Feasibility and Utilization of Electronic Patient-Reported Outcome Measures for Lung Cancer in Routine Clinical Care

11107

Ethan Basch, MD[1], Huamao M. Lin, PhD[2], Mary Lynn Cala, MS[2], Meni Styliadou, LLM[2], Aaron Galaznik, MD[3], Debra Wujcik, PhD, RN[3], Emelly Rusli, MPH[3], Nicholas Coombs, PhD, MSTAT[4], Emily Beamon, PhD, MA, MPH[4]
[1]University of North Carolina, [2]Takeda Pharmaceuticals, [3]Carevive Systems, Inc., [4]Piedmont Research Strategies

BACKGROUND

- Prior studies have shown the value of Patient-Reported Outcomes (PRO) in cost burden, quality of life (QoL), and survival in cancer care[1-4].
- Technology advances allow capture of PRO data from home using devices such as smart phones, sending PRO results directly to the electronic medical record (EMR), allowing clinical professionals to monitor patient-reported status in real time and longitudinally.
- To facilitate focus on patient-centricity and value-based care, the European Health Outcomes Observatory (H2O) developed a Core Outcomes Set (COS) in lung cancer[5].
- We explored the feasibility of deploying standardized PRO measures among patients with lung cancer in US clinical practice.

METHODS

- A retrospective database study, with IRB waiver, was conducted in 207 adult lung cancer patients on active treatment across 9 US cancer centers from 9/2020 to 5/2023 using Carevive PROmpt®, an electronic PRO (ePRO) remote symptom monitoring platform with weekly surveys comparable in scope to H2O COS using:
 - The PRO-CTCAE to assess symptomatic adverse events
 - The PROMIS 4A Physical Function to assess physical function
 - Items 29 and 30 from the EORTC QLQ-C30 to assess global health status/quality of life
- Moderate and severe PRO-CTCAE symptoms scores triggered alerts to the clinical team (e.g., nurses, Advanced Practitioner nurses (AP), and physicians).
- Patient engagement was measured by survey compliance, follow-up time, and data completeness.
 - Survey compliance was defined as the number of surveys completed over the number of surveys assigned per patient.
 - Follow-up time was defined as the number of weeks between the patient's first and last completed survey.
 - Data completeness was measured for each clinical and PRO domain and was defined as the proportion of patients (or surveys, for longitudinal PRO variables) with data available for a given domain at any time during the study period. Median completion was summarized over the number of domains assessed.
- Clinical team engagement was measured by platform utilization time, time to alert response, and clinical action taken.

RESULTS

- Participants were 50% female, 86% White, 12% African American, median age 68 years, 81% with NSCLC, and 26% with metastatic disease (Table 1).

Table 1: Demographic and clinical characteristics

Variable	All Patients (n = 207)	NSCLC (n = 168)	SCLC (n=39)
Female, N (%)	103 (50)	80 (48)	23 (59)
Age at enrollment (years), Mean (SD)	66.5 (0.69)	66.7 (0.78)	65.5 (1.49)
Age at enrollment (years), Median	68	68	66
Age group, N (%)			
<30 years old	1 (1)	1 (1)	0 (0)
31-40 years old	2 (1)	1 (1)	1 (3)
41-50 years old	11 (5)	9 (5)	2 (5)
51-60 years old	31 (15)	25 (15)	6 (15)
61-70 years old	87 (42)	69 (41)	18 (46)
71-80 years old	63 (30)	51 (30)	12 (31)
>80 years old	12 (6)	12 (7)	0 (0)
Race, n (%)			
White	178 (86)	143 (85)	35 (90)
Black or African American	24 (12)	22 (13)	2 (5)
Asian	2 (1)	1 (1)	1 (3)
Not Specified	2 (1)	1 (0)	1 (3)
American Indian or Alaska Native	1 (0)	1 (1)	0 (0)
Ethnicity, N (%)			
Not Hispanic or Latino	107 (52)	90 (54)	17 (44)
Not Specified	100 (48)	78 (46)	22 (56)
Currently deceased, N (%)	10 (5)	5 (3)	5 (13)
Stage, n (%)			
I	11 (5)	7 (4)	4 (10)
II	16 (8)	14 (8)	2 (5)
III	51 (25)	46 (27)	5 (13)
IV	54 (26)	46 (28)	8 (21)
Extensive	4 (2)	0 (0)	4 (10)
Not Specified	71 (34)	55 (33)	16 (41)

NSCLC: Non-Small Cell Lung Cancer; SCLC: Small Cell Lung Cancer

QUESTION

Is it feasible to deploy standardized PRO measures among patients with lung cancer in US clinical practice?

STUDY DESIGN

A retrospective database study was conducted in adult lung cancer patients using Carevive PROmpt®, an ePRO remote symptom monitoring platform across 9 US cancer centers from 9/2020 to 5/2023.

RESULTS

- Patient engagement was high, as indicated by survey compliance and completeness, particularly in the first 5 weeks of enrollment. Median survey follow-up was 8 weeks with 85% median compliance over that time (Figure 2).
- Of 178 patients triggering >1 ePRO symptom alert, 87% were within 4 weeks and 46% of alerts include patients requesting a callback to discuss symptoms with the clinical team (Figure 3).

Figure 2: Patient engagement with Carevive ePRO platform indicated by survey compliance and follow-up

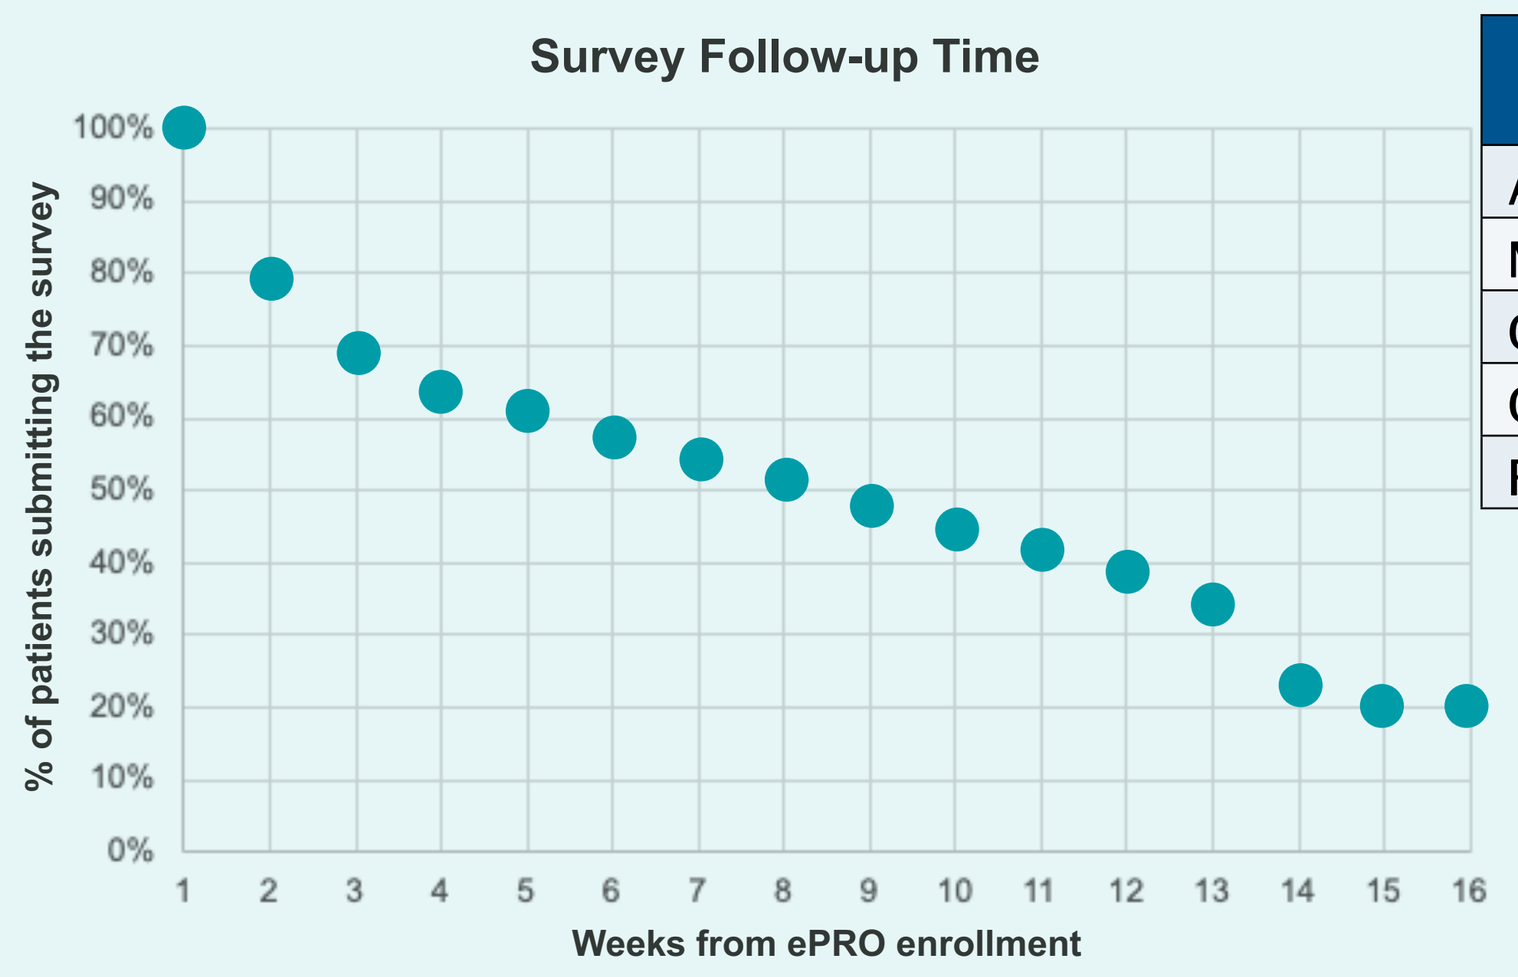
Survey Follow-up Time
% of patients submitting the survey
Weeks from ePRO enrollment

Summary statistics of survey compliance*	
Average (SD)	70% (32%)
Median	85%
Q1	37%
Q3	100%
Range (Min, Max)	8%, 100%

*Number of surveys completed over the number of surveys assigned per patient.

Figure 3: ePRO System Alerts Overview

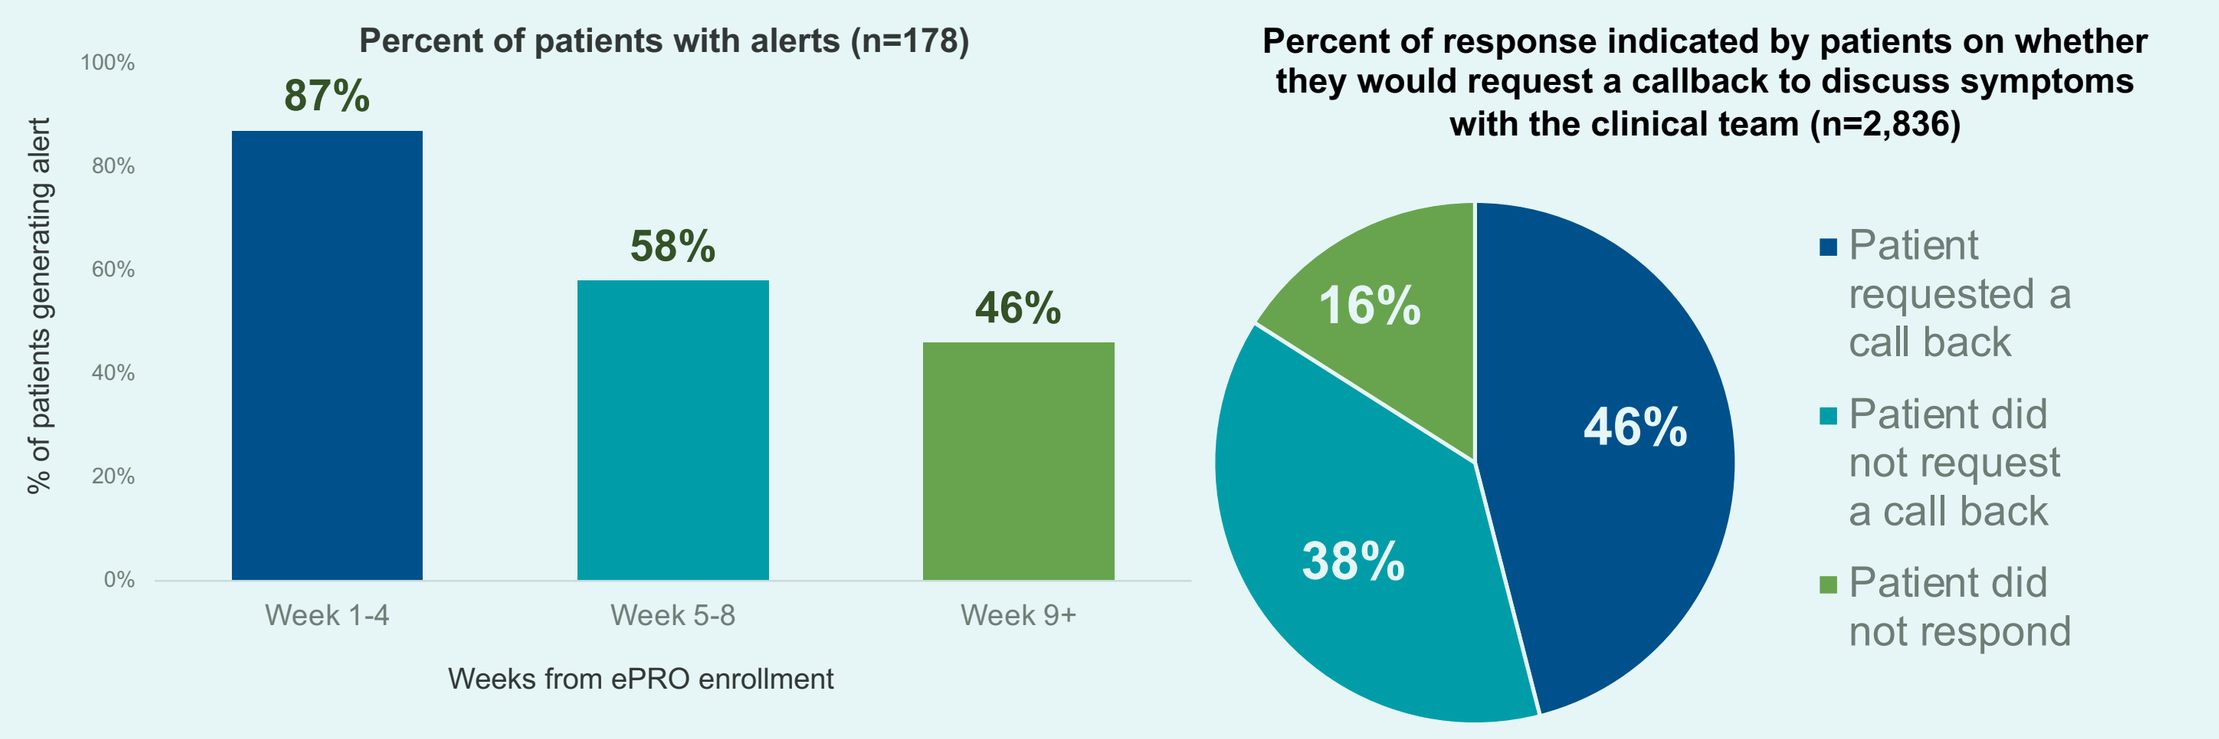
Percent of patients with alerts (n=178)
% of patients generating alert
87%
58%
46%
Week 1-4
Week 5-8
Week 9+
Weeks from ePRO enrollment
Percent of response indicated by patients on whether they would request a callback to discuss symptoms with the clinical team (n=2,836)
46%
38%
16%
Patient requested a call back
Patient did not request a call back
Patient did not respond

Symptoms with most frequent call back requested
69% depression/sadness, 57% anxiety 54% decreased appetite

KEY TAKEAWAY

It is feasible to integrate standardized ePRO measures into routine lung cancer care across multiple US cancer sites.

- More than half (58%) of domains (n=37) overlapped with the H2O COS. This included 15 clinical outcomes and 22 PRO domains (Figure 1a, 1b). Median completion rates of clinical and PRO domains were 60% and 98%, respectively.
- About 30% (n=19) of overlapping domains were measured longitudinally with a median completion of 76% (Figure 1c).

Figure 1: Overlapping H2O Domains with Carevive Data

A: H2O COS Clinical Domains (n=15)

Median data completeness: 60%

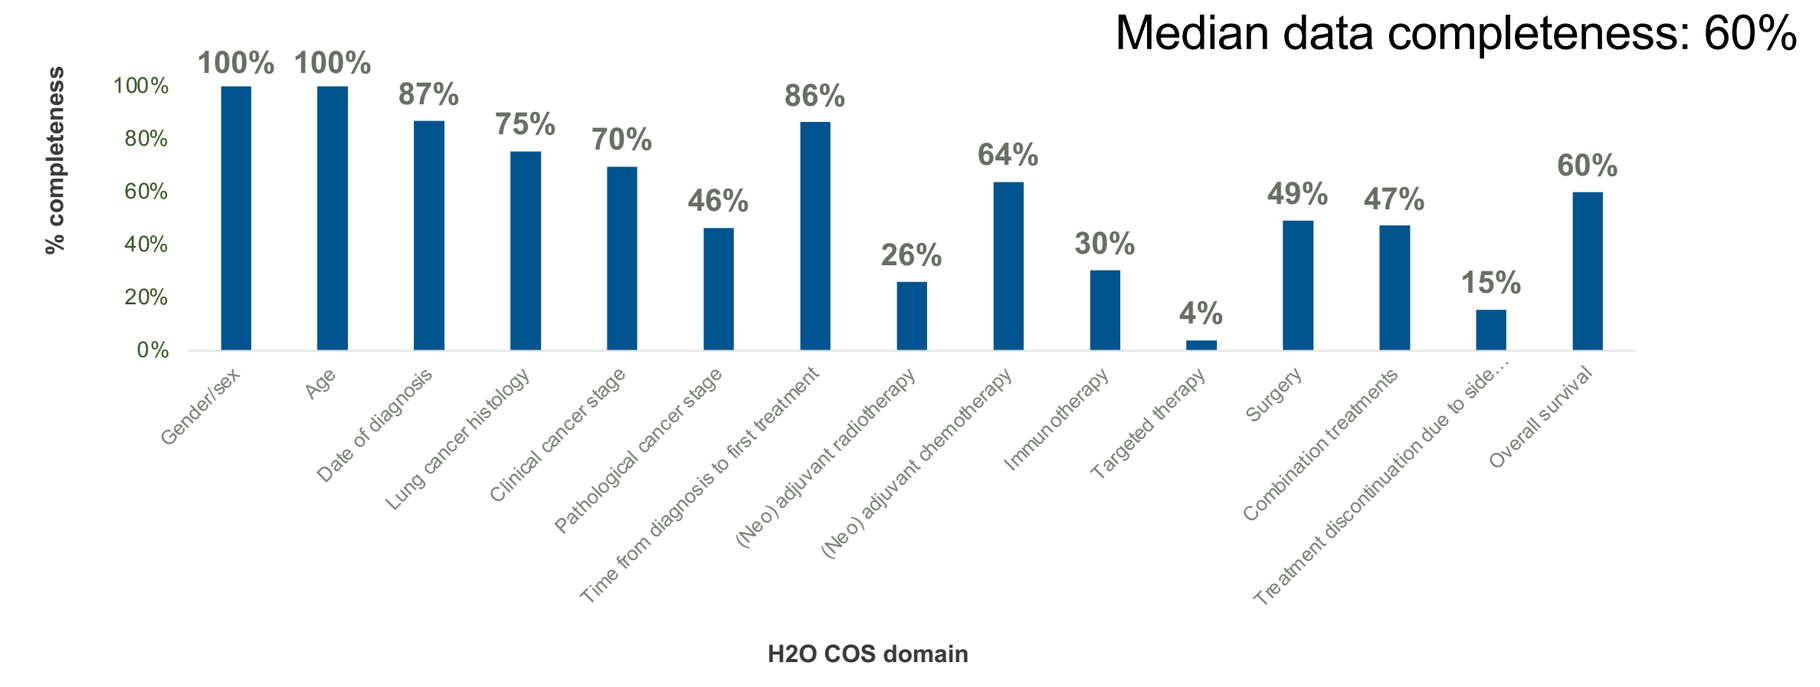
% completeness
Gender/sex 100%
Age 100%
Date of diagnosis 87%
Lung cancer histology 75%
Clinical cancer stage 70%
Pathological cancer stage 46%
Time from diagnosis to first treatment 86%
(Neo) adjuvant radiotherapy 26%
(Neo) adjuvant chemotherapy 64%
Immunotherapy 30%
Targeted therapy 4%
Surgery 49%
Combination treatments 47%
Treatment discontinuation due to side... 15%
Overall survival 60%
H2O COS domain

B: H2O COS PRO Domains (n=22)

Median data completeness: 98%

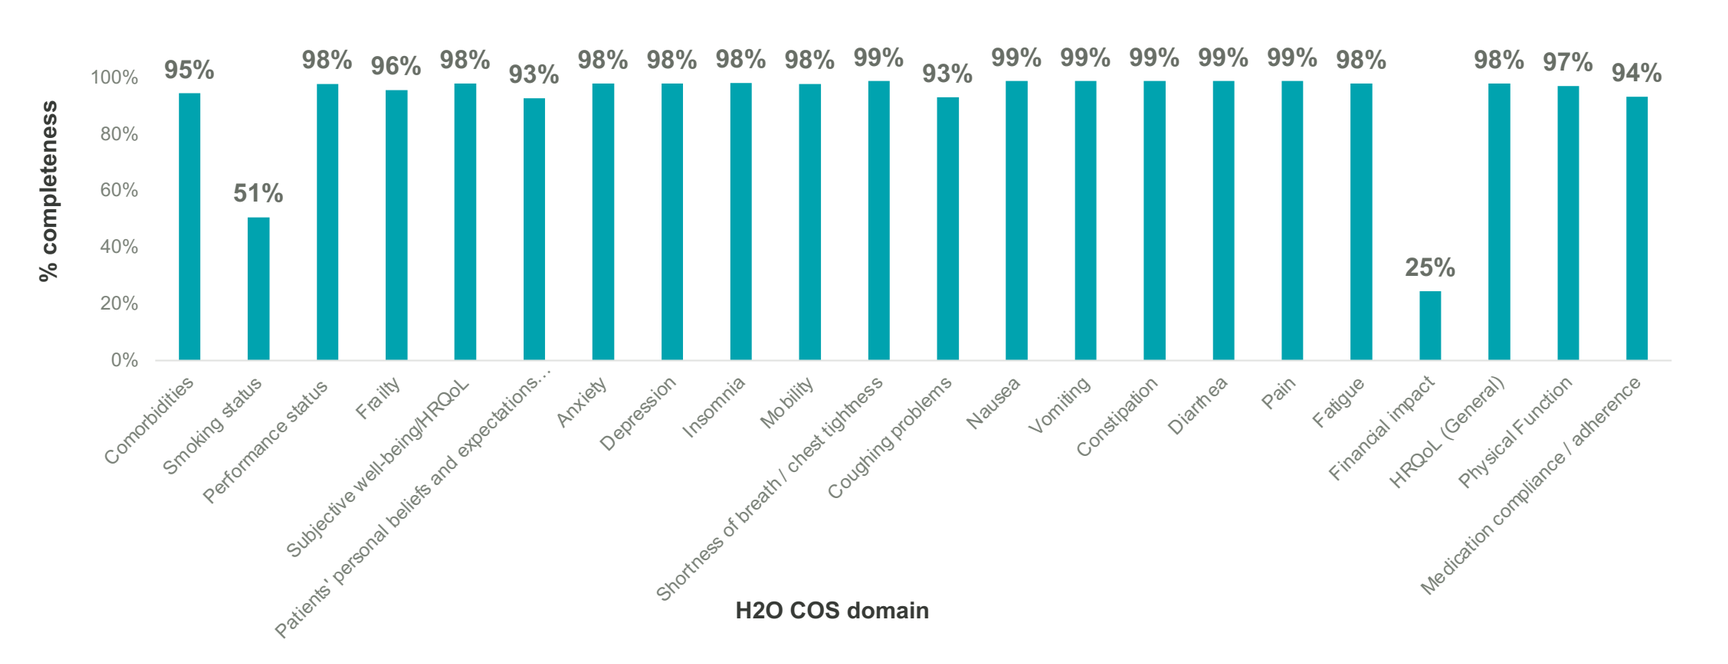
% completeness
Comorbidities 95%
Smoking status 51%
Performance status 98%
Frailty 96%
Subjective well-being/HRQoL 98%
Patients' personal beliefs and expectations... 93%
Anxiety 98%
Depression 98%
Insomnia 98%
Mobility 98%
Shortness of breath / chest tightness 99%
Coughing problems 93%
Nausea 99%
Vomiting 99%
Constipation 99%
Diarrhea 99%
Pain 99%
Fatigue 98%
Financial impact 25%
HRQoL (General) 98%
Physical Function 97%
Medication compliance / adherence 94%
H2O COS domain

C: H2O COS longitudinal PRO Domain (n=19)

Median data completeness: 76%

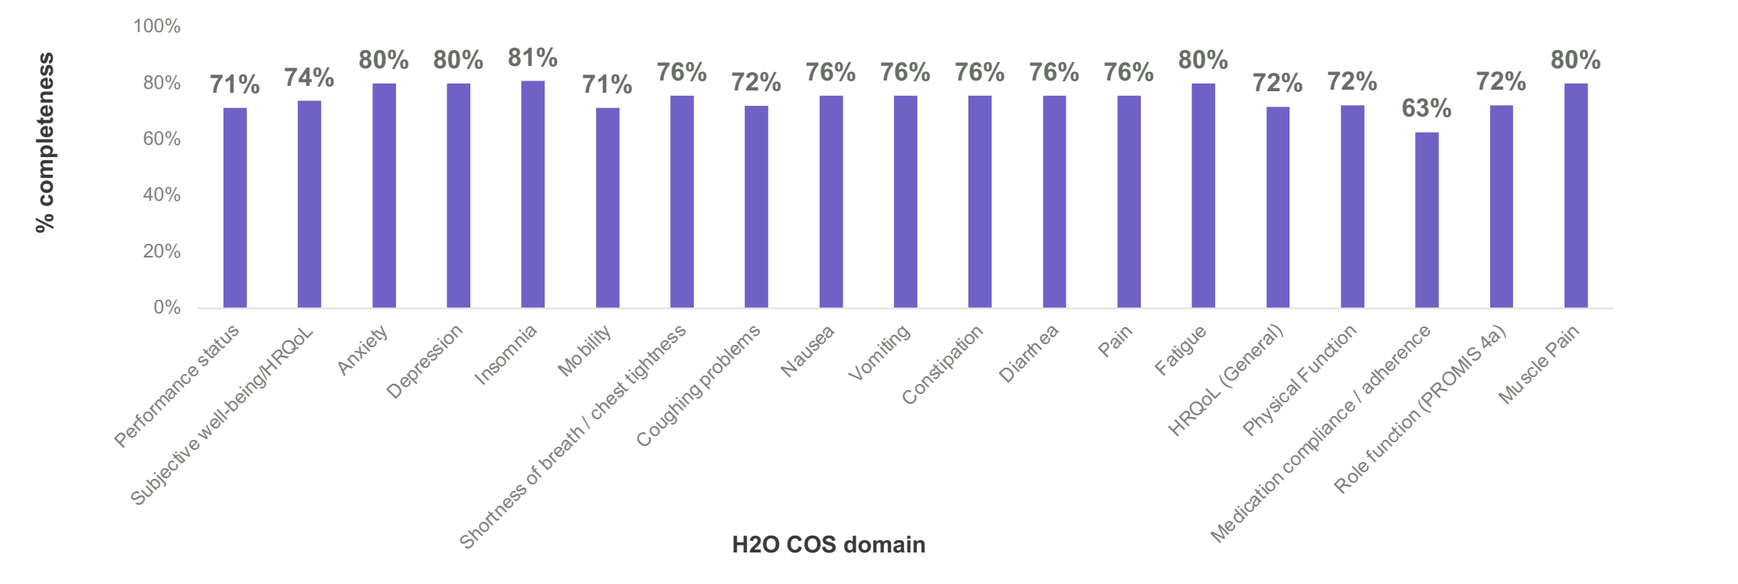
% completeness
Performance status 71%
Subjective well-being/HRQoL 74%
Anxiety 80%
Depression 80%
Insomnia 81%
Mobility 71%
Shortness of breath / chest tightness 76%
Coughing problems 72%
Nausea 76%
Vomiting 76%
Constipation 76%
Diarrhea 76%
Pain 76%
Fatigue 80%
HRQoL (General) 72%
Physical Function 72%
Medication compliance / adherence 63%
Role function (PROMIS 4a) 72%
Muscle Pain 80%
H2O COS domain

- Median clinical team response time to alerts (moderate and severe symptom reports) was 20 hours, with 69.3% addressed in less than 2 days (Figure 4).

Figure 4: Clinical team time to resolve ePRO system alerts

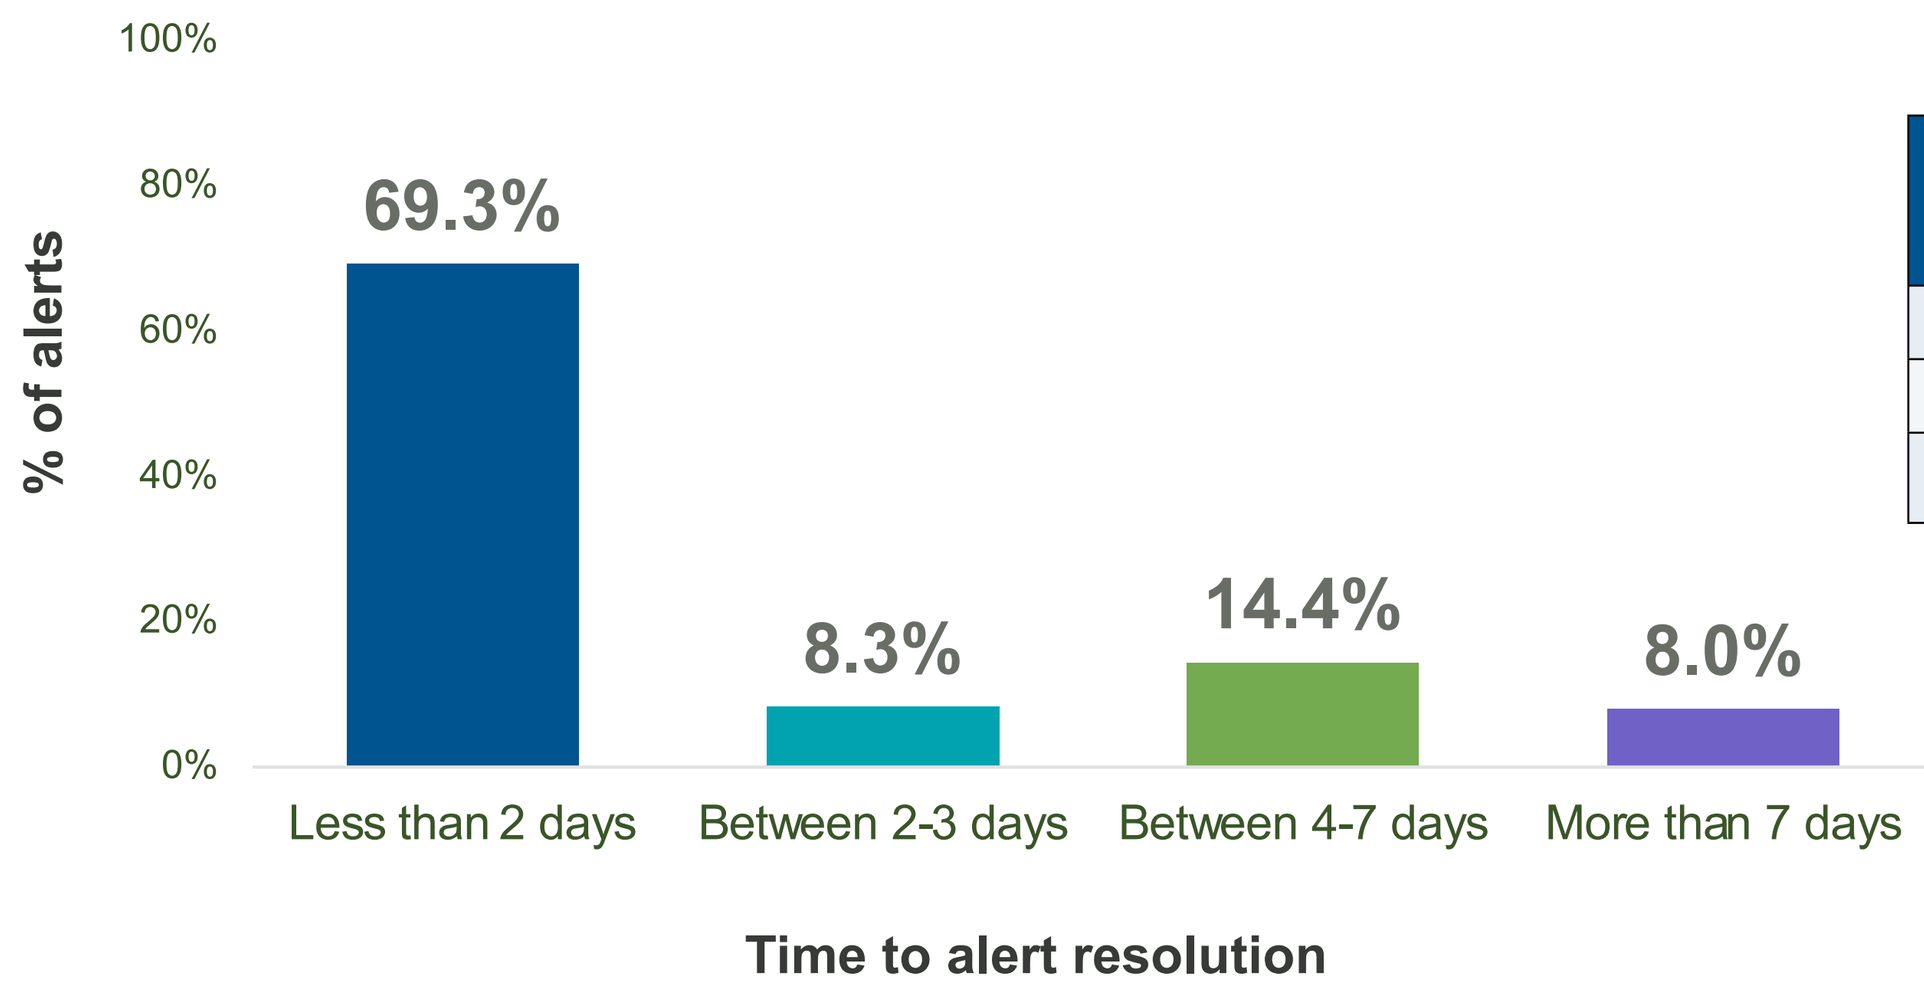
% of alerts
69.3%
8.3%
14.4%
8.0%
Less than 2 days
Between 2-3 days
Between 4-7 days
More than 7 days
Time to alert resolution

Clinical team time to resolve ePRO alert (days)	
Average (SD)	2.48 (7.78)
Median	0.84 (~20.2 hours)
Q1, Q3	0.15, 2.43

- Clinical professionals accessed the platform 2-3 times weekly, for median 2 minutes (range: 1-14 minutes) per login for nurses and Advanced Practice Nurses (APN). 89% of alerts between Week 1-4 indicated a response from nurses and APN (Figure 5).

Figure 5: Distribution of clinical professionals who responded to ePRO system alerts

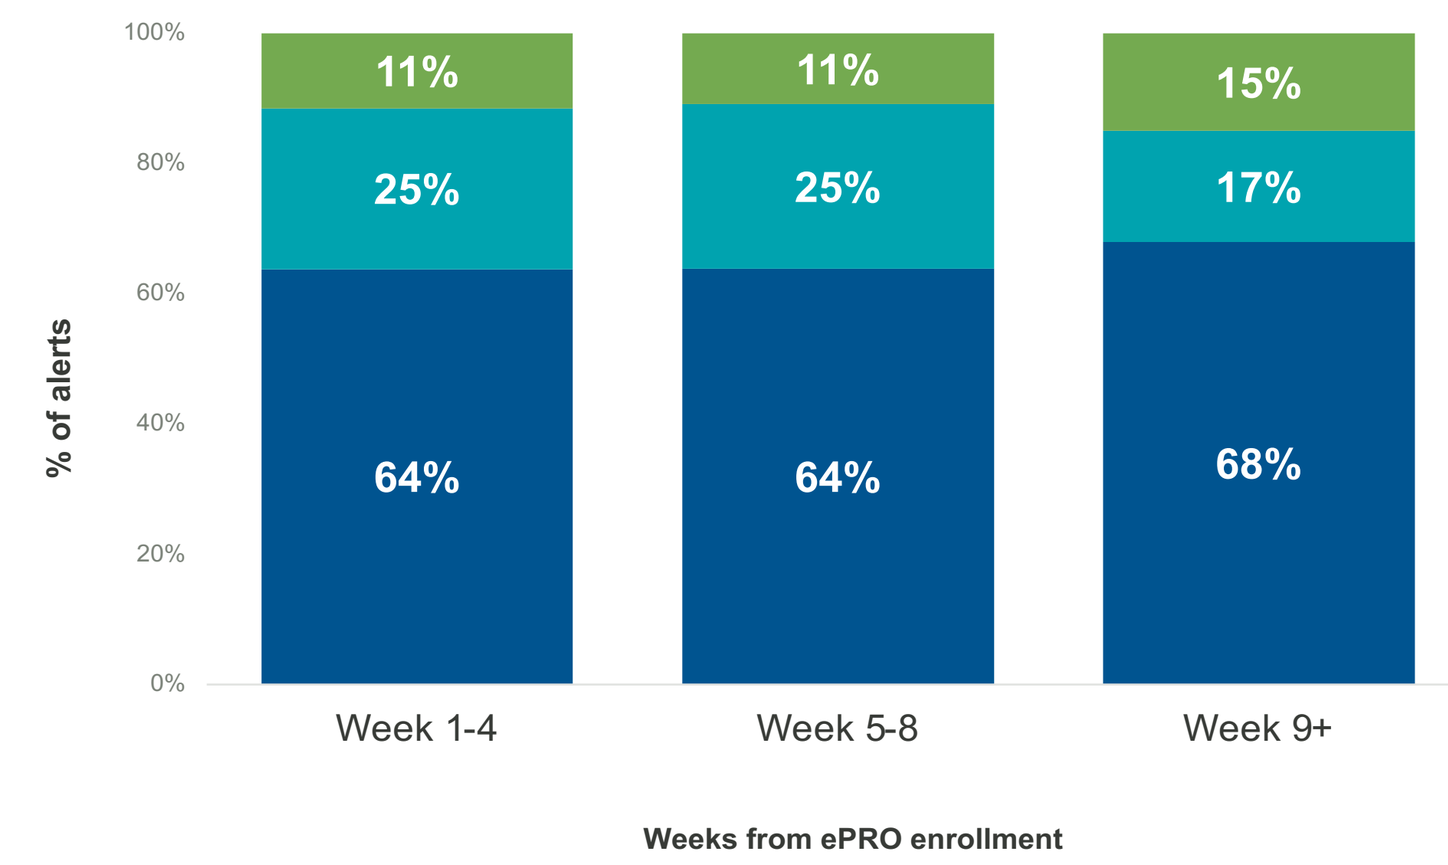
% of alerts
Week 1-4: 11% 25% 64%
Week 5-8: 11% 25% 64%
Week 9+: 15% 17% 68%
Weeks from ePRO enrollment
Non-Medical
Advanced Practice
Nurse

Non-medical team refers to oncology research coordinators. In this case, clinical response would have been made in collaboration and supervised by clinical providers (oncology nurse and/or physicians on the site).

Nurses and Advanced Practitioners' frequency of login/week to Carevive platform	
Average	2.3 times

Summary statistics of the time clinical professionals took to resolve ePRO alert (days)	
Median	2
Range (Min, Max)	1, 13.5

- Out of 3,476 alert responses made by clinical care team, 66% advised continuing treatment, 12% led to increased monitoring and were escalated to the clinical team, and 1% resulted in hospitalization or emergency room visits (Figure 6).

Figure 6: Clinical recommendations to ePRO system alerts

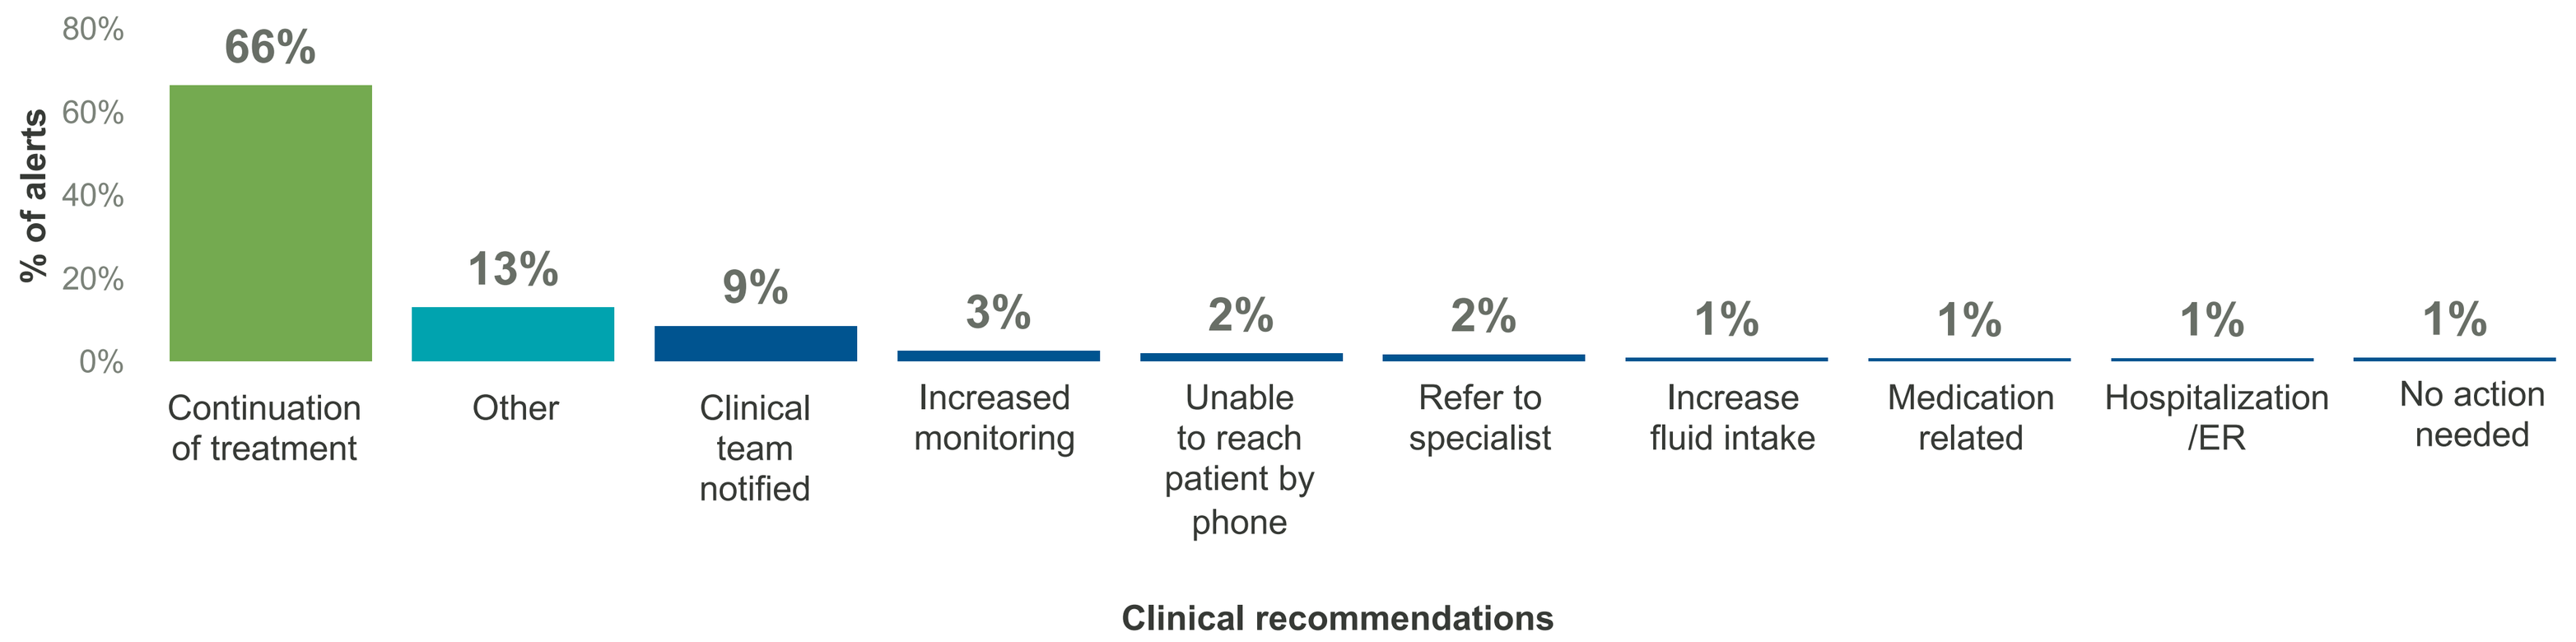
% of alerts
Continuation of treatment 66%
Other 13%
Clinical team notified 9%
Increased monitoring 3%
Unable to reach patient by phone 2%
Refer to specialist 2%
Increase fluid intake 1%
Medication related 1%
Hospitalization /ER 1%
No action needed 1%
Clinical recommendations

"Other" category may include: offered psychology services during the infusion process, CT chest PE protocol, etc.

CONCLUSION

- Results support the feasibility of integrating a standard set of multi-domain ePRO measures, comparable in breadth and scope to the H2O COS, into routine cancer care at multiple sites with consistent patient engagement and provider utilization.
- Findings support value of remote symptom alerts, when integrated into clinical workflow, for enhancing timeliness patient and care team communications and timely addressing of symptoms to optimize care.
- Patient engagement was high (>60%) in the first 5 weeks but decreased afterwards. Future study should investigate factors that may contribute to the decline.
- One limitation to the study is that clinical and financial impact were not directly assessed. Additionally, the small proportion of non-White, non-Hispanic/Latino patients may impact generalizability to these patients.
- Future research should explore direct clinical team and patient feedback on user experience, drivers of successful systematic implementation of PRO measures, and impacts on patient outcomes, resource utilization, and health systems.

1. Basch E, Deal AM, Kris MG, et al. Symptom monitoring with patient-reported outcomes during routine cancer treatment: A randomized controlled trial. *J Clin Oncol.* 2016; 34: 557-65.
2. Basch E, Reeve BB, Mitchell SA, Clauser SB, Minasian LM, Dueck AC, et al. Development of the National Cancer Institute's Patient-reported Outcomes Version of the Common Terminology Criteria for Adverse Events (PRO-CTCAE). *J Natl Cancer* Inst. 2014;106(9).
3. Basch E. Patient-reported outcomes— Harnessing patient'' voices to improve clinical are. *N Engl J Med*. 2017a. 376(2):105-108.
4. Basch E., Deal AM, Dueck, AC, et al. Overall survival results of a trial assessing patient-reported outcomes for symptom monitoring during routine cancer treatment. *JAMA*. 2017b. 318(2):197–198.
5. deRooij BH, van den Hurk C, Smaardijk V, et al. Development of an updated, standardized, patient-centered outcome set for lung cancer. *Lung Cancer* 2022. 173:5-13.

Disclosures
This study was sponsored by Takeda Pharmaceuticals.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® or the author of this poster.

Email for questions or comments Aaron Galaznik MD, **aaron.galaznik@carevive.com.** Presented at 2024 ASCO Annual Meeting, Chicago, USA, June 2024.