Phase 1a/1b study design of the novel STING agonist immune stimulating antibody conjugate (ISAC) TAK-500, with or without pembrolizumab in patients with advanced solid tumors

TPS2690

Jennifer R. Diamond,[1] Jason T. Henry,[2] Gerald S. Falchook,[2] Anthony J. Olszanski,[3] Harshabad Singh,[4] E. Jane Leonard,[5] Richard C. Gregory,[5] Vicky A. Appleman,[5]
John P. Gibbs,[5] Carole E. Harbison,[5] Cong Li,[5] Jessica M. Sapiro,[5]* Tomoki Yoneyama,[5] Alexander Parent,[5] Vincent Chung[6]

[1]University of Colorado Anschutz Medical Campus, Aurora, CO, USA; [2]Sarah Cannon Research Institute at HealthONE, Denver, CO, USA; [3]Fox Chase Cancer Center, Philadelphia, PA, USA; [4]Dana-Farber Cancer Institute, Boston, MA, USA;
[5]Takeda Development Center Americas, Inc. (TDCA), Lexington, MA, USA; [6]City of Hope National Medical Center, Duarte, CA, USA; *Current affiliation: Blueprints Medicines Corporation, Cambridge, MA, USA

Background

- Immuno-oncology has emerged as a major driver of anticancer therapeutics, including immune checkpoint inhibitors (CPIs); however, resistance to CPIs remains a challenge[1–3]
 - Reduced interferon (IFN) signaling, immune escape, altered antigen presentation, and immunosuppressive tumor phenotypes have been proposed as resistance mechanisms[1–3]
- Accordingly, a possible strategy to overcome resistance to CPIs is to stimulate innate immune cells in the tumor microenvironment
- STimulator of INterferon Genes (STING) is a cytosolic protein critical for induction of Type I IFN-dependent innate immunity[4]
- Cysteine-cysteine chemokine receptor Type II (CCR2) is expressed by tumor-infiltrating myeloid cells, including tumor-associated macrophages, which promote immune escape by limiting CD8+ T-cell infiltration, thereby decreasing the efficacy of immunotherapy[5,6]

Study design

- The study will proceed in two main sections:
 - A phased dose escalation of each treatment arm; TAK-500 as a single-agent and in combination with pembrolizumab
 - A subsequent dose expansion phase that will evaluate TAK-500 in combination with pembrolizumab

Dose escalation phase (N~72)

TAK-500

- Patients will receive single-agent intravenous (IV) TAK-500 (escalating doses) administered once every 3 weeks (Q3W) in 21-day cycles to determine the pharmacologically active dose (PAD) range
- The proposed dose range of TAK-500 is 8–480 µg/kg guided by the Bayesian Optimal Interval (BOIN) design

TAK-500 + pembrolizumab 200 mg

- An additional escalation cohort will receive IV TAK-500 in combination with IV pembrolizumab 200 mg Q3W in 21-day cycles
- The initial TAK-500 dose level (DL) will be 1–2 DLs below the single-agent predicted PAD range; dose level will be assigned at the time of enrollment

Dose expansion phase (N~34)

- Identification of minimum PAD and PAD+1 DLs in the combination arm of the dose escalation phase will trigger enrollment in the expansion cohort(s)
- Patients will receive IV TAK-500 in combination with IV pembrolizumab 200 mg, with possible exploration of Q3W versus once every 2 weeks (Q2W) administration of TAK-500

- The tumor types selected for this study were chosen based on clinical unmet need and the presence of the following biomarkers: CCR2 expression, monocyte and macrophage tumor infiltration, and STING gene signature expression as analyzed via publicly available databases

TAK-500 structure and mechanism of action

- TAK-500 is an ISAC that consists of three parts:
 - A synthetic analog of cyclic guanosine monophosphate-adenosine monophosphate (cGAMP) STING agonist payload based on TAK-676 (a potent early clinical stage STING agonist currently under phase 1 clinical evaluation [NCT04420884; NCT04879849][7,8])
 - A humanized immunoglobulin G1 anti-CCR2 antibody
 - A self-immolating maleimide-containing protease-cleavable peptide linker
- By targeting the STING pathway, and CCR2-expressing myeloid cells, TAK-500 may offer enhanced potency via improved pharmacokinetics (PK) and selective delivery. As such, TAK-500 has three possible mechanisms of action:
 - Activation of IFN response
 - Reprogramming of suppressive intratumoral CCR2+ myeloid cells to an inflammatory phenotype
 - Blockade of suppressive tumor-associated macrophage recruitment
- Through these mechanisms, TAK-500 has the potential to overcome resistance to CPIs in both CPI-refractory and immunologically cold tumors

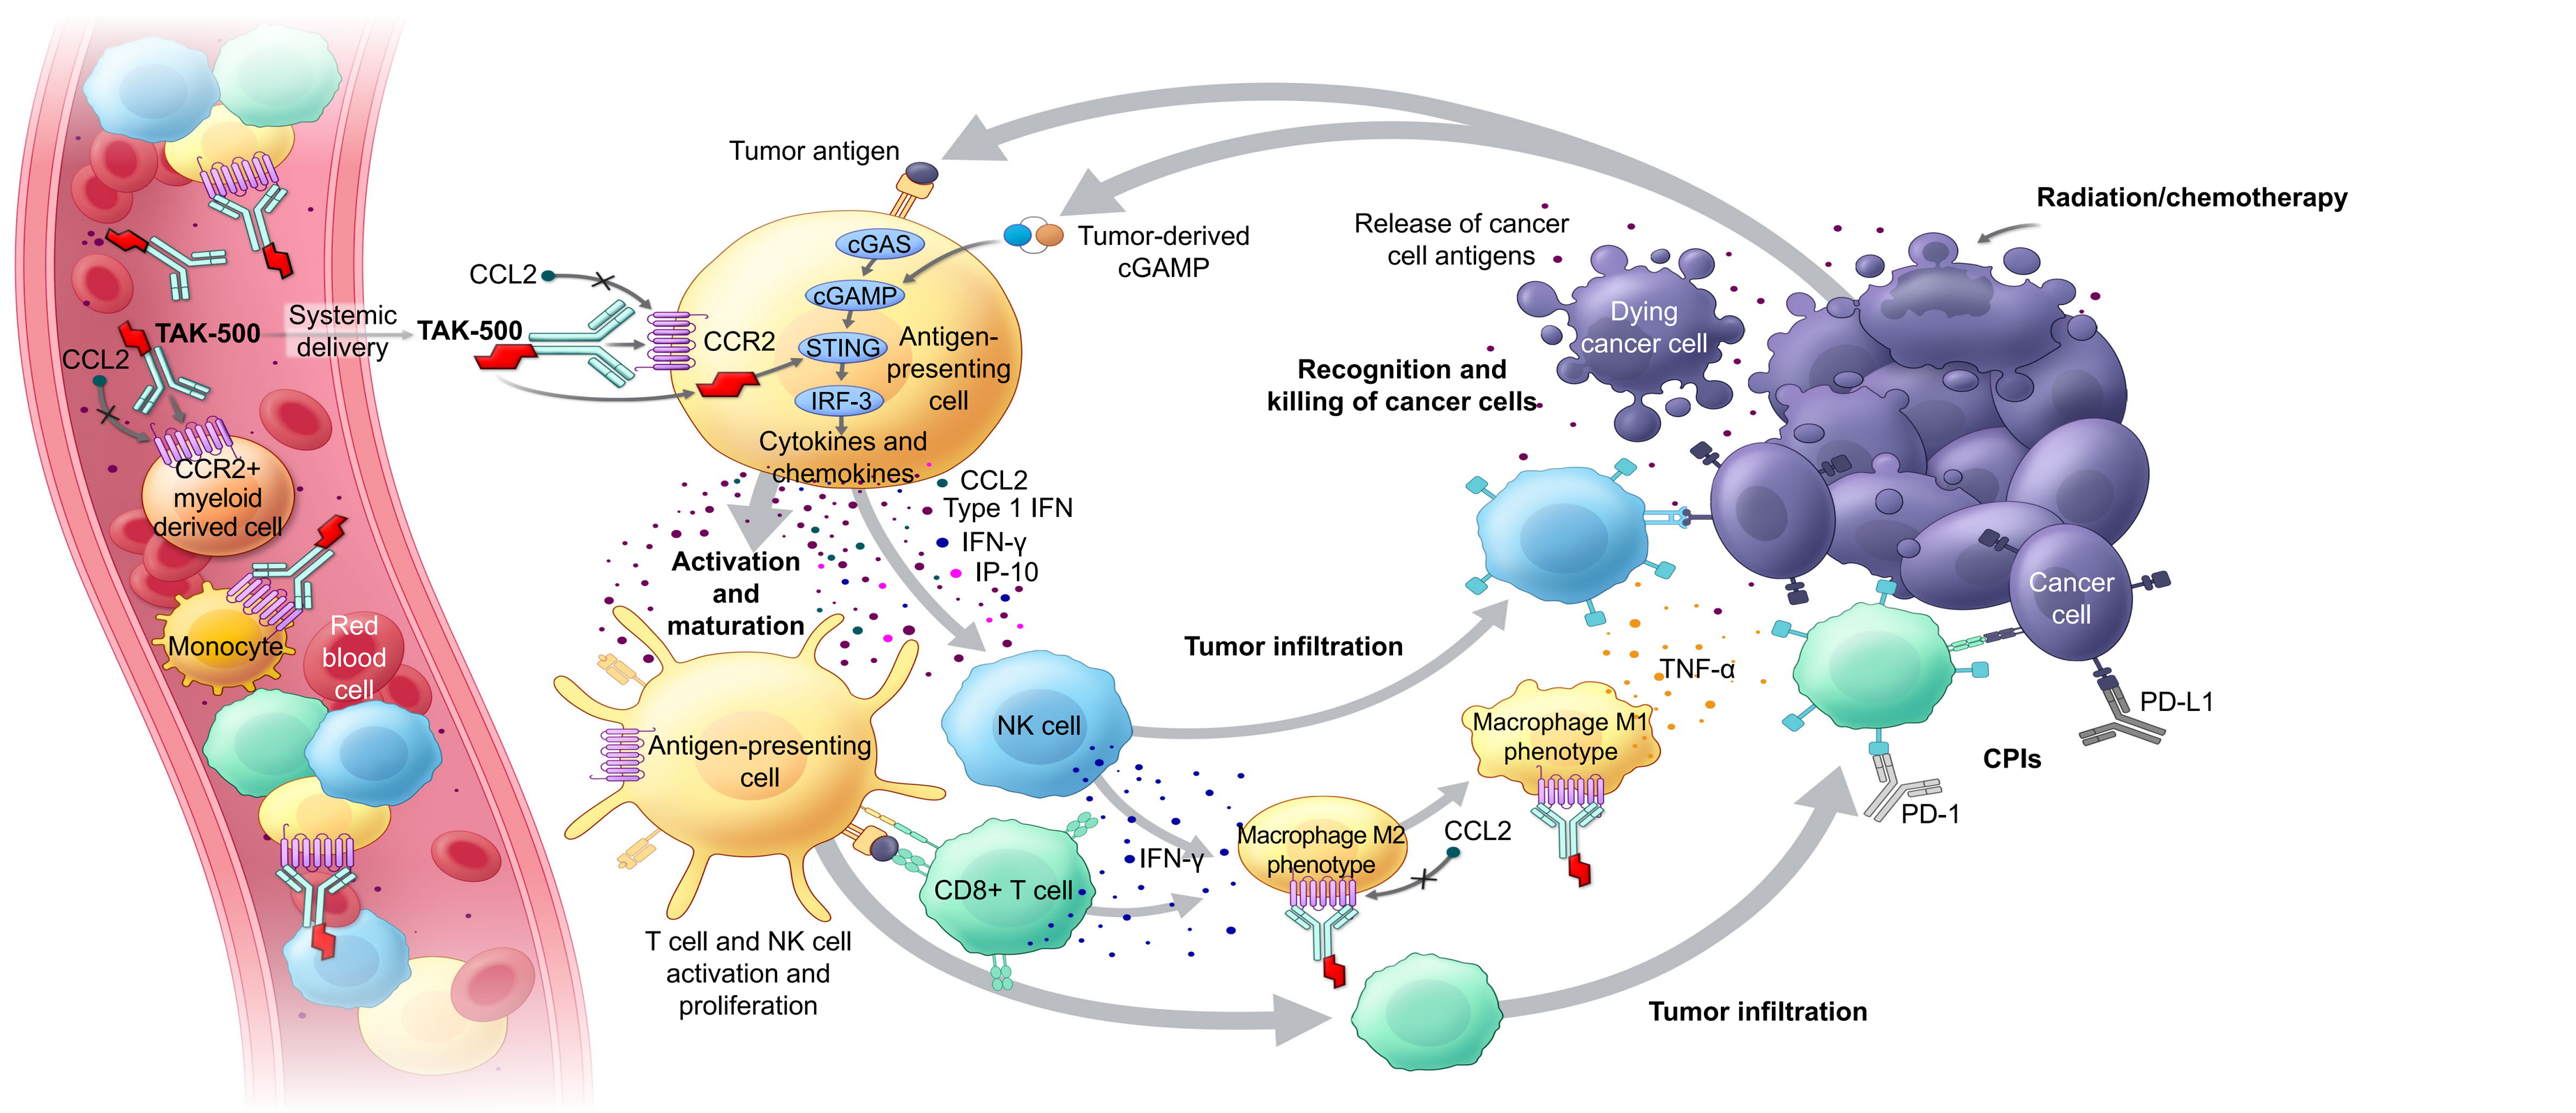
Tumor antigen
Tumor-derived cGAMP
Release of cancer cell antigens
Radiation/chemotherapy
Dying cancer cell
Recognition and killing of cancer cells
CCL2
TAK-500
Systemic delivery
TAK-500
CCR2
cGAS
cGAMP
STING
IRF-3
Antigen-presenting cell
Cytokines and chemokines
CCL2
Type 1 IFN
IFN-γ
IP-10
CCR2+ myeloid derived cell
Monocyte
Red blood cell
Activation and maturation
Antigen-presenting cell
NK cell
Tumor infiltration
TNF-α
Macrophage M1 phenotype
Cancer cell
PD-L1
CPIs
PD-1
Macrophage M2 phenotype
CCL2
IFN-γ
CD8+ T cell
T cell and NK cell activation and proliferation
Tumor infiltration

CCL2, cysteine-cysteine chemokine ligand 2; cGAS, cyclic guanosine monophosphate-adenosine monophosphate synthase; IFN-γ, interferon gamma; IP-10, interferon gamma induced protein 10; IRF-3, interferon regulatory transcription factor 3; NK, natural killer; PD-1, programmed cell death protein 1; PD-L1, programmed cell death-ligand 1; TNF, tumor necrosis factor.

Study design — **First-in-human, open-label, dose escalation and expansion, phase 1a/1b study of TAK-500, as a single-agent and in combination with pembrolizumab, in patients with select locally advanced or metastatic solid tumors**

Study objectives — **Primary:** Determine the safety and tolerability of TAK-500 when given alone or in combination with pembrolizumab
Secondary: Determine the PAD range, RP2D, antitumor activity, PK, immunogenicity, and pharmacodynamics of TAK-500 when given alone or in combination with pembrolizumab

Eligible conditions

This study is enrolling patients (N~106) with the following pathologically confirmed tumor types, whose disease has progressed on, or are intolerant to, all standard therapy:

- Gastroesophageal adenocarcinoma
- Pancreatic adenocarcinoma
- Hepatocellular carcinoma
- Non-squamous non-small cell lung cancer
- Squamous cell carcinoma of the head and neck
- Mesothelioma
- Triple-negative breast cancer

Escalation phase

n~40: **TAK-500 single-agent** (escalating doses)† IV Q3W in 21-day cycles

Determination of the minimum PAD will trigger initiation of the combination dose escalation

n~32*: **TAK-500** (escalating doses)† IV Q3W in 21-day cycles **+ pembrolizumab 200 mg** IV Q3W in 21-day cycles

Dose expansion phase

n~34: **TAK-500** IV **Q3W and/or Q2W in 42-day cycles + pembrolizumab 200 mg** IV Q3W in 42-day cycles in single tumor type expansion cohorts

Patients will receive treatment for up to 1 year

ClinicalTrials.gov identifier: NCT05070247

*In the combination arm, the number of patients will depend on the starting DL. †Dose escalation in both single-agent and combination cohorts will be guided by the BOIN design.

Study objectives and endpoints

Study objectives

Primary

- Safety and tolerability of TAK-500 as a single-agent or in combination with pembrolizumab

Secondary

- Determine PAD range and recommended phase 2 dose (RP2D)
- Characterize single- and multiple-doses of PK
- Evaluate preliminary antitumor activity
- Evaluate dose-responsive impact on tumor immune cell infiltration and activation
- Determine immunogenicity of TAK-500

Primary endpoints

- Frequency of treatment-emergent adverse events (TEAEs), serious TEAEs, and TEAEs leading to dose modifications and discontinuations
- Number of patients with dose-limiting toxicities

Secondary endpoints

- PAD range and RP2D
- PK parameters of TAK-500
- Response assessments by the investigator per Response Evaluation Criteria in Solid Tumors (RECIST) version 1.1 for both single-agent TAK-500 and in combination with pembrolizumab
- Immune cell infiltration and activation by immunohistochemistry or in-situ hybridization
- Changes in gene expression, including STING agonism and IFN gene signatures, between pre-treatment and on-treatment tumor biopsies
- Incidence of patients who are antidrug antibody-positive and have acquired immunogenicity

Patient eligibility

Key inclusion criteria

- Male or female participants aged ≥18 years
- Eastern Cooperative Oncology Group performance status of 0 or 1
- Patients with select pathologically confirmed (cytological diagnosis is adequate), locally advanced or metastatic solid tumors, whose disease has progressed on, or are intolerant to, all standard therapy
- At least one RECIST version 1.1 evaluable lesion
- Adequate bone marrow, renal, hepatic, pulmonary, and cardiac functions

Key exclusion criteria

- Treated with other STING agonists/antagonists, Toll-like receptor agonists, or CCR2 agonists/antagonists within the past 6 months
- Active vaping and/or smoking within 90 days of the first dose of study drug
- Treatment with any investigational products or other anticancer therapy (including chemotherapy, targeted agents, and immunotherapy) within 14 days or 5 half-lives, whichever is shorter, before the first dose of study drug
- Radiation therapy within 14 days (42 days for radiation to the lungs) and/or systemic treatment with radionuclides within 42 days before the first dose of study drug. Patients with clinically relevant ongoing pulmonary complications from prior radiation therapy are not eligible
- History of brain metastasis, unless:
 - Clinically stable following prior surgery, whole-brain radiation, or stereotactic radiosurgery
 - Off corticosteroids for central nervous system involvement
- Grade ≥2 fever of malignant origin
- Patients with either/both of:
 - Uncontrolled, known, or suspected, autoimmune disease
 - A diagnosis of congenital or acquired immunodeficiency

Enrollment and study timelines

- Recruitment is ongoing
- The study is planned to enroll ~106 patients across 7 to 10 sites in the United States
- Start date: February 1, 2022
- Estimated study completion date: April 8, 2025

References

1. Jenkins RW, et al. Br J Cancer 2018;118:9–16
2. Sharma P, et al. Cell 2017;168:707–23
3. Liu D, et al. Am J Clin Dermatol 2019;20:41–54
4. Ishikawa H, et al. Nature 2009;461:788–92
5. Kim IS, et al. Nat Cell Biol 2019;21:1113–26
6. Lesokhin AM, et al. Cancer Res 2012;72:876–86
7. Falchook GS, et al. J Clin Oncol 2021;39 (Suppl 15): Abstract #TPS2670
8. Cooper BT, et al. AACR 2022: Abstract #CT243

Disclosures

J.R. Diamond: consultancy for Immunomedics Gilead, OnKure Therapeutics; leadership role with and stock/other ownership interests for OnKure; research funding (to institution) from Millennium, Bayer, MedImmune, Taiho Pharmaceuticals, Rexahn Pharmaceuticals, Immunomedics, OnKure, Adlai, Astellas, BMS, Deciphera, Genentech, Hutchison, Merck, Takeda. **J.T. Henry:** travel (self, for work or research related to institution) for Sarah Cannon Research Institute; research funding (to institution) for any trial for which I have been the principal investigator (ever) or sub-investigator (minimum last 4 years) from Abbiscko Therapeutics, ABL Bio, Accutar Biotech, ADC Therapeutics, Aileron Therapeutics, Amgen, Artios, AstraZeneca/Merck, Bicycle Therapeutics, BioAlta, BioInvent Pharma, Biosplice Therapeutics, Boehringer Ingelheim, Cyteir, Daiichi Sankyo, Lilly, Epizyme, Erasca Inc, Exelixis, Fujifilm, GlaxoSmithKline, Hutchison MediPharma, IgM Biosciences, Immunogen, Jacobio, Jounce Therapeutics, Loxo, Molecular Templates, Navire, NiKang Therapeutics, Oncorus, Prelude Therapeutics, Poseida Therapeutics, PureTech, Pyramid Biosciences, RasCal, Regeneron, Relay Therapeutics, Rgenix, Ribon Therapeutics, Sapience Therapeutics, Sarah Cannon Research Institute, Simcha Therapeutics, Sirna, Stingthera, Synthorx, Takeda, Tarveda Therapeutics, Teneothree, Tesaro, Turning Point Therapeutics, Xencor. **G.S. Falchook:** employment with Sarah Cannon Research Institute and HealthONE; research funding (to institution) for any trial for which I have been the principal investigator (ever) or sub-investigator (minimum last 4 years) from 3-V Biosciences, Abbisko, Abbvie, ABL Bio, ADC Therapeutics, Accutar, Aileron, American Society of Clinical Oncology, Amgen, ARMO/Eli Lilly, Artios, AstraZeneca, BeiGene, Bioatla, BioInvent, Biothera, Bicycle, Boehringer Ingelheim, Celldex, Celgene, Ciclomed, Curegenix, Curis, Cyteir, Daiichi, DelMar, eFFECTOR, EMD Serono, Epizyme, Erasca, Exelixis, Freenome, Fujifilm, Genmab, Hutchison MediPharma, IGM Biosciences, Ignyta, ImmunoGen/MacroGenics, Incyte, Jacobio, Jounce, Kolltan, MedImmune, Millennium, Merck, miRNA Therapeutics, Molecular Templates, Navire, NiKang, NovartLoxois, OncoMed, Oncorus,Poseida, Precision Oncology, Prelude, PureTech, Pyramid, RasCal, Regeneron, Relay, Rgenix, Ribon, Samumed, Sapience, Silicon, Simcha, Sirnaomics, Strategia, Syndax, Synthorx, Taiho, Takeda, Tarveda, Teneobio, Tesaro, Tocagen, Turning Point, Vegenics, Xencor; travel/accommodation/expenses (self, for work or research related to institution) for Bristol Myers Squibb, EMD Serono, Fujifilm, Millennium, Sarah Cannon Research Institute; royalties (self) from Wolters Kluwer; advisory role (to institution) for FujiFilm, Silicon, Navire, Turning Point, Predicine, Inspirna, Regeneron; advisory role (self) for EMD Serono; speakers honorarium (self) for CME from Total Health Conferencing, Rocky Mountain Oncology Society; patents/royalties/other intellectual property for Handbook of Targeted Cancer Therapy. **A.J. Olszanski:** consulting/advisory role for Merck, Pfizer, Bristol-Myers Squibb, Sanofi, Takeda, Eisai; research funding (to institution) from Takeda, Immunocore, Bristol-Myers Squibb, Checkmate Pharmaceuticals, Targovax, Alkermes, Astellas Pharma, GlaxoSmithKline, Kartos Therapeutics, Glyconex, Intensity Therapeutics, Kadmon, Nektar, OncoSec, Seattle Genetics, Sound Biologics, Spring Bank, Gan & Lee, Lovance Biotherapeutics, Regeneron, Navire, Sumitomo Dainippon Pharma; uncompensated relationships with Takeda. **H. Singh:** none. **E.J. Leonard:** employment with Takeda Development Center Americas, Inc. (TDCA); stock/other ownership interests for Takeda and Pfizer. **R.C. Gregory:** employment with Takeda Development Center Americas, Inc. (TDCA); stock/other ownership interests for Takeda; research funding from Takeda; travel/accommodation/expenses for Takeda. **V.A. Appleman:** employment with Takeda Development Center Americas, Inc. (TDCA); stock/other ownership interests for Takeda; research funding from Takeda; patents/royalties/other intellectual property for Takeda; travel/accommodation/expenses for Takeda. **J.P. Gibbs, C.E. Harbison, J.M. Sapiro, A. Parent:** employment with Takeda Development Center Americas, Inc. (TDCA); stock/other ownership interests for Takeda. **C. Li:** employment with Takeda Development Center Americas, Inc. (TDCA); stock/other ownership interests for Takeda; patents/royalties/other intellectual property for Takeda. **T. Yoneyama:** employment with Takeda Development Center Americas, Inc. (TDCA). **V. Chung:** speakers bureau for Ipsen, AstraZeneca, Coherus; research funding (to institution) from Merck; consulting for Pethera, Pfizer. **For questions or comments, please contact Dr Vincent Chung: VChung@coh.org.**

Acknowledgments

- This study is funded by Takeda Development Center Americas, Inc. (TDCA)
- Medical writing support for the development of this poster, under the direction of the authors, was provided by Beth Jamieson, BSc, and Georgia, Pangratiou, PhD of Ashfield MedComms, an Ashfield Health company, funded by Takeda Pharmaceuticals U.S.A., Inc., and complied with the Good Publication Practice-3 (GPP3) guidelines (Battisti WP, et al. Ann Intern Med 2015;163:461–4)

To view an electronic version of this poster, scan this QR code or visit: **https://tqr.bz/p9if2**

Copies of the poster obtained through the Quick Response (QR) codes are for personal use only and may not be reproduced without permission from ASCO® and the author of this poster.

Presented at the American Society of Clinical Oncology (ASCO) Annual Meeting, Chicago, IL, USA, June 3–7, 2022