A phase 1/2, first-in-human, open-label, dose-escalation study of TAK-280, an investigational B7-H3 x CD3ε Conditional Bispecific Redirected Activation (COBRA) T-cell engager, in adult patients with unresectable, locally advanced, or metastatic solid tumors

TPS2684

Gary Richardson,[1] Ganessan Kichenadasse,[2] Nicholas Zorko,[3] Wen Wee Ma,[4] Hui-Zi Chen,[5] David Starks,[6] Sarah Sutherland,[7] Sujana Kamasani,[8] Antari Khot,[8] Xin Ma,[8] William L. Trepicchio,[8] Yefei Zhang,[8] Elizabeth Ahern[9]

[1]Cabrini Research, Melbourne, Australia; [2]Southern Oncology Clinical Research Unit Pty Ltd, Adelaide, Australia; [3]University of Minnesota Masonic Cancer Center, Minneapolis, MN, USA; [4]Cleveland Clinic, Cleveland, OH, USA; [5]Medical College of Wisconsin, Milwaukee, WI, USA; [6]Avera Cancer Institute, Sioux Falls, SD, USA; [7]Chris O'Brien Lifehouse, Sydney, Australia; [8]Takeda Development Center Americas, Inc. (TDCA), Lexington, MA, USA; [9]Monash Health, Melbourne, Australia

Trial design First-in-human, open-label, phase 1/2 study of TAK-280 in patients with unresectable, locally advanced or metastatic cancers

Study objectives

Primary: To characterize the **safety**, **tolerability**, and **DLTs** of TAK-280 to determine its **MTD** (if any) and **RP2D**

Secondary: To characterize the **PK** and **potential immunogenicity,** and to evaluate the **preliminary anti-tumor activity** of TAK-280

Key inclusion criteria

- Unresectable, locally advanced, or metastatic solid tumor
- Ineligible for, or intolerant of, standard therapies
- ECOG PS ≤1
- Measurable disease per RECIST v1.1

N~182

Phase 1: dose escalation

N~42

TAK-280 IV QW in 28-day cycles*
Dose-escalation† at 8 dose levels following a BOIN design

2 RDEs selected for 1:1 randomization (in 1 of the cohorts)

Phase 2: cohort expansion‡

N~100–140

Cohort 1

Cohort 2

Cohort 3

Patients will be followed-up for survival every 12 weeks for ≤48 weeks after the last dose of study drug

Patients who do not experience dose-limiting or other unacceptable toxicities may receive ≤14 treatment cycles depending on response§; patients are treated until disease progression, unacceptable toxicity, or withdrawal from study

Clinicaltrials.gov ID: NCT05220098

*Initial infusion of 60 minutes, may be adjusted to shorter or longer time (but never less than 30 minutes). †Only one patient will be enrolled for the first two dose levels. ‡Indications to be confirmed based on data from the escalation phase.
§Patients who, in the opinion of the investigator, continue to receive clinical benefit by the end of the 14 cycles with acceptable toxicity and no alternative treatment option, may continue therapy on a case-by-case basis.
BOIN, Bayesian Optimal Interval design; DLT, dose-limiting toxicity; ECOG PS, Eastern Cooperative Oncology Group performance status; IV, intravenous; MTD, maximum tolerated dose; QW, once weekly; PK, pharmacokinetic; RDE, recommended dose for expansion; RECIST, Response Evaluation Criteria in Solid Tumors; RP2D, recommended phase 2 dose.

Background

- The COnditional Bispecific Redirected Activation (COBRA) platform is designed to allow the targeting of antigens widely expressed in solid tumors that are typically not targetable with traditional T-cell engagers (TCEs)[1–3]
- Conditional T cell engagement has the potential to deliver wider therapeutic margin to treat solid tumors[1,4,5]
- TAK-280 is a novel COBRA TCE that targets the transmembrane protein B7 homolog 3 protein (B7-H3; also known as CD276)
- B7-H3 is known to be overexpressed in a range of solid tumors, including metastatic castration-resistant prostate cancer (mCRPC) and non–small cell lung cancer[6,7]

Figure 1. TAK-280 prodrug molecular design

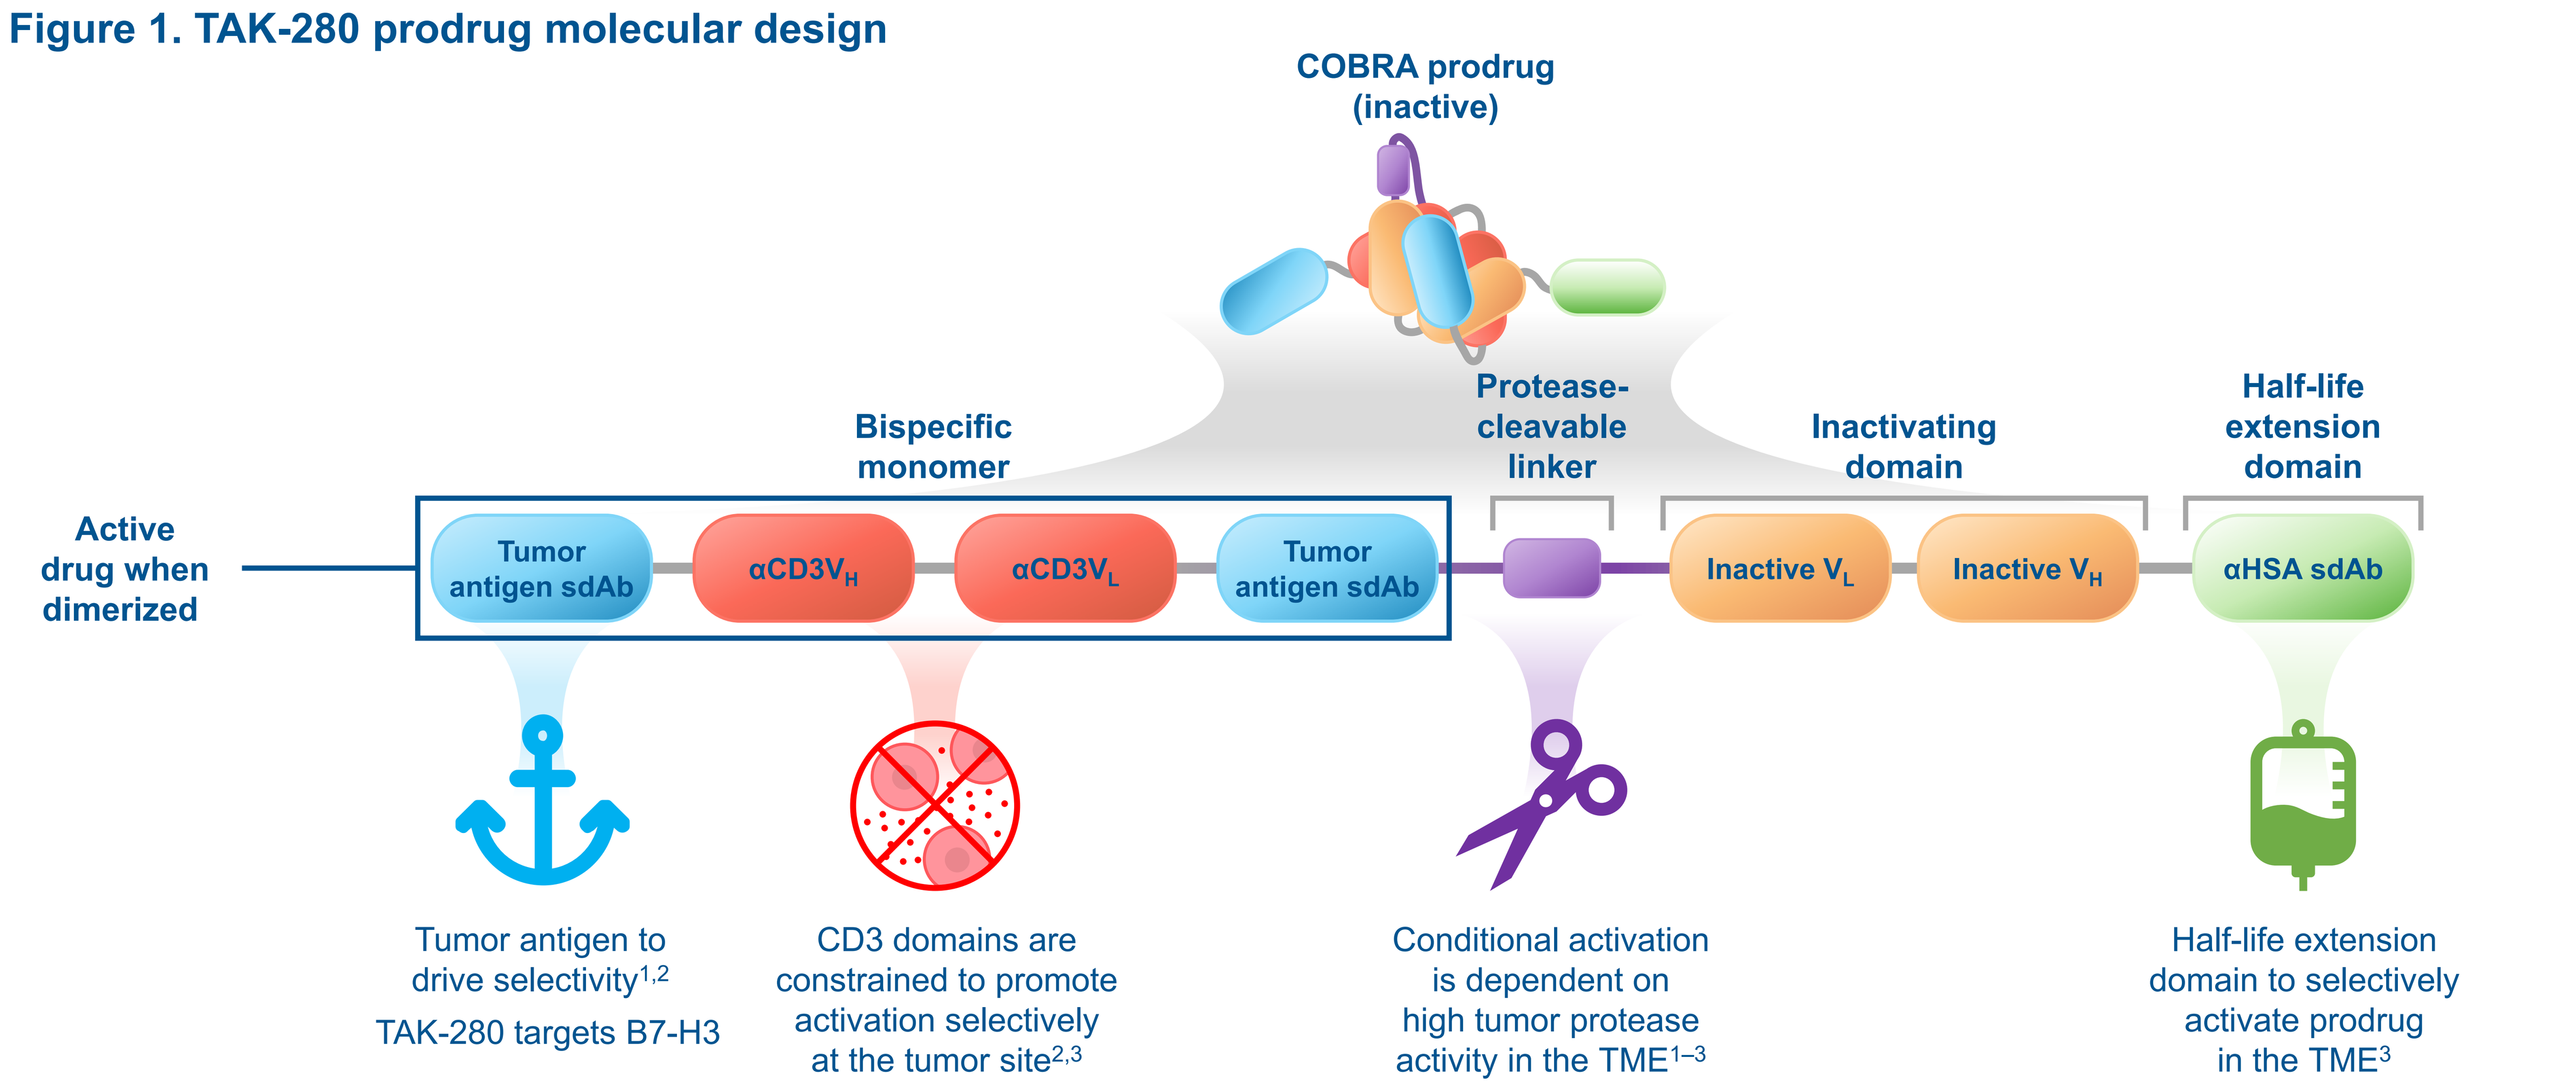
COBRA prodrug (inactive)
Bispecific monomer
Protease-cleavable linker
Inactivating domain
Half-life extension domain
Active drug when dimerized
Tumor antigen sdAb
αCD3V_H
αCD3V_L
Tumor antigen sdAb
Inactive V_L
Inactive V_H
αHSA sdAb
Tumor antigen to drive selectivity[1,2]
TAK-280 targets B7-H3
CD3 domains are constrained to promote activation selectively at the tumor site[2,3]
Conditional activation is dependent on high tumor protease activity in the TME[1–3]
Half-life extension domain to selectively activate prodrug in the TME[3]

HSA, human serum albumin; sdAb, single-domain antibody; V_H, variable domain of the heavy chain; V_L, variable domain of the light chain.

Figure 2. Conditional activation of TAK-280

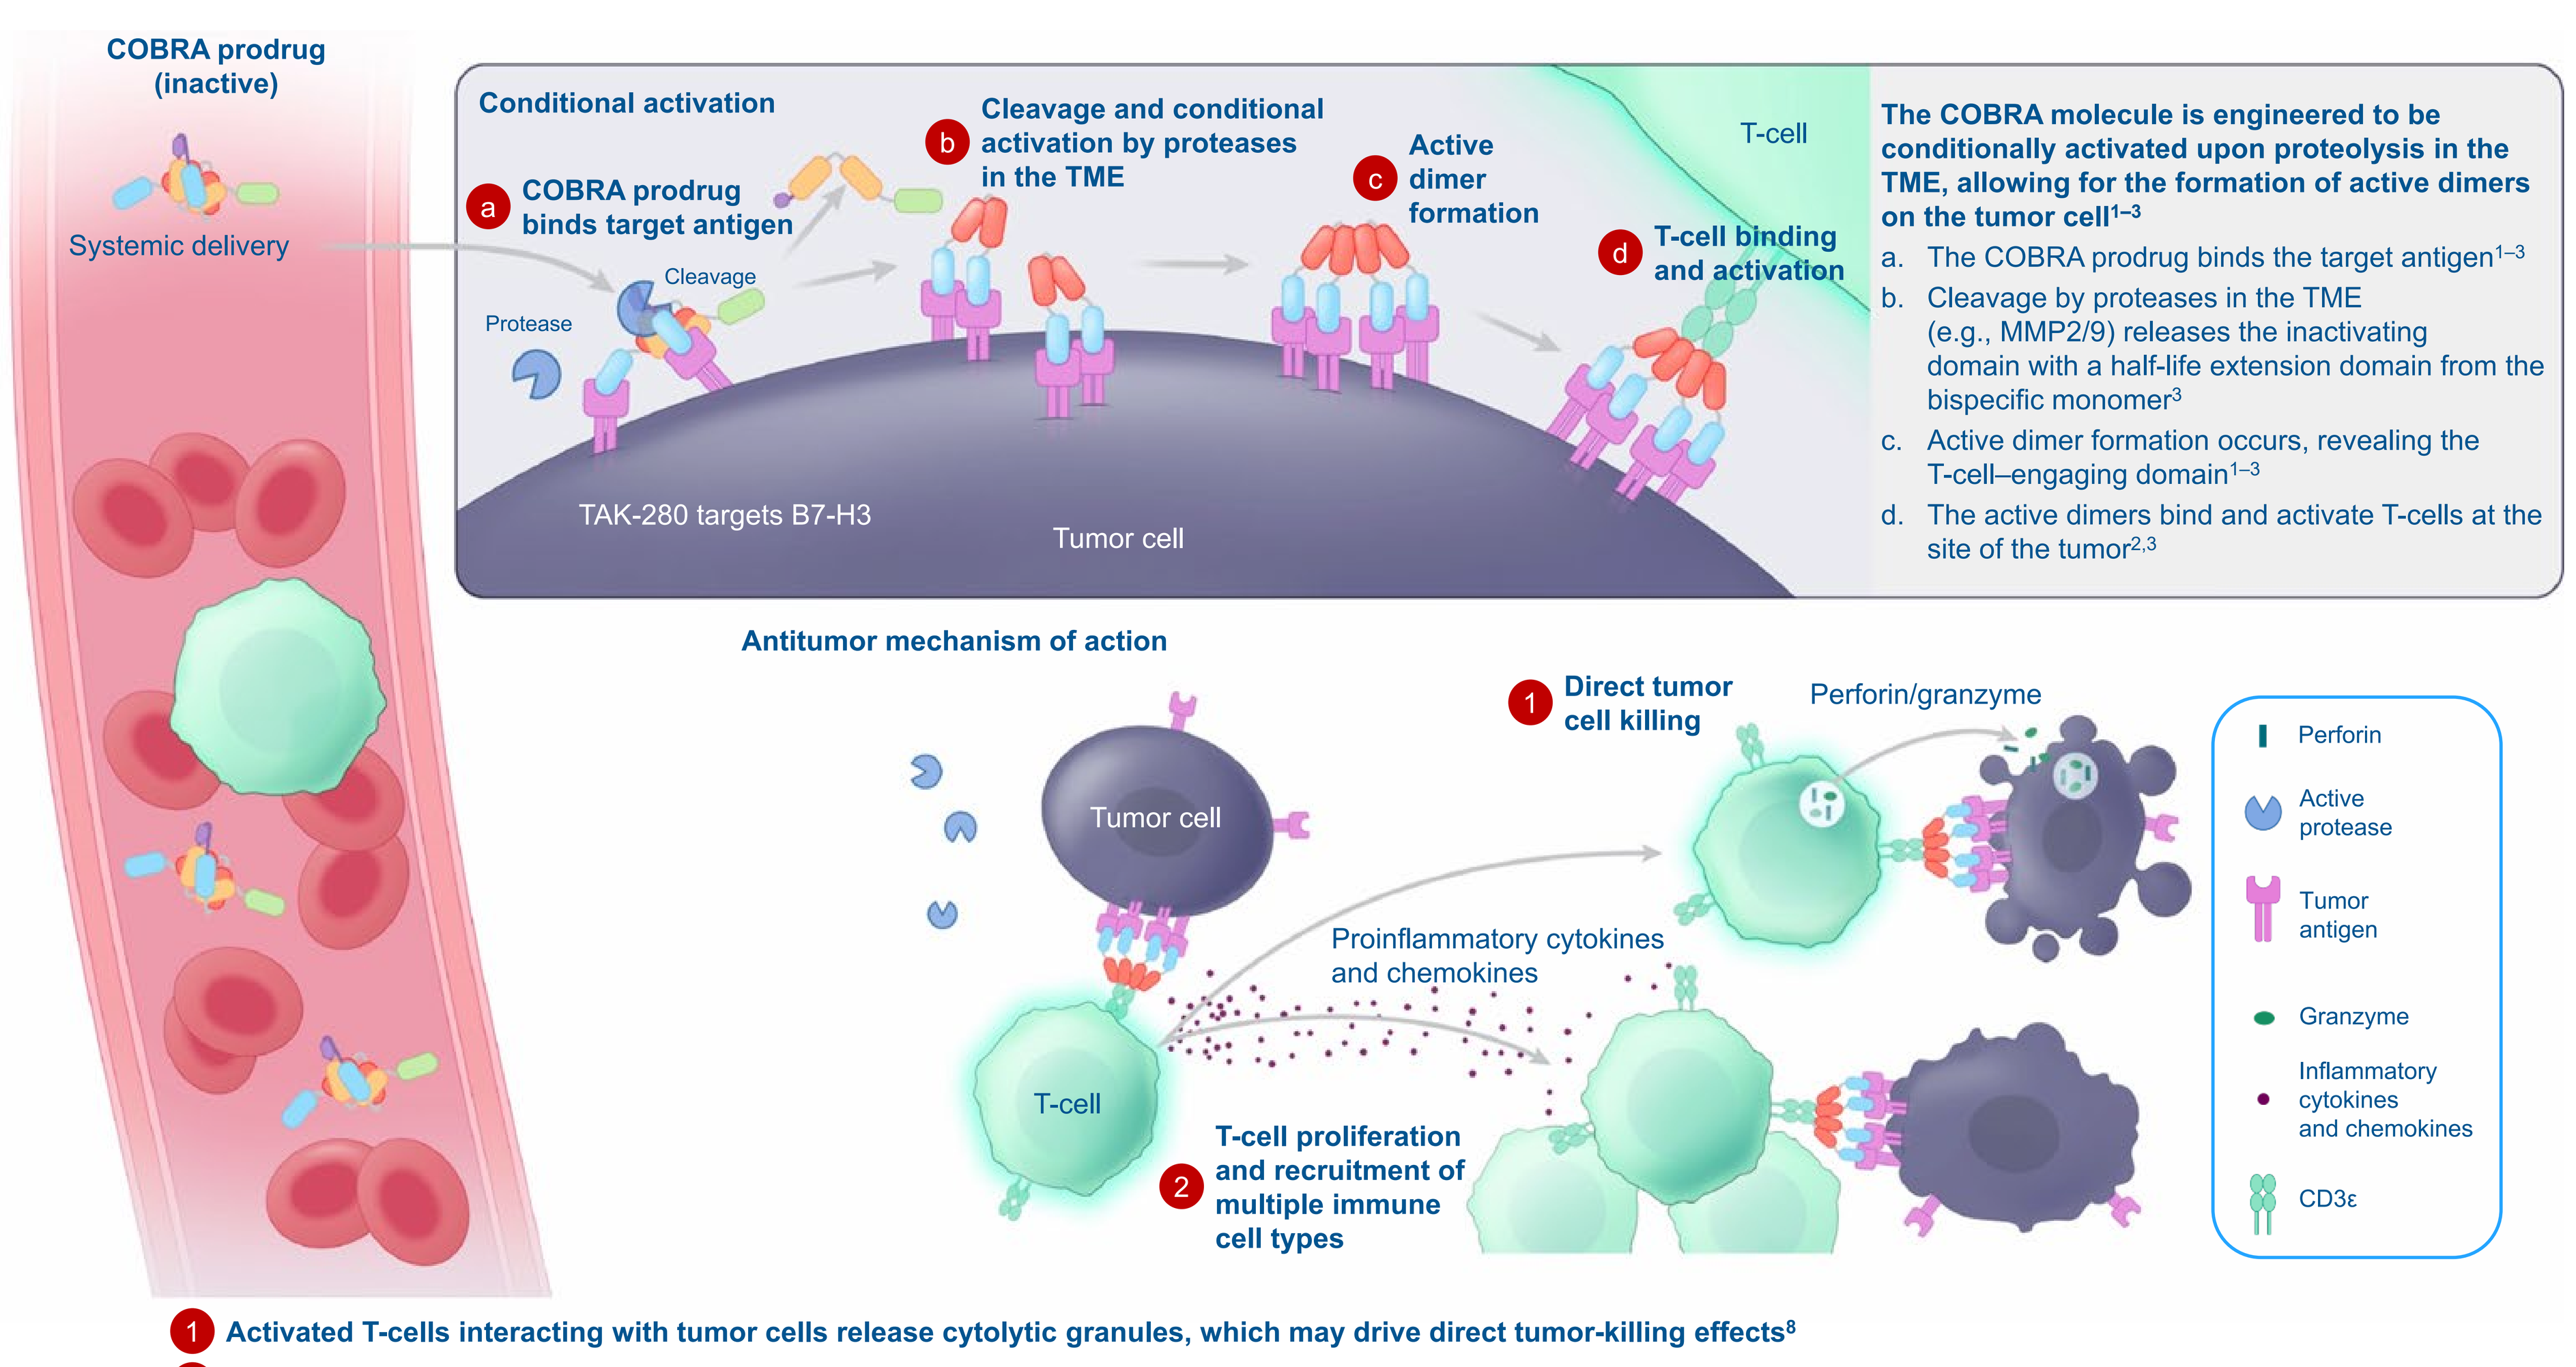
COBRA prodrug (inactive)
Systemic delivery
Conditional activation
a COBRA prodrug binds target antigen
Protease
Cleavage
b Cleavage and conditional activation by proteases in the TME
c Active dimer formation
d T-cell binding and activation
T-cell
TAK-280 targets B7-H3
Tumor cell
The COBRA molecule is engineered to be conditionally activated upon proteolysis in the TME, allowing for the formation of active dimers on the tumor cell[1–3]
a. The COBRA prodrug binds the target antigen[1–3]
b. Cleavage by proteases in the TME (e.g., MMP2/9) releases the inactivating domain with a half-life extension domain from the bispecific monomer[3]
c. Active dimer formation occurs, revealing the T-cell–engaging domain[1–3]
d. The active dimers bind and activate T-cells at the site of the tumor[2,3]
Antitumor mechanism of action
1 Direct tumor cell killing
Perforin/granzyme
Tumor cell
Proinflammatory cytokines and chemokines
T-cell
2 T-cell proliferation and recruitment of multiple immune cell types
Perforin
Active protease
Tumor antigen
Granzyme
Inflammatory cytokines and chemokines
CD3ε

1 Activated T-cells interacting with tumor cells release cytolytic granules, which may drive direct tumor-killing effects[8]
2 Activated T-cells release cytokines to drive T-cell proliferation and expend T-cells and other immune cells, potentially amplifying the tumor-killing effect[9,10]

- In its prodrug form, TAK-280 binds to B7-H3 but not to CD3ε on T-cells (**Figure 1**)
- Once in the protease-rich tumor microenvironment (TME), TAK-280 undergoes protease-mediated activation
 – Formation of active dimers through CD3ε-binding, and co-engagement of B7-H3 expressing cells is postulated to result in cytotoxic anti-tumor response[1] (**Figures 1** and **2**)
- In preclinical studies, COBRA molecules demonstrated cleavage-dependent conditionality, engagement of tumor target antigen, and induction of T-cell mediated tumor killing[3]

Study eligibility

- This phase 1/2 study is enrolling adult patients with unresectable, locally advanced, or metastatic solid tumors described in the literature to have enhanced B7-H3 expression[6] (**Summary panel** and **Table 1**)

Table 1. Key inclusion and exclusion criteria

Inclusion criteria	Exclusion criteria
≥18 years old	History of known autoimmune disease
Histological or pathological confirmation of unresectable, locally advanced, or metastatic solid tumor described in the literature to have enhanced B7-H3 expression	Major surgery, or traumatic injury within 8 weeks before the first dose of TAK-280
Ineligible for or intolerant of standard therapies, have no approved therapy with demonstrated benefit or have exhausted all available therapies	Vaccination ≤2 weeks (or ≤4 weeks for live virus vaccine) of first dose of TAK-280
ECOG PS ≤1	Ongoing or active infection of grade ≥2, or inflammatory process unresolved for ≥4 weeks before first dose of TAK-280
Measurable disease per RECIST v1.1. (except for patients with mCRPC and bone lesions only)	Clinically significant cardiac or gastrointestinal disorders ≤6 months prior to receiving the first dose of TAK-280
Acceptable laboratory parameters	Bone marrow or solid organ transplant, or use of immunosuppressive agents, within the past 5 years
Appropriate measures taken to prevent pregnancies in female patients of child-bearing potential or in male patients with partners of child-bearing potential	Known hypersensitivity to TAK-280 or any excipient
Treated symptomatic or asymptomatic ≥14 days central nervous system (CNS) metastases with no concurrent treatment for CNS disease or leptomeningeal disease or cord compression	Second primary invasive malignancy not in remission for ≥3 years

Key study endpoints

Primary endpoint

1. Incidence of DLTs and other toxicities* during dose escalation

Secondary endpoints

1. PK parameters
2. Response assessment by the investigator (using RECIST v1.1)
3. Confirmed overall response rate (defined as rate of complete responses + partial responses)
4. Duration of response
5. Disease control rate
6. Overall survival and progression-free survival
7. Immunogenicity characterization (presence, titer, and impact assessment on exposure, safety, and efficacy)
8. Prostate-specific antigen (PSA) assessment (for patients with mCRPC only): PSA response, duration of response, time to PSA progression, and proportion of patients with PSA reduction ≥6

*Safety endpoints are evaluated according to National Cancer Institute (NCI) Common Terminology Criteria for Adverse Events (CTCAE) v5.0 for non-cytokine-release syndrome (CRS) toxicities, and the American Society for Transplantation and Cellular Therapy (ASTCT) CRS consensus grading for CRS/infusion-related reaction toxicities.[11]

Study status

- Patients are being enrolled in ~16 study sites in the United States, Australia, Canada, and Spain[12]
- As of the data cutoff date of Jan 10, 2024, 18 patients have received the study drug
- Study completion is estimated for 2026[11]

References

1. Panchal A, et al. *mAbs*. 2020;12:1792130
2. Krakow J, et al. *Antib Ther*. 2022;5:1–10
3. Dettling DE et al. *J Immunother Cancer*. 2022;10:e004336
4. Chen TT. *Exp Opin Biol Ther*. 2022;8:955–63
5. Vafa O and Trinklein ND. *Front Oncol*. 2020;10:446
6. Yang S, et al. *Int J Biol Sci*. 2020;16:1767–73
7. Guo C, et al. *Eur Urol*. 2023;83:224–38
8. Waldman AD, et al. *Nat Rev Immunol*. 2020;20:651–68
9. Huehls AM, et al. *Immunol Cell Biol*. 2015;93:290–6
10. Kamakura D, et al. *Pharmaceuticals*. 2021;14:1172
11. Lee DW, et al. *Biol Blood Marrow Transplant*. 2019;25:625–38
12. ClinicalTrials.gov entry for NCT05220098. Available at: https://clinicaltrials.gov/study/NCT05220098. Accessed on April 11, 2024

Acknowledgments

This study was funded by Takeda Development Center Americas, Inc. (TDCA), Lexington, MA, USA. Medical writing support for the development of this poster, under the direction of the authors, was provided by Emily Ruban-Fell, PhD, of Ashfield MedComms, an Inizio Company, funded by Takeda Pharmaceuticals U.S.A., Inc., Lexington, MA, and complied with the Good Publication Practice (GPP) guidelines (DeTora LM, et al. Ann Intern Med. 2022;175:1298–304).

Disclosures

GR: Research Funding: Bristol-Myers Squibb, Roche/Genentech, AstraZeneca, Merck, Takeda, Beigene, Pfizer, CBT Pharmaceuticals, Corvus Pharmaceuticals, Novotech, Fosun Pharma, Shangai Henlius Biotech, Five Prime Therapeutics, Suzhou Alphamab, Boehringer Ingelheim, Adagene, Bio-Thera Solutions, ChemoCentryx, Curon Biopharmaceutical, D3 Bio, InventisBio, Senz Oncology, Genfleet Therapeutics, Gene Quantum, Keythera Pharmaceuticals, LaNova Australia, Medicenna, Minghui Pharmaceutical, Neoleukin Therapeutics, PharmAbcine, RemeGen, Seagen, Surface Oncology, Eucure Biopharma, Janssen Oncology, ImmunGen Inc, Imugene, Therapim, Zentalis, Agenus. **NZ:** Travel, Accommodations, Expenses: Telix Pharmaceuticals, Caris Life Sciences. **WWM:** Consulting or Advisory Role: Kodikaz Therapeutic Solutions; Research Funding: Athenex, Bristol-Myers Squibb, Ipsen, Actuate Therapeutics, Sun Pharma, BioMed Valley Discoveries, Genentech, Elucida Oncology, Cardiff Oncology, Jacobio, CUE Biopharma, Repare Therapeutics, Merus. **HC:** Speakers' Bureau: Jazz Pharmaceuticals. **SK:** Employment: Takeda; Stock and Other Ownership Interests: Takeda; **AK:** Employment: Takeda; Stock and Other Ownership Interests: Takeda. **XM:** Employment: Takeda. **WLT:** Employment: Takeda; Leadership: Takeda; Stock and Other Ownership Interests: Takeda. **YZ:** Employment: Takeda; Stock and Other Ownership Interests: Takeda. **EA:** Travel, Accommodations, Expenses: Amgen, Bristol-Myers Squibb; Patents, Royalties, Other Intellectual Properties: U.S. Provisional application n. 62/775803; Honoraria: MSD Oncology; Research Funding: Amgen. **GK, DS, SS:** no COI to disclose.

To view a copy of this poster, scan this quick response (QR) code or visit: https://tiny.one/7J21479yaU.

Copies of this poster obtained through QR Code are for personal use only and may not be reproduced without permission from the author of this presentation.

Presented at the American Society of Clinical Oncology (ASCO) 2024 annual meeting Chicago, IL, USA, May 31−June 4, 2024. For questions or comments, please contact Dr Gary Richardson: gary.richardson@ocv.net.au