Subgroup analyses of safety and efficacy by number and types of prior lines of treatment in FRESCO-2, a global phase 3 study of fruquintinib in patients with refractory metastatic colorectal cancer

Poster 304

Arvind Dasari,[1] Sara Lonardi,[2] Rocio Garcia-Carbonero,[3] Elena Elez,[4] Takayuki Yoshino,[5] Alberto Sobrero,[6] James Yao,[1] Pilar García-Alfonso,[7] Judit Kocsis,[8] Antonio Cubillo Gracian,[9] Andrea Sartore-Bianchi,[10] Taroh Satoh,[11] Violaine Randrian,[12] Jiri Tomasek,[13] Geoff Chong,[14] Zhao Yang,[15] Rajash Pallai,[15] Iris Carton,[15] Josep Tabernero,[4] Cathy Eng[16]

[1]Department of Gastrointestinal Medical Oncology, University of Texas MD Anderson Cancer Center Houston, TX, USA; [2]Medical Oncology Unit 1, Veneto Institute of Oncology IOV - IRCCS, Padua, Italy; [3]Oncology Department, Hospital Universitario 12 de Octubre, Imas 12, UCM, Madrid, Spain; [4]Vall d'Hebron Barcelona Hospital Campus, Vall d'Hebron Institute of Oncology, Barcelona, Spain; [5]Department of Gastroenterology and Gastrointestinal Oncology, National Cancer Center Hospital East, Kashiwa, Chiba, Japan; [6]Department of Medical Oncology, Azienda Ospedaliera San Martino, Genoa, Italy; [7]Medical Oncology, Hospital Universitario Gregorio Marañón, Madrid, Spain; [8]Department of Oncoradiology, Bács-Kiskun Megyei Oktatókórház, Kecskemét, Hungary; [9]Medical Oncology, HM Universitario Sanchinarro Centro Integral Oncológico Clara Campal, Madrid, Spain; [10]Department of Oncology and Hemato-Oncology, Università degli Studi di Milano, Milan, Italy; [11]Department of Gastroenterological Surgery, Graduate School of Medicine, Osaka University, Suita, Japan; [12]Hepato-Gastroenterology Department, Poitiers University Hospital, Poitiers, France; [13]Department of Complex Oncology Care, Masaryk Memorial Cancer Institute, Brno, Czech Republic; [14]Olivia Newton John Cancer & Wellness Centre, Austin Hospital, Heidelberg, VIC, Australia; [15]HUTCHMED International Corporation., Florham Park, NJ, USA; [16]Department of Medicine, Division of Hematology and Oncology, Vanderbilt Ingram Cancer Center, Nashville, TN, USA

BACKGROUND

- Effective treatment options are limited for patients with refractory metastatic colorectal cancer (mCRC)
- Fruquintinib is a highly selective, potent, oral tyrosine kinase inhibitor of all 3 VEGF receptors (VEGFR-1, -2, and -3) with weak or no inhibitory effect on other receptor kinases[1]
- A statistically significant and clinically meaningful improvement in overall survival (OS) and progression-free survival (PFS) with an acceptable and tolerable safety profile was demonstrated with fruquintinib + best supportive care (BSC) vs placebo + BSC in the global phase 3 FRESCO-2 study (NCT04322539)[2]
- Here we report subgroup analyses of efficacy and safety by prior lines of treatment (LOT) and types of anti-cancer treatment for metastatic disease

METHODS

- FRESCO-2 was conducted in the US, Europe, Japan, and Australia, comparing fruquintinib + BSC vs placebo + BSC in patients with mCRC
- Eligible patients had received all standard treatments, including fluoropyrimidine-, oxaliplatin- and irinotecan-based chemotherapy, anti-VEGF therapy, anti-EGFR therapy (if RAS wild type), and trifluridine/tipiracil or regorafenib
 - Patients with MSI-high or MMR-deficient tumors or BRAFV600E-mutant tumors must have also received an immune checkpoint inhibitor or BRAF inhibitor, respectively
- Randomization was stratified by prior treatment (trifluridine/tipiracil vs regorafenib vs both), RAS mutation status (wild type vs mutant), and duration of metastatic disease (≤18 months vs >18 months)
- Patients were grouped according to number of prior LOT (≤4 vs >4, based on a median of 4 prior LOT); efficacy and safety data were analyzed according to these subgroups, as well as according to type of prior anti-cancer treatment received (anti-VEGF, anti-EGFR, regorafenib, TAS-102)

RESULTS

Patients and prior anti-cancer treatment

- A total of 691 patients were randomized and 686 patients received at least one dose of study drug
- The median number of prior lines of anti-cancer treatment for metastatic disease was 4 (range 2–16) for patients in the fruquintinib + BSC arm and 4 (range 2–12) for patients in the placebo + BSC arm
- Baseline characteristics were balanced between arms in patients who had received ≤4 prior LOT and in patients who had received >4 prior LOT **(Table 1)**

Table 1. Patient demographics and baseline characteristics by number of prior lines of anti-cancer treatment for metastatic disease (ITT population)

Characteristic, n (%)		≤4 prior LOT (n=370) Fruquintinib + BSC n=247	≤4 prior LOT (n=370) Placebo + BSC n=123	>4 prior LOT (n=321) Fruquintinib + BSC n=214	>4 prior LOT (n=321) Placebo + BSC n=107
Age	Median years (range)	64 (25–82)	63 (30–82)	63 (31–81)	65 (35–86)
	≥65 years	114 (46.2)	55 (44.7)	100 (46.7)	56 (52.3)
Sex	Female	127 (51.4)	45 (36.6)	89 (41.6)	45 (42.1)
	Male	120 (48.6)	78 (63.4)	125 (58.4)	62 (57.9)
ECOG PS	0	111 (44.9)	63 (51.2)	85 (39.7)	39 (36.4)
	1	136 (55.1)	60 (48.8)	129 (60.3)	68 (63.6)
Primary site at 1st diagnosis	Colon, left	102 (41.3)	49 (39.8)	90 (42.1)	43 (40.2)
	Colon, right	57 (23.1)	33 (26.8)	40 (18.7)	20 (18.7)
	Colon, left and right	3 (1.2)	1 (0.8)	1 (0.5)	1 (0.9)
	Colon, unknown	14 (5.7)	6 (4.9)	11 (5.1)	7 (6.5)
	Rectum only	71 (28.7)	34 (27.6)	72 (33.6)	36 (33.6)
Liver metastases	Yes	181 (73.3)	83 (67.5)	158 (73.8)	73 (68.2)
Duration of metastatic disease	≤18 months	37 (15.0)	12 (9.8)	0	1 (0.9)
	>18 months	210 (85.0)	111 (90.2)	214 (100)	106 (99.1)
RAS status	Wild type	77 (31.2)	36 (29.3)	93 (43.5)	49 (45.8)
	Mutant	170 (68.8)	87 (70.7)	121 (56.5)	58 (54.2)
Type of prior anti-cancer treatment	VEGF biologics	233 (94.3)	115 (93.5)	212 (99.1)	106 (99.1)
	EGFR biologics	77 (31.2)	36 (29.3)	103 (48.1)	52 (48.6)
	Regorafenib	31 (12.6)	14 (11.4)	9 (4.2)	4 (3.7)
	TAS-102	162 (65.6)	88 (71.5)	78 (36.4)	33 (30.8)
	Regorafenib and TAS-102	54 (21.9)	21 (17.1)	127 (59.3)	70 (65.4)

ECOG PS, Eastern Cooperative Oncology Group performance status; ITT, intent-to-treat

Question

Did patients with mCRC treated with fruquintinib + BSC benefit regardless of prior number of lines and/or type of anti-cancer treatment?

Study design

FRESCO-2 (NCT04322539)
Global, multicenter, randomized, placebo-controlled phase 3 study

2:1

Fruquintinib 5 mg PO QD (3 weeks on, 1 week off) + BSC

Placebo 5 mg PO QD (3 weeks on, 1 week off) + BSC

Endpoints:
- **Primary:** OS - **Key secondary:** PFS
- **Other secondary:** ORR, DCR, safety

Subgroup analyses for efficacy and safety were performed according to the number of prior LOT (≤4 vs >4) and by types of prior treatment

Key results for OS (ITT population)

Figure 1A: OS with fruquintinib + BSC vs placebo + BSC by ≤4 vs >4 prior LOT

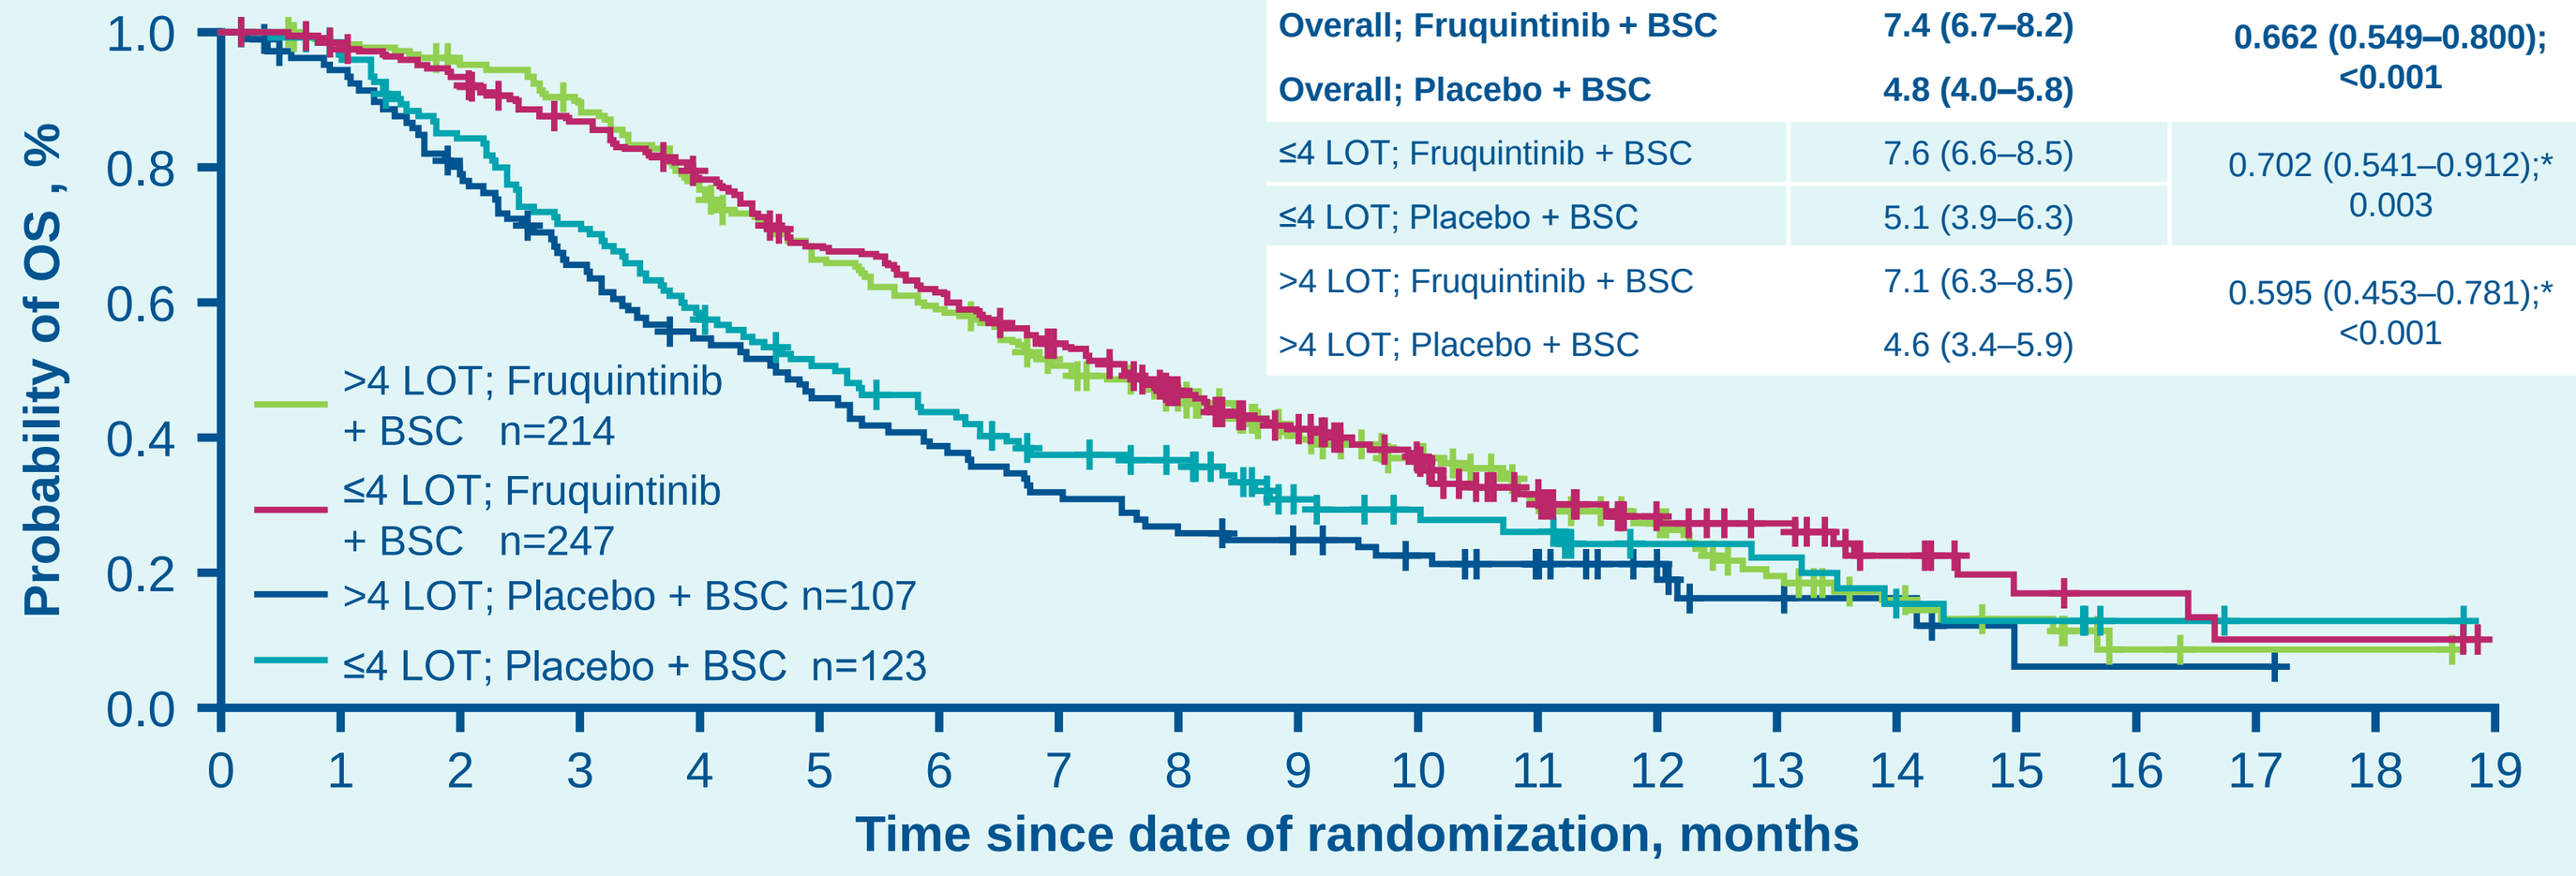
Probability of OS , %
Time since date of randomization, months
>4 LOT; Fruquintinib + BSC n=214
≤4 LOT; Fruquintinib + BSC n=247
>4 LOT; Placebo + BSC n=107
≤4 LOT; Placebo + BSC n=123

	Median OS (95% CI)	HR (95% CI); *P* value
Overall; Fruquintinib + BSC	**7.4 (6.7–8.2)**	**0.662 (0.549–0.800); <0.001**
Overall; Placebo + BSC	**4.8 (4.0–5.8)**	
≤4 LOT; Fruquintinib + BSC	7.6 (6.6–8.5)	0.702 (0.541–0.912);* 0.003
≤4 LOT; Placebo + BSC	5.1 (3.9–6.3)	
>4 LOT; Fruquintinib + BSC	7.1 (6.3–8.5)	0.595 (0.453–0.781);* <0.001
>4 LOT; Placebo + BSC	4.6 (3.4–5.9)	

*Adjusted HR estimated using Cox's proportional hazards model with stratification factors (not including the factor of prior treatment) and treatment group as covariates. DCR, disease control rate; ORR, overall response rate

Figure 1B: Median OS with fruquintinib + BSC vs placebo + BSC by type of prior anti-cancer treatment

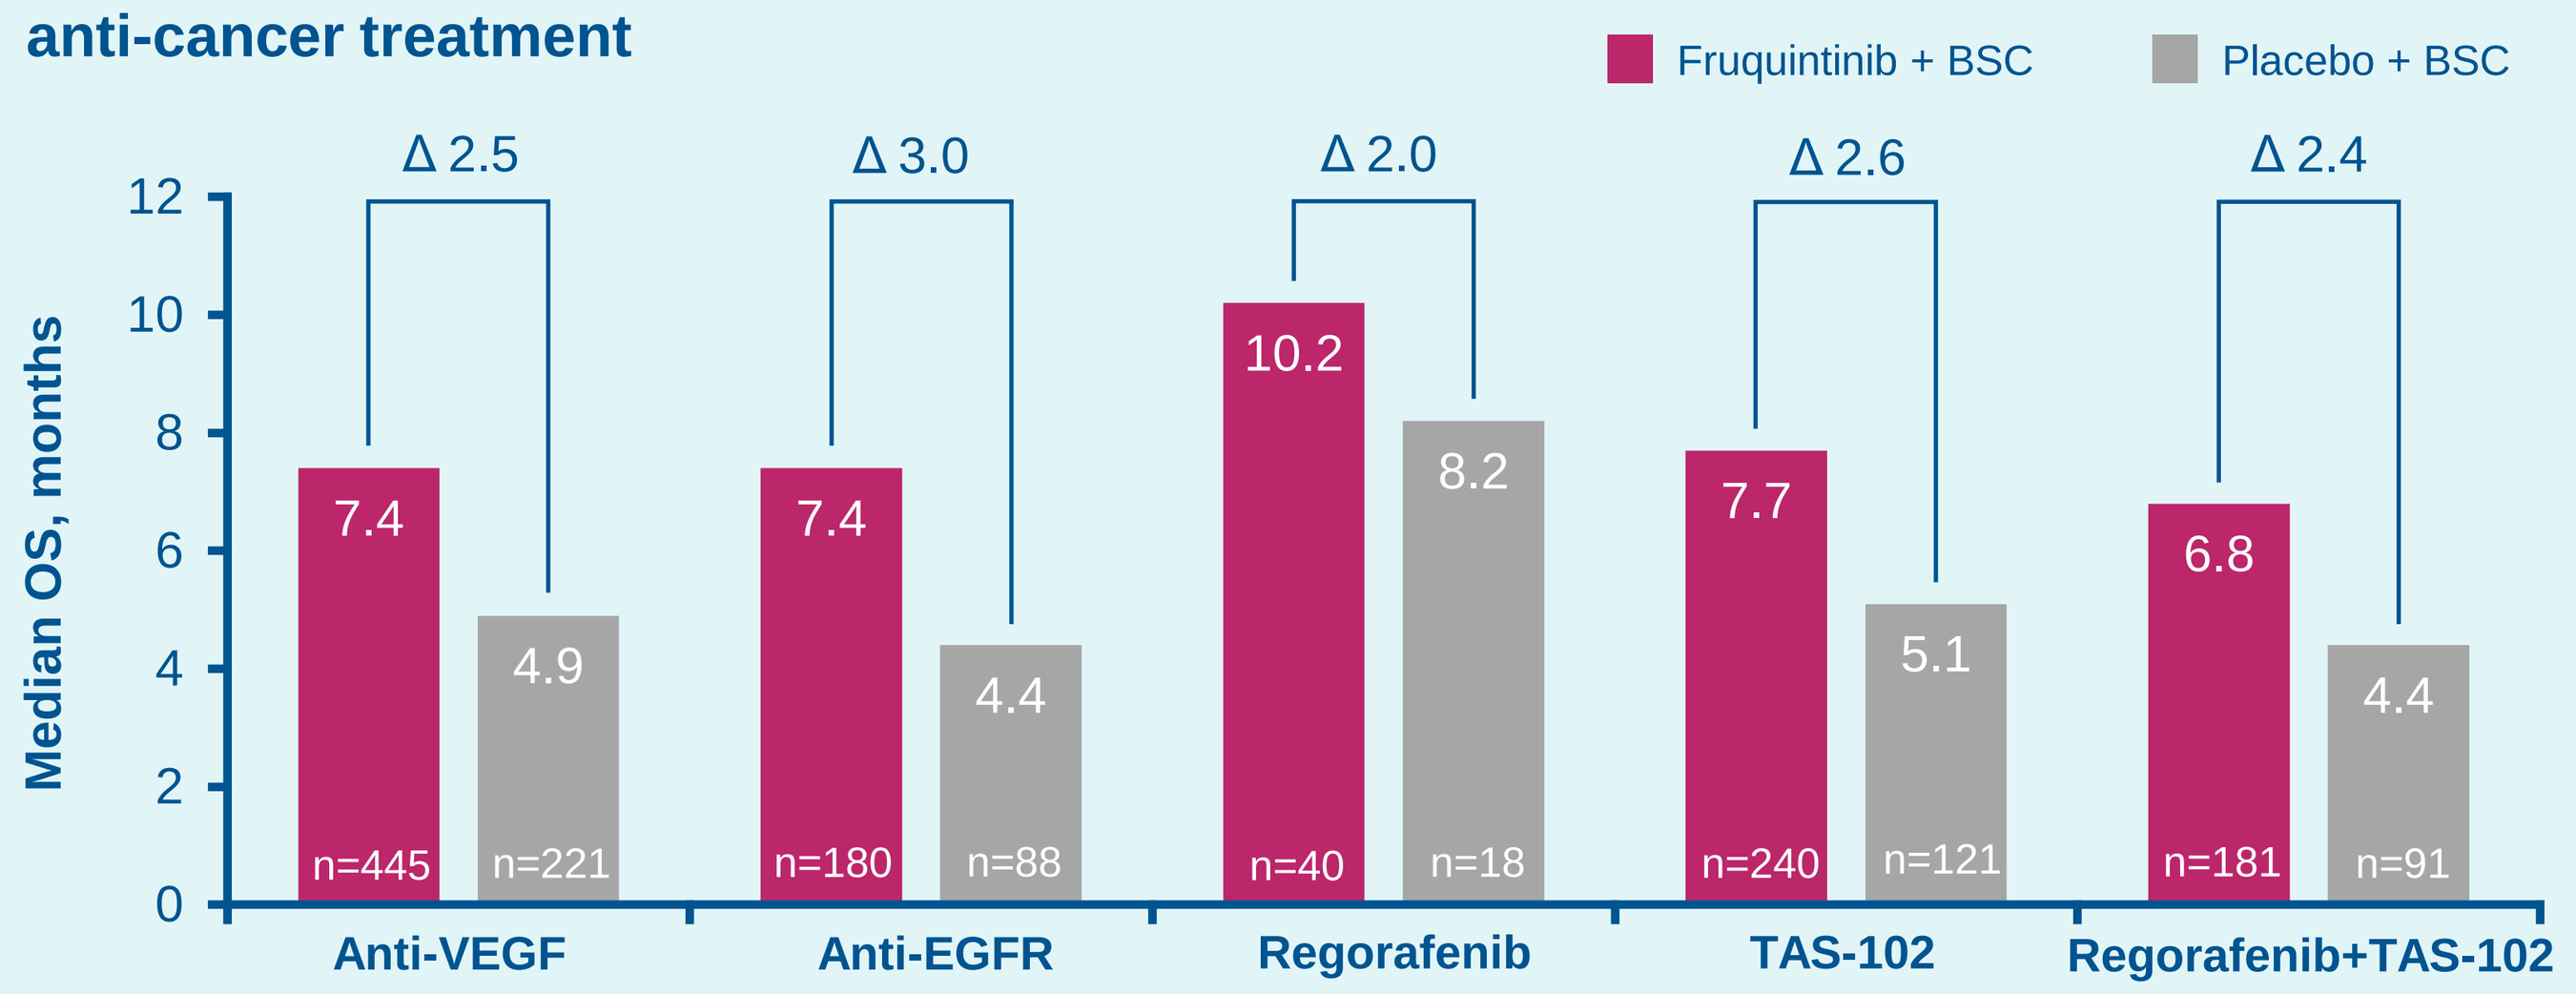
Fruquintinib + BSC
Placebo + BSC
Median OS, months
Δ 2.5
Δ 3.0
Δ 2.0
Δ 2.6
Δ 2.4
7.4
4.9
7.4
4.4
10.2
8.2
7.7
5.1
6.8
4.4
n=445
n=221
n=180
n=88
n=40
n=18
n=240
n=121
n=181
n=91
Anti-VEGF
Anti-EGFR
Regorafenib
TAS-102
Regorafenib+TAS-102

Δ=difference in median OS between fruquintinib + BSC and placebo + BSC arms.

Key take aways

Fruquintinib + BSC demonstrated improvement in OS and PFS vs placebo + BSC , irrespective of prior number of lines or type of treatment

Treatment exposure, OS, and PFS by prior number of lines and type of anti-cancer treatment

- The median duration of exposure of fruquintinib + BSC vs placebo + BSC was
 - 2.76 months (range 0.4–19.1) vs 1.84 months (range 0.4–5.8) in patients who received ≤4 prior LOT, respectively
 - 3.45 months (range 0.3–17.6) vs 1.84 months (range 0.3–12.0) in patients who received >4 prior LOT, respectively
- Fruquintinib + BSC demonstrated consistent OS and PFS benefit compared with placebo + BSC in patients who had been previously treated with anti-VEGF, regorafenib, or TAS-102 **(Table 2)**

Table 2: Median duration of prior anti-cancer treatment, median duration of study treatment, and median OS and PFS by prior number of LOT (ITT population)

	Anti-VEGF*				Regorafenib (any†)				TAS-102 (any‡)			
	≤4 prior LOT		>4 prior LOT		≤4 prior LOT		>4 prior LOT		≤4 prior LOT		>4 prior LOT	
	F	P	F	P	F	P	F	P	F	P	F	P
Median prior treatment, months (range)	n=234 11.47 (0–57.7)	n=114 11.24 (1.8–36.9)	n=212 14.55 (0–58.0)	n=105 14.52 (0.7–77.5)	n=85 3.02 (0.3–15.9)	n=35 2.89 (0.3–8.9)	n=136 3.29 (0.1–31.7)	n=74 3.24 (0.2–28.0)	n=216 3.09 (0–18.6)	n=109 3.15 (0–14.1)	n=205 3.58 (0–27.8)	n=102 3.02 (0.4–31.0)
Median OS, months HR (95% CI)	7.6 vs 5.2 0.715 (0.545–0.938)		7.1 vs 4.6 0.600 (0.456–0.791)		7.2 vs 5.2 0.745 (0.463–1.196)		7.0 vs 4.6 0.560 (0.403–0.777)		7.4 vs 5.1 0.707 (0.537–0.930)		7.1 vs 4.4 0.601 (0.456–0.792)	
Median PFS, months HR (95% CI)	3.4 vs 1.9 0.352 (0.272–0.457)		3.7 vs 1.8 0.301 (0.229–0.395)		3.6 vs 1.9 0.319 (0.202–0.502)		3.7 vs 1.8 0.249 (0.177–0.350)		3.5 vs 1.9 0.329 (0.252–0.431)		3.7 vs 1.8 0.315 (0.241–0.412)	
Median study treatment, months (range)	n=233 2.76 (0.4–19.1)	n=112 1.84 (0.4–5.8)	n=210 3.37 (0.3–17.6)	n=105 1.84 (0.2–12.0)	n=84 3.55 (0.7–14.7)	n=34 1.84 (0.7–5.8)	n=136 3.48 (0.3–17.6)	n=74 1.84 (0.3–7.4)	n=215 2.76 (0.4–19.1)	n=107 1.84 (0.4–5.8)	n=203 3.45 (0.3–17.6)	n=102 1.84 (0.2–12.0)

*The number of patients included in this analysis was based on prior anti-cancer treatment, and may differ to the number of patients shown in Figures 1B and 2B, for which data were collected from oncology history; †Alone or in combination with TAS-102; ‡Alone or in combination with regorafenib. CI, confidence interval; F, fruquintinib + BSC; HR, hazard ratio; P, placebo + BSC

- The median follow-up was 11.3 months with fruquintinib + BSC and 11.2 months with placebo + BSC
- Fruquintinib + BSC improved OS **(Figure 1)** and PFS **(Figure 2)** compared with placebo + BSC irrespective of number of prior LOT or prior types of anti-cancer treatment

Figure 2: PFS with fruquintinib + BSC vs placebo + BSC by prior (A) ≤4 vs >4 LOT and (B) types of anti-cancer treatment (ITT population)

A

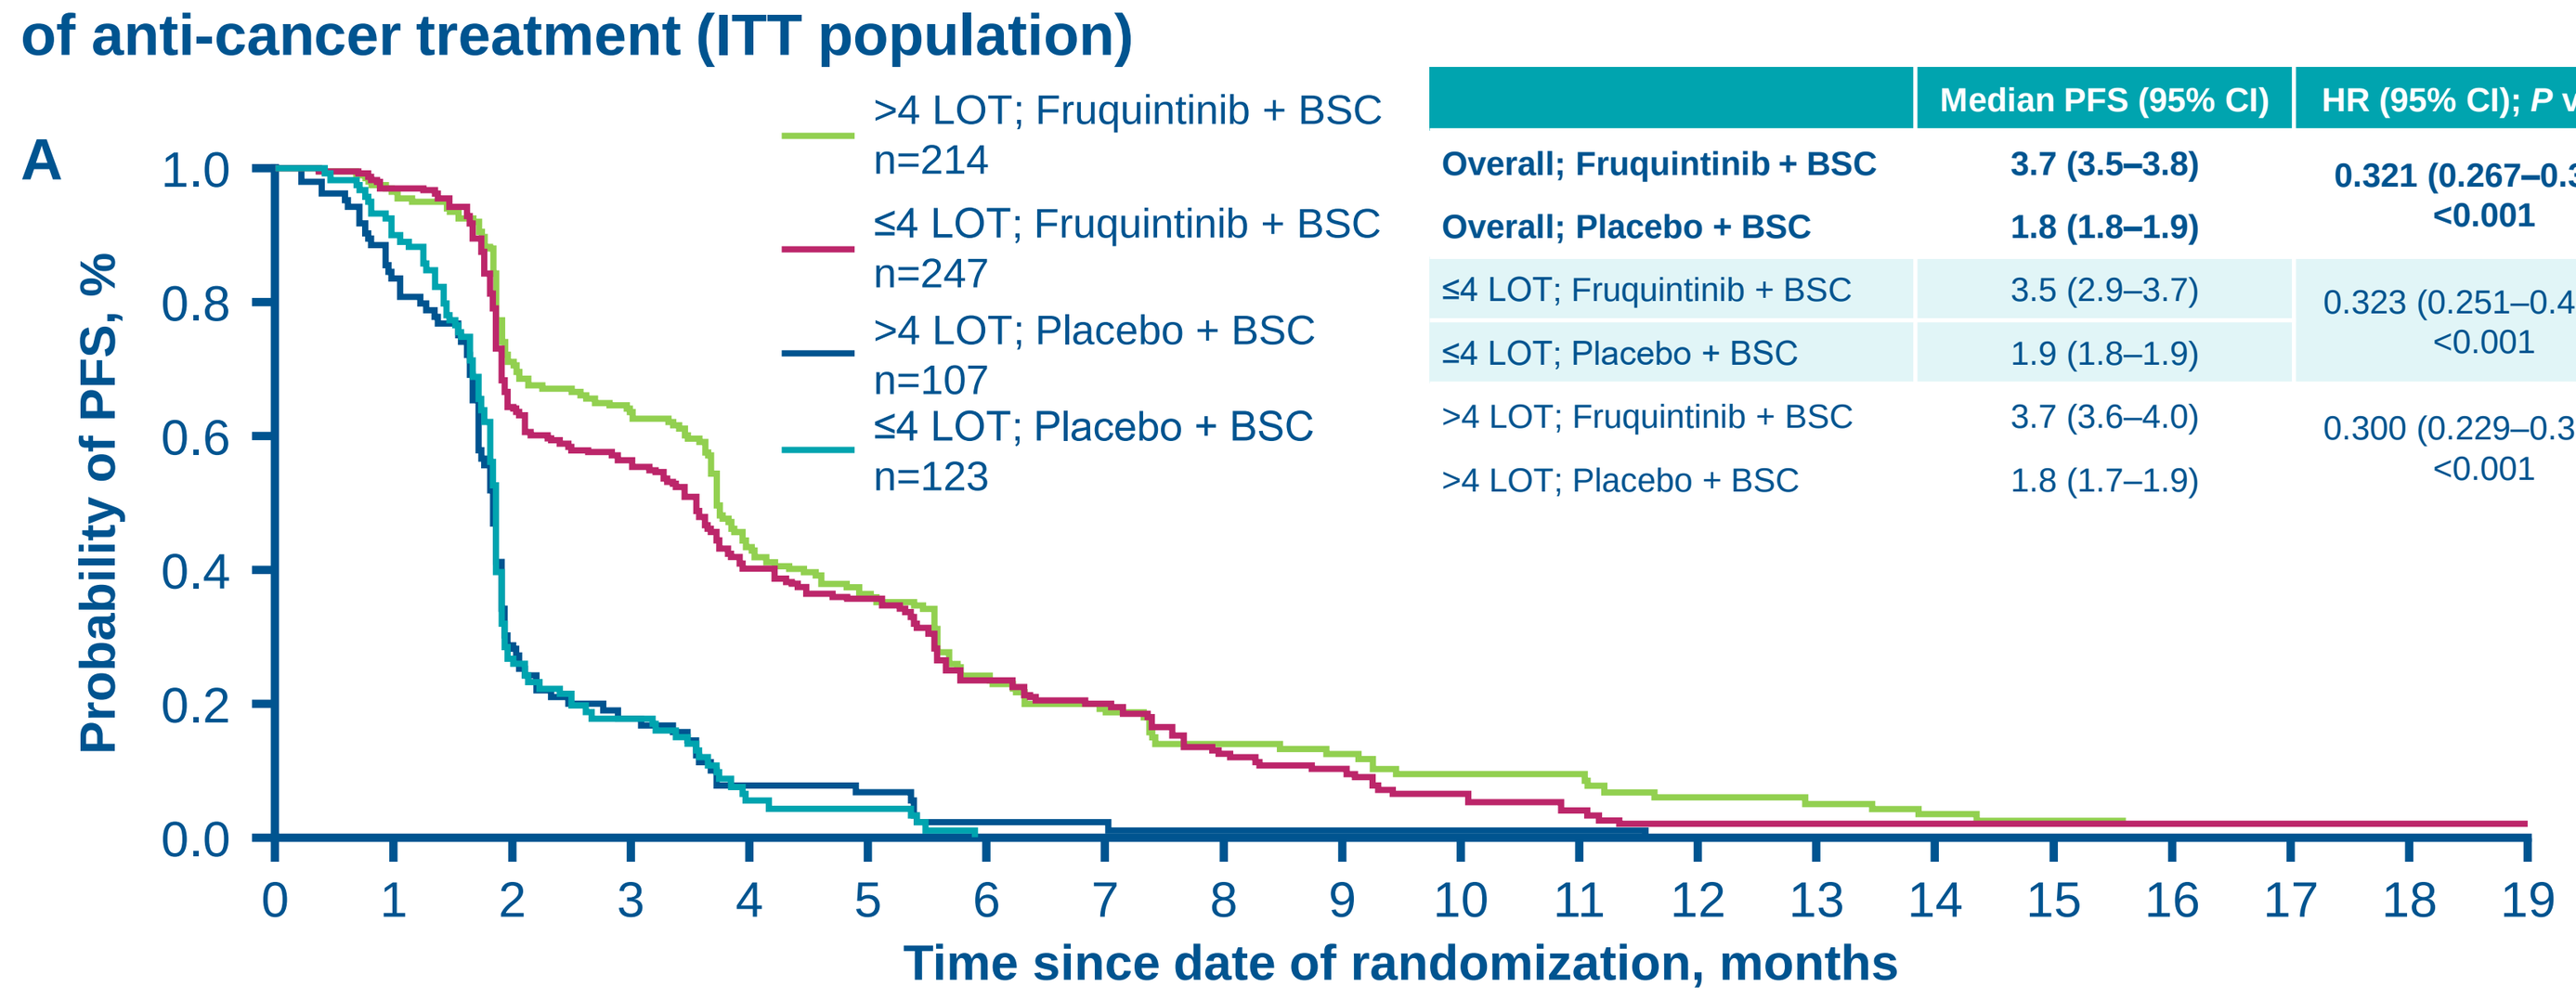
Probability of PFS, %
Time since date of randomization, months
>4 LOT; Fruquintinib + BSC n=214
≤4 LOT; Fruquintinib + BSC n=247
>4 LOT; Placebo + BSC n=107
≤4 LOT; Placebo + BSC n=123

	Median PFS (95% CI)	HR (95% CI); *P* value
Overall; Fruquintinib + BSC	**3.7 (3.5–3.8)**	**0.321 (0.267–0.386) <0.001**
Overall; Placebo + BSC	**1.8 (1.8–1.9)**	
≤4 LOT; Fruquintinib + BSC	3.5 (2.9–3.7)	0.323 (0.251–0.417);* <0.001
≤4 LOT; Placebo + BSC	1.9 (1.8–1.9)	
>4 LOT; Fruquintinib + BSC	3.7 (3.6–4.0)	0.300 (0.229–0.393);* <0.001
>4 LOT; Placebo + BSC	1.8 (1.7–1.9)	

B

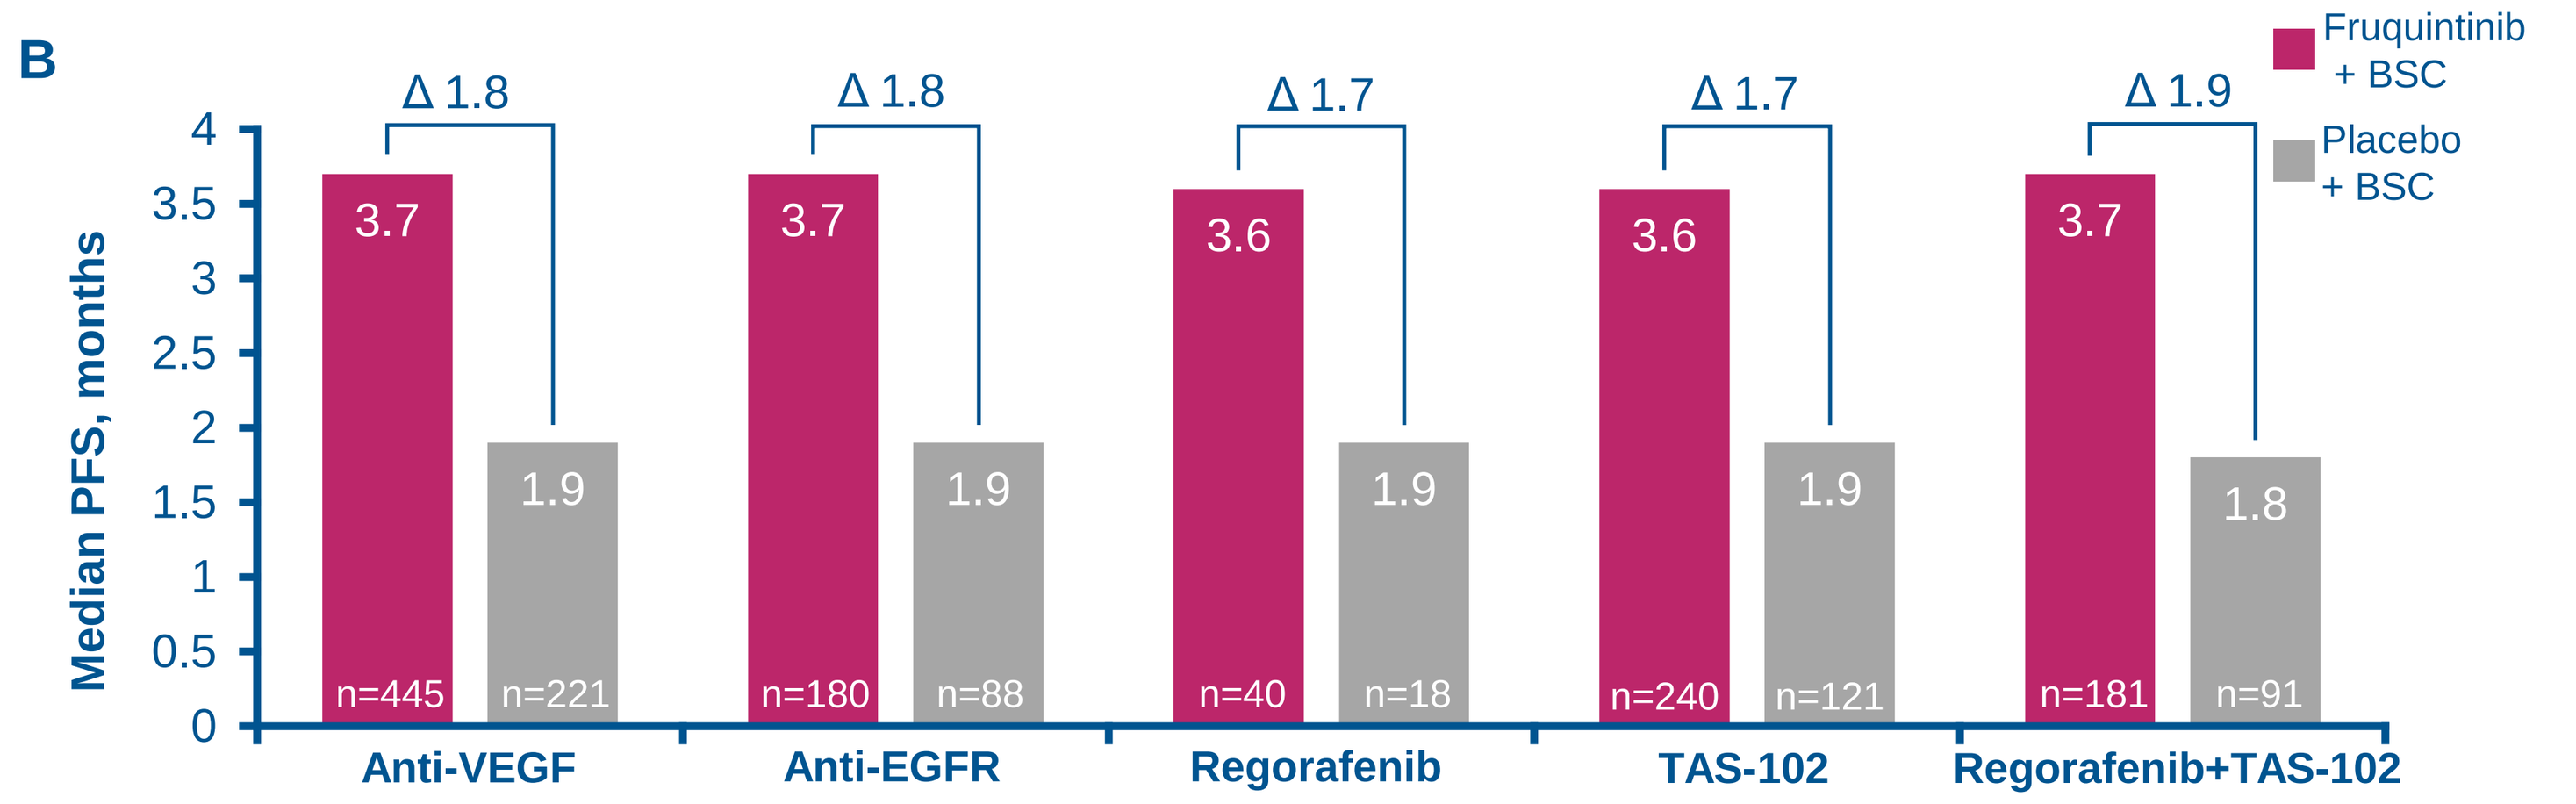
Fruquintinib + BSC
Placebo + BSC
Median PFS, months
Δ 1.8
Δ 1.8
Δ 1.7
Δ 1.7
Δ 1.9
3.7
1.9
3.7
1.9
3.6
1.9
3.6
1.9
3.7
1.8
n=445
n=221
n=180
n=88
n=40
n=18
n=240
n=121
n=181
n=91
Anti-VEGF
Anti-EGFR
Regorafenib
TAS-102
Regorafenib+TAS-102

*Adjusted HR estimated using Cox's proportional hazards model with stratification factors (not including the factor of prior treatment) and treatment group as covariates; Δ=difference in median PFS between fruquintinib + BSC and placebo + BSC arms.

Safety

- Occurrences of treatment-emergent adverse events (TEAEs) were generally balanced between the fruquintinib + BSC vs placebo + BSC arms among patients with both ≤4 and >4 prior LOT **(Table 3)**

Table 3: Overall TEAE safety profile in the safety population*

Category, n (%)	≤4 prior LOT (n=367) Fruquintinib + BSC n=245	≤4 prior LOT (n=367) Placebo + BSC n=122	>4 prior LOT (n=319) Fruquintinib + BSC n=211	>4 prior LOT (n=319) Placebo + BSC n=108
Any TEAE	241 (98.4)	111 (91.0)	210 (99.5)	102 (94.4)
Grade ≥3	148 (60.4)	55 (45.1)	138 (65.4)	61 (56.5)
Treatment-related	214 (87.3)	69 (56.6)	181 (85.8)	61 (56.5)
Treatment-related grade ≥3	91 (37.1)	12 (9.8)	73 (34.6)	14 (13.0)
Leading to dose reduction	61 (24.9)	5 (4.1)	49 (23.2)	4 (3.7)
Leading to dose interruption	103 (42.0)	31 (25.4)	110 (52.1)	30 (27.8)
Leading to treatment discontinuation	50 (20.4)	19 (15.6)	20 (9.5)	3 (2.8)
Leading to death	25 (10.2)	22 (18.0)	23 (10.9)	23 (21.3)

*Of 5 patients assigned to fruquintinib + BSC, 3 did not receive fruquintinib, and 2 patients received placebo instead. In the placebo + BSC group, 2 patients did not receive treatment.

- The grade ≥3 treatment-related TEAEs in ≥5% of patients with ≤4 or >4 prior LOT in the fruquintinib + BSC arm were hypertension, palmar plantar erythrodysesthesia (PPE), and asthenia **(Table 4)**

Table 4: Grade ≥3 treatment-related TEAEs in ≥1% of patients in either arm (safety population)*

Preferred term, n (%)	≤4 prior LOT (n=367) Fruquintinib + BSC n=245	≤4 prior LOT (n=367) Placebo + BSC n=122	>4 prior LOT (n=319) Fruquintinib + BSC n=211	>4 prior LOT (n=319) Placebo + BSC n=108
Hypertension	30 (12.2)	2 (1.6)	19 (9.0)	0
PPE	14 (5.7)	0	14 (6.6)	0
Asthenia	12 (4.9)	1 (0.8)	12 (5.7)	2 (1.9)
Fatigue	7 (2.9)	1 (0.8)	8 (3.8)	0
Diarrhea	5 (2.0)	0	10 (4.7)	0
Proteinuria	5 (2.0)	1 (0.8)	2 (0.9)	0
Stomatitis	3 (1.2)	0	4 (1.9)	0
Decreased appetite	2 (0.8)	1 (0.8)	4 (1.9)	1 (0.9)
Increased ALT	2 (0.8)	0	3 (1.4)	0
Anemia	0	1 (0.8)	1 (0.5)	3 (2.8)
Abdominal pain	0	1 (0.8)	0	2 (1.9)

*Of 5 patients assigned to fruquintinib + BSC, 3 did not receive fruquintinib, and 2 patients received placebo instead. In the placebo + BSC group, 2 patients did not receive treatment. ALT, alanine aminotransferase

Conclusions

- Fruquintinib + BSC demonstrated longer OS and PFS vs placebo + BSC in patients with mCRC irrespective of prior number of LOT and type of prior treatment
- The overall safety profile was consistent in patients who received ≤4 prior LOT or >4 prior LOT
- These results were consistent with the effect observed in the overall population, supporting fruquintinib as a treatment option for patients with refractory mCRC, irrespective of prior number of LOT or type of prior anti-cancer treatment

References

1. Sun Q, et al. *Cancer Biol Ther* 2014;15(12):1635-45
2. Dasari A, et al. *Annals of Oncology* 2022;33(7):S1391–2

Acknowledgments

The FRESCO-2 study was sponsored by HUTCHMED. Medical writing support for the development of this poster, under the direction of the authors, was provided by Emily Ruban-Fell, PhD, of Ashfield MedComms, an Inizio Company, funded by Takeda Pharmaceuticals U.S.A., Inc., Lexington, MA and complied with the Good Publication Practice (GPP) guidelines (DeTora LM, et al. Ann Intern Med 2022;175:1298–1304).

To view an electronic version of this poster, scan this Quick Response (QR) code or visit: https://tinyurl.com/2f8bnapm.

Copies of this poster obtained through QR code are for personal use only and may not be reproduced without permission from the congress and the author of this poster.

Presented at the American Society of Oncology (ASCO) Annual Meeting, Chicago, IL, USA, June 2–6, 2023. For questions or comments, please contact Dr Arvind Dasari: ADasari@mdanderson.org.