P1.23-01

ctDNA positivity is a clinical prognostic marker in patients with *EGFR* ex20ins+ NSCLC

Sampurna Chatterjee,[1] Xin Tong,[1] Zhenqiang Su,[1] Ying Li,[1] Minakshi Guha,[1] Eric N Churchill,[1] Robert J Fram,[1] Sylvie Vincent[1]

[1]Takeda Development Center Americas, Inc., Lexington, MA, USA

Background

- Exon 20 insertion (ex20ins) mutations in the epidermal growth factor receptor (*EGFR*) gene occur in approximately 1%–3% of all patients with non–small cell lung cancer (NSCLC)[1]
- Mobocertinib is a potent, irreversible, oral EGFR tyrosine kinase inhibitor that selectively targets *EGFR* ex20ins mutations[2,3] **(Figure 1)**

Figure 1: Mobocertinib mechanism of action

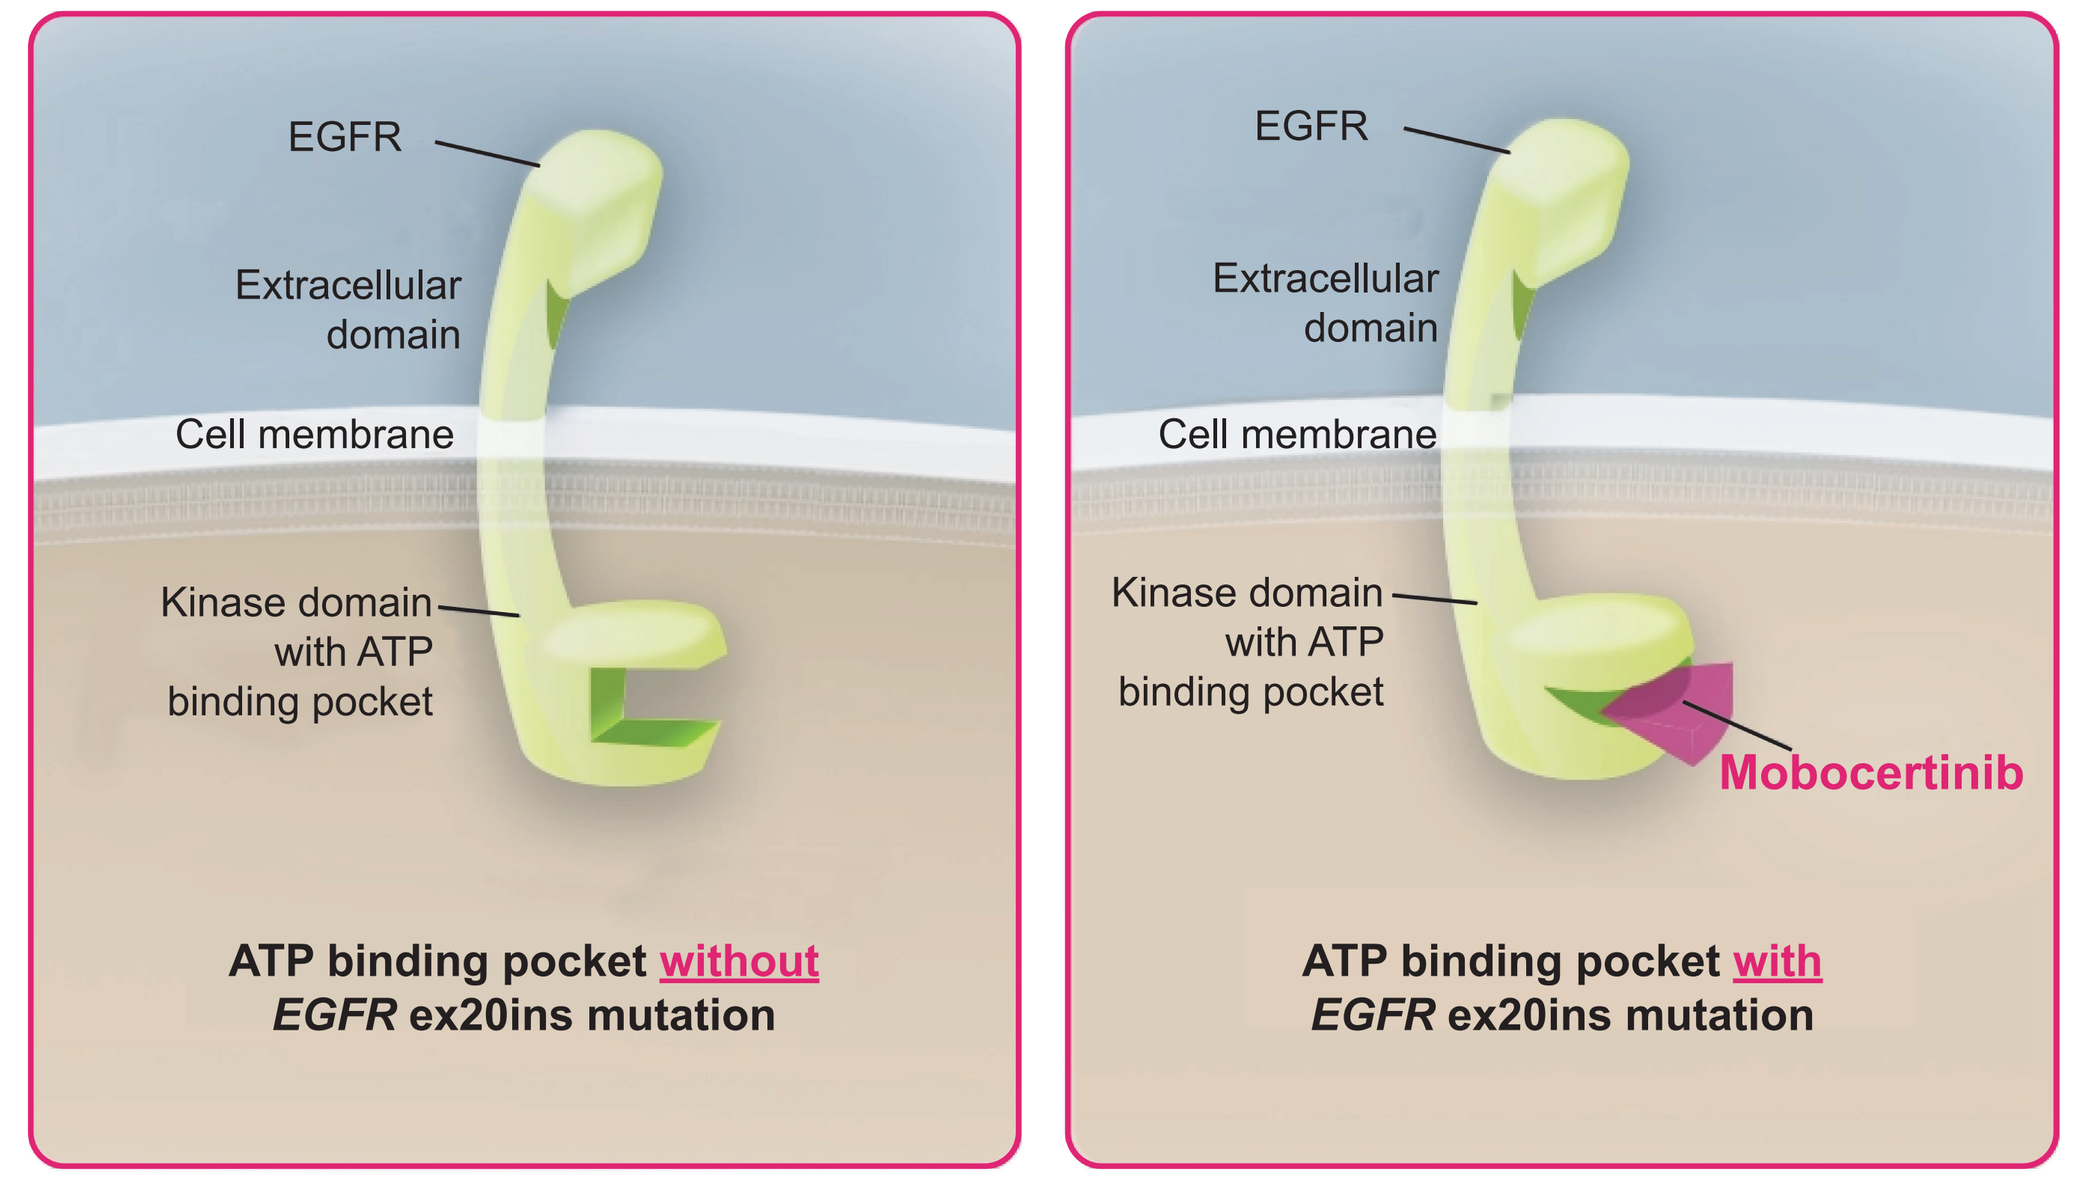
EGFR
Extracellular domain
Cell membrane
Kinase domain with ATP binding pocket
ATP binding pocket without
EGFR ex20ins mutation
EGFR
Extracellular domain
Cell membrane
Kinase domain with ATP binding pocket
Mobocertinib
ATP binding pocket with
EGFR ex20ins mutation

ATP, adenosine triphosphate

- Mobocertinib is approved for patients with *EGFR* ex20ins+ locally advanced or metastatic NSCLC whose disease has progressed on or after platinum-based chemotherapy[4] based on findings from a phase 1/2 trial (NCT02716116)[5,6]
 - At the November 1, 2021 data cutoff date, treatment with mobocertinib 160 mg/day once daily (qd) resulted in an independent review committee (IRC)–assessed overall response rate of 28% (95% confidence interval [CI]: 20%–37%) and a median duration of response of 15.8 months (95% CI: 7.4–19.4)[6]
 - Median progression-free survival (PFS) was 7.3 months (95% CI: 5.5–9.2),[6] and median overall survival was 20.2 months (95% CI: 14.9–25.3)
- Circulating tumor DNA (ctDNA) is a molecular predictor of response, treatment resistance, survival, and relapse in various cancers, including NSCLC[7–9]
- In a prior analysis, patients in the phase 1/2 trial of mobocertinib who had detectable *EGFR* ex20ins ctDNA at baseline (n=55) demonstrated a confirmed overall response rate (ORR) of 34.5%, which was comparable to the ORR of 28% observed among all platinum-pretreated patients (n=114) in the trial[10]
- The prognostic relevance of ctDNA in patients with *EGFR* ex20ins+ NSCLC requires further investigation
- This study evaluated the impact of baseline ctDNA positivity on PFS in patients with *EGFR* ex20ins+ NSCLC treated with mobocertinib after prior platinum-based chemotherapy

Objective

- To explore the dynamics of *EGFR* ex20ins ctDNA and its association with PFS in patients with *EGFR* ex20ins+ NSCLC treated with mobocertinib after prior platinum-based chemotherapy

Methods

- ctDNA was assessed in plasma samples from patients with *EGFR* ex20ins+ locally advanced or metastatic NSCLC treated with prior platinum-based chemotherapy who received once-daily oral mobocertinib (160 mg) in a phase 1/2 trial (NCT02716116; data cutoff: November 1, 2021; **Figure 2**)

Figure 2: Analysis of ctDNA by next-generation sequencing

1 Plasma collection (n=77) — Baseline →
- **Pretreatment (baseline) plasma samples were obtained**
- **ctDNA was analyzed using the FoundationOne Liquid CDx assay**

2 Plasma collection (n=60) — C3D1 →
- **Posttreatment plasma samples were obtained at C3D1 of mobocertinib treatment**
- **ctDNA was analyzed using the Illumina TruSight Oncology 500 ctDNA assay**

C3D1, Cycle 3, Day 1

- Detection of *EGFR* ex20ins ctDNA was based on the variant allele frequency (VAF)
 - Limit of detection was 0.5%
 - Change was categorized as:
 - Increase: change >0
 - Decrease: change <0
 - Not detected at baseline and at C3D1 (NDND)
- Total clearance was defined as *EGFR* ex20ins+ VAF not detected by C3D1
- To determine PFS, patients in the platinum-pretreated patient (PPP) cohort (N=114) were assessed per Response Evaluation Criteria In Solid Tumors (RECIST) version 1.1 criteria
- The association between the change in ctDNA from baseline and IRC-assessed PFS was evaluated

Results

- There was a significant association between detection of *EGFR* ex20ins ctDNA at baseline and PFS **(Figure 3)**
 - Patients with no *EGFR* ex20ins ctDNA detected at baseline had longer median PFS than those with *EGFR* ex20ins ctDNA detected (14.69 vs 5.52 months, respectively; hazard ratio [HR]=0.432 [95% CI: 0.219–0.854]; log-rank *P*=0.0157)
 - Patients with no *EGFR* ex20ins ctDNA detected at baseline had a 43.2% lower risk of disease progression than patients with *EGFR* ex20ins ctDNA detected

Figure 3: Association between PFS and *EGFR* ex20ins ctDNA detected at baseline

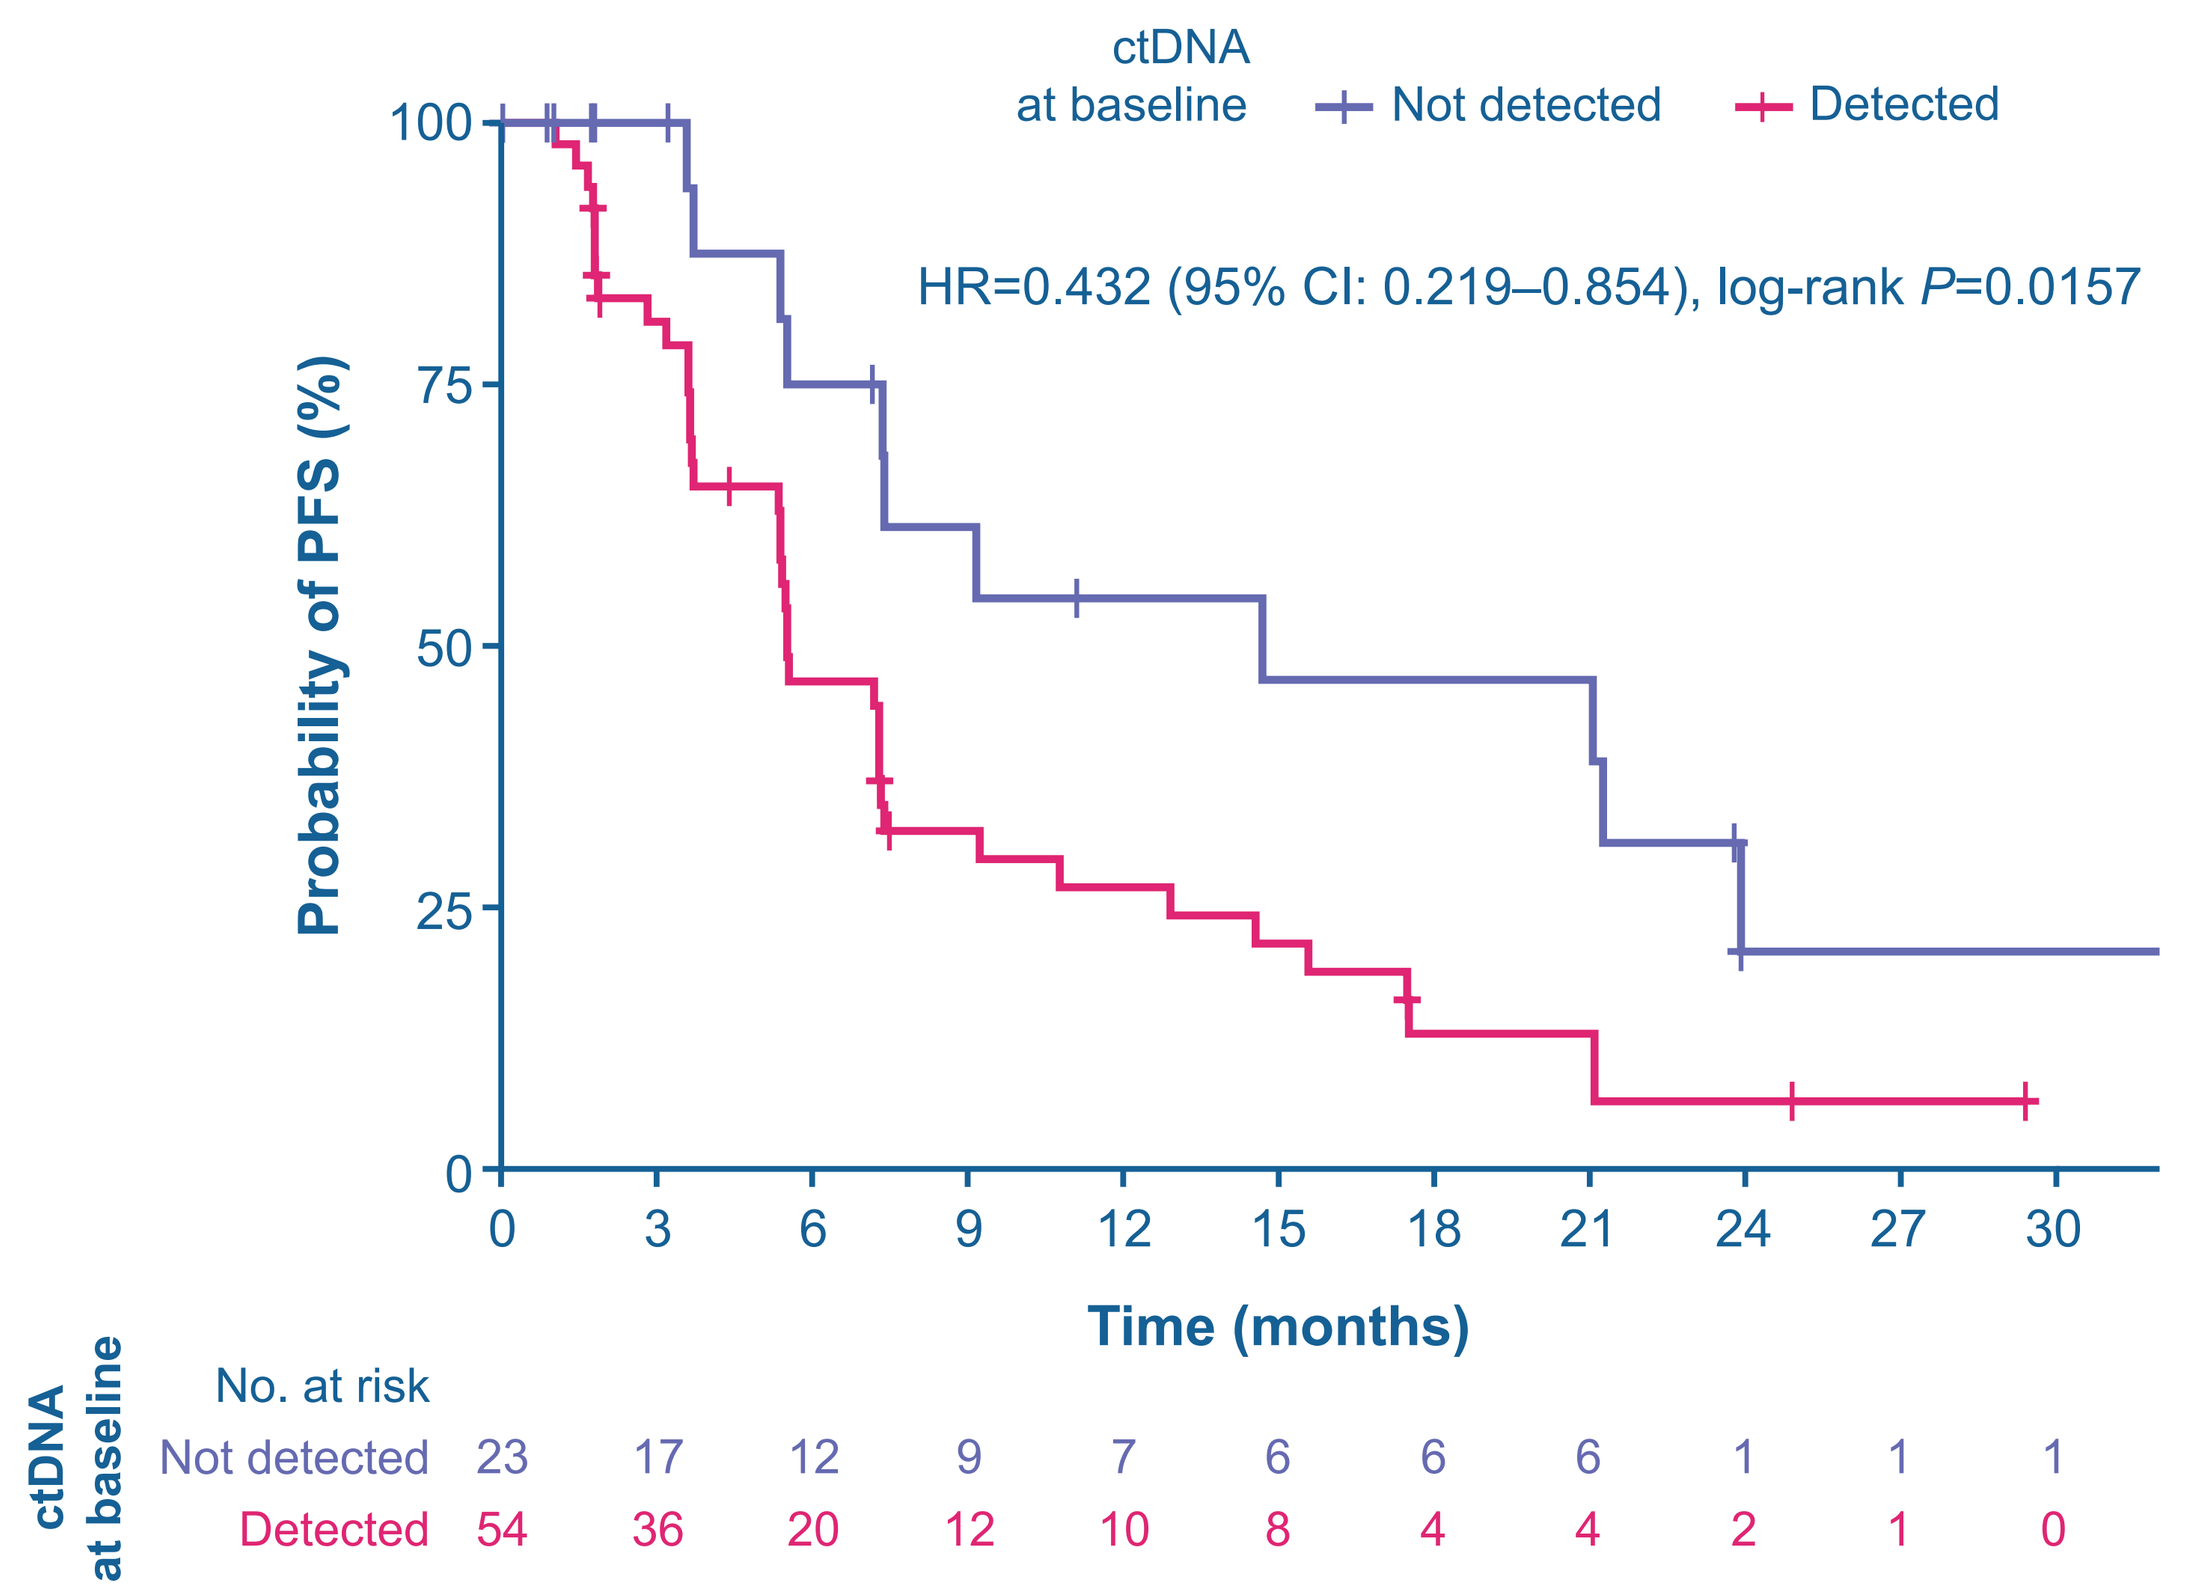
ctDNA at baseline
Not detected
Detected
HR=0.432 (95% CI: 0.219–0.854), log-rank P=0.0157
Probability of PFS (%)
100
75
50
25
0
0 3 6 9 12 15 18 21 24 27 30
Time (months)
ctDNA at baseline
No. at risk
Not detected 23 17 12 9 7 6 6 6 1 1 1
Detected 54 36 20 12 10 8 4 4 2 1 0

PPP Cohort (N=114)	Events (n)	Median PFS, months (95% CI)
All patients		**7.39 (5.52–14.69)**
ctDNA not detected	**12 (n=23)**	**14.69 (7.36–NR)**
ctDNA detected	**39 (n=54)**	**5.52 (5.39–7.39)**

NR, not reached

Question

What is the impact of *EGFR* ex20ins ctDNA status on PFS in patients with *EGFR* ex20ins+ NSCLC treated with mobocertinib after prior platinum chemotherapy?

Investigation

PPP cohort *EGFR* ex20ins+ NSCLC (N=114) → **Mobocertinib 160 mg oral qd** → ***EGFR* ex20ins ctDNA assessment baseline versus C3D1** → **PFS assessment**

Results

- A significant association was observed between the clearance of *EGFR* ex20ins ctDNA by C3D1 and PFS
- Patients with *EGFR* ex20ins ctDNA clearance by C3D1 had longer PFS than patients without *EGFR* ex20ins ctDNA clearance

Association between *EGFR* ex20ins ctDNA clearance at C3D1 of mobocertinib treatment and PFS

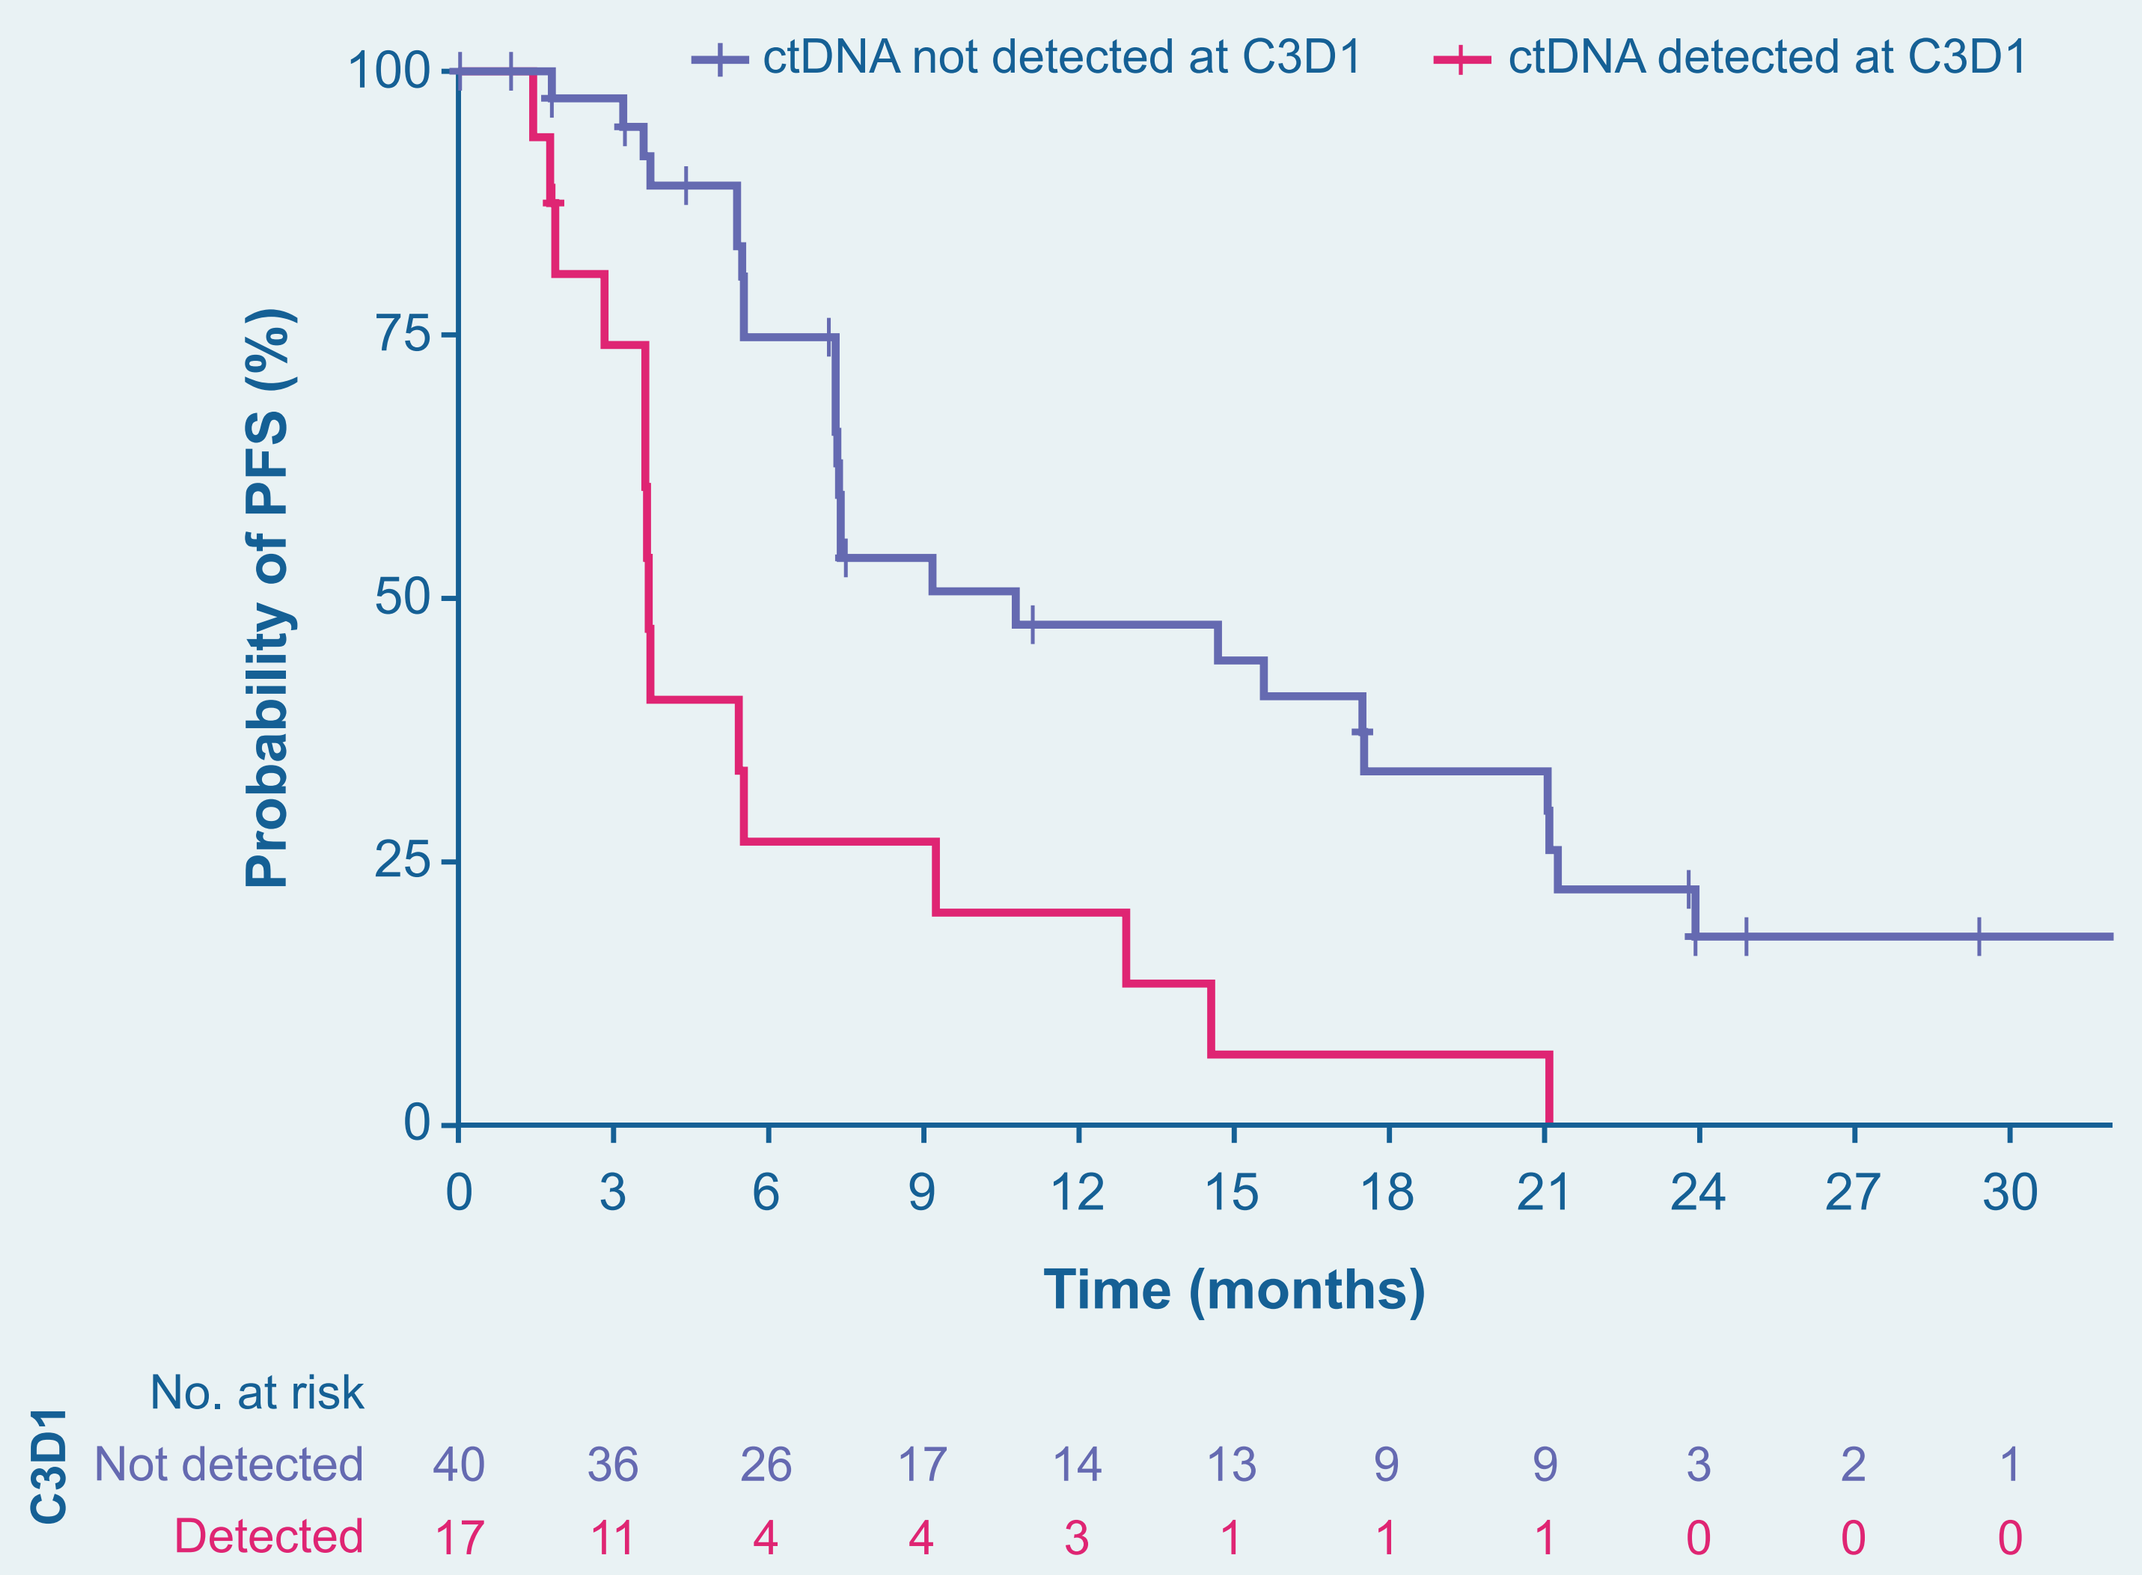
ctDNA not detected at C3D1
ctDNA detected at C3D1
Probability of PFS (%)
100
75
50
25
0
0 3 6 9 12 15 18 21 24 27 30
Time (months)
C3D1
No. at risk
Not detected 40 36 26 17 14 13 9 9 3 2 1
Detected 17 11 4 4 3 1 1 1 0 0 0

ctDNA detected versus not detected:
HR=0.317 (95% CI: 0.164–0.612);
log-rank *P*=0.0006

PPP Cohort (N=114)	Events (n)	Median PFS, months (95% CI)
All patients		**7.39 (5.52–14.69)**
ctDNA not detected at C3D1[a]	**27 (n=40)**	**10.78 (7.32–21.09)**
ctDNA detected at C3D1	**15 (n=17)**	**3.68 (3.61–12.91)**

[a]Total clearance was defined as *EGFR* ex20ins VAF not detected by C3D1

Key Takeaway

Patients with *EGFR* ex20ins ctDNA clearance at C3D1 of mobocertinib treatment achieved longer PFS than patients who had persistent *EGFR* ex20ins ctDNA or had an increase in *EGFR* ex20ins ctDNA

- Patients who had an increase in *EGFR* ex20ins VAF by C3D1 had a higher risk of disease progression than those who had a decrease in *EGFR* ex20ins VAF **(Figure 4)**
 - Patients with an increase in *EGFR* ex20ins ctDNA by C3D1 had a shorter mean PFS than those who remained negative for *EGFR* ex20ins ctDNA from baseline to C3D1 (4.53 vs 21.06 months; HR=10.46; 95% CI [2.60–42.05]; log-rank *P*=0.0009)
 - Patients with an increase in *EGFR* ex20ins VAF by C3D1 had a shorter mean PFS than those with a decrease in *EGFR* ex20ins ctDNA VAF (3.65 vs 7.29 months; HR= 3.69 [95% CI: 1.03–13.27]; log-rank *P*=0.045)

Figure 4: Association between change in median PFS and in *EGFR* ex20ins ctDNA status from baseline to C3D1 of mobocertinib treatment

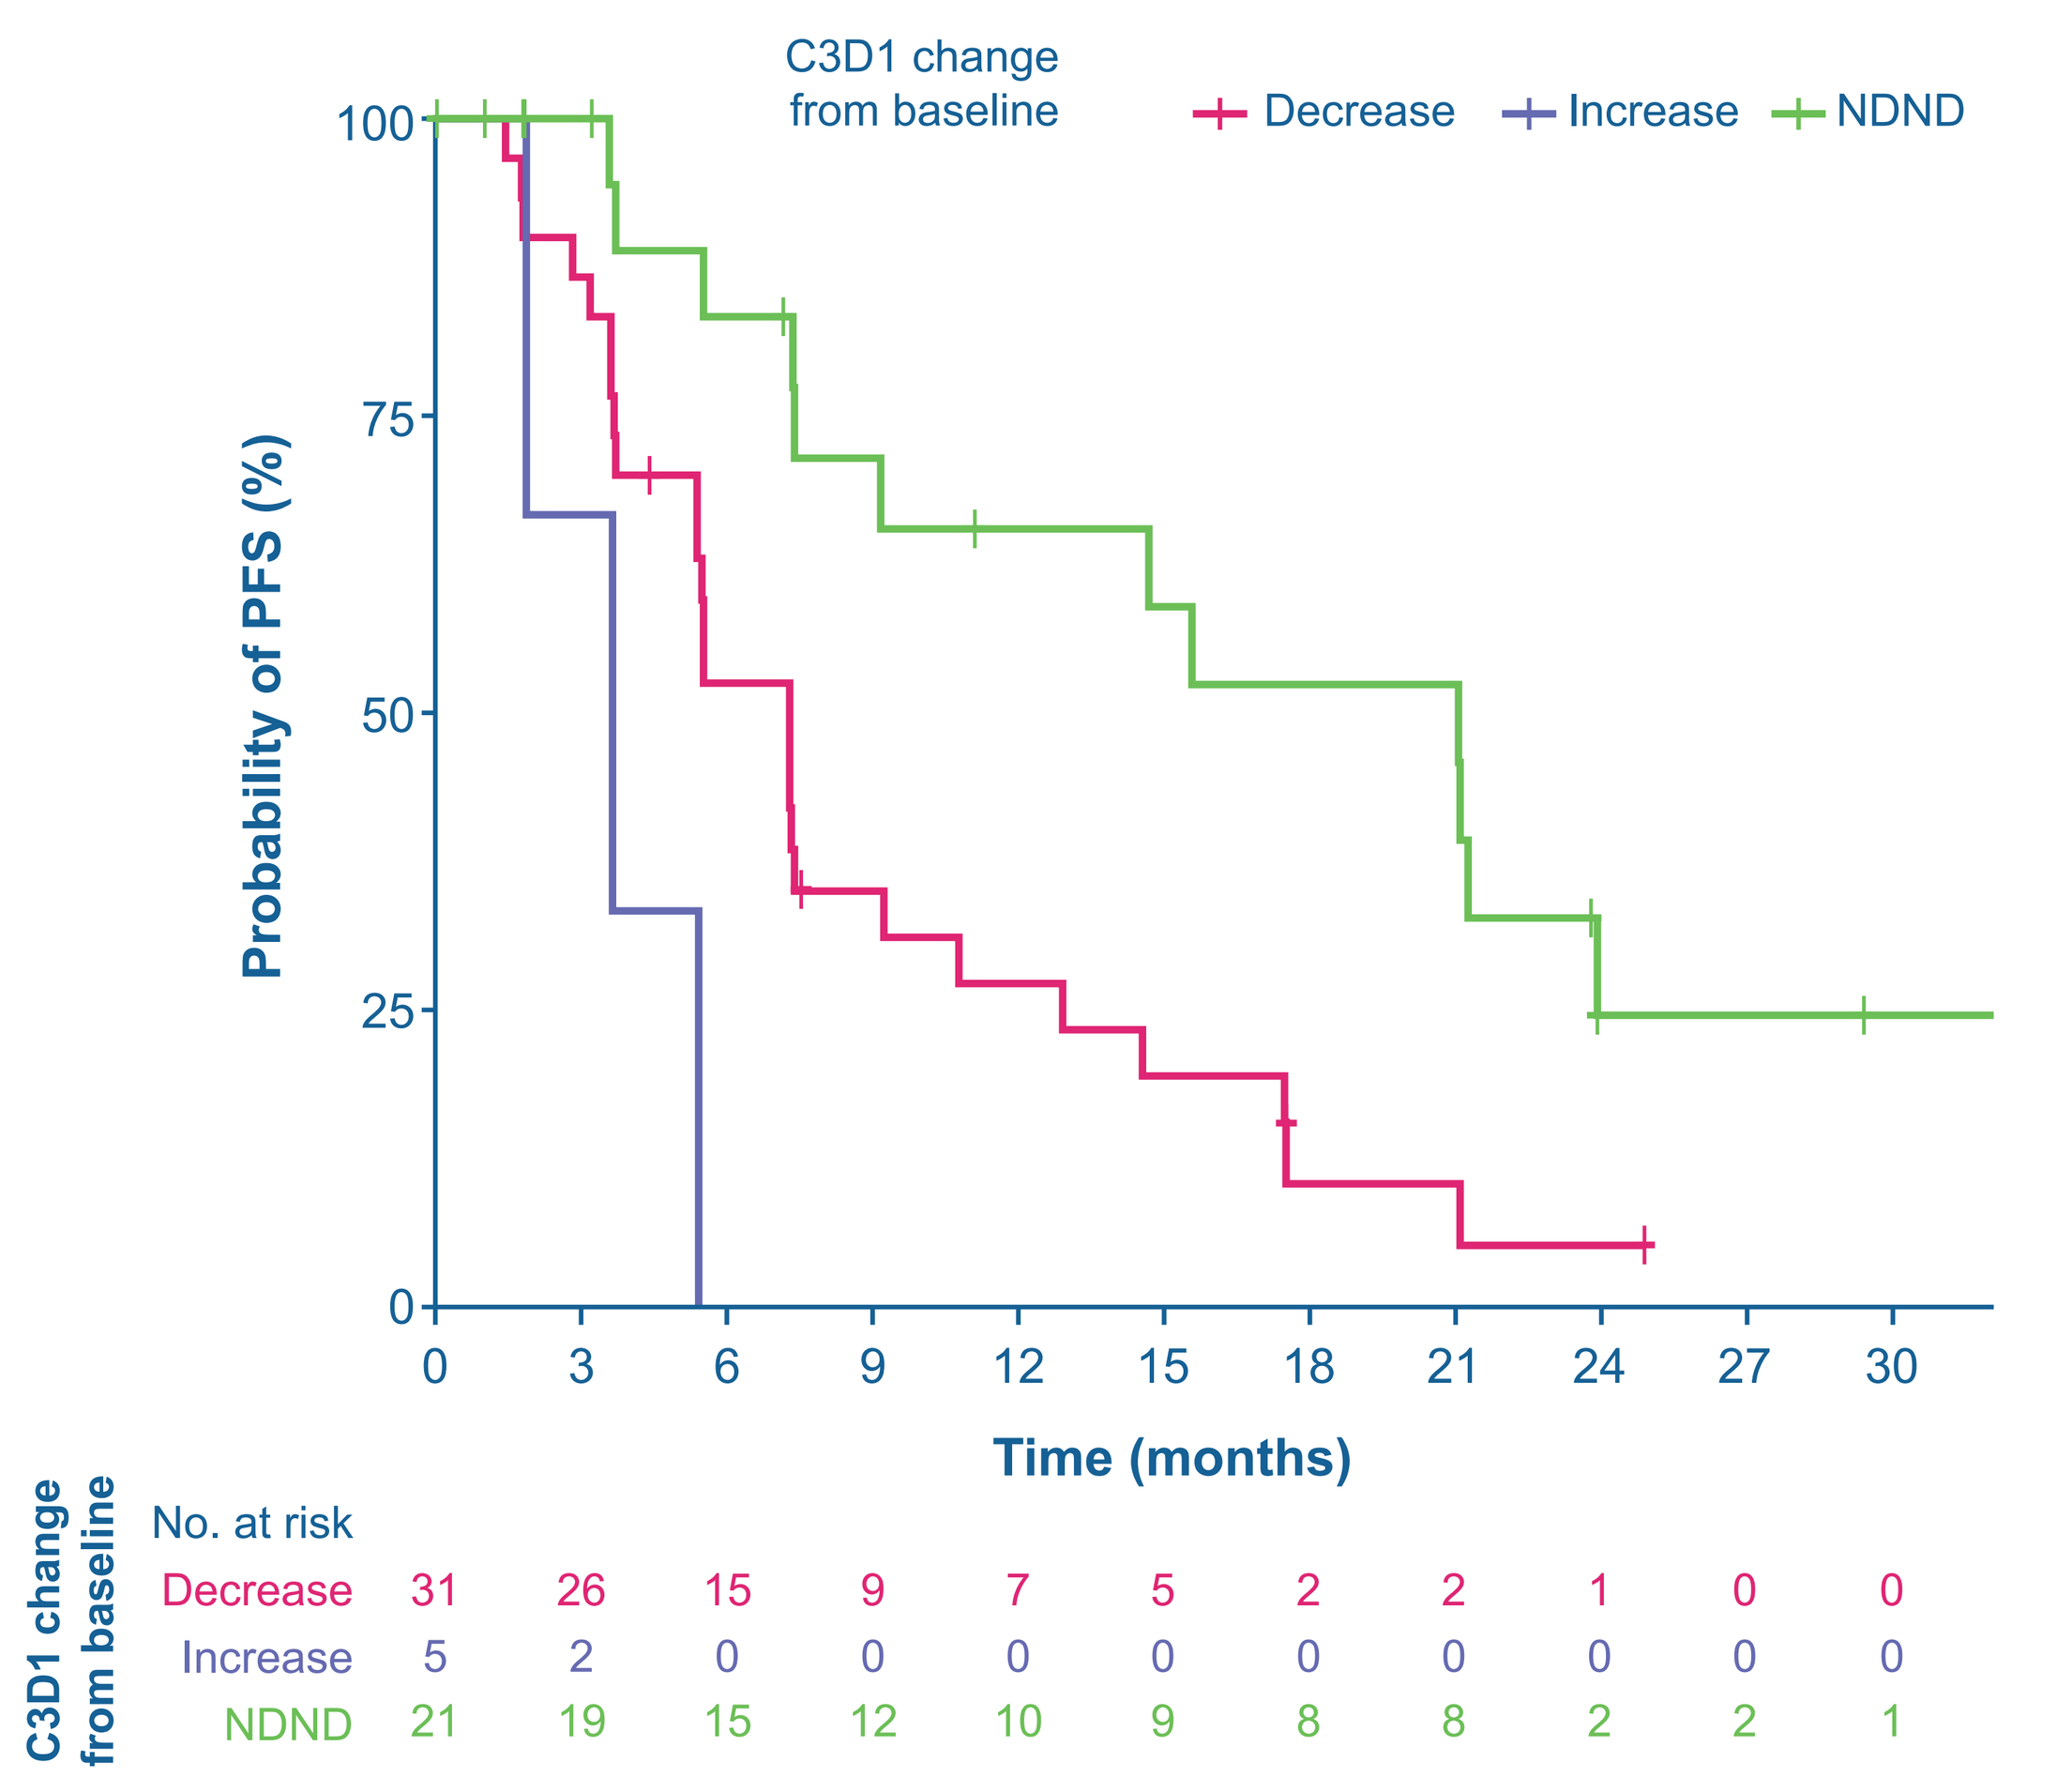
C3D1 change from baseline
Decrease
Increase
NDND
Probability of PFS (%)
100
75
50
25
0
0 3 6 9 12 15 18 21 24 27 30
Time (months)
C3D1 change from baseline
No. at risk
Decrease 31 26 15 9 7 5 2 2 1 0 0
Increase 5 2 0 0 0 0 0 0 0 0 0
NDND 21 19 15 12 10 9 8 8 2 2 1

Decrease vs NDND:
HR=2.83 (95% CI: 1.39–5.78);
log-rank *P*=0.0042

Increase vs NDND:
HR=10.46 (95% CI: 2.60–42.05);
log-rank *P*=0.0009

Increase vs Decrease:
HR=3.69 (95% CI: 1.03–13.27];
log-rank *P*=0.045

PPP Cohort (N=114)	Events (n)	Median PFS, months (95% CI)
All patients		**7.39 (5.52–14.69)**
ctDNA decrease	**26 (n=31)**	**7.29 (5.39–10.78)**
ctDNA increase	**3 (n=5)**	**3.65 (1.87–NR)**
ctDNA NDND	**13 (n=21)**	**21.06 (9.17–NR)**

NR, not reached

Conclusions

- Patients with no detectable *EGFR* ex20ins ctDNA at baseline had longer PFS than those with *EGFR* ex20ins ctDNA detected at baseline
- Patients with clearance of *EGFR* ex20ins ctDNA at C3D1 of mobocertinib treatment achieved longer PFS compared with those who had persistent *EGFR* ex20ins ctDNA or an increase in *EGFR* ex20ins VAF
 - Patients with no detectable *EGFR* ex20ins ctDNA both at baseline and at C3D1 had a median PFS of 21.06 months, which was markedly higher than the median PFS in patients with an increase (3.65 months) or decrease (7.29 months) from baseline in *EGFR* ex20ins VAFs on C3D1
- Further evaluation of *EGFR* ex20ins ctDNA status as a prognostic biomarker in patients with *EGFR* ex20ins+ NSCLC is warranted

References

1. Watanabe, et al. Ann Transl Med. 2022;10(23):1283
2. Gonzalvez F, et al. Cancer Discov. 2021;11(7):1672–87.
3. Riely GJ, et al. Cancer Discov. 2021;11(7):1688–99.
4. Exkivity (mobocertinib) [package insert]. (Takeda Pharmaceuticals America, Inc., Lexington, MA. 2023)
5. Zhou, et al. JAMA Oncol. 2021;7(12):e214761.
6. Ramalingam, et al. Presented at the Annual Meeting of the European Society of Medical Oncology; September 9–13, 2022; Paris, France.
7. Zhang, et al. Mol Cancer. 2019;18:36.
8. Guo, et al. Transl Lung Cancer Res. 2021;10(7):3213–25.
9. Pender, et al. Transl Lung Cancer Res. 2020;9(4):1084–92.
10. Neal, et al. J Clin Oncol. 2023;41(16_suppl): 9082-82.

Acknowledgments

The authors would like to thank the patients, their families, and their caregivers; the study investigators and their team members at each study site; and colleagues from Takeda and its subsidiaries. Medical writing support for the development of this poster, under the direction of the authors, was provided by Kalpana Vijayan, PhD of Peloton Advantage, LLC, an OPEN Health company; was funded by Takeda Development Center Americas, Inc., Lexington, MA, USA; and complied with the Good Publication Practice (GPP) guidelines (DeTora LM, et al. Ann Intern Med 2022;175:1298–1304). The study was sponsored by Takeda Development Center Americas, Inc.

Disclosures

SC, XT, ZS, YL, MG, and **ENC:** Employment with Takeda. **RJF:** Employment with Takeda; stock ownership with Baxter, Bristol Myers Squibb, GE Healthcare, Gilead, Johnson & Johnson, Medtronic, Pfizer, Takeda, Teva, Viatris, Zimmer Biomet, and Zimvie. **SV:** Employment with Takeda.

For an e-Print, scan this QR code.
Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the authors of this poster.

Presented at the World Conference on Lung Cancer (WCLC), September 9–12, 2023, Singapore

Email for questions or comments: sam.chatterjee@takeda.com