Treatment patterns and clinical outcomes among advanced NSCLC patients with *EGFR* exon 20 insertion mutations in China

Yan Wang[1], Huamao M. Lin[2], Ning Ning[3], Fen Du[4], Xin Gao[3], Qi Dong[2], Haiyan Xu[5], Yayi He[6], Caicun Zhou[6]

[1]Department of Medical Oncology, Cancer Hospital, Chinese Academy of Medical Sciences and Peking Union Medical College, Beijing, China; [2]Takeda Development Center Americas, Inc., Lexington, MA, USA; [3]OPEN Health, Bethesda, MD, USA; [4]OPEN Health, Shanghai, China; [5]Department of Comprehensive Oncology, Cancer Hospital, Chinese Academy of Medical Sciences and Peking Union Medical College, Beijing, China; [6]Department of Oncology, Shanghai Pulmonary Hospital, School of Medicine, Tongji University, Shanghai, China

WCLC
Singapore
EP10.01-17

Background

- In China, lung cancer is also the most common cancer with the incidence rate of 59.89 per 100,000 populations, leading to most cancer deaths with the mortality rate of 47.51 per 100,000 populations [1,2]
- Epidermal growth factor receptor (*EGFR*) is among the most important genes in non-small cell lung cancer (NSCLC), particularly in Asian populations where it is prevalent in up to 50% of patients [3,4]
- NSCLC patients with *EGFR* exon 20 insertion mutations (ex20Ins) have a poor prognosis with median overall survival ranging from 16.5 – 17.8 months [5,6,7]
- Mobocertinib is approved by FDA as the first oral therapy specifically designed for NSCLC patients with ex20Ins, and was recently approved by the China's National Medical Products Administration (NMPA) in 2023 [8,9]
- There are very few studies on real-world clinical efficacy and safety of available treatments and the unmet needs for NSCLC patients with ex20Ins in China

Methods

Patient sample

- Inclusion criteria
 - Patients aged ≥ 18 years at advanced NSCLC diagnosis
 - Had a pathologically and/or cytologically confirmed diagnosis of stage IIIB/IV NSCLC prior to June 1, 2019 (either primary or after relapse of initial non-metastatic disease)
 - Had a detection of an ex20Ins through genomic sequencing
 - Had documentation available for at least two visits
- Patient sampling
 - All eligible patients were consecutively entered into the study starting from the months prior to June 1, 2019
 - Enrolling consecutive eligible patients in each month prior to June 2019
 - Patients were followed from the diagnosis to the end of data availability or end of the study period (Jan 2022), whichever came first

Study outcomes

- Patient demographics, clinical characteristics, and treatment patterns
- Real-world progression-free survival (PFS) for each line of therapy (LOT)
- Confirmed objective response rates (ORR): defined as percentages of patients with 2 complete responses (CRs) or partial responses (PRs), as confirmed by the study lead clinicians

Statistical analysis

- Descriptive analyses were conducted to describe patient demographics and clinical characteristics, treatment patterns, and clinical outcomes
- Kaplan-Meier analyses were performed to estimate real-world PFS for each LOT

Results

Demographic and clinical characteristics (Table 1)

- 116 patients were included: 70 patients from SPH and 46 from CICAMS
- Average age of patients was 56.2 years (range 27.8 to 76.1)
- 46.6% were women
- 58.6% had intrathoracic metastases and 30.2% had brain metastases

Study objective

To evaluate real-world data on treatment patterns and clinical outcomes for advanced non-small cell lung cancer (NSCLC) patients with epidermal growth factor receptor (*EGFR*) exon 20 Insertion mutations (ex20Ins) in China

Study design

A retrospective chart review study was undertaken at 2 large state-level cancer hospitals in China:

- **Shanghai Pulmonary Hospital (SPH)**
- **Cancer Hospital, Chinese Academy of Medical Sciences and Peking Union Medical College in Beijing (CICAMS)**

Results

Treatment characteristics and Clinical outcomes

- All patients received 1st line of therapy (LOT)
 - 99 (85.3%) patients initiated 1st line platinum-based chemotherapy
 - 3 (2.6%) patients initiated EGFR tyrosine kinase inhibitors (TKIs)
- Among patients who received platinum-based chemotherapy as 1st LOT (N=99), only 59.6% of patients continued with 2nd LOT (N=59)
- Kaplan-Meier analyses showed
 - Median real-world PFS of 1st LOT was 6.2 months
 - Median real-world PFS of 2nd LOT was 4.2 months for patients with post platinum-based treatments
- Confirmed objective response rates
 - 14.8% (N=13) for patients with 1st LOT
 - 10.0% (N=3) for patients with post-platinum 2nd LOT

Figure 1. Real-world PFS of 1st LOT (N=116)

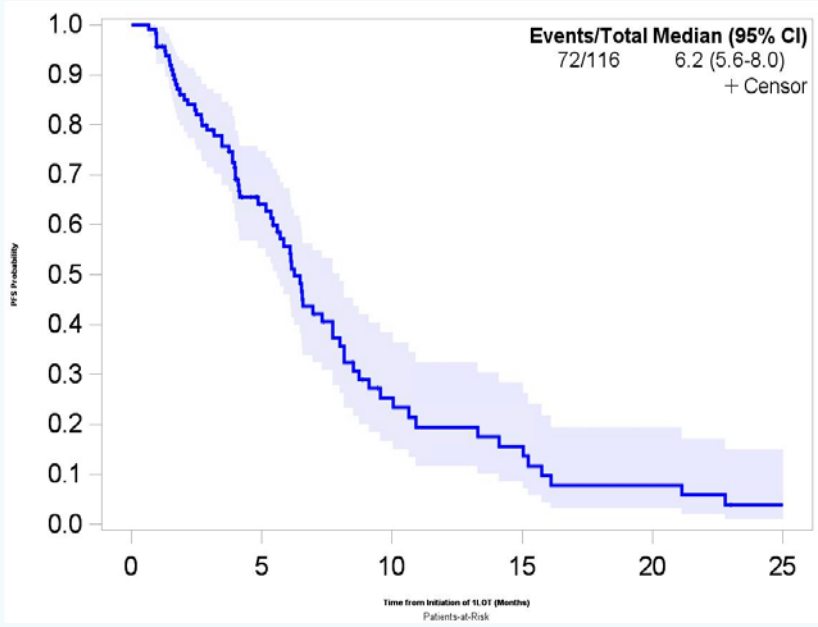

Events/Total Median (95% CI)
72/116 6.2 (5.6-8.0)
+ Censor
1.0
0.9
0.8
0.7
0.6
0.5
0.4
0.3
0.2
0.1
0.0
0
5
10
15
20
25

Figure 2. Real-world PFS of 2nd LOT for patients who used platinum therapy as 1st LOT (N=59)

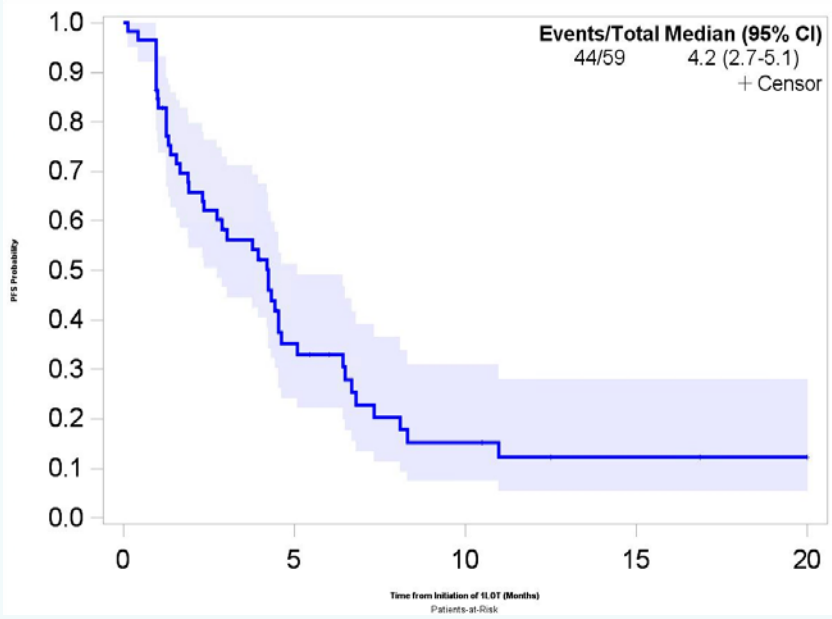

Events/Total Median (95% CI)
44/59 4.2 (2.7-5.1)
+ Censor
1.0
0.9
0.8
0.7
0.6
0.5
0.4
0.3
0.2
0.1
0.0
0
5
10
15
20

Key Take Aways

- **This study evaluated previous treatment patterns and clinical outcomes for advanced NSCLC patients with ex20Ins in China, addressing a critical gap in existing knowledge of those patients in real-world**
- **The findings of this study revealed that the prognosis of advanced NSCLC patients with *EGFR* ex20ins in China was poor, highlighting the unmet need for novel therapies in this patient population**
- **Future real-world data are warranted to evaluate the effectiveness of approved novel target therapy in China**

Table 1. Demographic and clinical characteristics of advanced NSCLC patients in the study

Demographic and clinical information	Total (N=116)
Site, % (n)	
SPH	60.3% (70)
CICAMS	39.7% (46)
Age, year	
Mean	56.2
Median	57.6
Range	27.8 – 76.1
Gender, % (n)	
Female	46.6% (54)
Male	53.4% (62)
Site of metastases, % (n)	
Intrathoracic	58.6% (68)
Brain metastases	30.2% (35)
Insurance type, % (n)	
Urban medical insurance	59.5% (69)
Rural medical insurance	9.5% (11)
Smoking status, % (n)	
Current	18.1% (21)
Previous	13.8% (16)
Non-smoker	62.1% (72)
Cancer stage at the time of diagnosis, % (n)	
IIIb	3.5% (4)
IVa	53.5% (62)
IVb	37.9% (44)
Not specified	5.2% (6)
ECOG at diagnosis, % (n)	
0	7.8% (9)
1	45.7% (53)
Not reported	46.6% (54)
Tested for PD-L1, % (n)	
Positive	5.7% (7)
Negative	3.5% (4)
Unknown	90.5% (105)

Table 2. Treatment patterns of advanced NSCLC patients

Treatments	Total (N=116)
Patients with LOTs, % (n)	
1st LOT	100% (116)
2nd LOT	57.8% (67)
Post-platinum 2nd LOT	50.9% (59)
1st LOT (N=116), % (n)	
Platinum-based chemotherapy	85.3% (99)
EGFR TKI	2.6% (3)
Immunotherapy	0.9% (1)
Other	11.2% (13)
2nd LOT (N=59), % (n)	
Platinum-based chemotherapy	18.6% (11)
EGFR TKI	13.6% (8)
Immunotherapy	3.4% (2)
Other	64.4% (38)
Docetaxel	23.7% (14)
Bevacizumab-based chemotherapy	16.9% (10)
Paclitaxel	8.5% (5)
Other	16.9% (10)

Table 3. Clinical outcomes of the advanced NSCLC patients

Clinical outcomes	Total (N=116)
TTD of 1st LOT	
Median in Months (95% CI)	4.0 (3.7, 4.8)
PFS of 1st LOT	
Median in Months (95% CI)	6.2 (5.6, 8.0)
Censored, % (n)	37.9% (44)
TTD of post-platinum 2nd LOT	
Median in Months (95% CI)	2.9 (1.9, 4.5)
PFS of post-platinum 2nd LOT	
Median in Months (95% CI)	4.2 (2.7, 5.1)
Censored, % (n)	25.4% (15)
Confirmed ORR, % (n)	
1st LOT (N=116)	14.8% (13)
Post-platinum 2nd LOT (N=59)	10.0% (3)

Discussion

Strengths of the study

- The study was conducted in two large state-level hospitals, ensuring that the findings are representative of the advanced NSCLC patient population with *EGFR* ex20ins in China, given the hospitals' extensive medical background and patient behaviors
- The study employed robust and validated methods to efficiently collect retrospective data, leveraging the best available data sources in China

Limitations of the study

- The study relied solely on data recorded in medical records, which may have resulted in missing data
- Complete death information was lacking from the current medical charts in China, which could have impacted the assessment of clinical outcomes
- Due to the nature of a chart review study, all clinical outcomes are subject to potential measurement errors

References

1. Siegel et al. CA Cancer J Clin. 2023;73(1):17-48
2. Zheng et al. Zhonghua Zhong Liu Za Zhi. 2023;45(3):212-220
3. Pi et al. Thorac Cancer. 2018;9(7):814-819
4. Zhang et al. Transl Lung Cancer Res. 2019;8(3):302-316.
5. Yang et al. Lung Cancer. 2021;152:39-48
6. Ou et al. American Society of Clinical Oncology. 2021
7. Christopoulos et al. European Lung Cancer Virtual Congress. 2021
8. Mobocertinib Approved by U.S. FDA. 2021
9. Mobocertinib Approved by China NMPA. 2023

Acknowledgments

The authors would like to thank all the patients who provided the electronic medical records and the data scientists who collected charts for this study. Additionally, the authors thank Christina DuVernay from OPEN Health for her editorial assistance as funded by Takeda, MA, US.

Abbreviations

ECOG, Eastern Cooperative Oncology Group; PD-L1, Programmed Cell Death Ligand 1; SPH, Shanghai Pulmonary Hospital; CICAMS, Cancer Hospital, Chinese Academy of Medical Sciences and Peking Union Medical College; EGFR, Epidermal Growth Factor Receptor; LOT, line of treatment; TKI, Tyrosine Kinase Inhibitors; CI, confidence interval; PFS, progression-free survival; TTD, time to treatment discontinuation.

Disclosures

This study is sponsored by Millennium Pharmaceuticals, Inc (Takeda), MA, US. YW: honoraria from Millennium Pharmaceuticals, Inc (Takeda), MA, US; employment with Chinese Academy of Medical Sciences and Peking Union Medical College; HML and QD: employment with Takeda, MA, US; XG, NN, FD: employment with OPEN Health, with funding from Millennium Pharmaceuticals, Inc (Takeda), MA, US.

Poster ID: #1903; Presenter Name: Yan Wang; Institution: Cancer Hospital, Chinese Academy of Medical Sciences and Peking Union Medical College; Country: China