bradley Hupf[10], Ahmet Melih Kurec[12], Eric N. Churchill[13], Robert J. Fram[10], Citadel Jungco Cabasag[14], Vishal Goriya[15], Yuzhe Zhao[16], Eric Gemmen[17], Maria Rosario García Campelo[18]

[1]Division of Hematology, Oncology Department of Medicine, Samsung Medical Center Sungkyunkwan University School of Medicine, Seoul, Republic of Korea; [2]IRCCS Istituto Romagnolo per lo Studio dei Tumori - "Dino Amadori" (IRST), Meldola, Forli - Cesena, Italy; [3]Guy's and St Thomas' NHS Foundation Trust, London, England, United Kingdom; [4]Department of Respiratory and Critical Care Medicine, Klinik Floridsdorf, Vienna, Austria; [5]Karl Landsteiner Institute of Lung Research and Pulmonary Oncology, Vienna, Austria; [6]Division of Chest Medicine, Department of Internal Medicine, Taichung Veterans General Hospital, Taichung, Taiwan; [7]National Taiwan University Hospital and National Taiwan University Cancer Center, Taipei, Taiwan; [8]National Cancer Center, Goyang, Republic of Korea; [9]Odense University Hospital, Odense C., Denmark; [10]Takeda Development Center Americas, Inc., Lexington, MA, USA; [11]Takeda Pharmaceuticals International AG, Glattpark-Opfikon (Zurich), Switzerland; [12]Takeda Pharmaceutical International Singapore Emerging Markets, Singapore; [13]Takeda Pharmaceuticals U.S.A. Inc., Lexington, MA, USA; [14]IQVIA RDS France, Courbevoie, France; [15]IQVIA RDS (India) Pvt Ltd, Thane, Maharashtra, India; [16]IQVIA RDS (Shanghai) Co., Ltd., Shanghai, China; [17]IQVIA Falls Church, VA, USA; [18]Medical Oncology Department, University Hospital A Coruña, and Biomedical Research Institute (INIBIC, A Coruña), A Coruña, Spain

*at the time of study

1144

Background

- Anaplastic lymphoma kinase-positive (*ALK*+) non-small cell lung cancer (NSCLC) accounts for approximately 5% of all NSCLC cases.[1,2]
- Targeted therapies for *ALK*+ NSCLC include the first-generation ALK tyrosine kinase inhibitor (TKI) crizotinib, and next-generation ALK TKIs including brigatinib.[3]
- In the phase III ALTA-1L trial, brigatinib exhibited superior clinical efficacy versus crizotinib for the first-line (1L) treatment of patients with *ALK*+ NSCLC.[4]
- A retrospective, non-interventional, multinational study previously reported real-world outcomes post-1L brigatinib, showing that patients treated with 1L brigatinib, followed by subsequent ALK TKIs, experienced prolonged clinical benefits.[5]
- The present analysis assessed additional subgroups, including those stratified by response to 1L brigatinib following discontinuation of 1L brigatinib in the ALTA-1L trial.

Objective

- To investigate the real-world treatment patterns and outcomes of subsequent treatments following discontinuation of 1L brigatinib in the ALTA-1L trial.

Methods

Study design and patients

- This was a retrospective, longitudinal, non-interventional, multinational, multisite-based chart review study.
- Patients were previously enrolled in the brigatinib arm of the ALTA-1L trial and discontinued 1L brigatinib during or after completing the trial (**Figure 1**).
- Patients were followed from last dose of brigatinib until end of follow-up or death.
- Data were extracted from electronic case report forms between November 4, 2022 and October 18, 2023 (database lock).

Study outcomes and statistical analyses

- Patient characteristics and treatment patterns were summarized using descriptive statistics.
- Study outcomes are defined in **Table 1**.
- Time-to-event analyses (real-world time to treatment discontinuation [rwTTD], real-world progression-free survival [rwPFS], rwPFS2, and overall survival [OS]) were performed using the Kaplan-Meier (KM) method.
- Analyses were performed using Statistical Analysis System version 9.4.
- Subgroups of interest were:
 - Patients who received any second-line (2L) ALK TKI;
 - Patients who received 2L lorlatinib or 2L alectinib;
 - Among patients who received any 2L ALK TKIs, patients who were responders (best of complete or partial response by blinded independent review committee) or non-responders to 1L brigatinib.

Table 1. Definitions of study outcomes

Outcome	Definition
rwTTD	Time from initiation to discontinuation of 2L treatment for any reason
rwPFS	Time elapsed from initiation of 2L treatment to disease progression or death
rwPFS2	Time from randomization in ALTA-1L trial to first documented disease progression on 2L treatment or death due to any cause among those patients who initiated subsequent systemic anticancer treatments
OS	Time from the date of randomization in the ALTA-1L trial until date of death
rwORR	Proportion of patients on a treatment line who achieved rwCR or rwPR assessment determination as their best response per treatment line
rwDCR	Proportion of patients who had a rwSD, rwPR, rwCR assessment during the course of a line of therapy, among all patients in that cohort

2L, second-line; OS, overall survival; rwCR, real-world complete response; rwDCR, real-world disease control rate; rwORR, real-world overall response rate; rwPFS, real-world progression-free survival; rwPR, real-world partial response; rwSD, real-world stable disease; rwTTD, real-world time to treatment discontinuation.

Results

Patients

- As of October 18, 2023, 48 patients across 20 sites in 10 countries (Hong Kong, South Korea, Taiwan, United Kingdom, Spain, Italy, Netherlands, France, Denmark, and Austria) were included in the enrolled set (ENR).

Treatment patterns

- A total of 40 (83.3%) of the 48 patients in the ENR received subsequent systemic anticancer therapies.
- Of these, 30 (75.0%) patients received a 2L ALK TKI and were included in this analysis (**Table 2**); median follow-up was 17.0 months.
 - Median age of patients who received a 2L ALK TKI was 57.5 years, 46.7% were White, 60.0% were female, and 50.0% were Asian.
 - 16 (53.3%) patients received 2L lorlatinib and 8 (26.7%) received 2L alectinib.
 - There were 23 (76.7%) responders and 7 (23.3%) non-responders to 1L brigatinib, consistent with ALTA-1L results.

Tumor response

- Real-world overall response rate (rwORR) was 33.3% for 2L ALK TKIs, 30.8% for 2L lorlatinib, and 57.1% for 2L alectinib.
- Real-world disease control rate (rwDCR) was 70.8% for 2L ALK TKIs, 76.9% for 2L lorlatinib, and 85.7% for 2L alectinib.
- For 1L responders receiving 2L ALK TKIs, rwORR was 33.3% and rwDCR was 66.7%; for 1L non-responders, rwORR was 33.3% and rwDCR was 83.3%.

Question

What are the real-world treatment patterns and outcomes of subsequent systemic anticancer therapies post-1L brigatinib in patients with *ALK*+ NSCLC, including those stratified by response to 1L brigatinib following discontinuation of 1L brigatinib in the ALTA-1L trial?

Study design

Figure 1. Study design flow chart

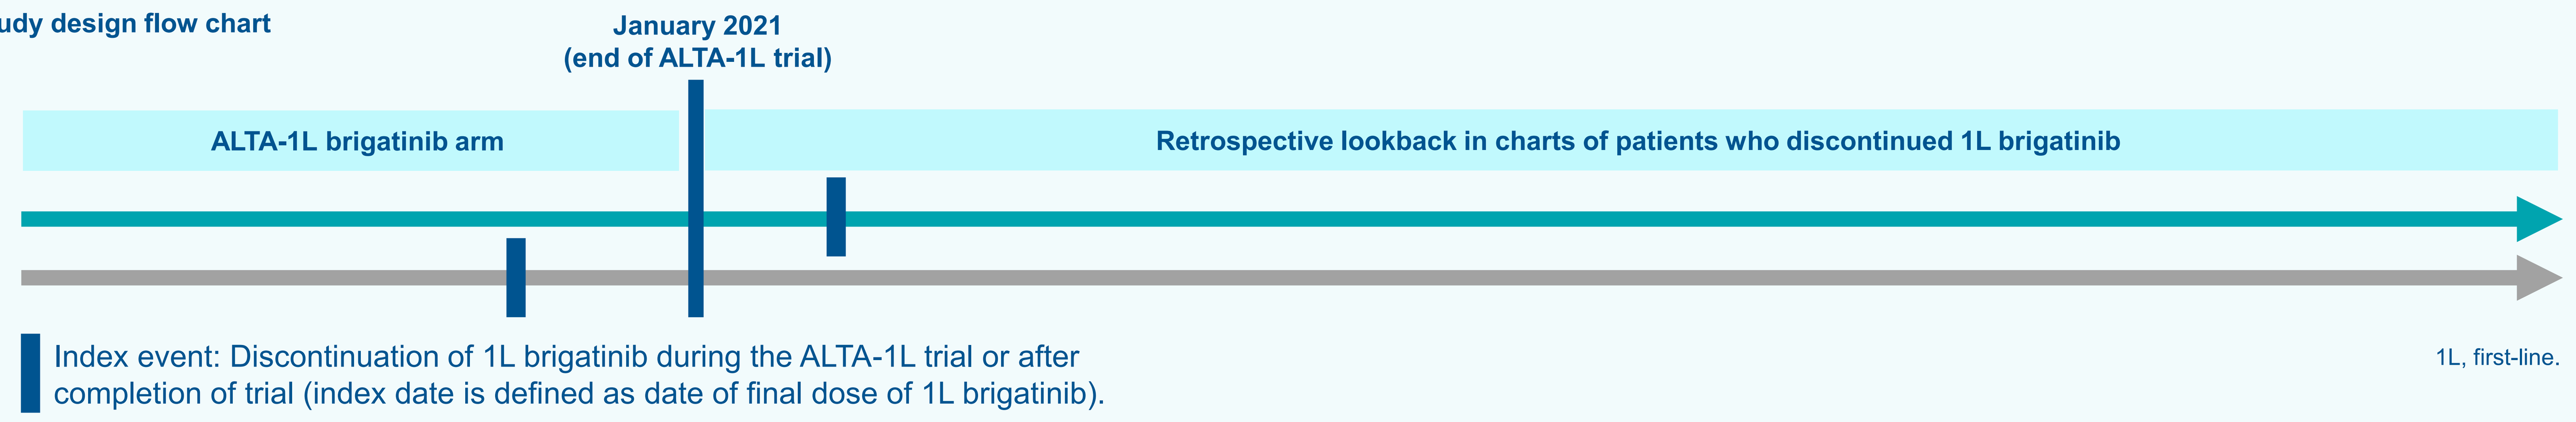
January 2021
(end of ALTA-1L trial)
ALTA-1L brigatinib arm
Retrospective lookback in charts of patients who discontinued 1L brigatinib
Index event: Discontinuation of 1L brigatinib during the ALTA-1L trial or after completion of trial (index date is defined as date of final dose of 1L brigatinib).

1L, first-line.

Results

Figure 2. rwTTD for 2L ALK TKIs, 2L alectinib, and 2L lorlatinib

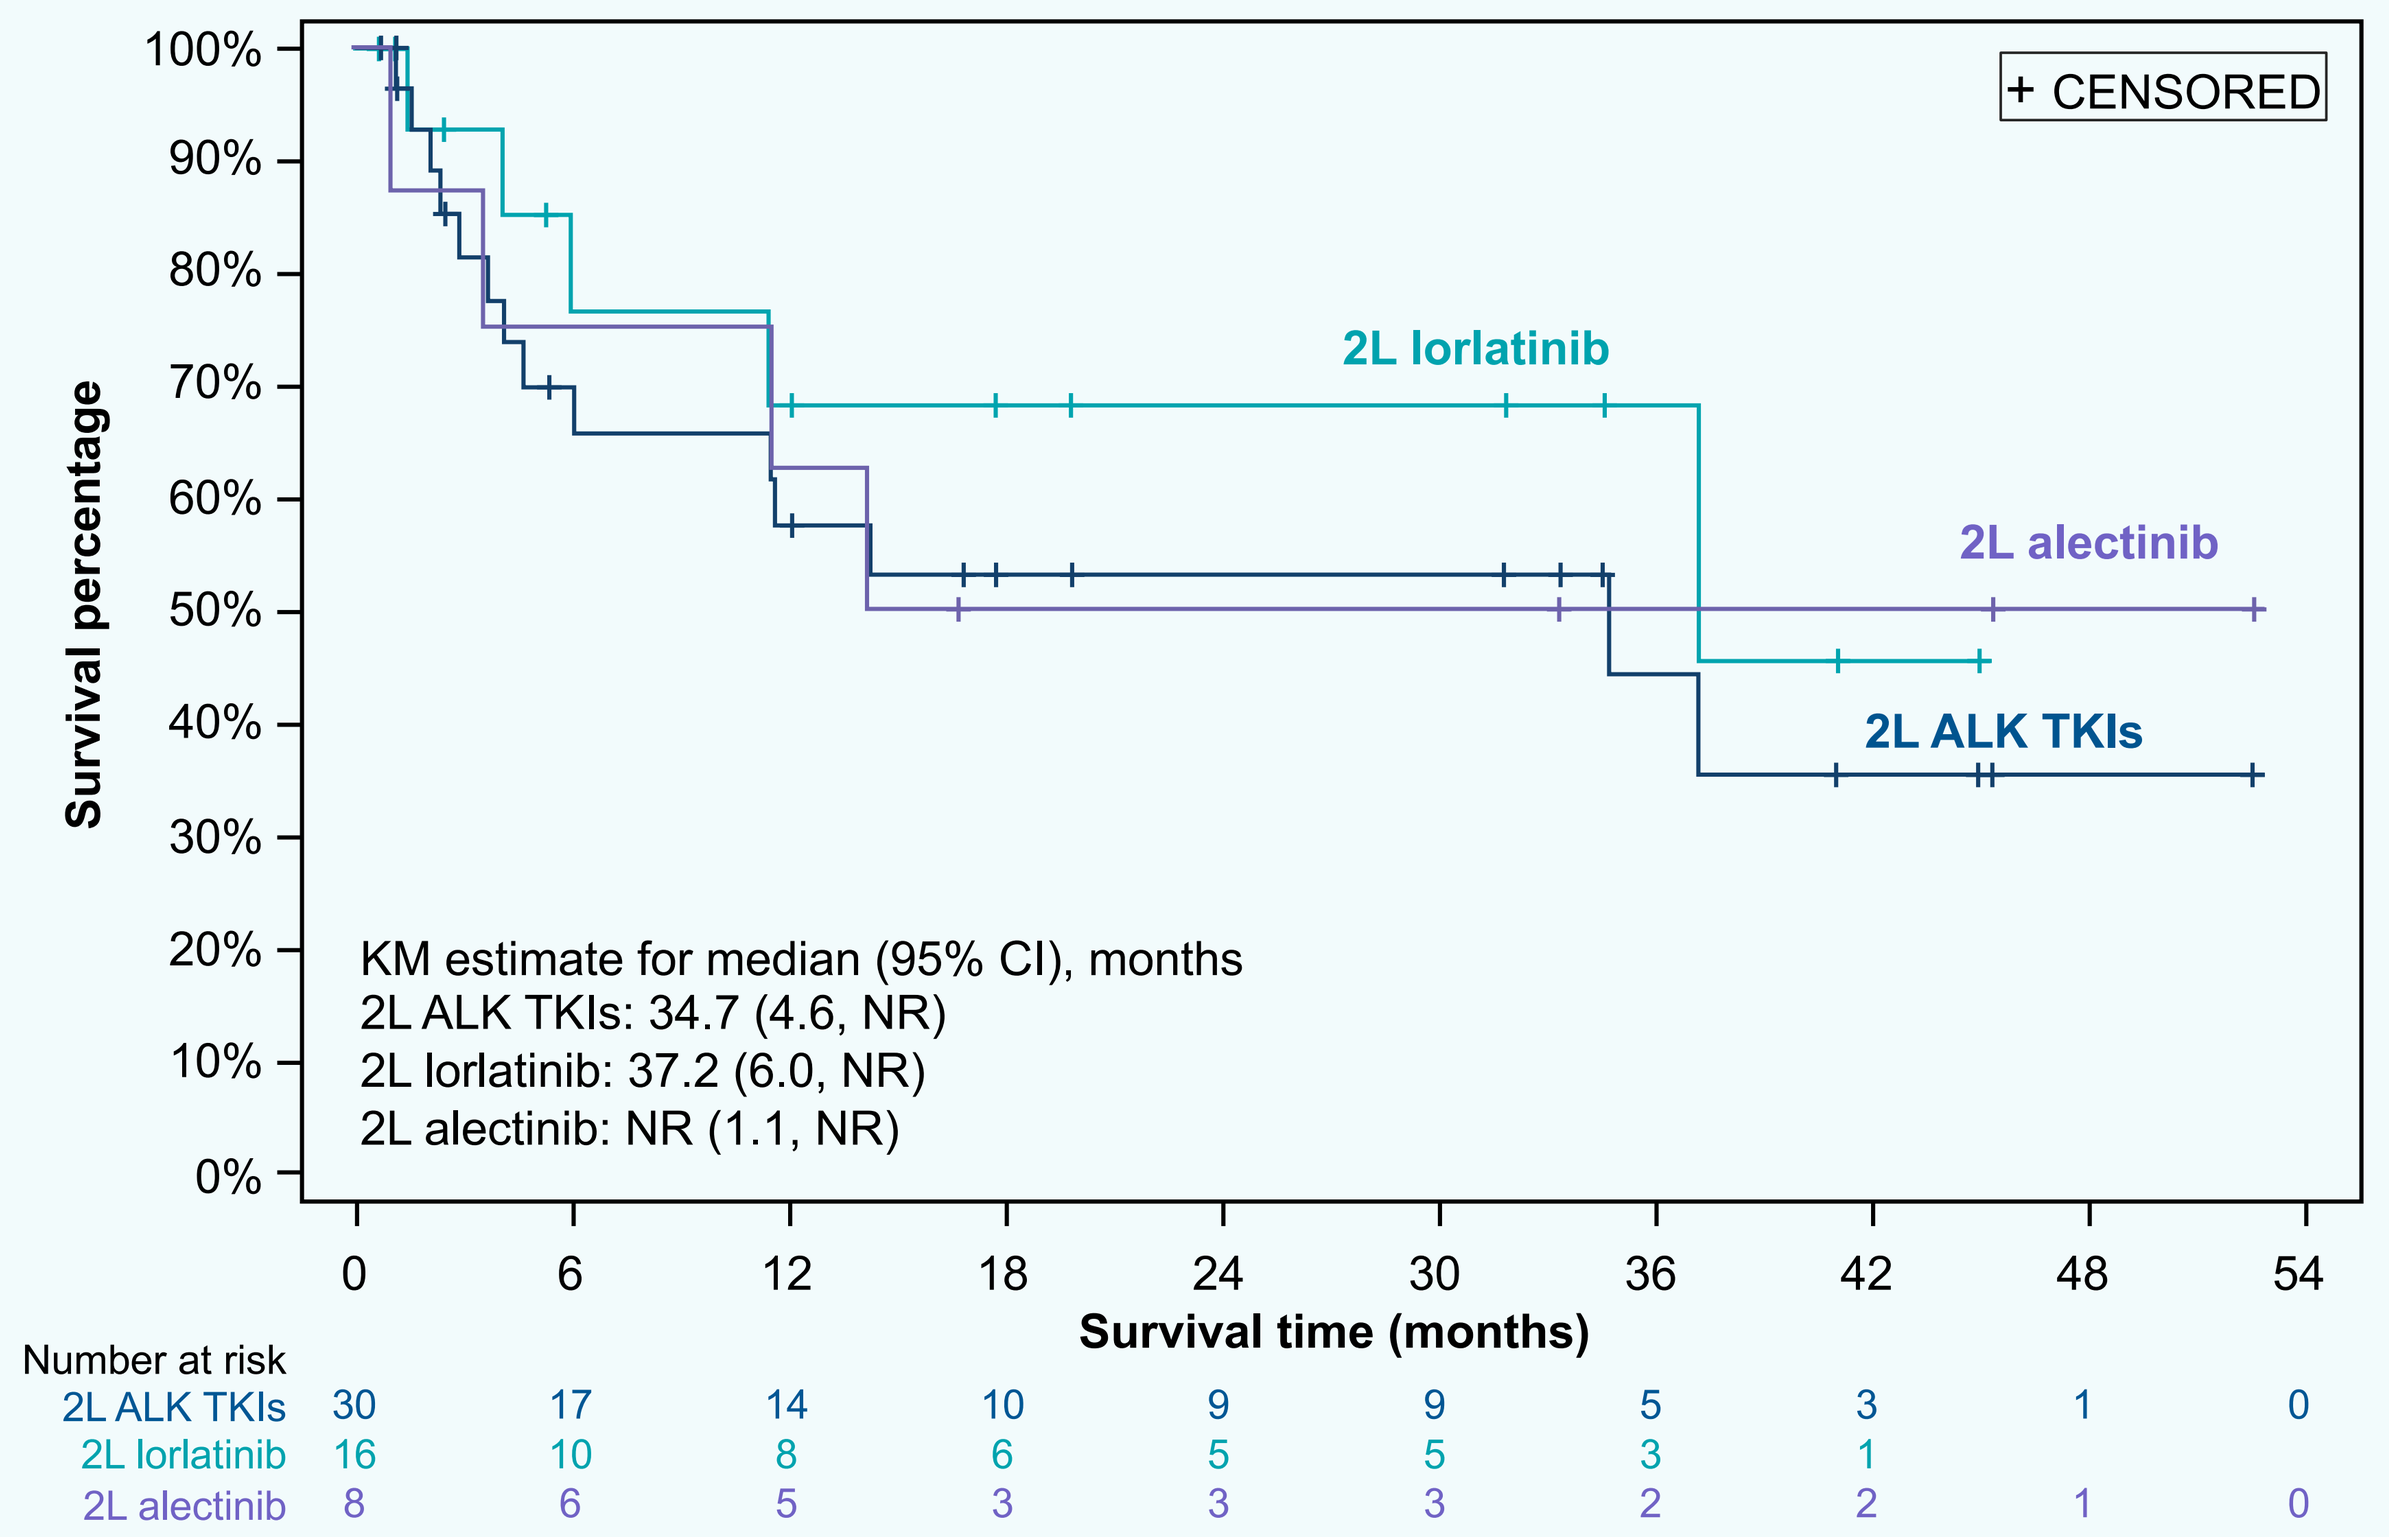
+ CENSORED
2L lorlatinib
2L alectinib
2L ALK TKIs
Survival percentage
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
KM estimate for median (95% CI), months
2L ALK TKIs: 34.7 (4.6, NR)
2L lorlatinib: 37.2 (6.0, NR)
2L alectinib: NR (1.1, NR)
0 6 12 18 24 30 36 42 48 54
Survival time (months)
Number at risk
2L ALK TKIs 30 17 14 10 9 9 5 3 1 0
2L lorlatinib 16 10 8 6 5 5 3 1
2L alectinib 8 6 5 3 3 3 2 2 1 0

2L, second-line; ALK, anaplastic lymphoma kinase; CI, confidence interval; KM, Kaplan-Meier; NR, not reached; rwTTD, real-world time to treatment discontinuation; TKIs, tyrosine kinase inhibitors.

Figure 3. rwPFS for 2L ALK TKIs, 2L alectinib, and 2L lorlatinib

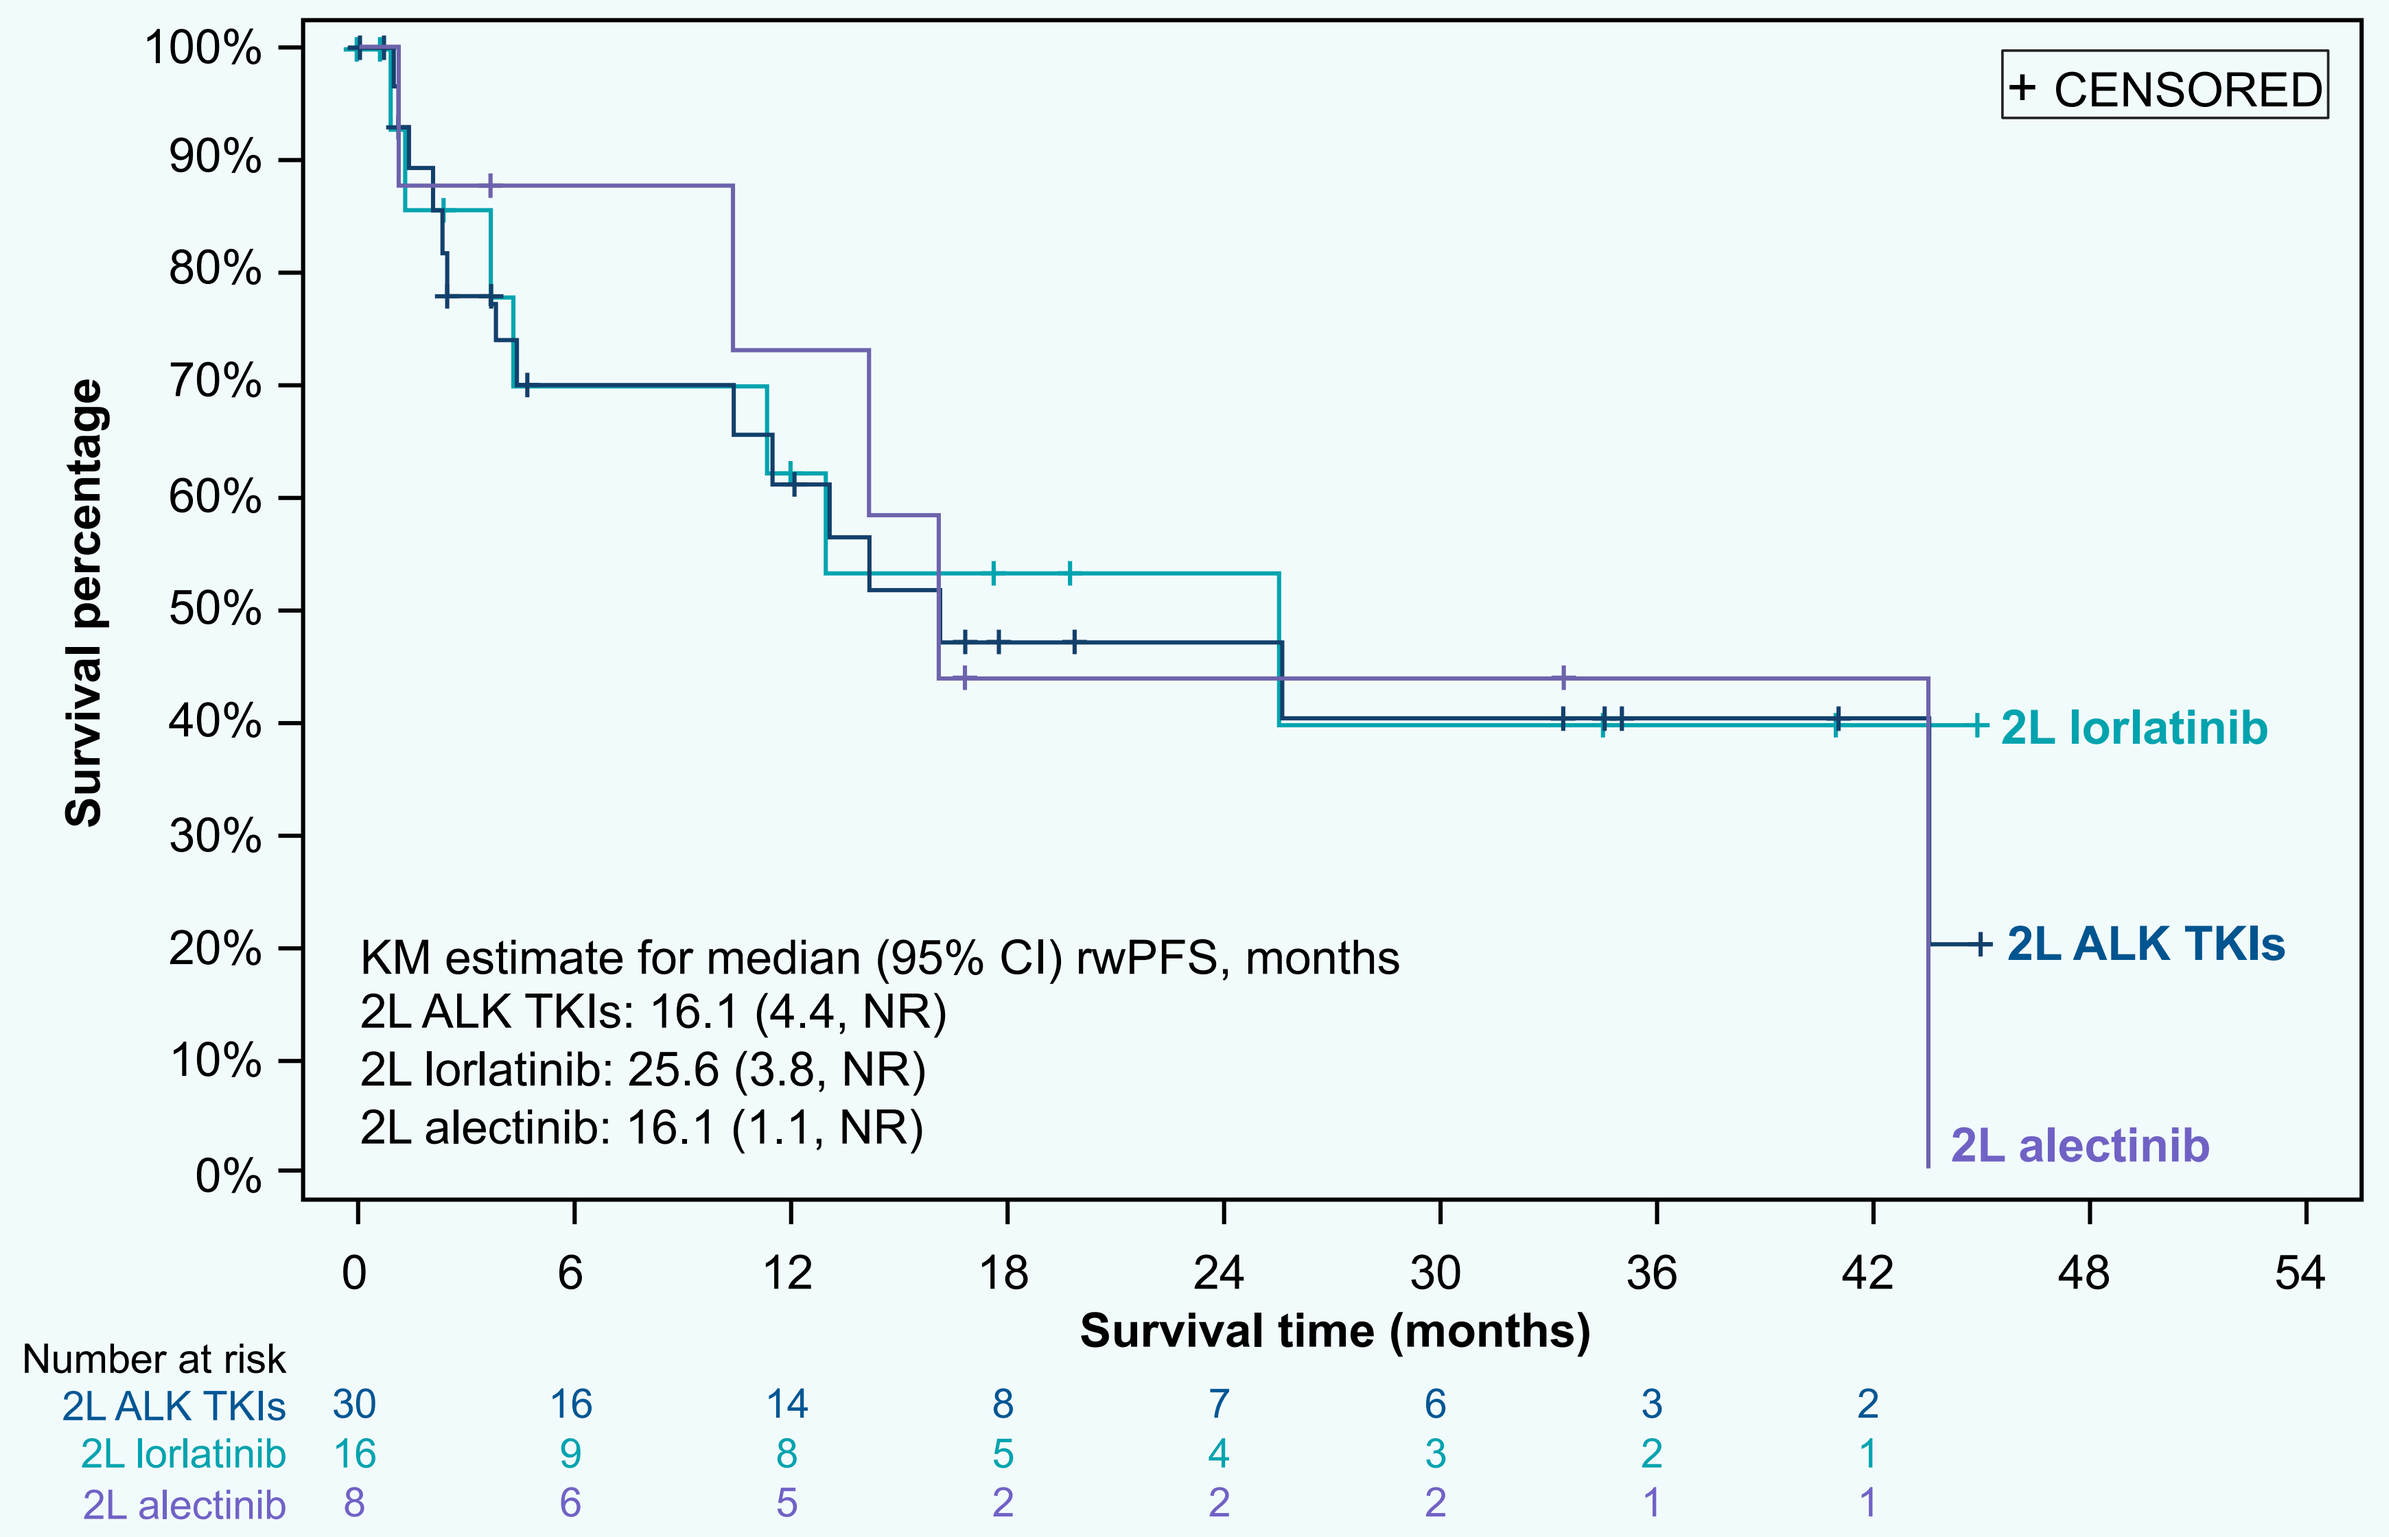
+ CENSORED
2L lorlatinib
2L ALK TKIs
2L alectinib
Survival percentage
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
KM estimate for median (95% CI) rwPFS, months
2L ALK TKIs: 16.1 (4.4, NR)
2L lorlatinib: 25.6 (3.8, NR)
2L alectinib: 16.1 (1.1, NR)
0 6 12 18 24 30 36 42 48 54
Survival time (months)
Number at risk
2L ALK TKIs 30 16 14 8 7 6 3 2
2L lorlatinib 16 9 8 5 4 3 2 1
2L alectinib 8 6 5 2 2 2 1 1

2L, second-line; ALK, anaplastic lymphoma kinase; CI, confidence interval; KM, Kaplan-Meier; NR, not reached; rwPFS, real-world progression-free survival; TKIs, tyrosine kinase inhibitors.

Key Take Away

Patients who received subsequent ALK TKIs after 1L brigatinib had prolonged clinical benefit.

rwTTD

- Median (95% confidence interval [CI]) rwTTD was 34.7 (4.6, not reached [NR]) months for 2L ALK TKIs and 37.2 (6.0, NR) months for 2L lorlatinib (**Table 3, Figure 2**).
 - At 24 months, rwTTD (95% CI) was 53.1% (32.2, 70.2) for 2L ALK TKIs and 68.1% (35.4, 86.8) for 2L lorlatinib.
- rwTTD for the additional subgroups of interest including 2L alectinib, 1L responders, and non-responders are shown in **Table 2.**

rwPFS, rwPFS2, and OS

- Median (95% CI) rwPFS was 16.1 (4.4, NR) months for 2L ALK TKIs and 25.6 (3.8, NR) months for 2L lorlatinib (**Table 3, Figure 3**).
 - Estimated 24-month rwPFS (95% CI) was 47.0% (26.2, 65.3) for 2L ALK TKIs and 53.4% (23.9, 76.0) for 2L lorlatinib.
- Median (95% CI) rwPFS2 was 51.6 (25.9, NR) months for 2L ALK TKIs and 74.7 (25.9, NR) months for 2L lorlatinib (**Table 3**).
 - Estimated 24-month rwPFS2 (95% CI) was 78.4% (58.1, 89.7) for 2L ALK TKIs and 86.7% (56.4, 96.5) for 2L lorlatinib.
- Median (95% CI) OS was 74.7 (30.0, NR) months for 2L ALK TKIs and 74.7 (30.0, NR) months for 2L lorlatinib (**Table 3**).
 - Estimated 36-month OS (95% CI) was 66.7% (46.9, 80.5) for 2L ALK TKIs and 75.0% (46.3, 89.8) for 2L lorlatinib.
- The outcomes data for the remaining subgroups are presented in **Table 3**.

Table 2. Baseline characteristics of patients receiving 2L ALK TKIs post-1L brigatinib

Baseline characteristics		ENR N=48	2L ALK TKIs n=30	2L Lorlatinib n=16	2L Alectinib n=8	2L ALK TKIs	
						1L Responder n=23	1L Non-Responder n=7
Age, median (range) (years)		58.0 (33–85)	57.5 (34–82)	52.5 (40–77)	59.5 (34–71)	55.0 (34–81)	64.0 (51–82)
Female sex, n (%)		26 (54.2)	18 (60.0)	10 (62.5)	5 (62.5)	14 (60.9)	4 (57.1)
Race, n (%)	Native Asian	26 (54.2)	15 (50.0)	7 (43.8)	4 (50.0)	11 (47.8)	4 (57.1)
	White	21 (43.8)	14 (46.7)	8 (50.0)	4 (50.0)	11 (47.8)	3 (42.9)
	Unknown	1 (2.1)	1 (3.3)	1 (6.3)	0	1 (4.3)	0
Geographic region, n (%)	Asia Pacific	25 (52.1)	15 (50.0)	7 (43.8)	4 (50.0)	11 (47.8)	4 (57.1)
	Europe	23 (47.9)	15 (50.0)	9 (56.3)	4 (50.0)	12 (52.2)	3 (42.9)
Cigarette smoking, n (%)	Never	30 (62.5)	20 (66.7)	12 (75.0)	3 (37.5)	16 (69.6)	4 (57.1)
	Current	2 (4.2)	0	0	0	0	0
	Former	13 (27.1)	8 (26.7)	3 (18.8)	4 (50.0)	5 (21.7)	3 (42.9)
	Unknown	3 (6.3)	2 (6.7)	1 (6.3)	1 (12.5)	2 (8.7)	0
Brain metastasis, n (%)		20 (41.7)	16 (53.3)	10 (62.5)	4 (50.0)	13 (56.5)	3 (42.9)
Follow-up, median (IQR) (months)		12.4 (3.7, 27.0)	17.0 (6.1, 36.0)	14.8 (2.7, 33.2)	28.1 (14.8, 40.8)	17.7 (2.8, 41.1)	12.7 (7.0, 31.9)

1L, first-line; 2L, second-line; ALK, anaplastic lymphoma kinase; ENR, enrolled set; IQR, interquartile range; TKIs, tyrosine kinase inhibitors.

Table 3. Real-world time to event outcomes in patients receiving 2L ALK TKIs post-1L brigatinib

Outcome	2L ALK TKIs n=30	2L Lorlatinib n=16	2L Alectinib n=8	2L ALK TKIs	
				1L Responder n=23	1L Non-Responder n=7
rwTTD					
Median, mo (95% CI)	34.7 (4.6, NR)	37.2 (6.0, NR)	NR (1.1, NR)	37.2 (3.6, NR)	11.6 (2.8, NR)
Receiving 2L at 24 mo, % (95% CI)	53.1 (32.2, 70.2)	68.1 (35.4, 86.8)	50.0 (15.2, 77.5)	57.3 (32.1, 76.2)	42.9 (9.8, 73.4)
rwPFS					
Median, mo (95% CI)	16.1 (4.4, NR)	25.6 (3.8, NR)	16.1 (1.1, NR)	25.6 (3.8, NR)	13.0 (2.4, NR)
24-mo rwPFS, % (95% CI)	47.0 (26.2, 65.3)	53.4 (23.9, 76.0)	43.8 (10.1, 74.2)	51.0 (26.4, 71.2)	34.3 (4.8, 68.5)
rwPFS2					
Median, mo (95% CI)	51.6 (25.9, NR)	74.7 (25.9, NR)	47.2 (12.6, NR)	68.8 (25.9, NR)	51.6 (3.5, NR)
24-mo rwPFS2, % (95% CI)	78.4 (58.1, 89.7)	86.7 (56.4, 96.5)	72.9 (27.6, 92.5)	81.3 (57.4, 92.6)	68.6 (21.3, 91.2)
OS					
Median, mo (95% CI)	74.7 (30.0, NR)	74.7 (30.0, NR)	NR (13.8, NR)	NR (30.0, NR)	74.7 (8.2, NR)
36-mo OS, % (95% CI)	66.7 (46.9, 80.5)	75.0 (46.3, 89.8)	75.0 (31.5, 93.1)	69.6 (46.6, 84.2)	57.1 (17.2, 83.7)

1L, first-line; 2L, second-line; ALK, anaplastic lymphoma kinase; CI, confidence interval; mo, months; NR, not reached; OS, overall survival; rwPFS, real-world progression-free survival; rwTTD, real-world time to treatment discontinuation; TKIs, tyrosine kinase inhibitors.

Limitations

- The subset of included sites may have differed from the brigatinib arm or the overall ALTA-1L trial population; sites may not be representative of all sites in a given country.
- Patients who discontinued 1L brigatinib and received subsequent 2L ALK TKIs may have differed from the overall cohort of patients enrolled in brigatinib arm of ALTA-1L.
- The timing between patients discontinuing 1L brigatinib and receiving subsequent 2L ALK TKIs may have differed within the cohort of patients included in the study.
- The use of retrospective chart review data may be associated with systematic under-reporting of information.
- This study was limited by small sample size. Hence, KM estimates of median require cautious interpretation, particularly given the wide CIs; this study was not powered to detect differences between subgroups.

Conclusions

- This is the first long-term study evaluating real-world outcomes post-1L brigatinib in patients with *ALK*+ NSCLC.
- Following 1L brigatinib discontinuation, most patients started another ALK TKI for 2L treatment and had prolonged clinical benefit.
- Time on subsequent 2L ALK TKI was observed to be longer for patients who responded to 1L brigatinib compared with 1L non-responders.

References

1. Barlesi F, et al. *The Lancet.* 2016;387(10026):1415-1426.
2. Solomon B, et al. *J Thorac Oncol.* 2009;4(12):1450-1454.
3. Spagnuolo A, et al. *Expert Opin Emerg Drugs.* 2018;23(3):231-241.
4. Camidge DR, et al. *J Thorac Oncol.* 2021;16(12):2091-2108.
5. Delmonte A. Presented at ELCC 2024; Poster #38P.

Disclosures

Myung-Ju Ahn reports honoraria from AstraZeneca, Merck Sharp & Dohme, Roche, Bristol Myers Squibb, Merck KGaA, and Alpha Pharmaceuticals, outside of the submitted work.

Acknowledgements

This study was funded by Takeda Development Center Americas, Inc. (TDCA). Medical writing and editorial support was provided by SNELL Medical Communication, Inc. and funded by TDCA.

Presented at WCLC 2024, San Diego, USA, 7–10 September 2024; for any questions or comments please contact Myung-Ju Ahn (silkahn@skku.edu).